Engineered human IL-2/IL-2Rβ orthogonal pairs selectively enhance anti-GPC3 CAR T cells to drive complete responses in solid epithelial tumor models

Paul-Joseph Aspuria, Marie Semana, Mahalaksmi Ramadass, Navneet Ratti, Deepti Rokkam, Ivan Cheng, Tina Kochel, Ryan Burgess, George Zheng, Michele Bauer, Mohammed Ali, Henry Rosas, Sandro Vivona, Rob A. Kastelein, Patrick J. Lupardus, and Martin Oft
Synthekine, Menlo Park, CA

Synthekine

Abstract

CAR T cell therapy has demonstrated clinical efficacy against relapsed and refractory hematological malignancies. However, prominent barriers including poor T cell effector function, lack of proliferation, and limited CAR T cell persistence have prevented CAR T cell therapies from reaching their full curative potential, especially in solid tumors. Interleukin-2 (IL-2) is a potent stimulator of T cell proliferation, survival, and cytotoxic function, thereby making it an attractive cytokine to support CAR T cell therapy. However, therapeutic use of IL-2 is limited by systemic toxicity due its promiscuous activation of undesired immune cell populations, including non-tumor reactive T cells and NK cells.

To facilitate selective ex vivo and in vivo expansion of engineered T cells we have developed a human orthogonal (ortho) ligand/receptor system consisting of a IL-2 mutein (STK-009) that does not significantly stimulate cells expressing wild type IL-2 receptor and a mutated IL-2 Receptor Beta (orthoIL-2Rβ) that responds to STK-009 but not wild type IL-2. This system enables *in vivo* IL-2 signaling in engineered cells that express the orthoIL-2Rβ while avoiding stimulation of native T cells and NK cells. Previously, we demonstrated the ability of the STK-009/orthoIL-2Rβ receptor pair to selectively enhance the anti-tumor efficacy of orthoIL-2Rβ (hoRb) expressing CD19 CAR T cells (SYNCAR-001) in preclinical lymphoma mouse models[1]. We also demonstrated that STK-009 is selective for the orthoIL-2Rβ expressing cells and therefore in a non-human primate model does not stimulate native T or NK cells[1].

Here, we demonstrate the ability of the STK-009/hoRb system to enhance the anti-tumor activity and persistence of anti-glypican 3 (GPC3) CAR T cells. GPC3 overexpression is associated with various malignancies such as hepatocellular carcinoma (HCC), pediatric sarcomas, and non small cell lung carcinoma (NSCLC). Clinical trials of GPC3 CAR T therapy are ongoing, but early data suggests a need to boost CAR T cell function and persistence to achieve significant clinical responses. We incorporated the hoRb downstream of a second generation anti-GPC3 CAR via a T2A cleavage peptide (SYNCAR-002) to allow for bicistronic expression from one lentiviral construct. In vivo, STK-009 administration enhanced the anti-tumor efficacy of SYNCAR-002 in highly aggressive subcutaneous and intraperitoneal HCC models. STK-009 treatment resulted in significant expansion of SYNCAR-002 in the peripheral blood and drove infiltration of SYNCAR-002 into tumors. STK-009 treatment also induced intratumoral granzyme B and IFN-γ production by SYNCAR-002 indicating activation of effector T cell function.

These findings validate that the orthogonal IL-2 platform has the potential to improve the efficacy and durability of CAR T therapy for solid tumor targets such as GPC3 by selectively expanding CAR-T cells in vivo, driving CAR-T cells into the tumor, and activating CAR-T cells in the tumor microenvironment.

Orthogonal Cytokine + Cell Therapy: A Lock and Key System to Stimulate ACTs Selectively In Vivo

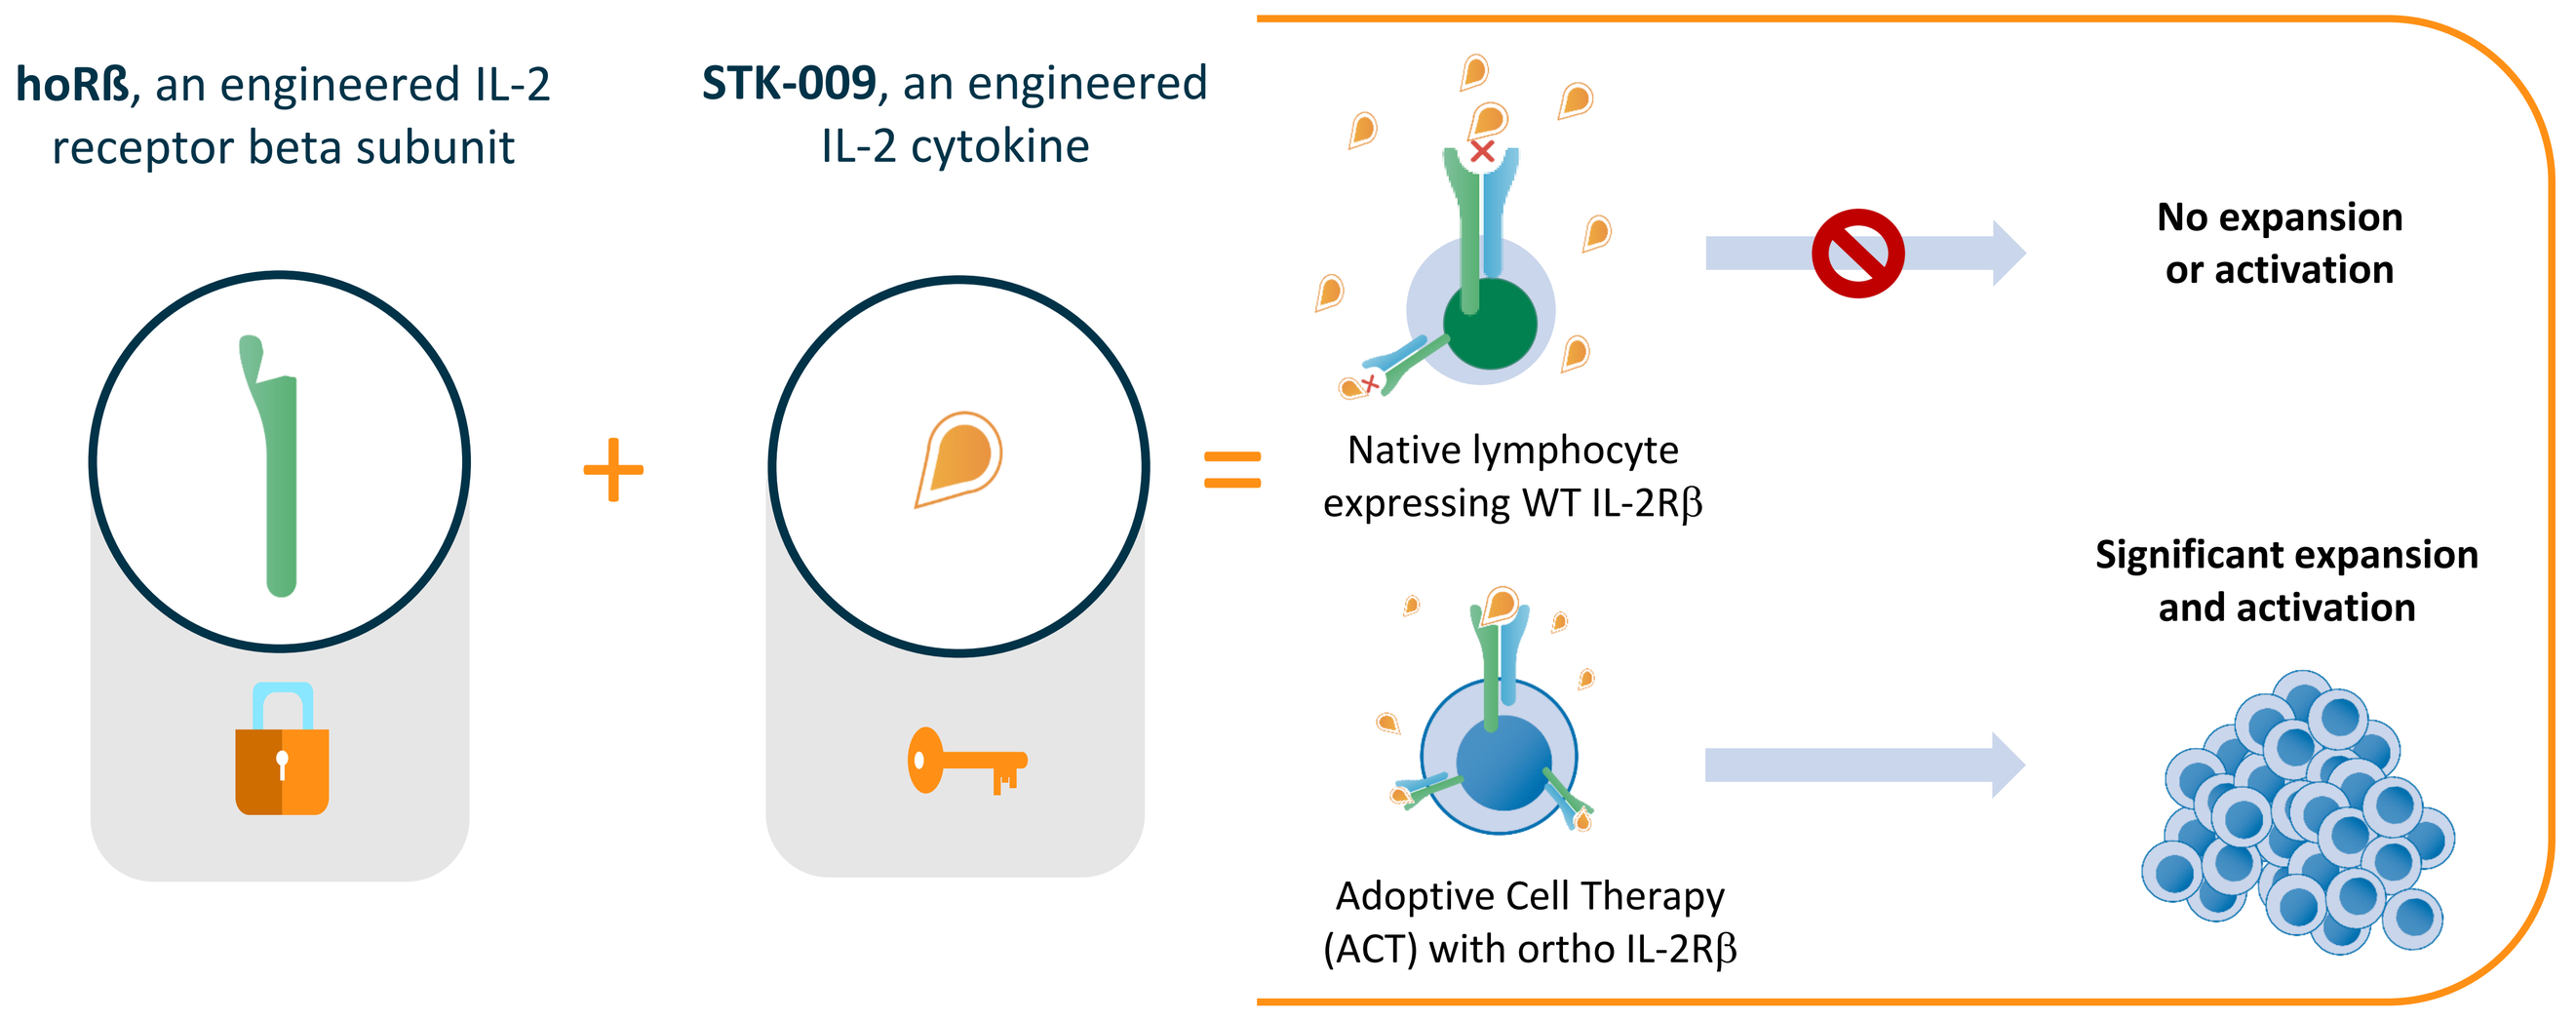
hoRß, an engineered IL-2 receptor beta subunit
STK-009, an engineered IL-2 cytokine
+
=
Native lymphocyte expressing WT IL-2Rβ
No expansion or activation
Adoptive Cell Therapy (ACT) with ortho IL-2Rβ
Significant expansion and activation

*Ortho*IL-2Rβ receptors (hoRb) exhibit significant preference for their cognate ligand, STK-009, a pegylated orthogonal IL-2. Therefore, engineered T cells expressing the *ortho* receptor will respond selectively to STK-009, thereby allowing specific expansion and enhancement of engineered T cell activity.

STK-009 demonstrates extended PK and no native lymphocyte activation in Non-Human Primates

- **STK-009 had high, sustained exposures in NHP**
 - PEG: decreased renal clearance
 - Ortho: decreased target mediated clearance
- **No clinical or pathological changes observed**
 - No increase in eosinophils
 - No change in lymphocyte / white blood cells
- **STK-009 treatment for up to 2 weeks did not induce IL-2 related cellular or cytokine/chemokine changes**
 - No induction of STAT-5 phosphorylation
 - No NK cell proliferation
 - No change to T cell populations
- **STK-009 does not activate the host IL-2 pathway in the absence of ortho-IL-2Rβ**

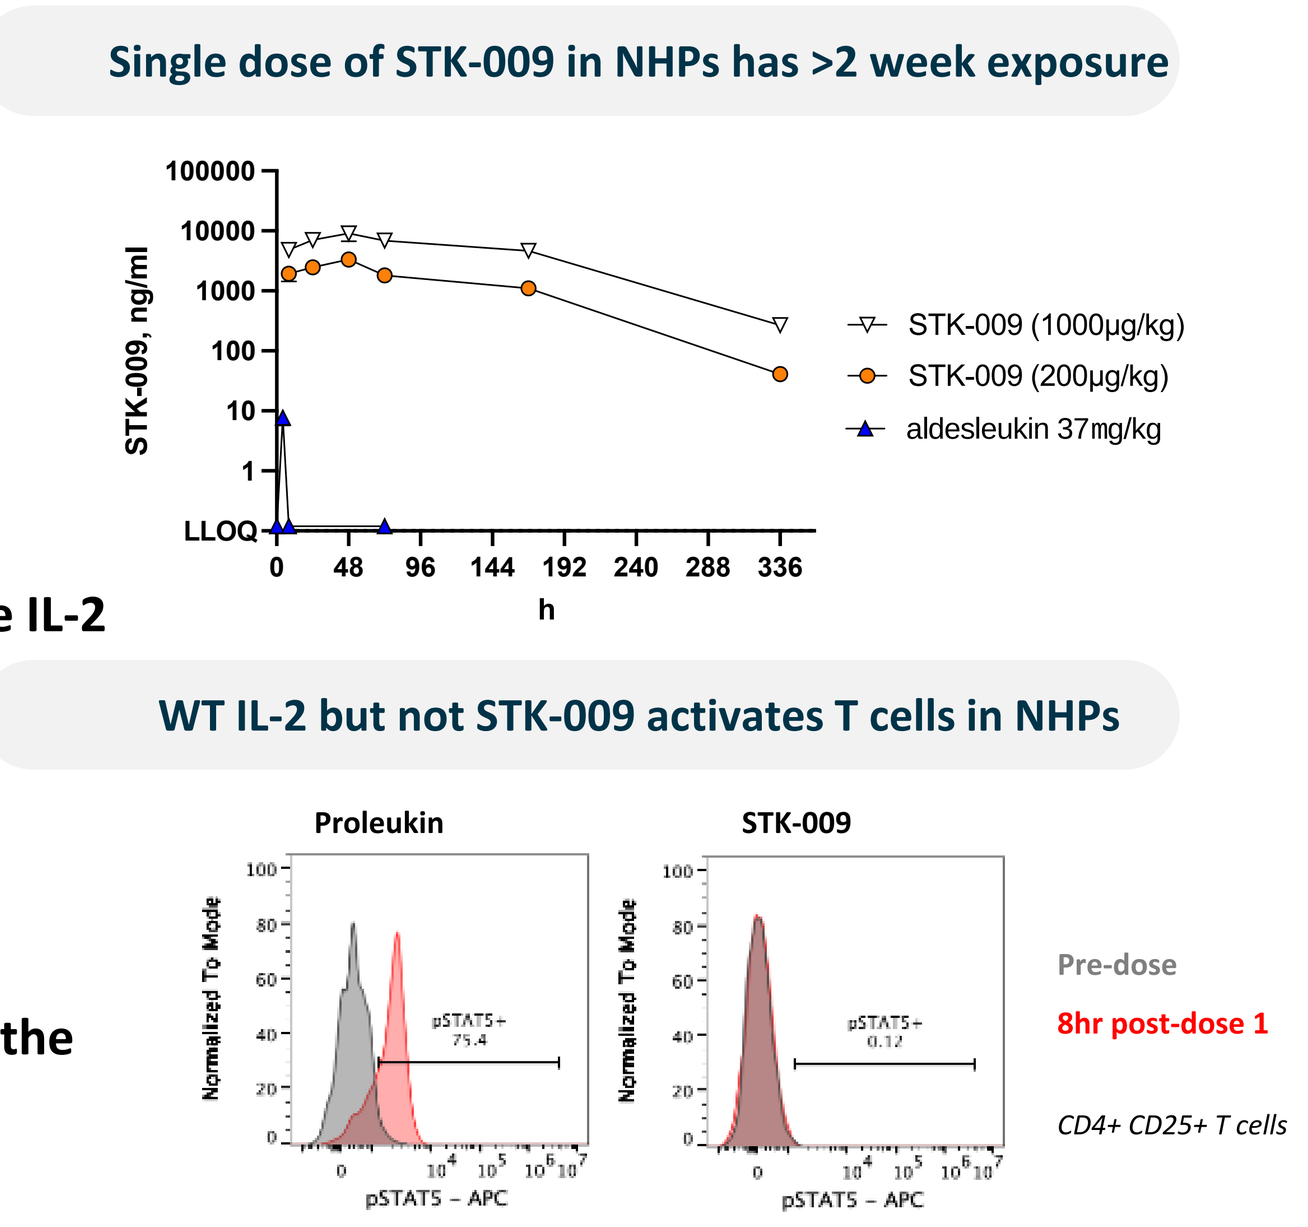
Single dose of STK-009 in NHPs has >2 week exposure
STK-009, ng/ml
100000
10000
1000
100
10
1
LLOQ
0
48
96
144
192
240
288
336
h
STK-009 (1000µg/kg)
STK-009 (200µg/kg)
aldesleukin 37mg/kg
WT IL-2 but not STK-009 activates T cells in NHPs
Proleukin
STK-009
Normalized To Mode
pSTAT5 – APC
Pre-dose
8hr post-dose 1
CD4+ CD25+ T cells

GPC3 is overexpressed in HCC and largely devoid in normal adult tissues

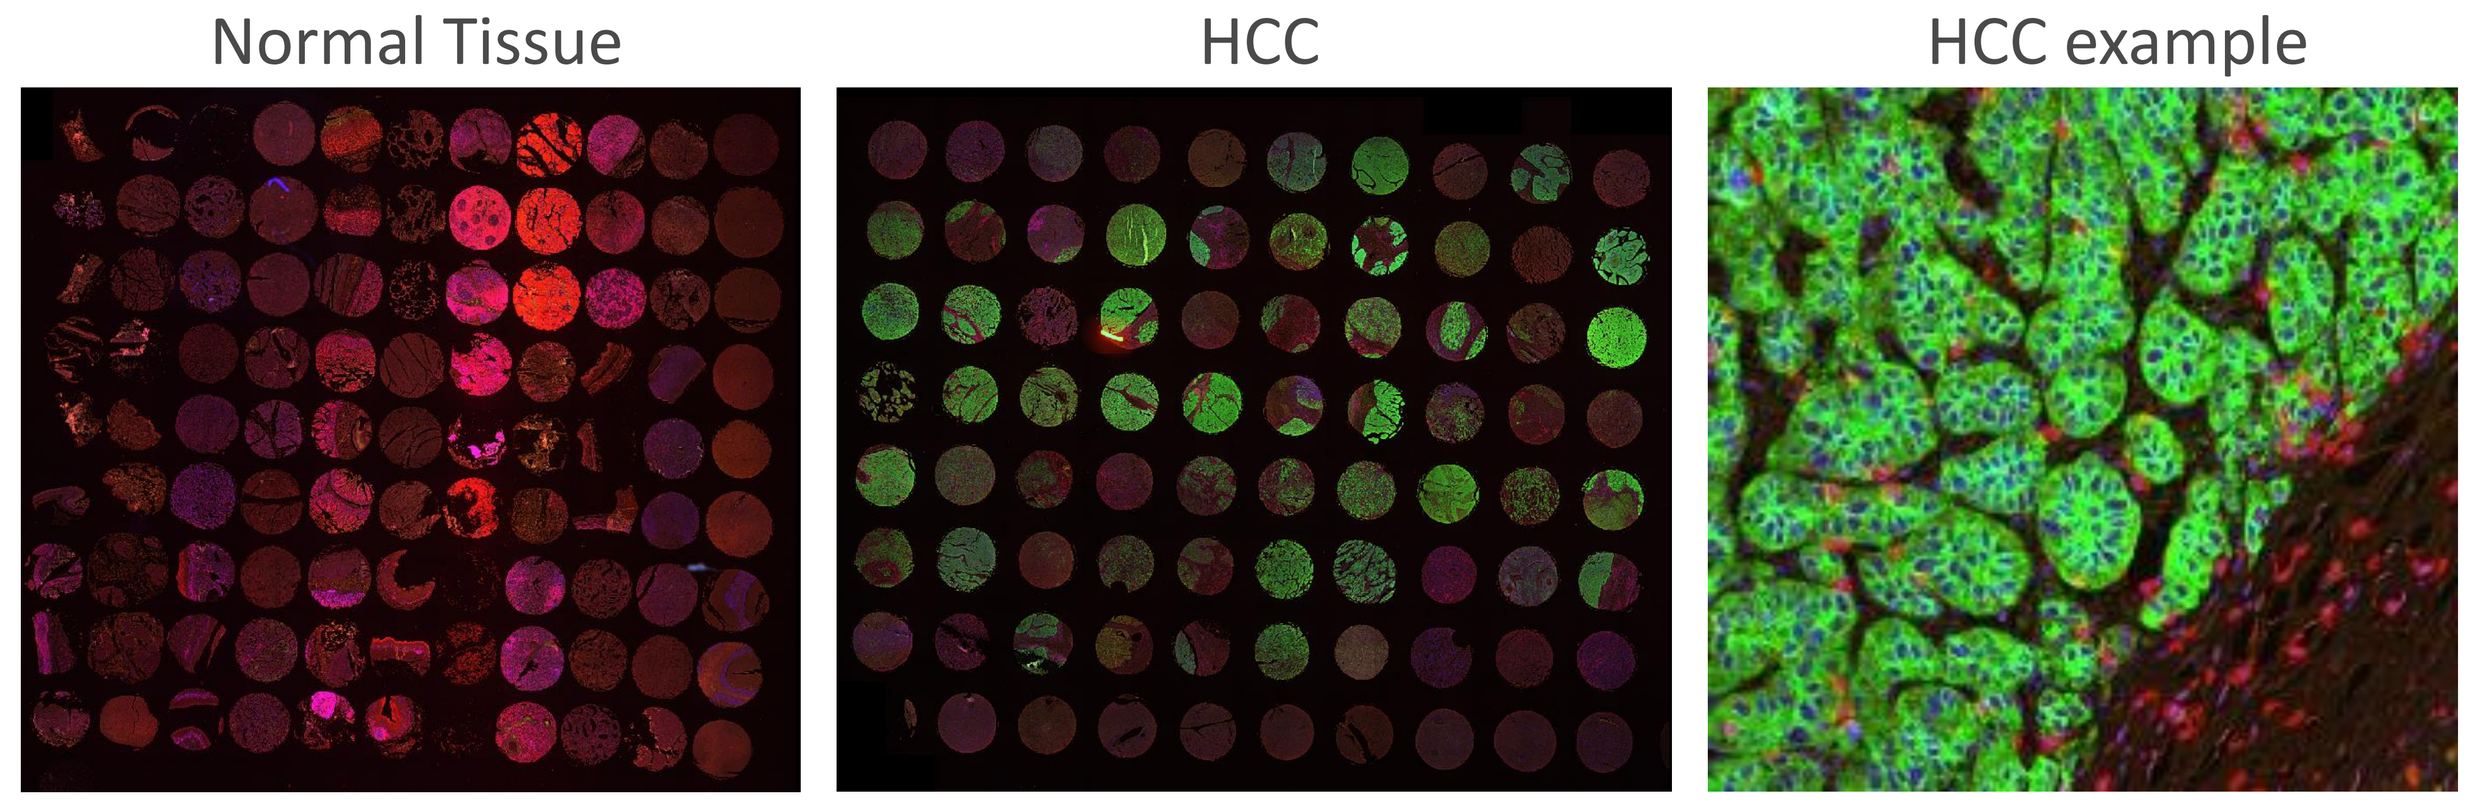
Normal Tissue
HCC
HCC example

Immunofluorescent staining of GPC3 (green) and CD3 T cells (red). Normal tissue microarray consisting of 90 cores representative of adult healthy tissues. 2/3 of HCC cores stained positive for membranous GPC3 expression. Zoomed in example of a GPC3 positive core.

SYNCAR-002: Anti-GPC3 CAR T cells co-expressing hoRb that specifically respond to STK-009 and maintain response to WT IL-2

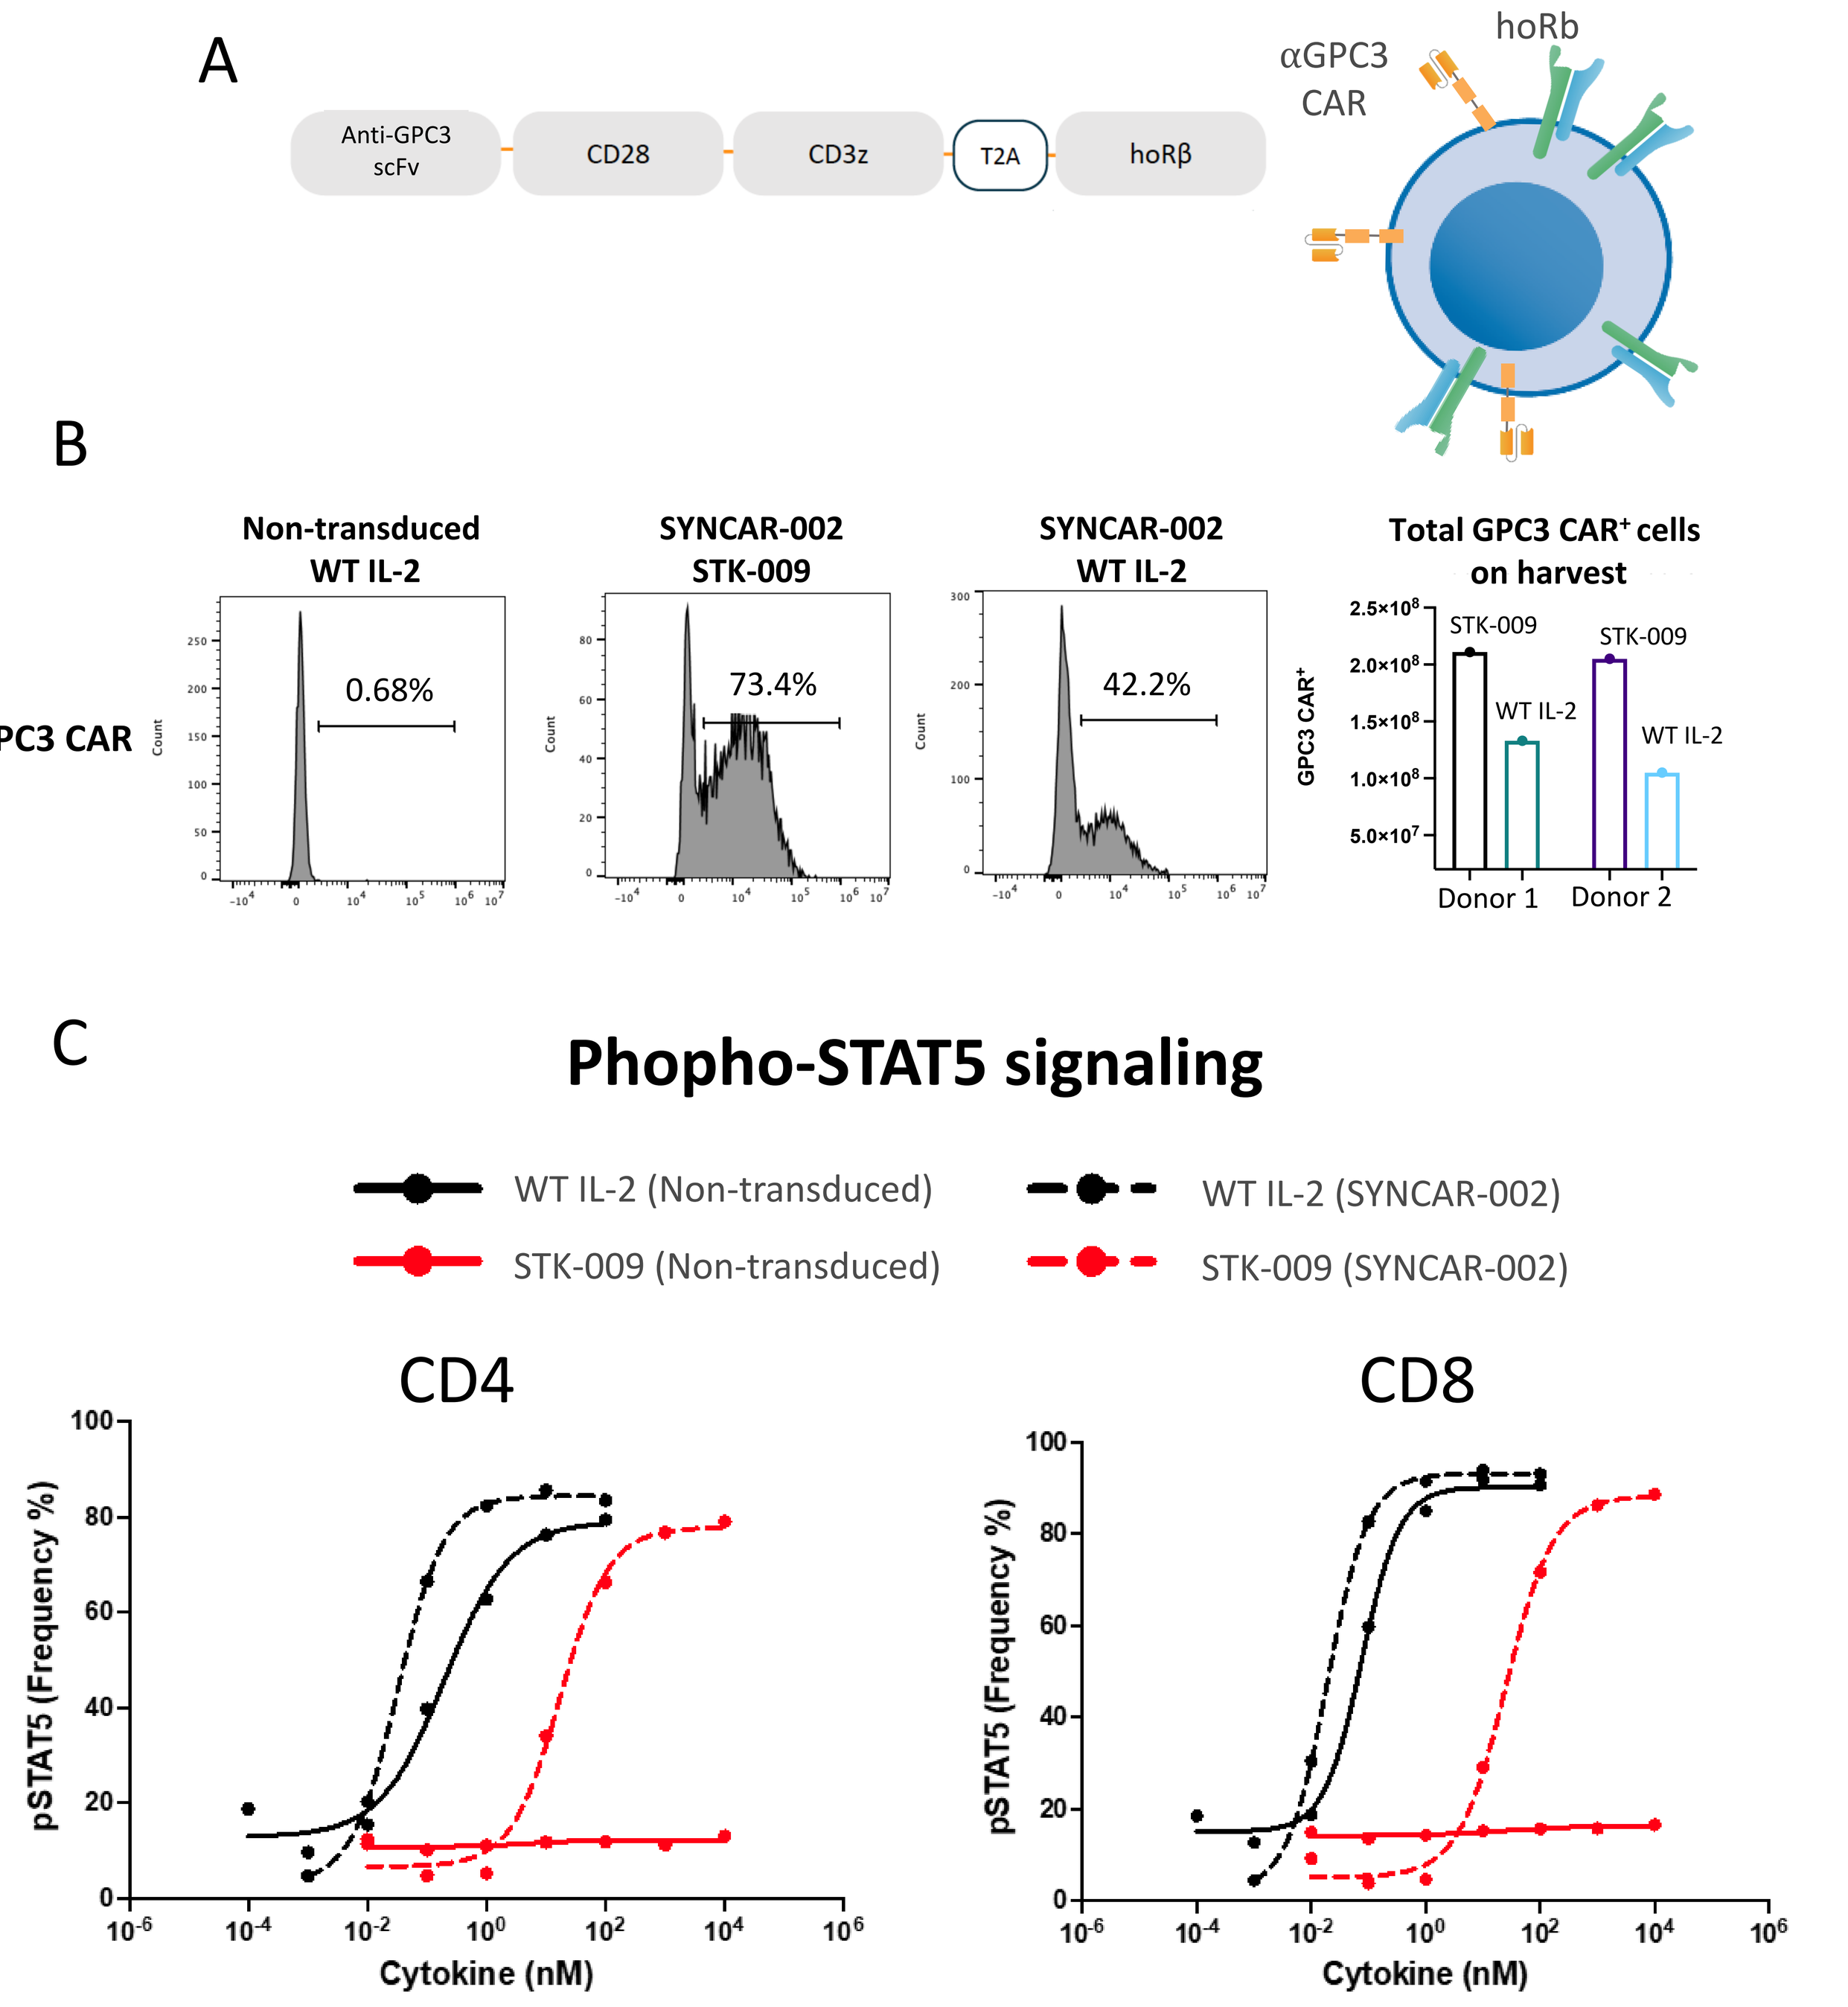
A
Anti-GPC3 scFv
CD28
CD3z
T2A
hoRβ
αGPC3 CAR
hoRb
B
Non-transduced WT IL-2
SYNCAR-002 STK-009
SYNCAR-002 WT IL-2
Total GPC3 CAR+ cells on harvest
GPC3 CAR
0.68%
73.4%
42.2%
STK-009
WT IL-2
Donor 1
Donor 2
C
Phopho-STAT5 signaling
WT IL-2 (Non-transduced)
STK-009 (Non-transduced)
WT IL-2 (SYNCAR-002)
STK-009 (SYNCAR-002)
CD4
CD8
pSTAT5 (Frequency %)
Cytokine (nM)

SYNCAR-002 architecture and specific response to STK-009. (A) Lentiviral construct containing the anti-GPC3 CAR, cleavage peptide T2A, and human orthoIL-2Rβ (hoRb) that immediately follows the CAR construct are expressed as a single mRNA. Expression is regulated via an EF1α promoter. **(B)** GPC3 CAR transduction assessed by flow cytometry. SYNCAR-002 cells manufactured in either WT IL-2 or STK-009 and total CAR⁺ T cells upon day of harvest. **(C)** Phospho-STAT5 signaling assay. Non-transduced and SYNCAR-002 cells were treated with either WT IL-2 or STK-009 for 20 min and processed for pSTAT5 analysis via flow cytometry. pSTAT5 frequency of CD4 and CD8 cells is displayed.

STK-009 administration enhances the anti-tumor efficacy of a suboptimal dose of SYNCAR-002 T cells in subcutaneous and intraperitoneal GPC3 expressing HCC mouse models

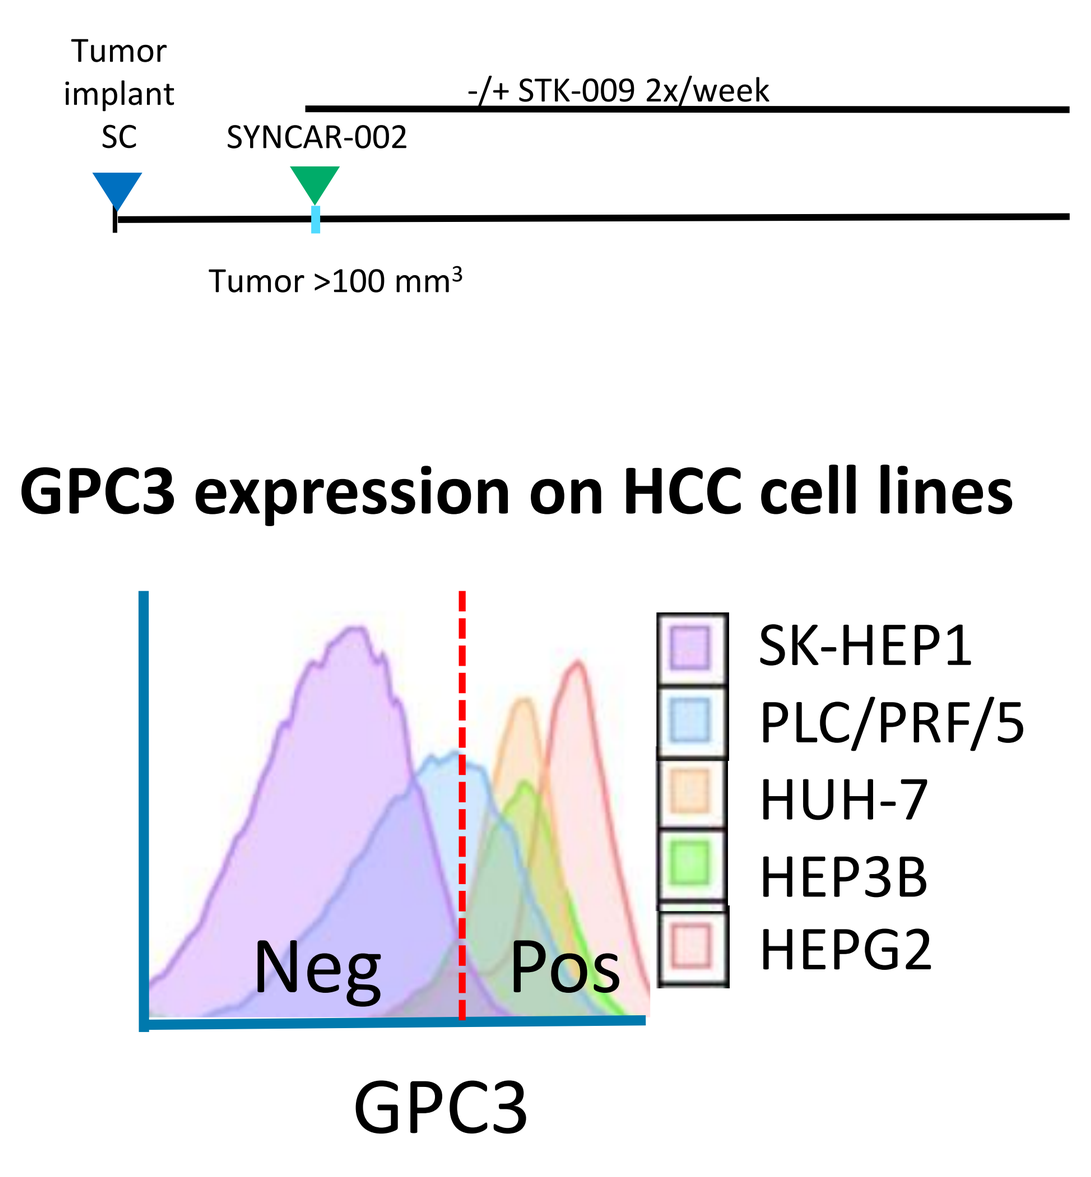
Tumor implant SC
SYNCAR-002
-/+ STK-009 2x/week
Tumor >100 mm³
GPC3 expression on HCC cell lines
Neg
Pos
GPC3
SK-HEP1
PLC/PRF/5
HUH-7
HEP3B
HEPG2

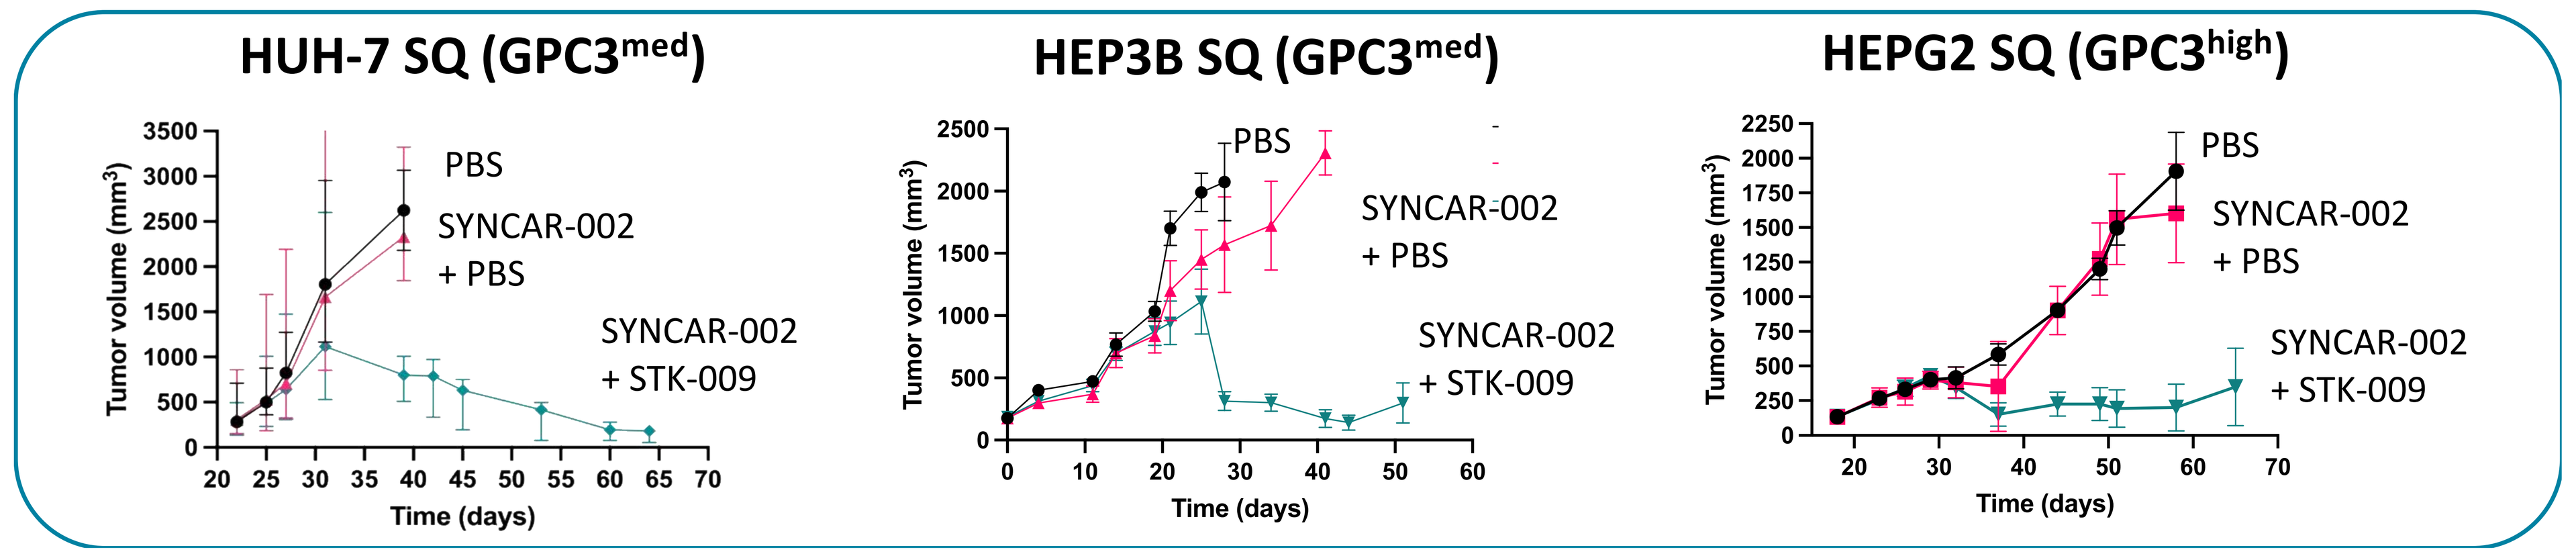
HUH-7 SQ (GPC3med)
HEP3B SQ (GPC3med)
HEPG2 SQ (GPC3high)
Tumor volume (mm³)
Time (days)
PBS
SYNCAR-002 + PBS
SYNCAR-002 + STK-009

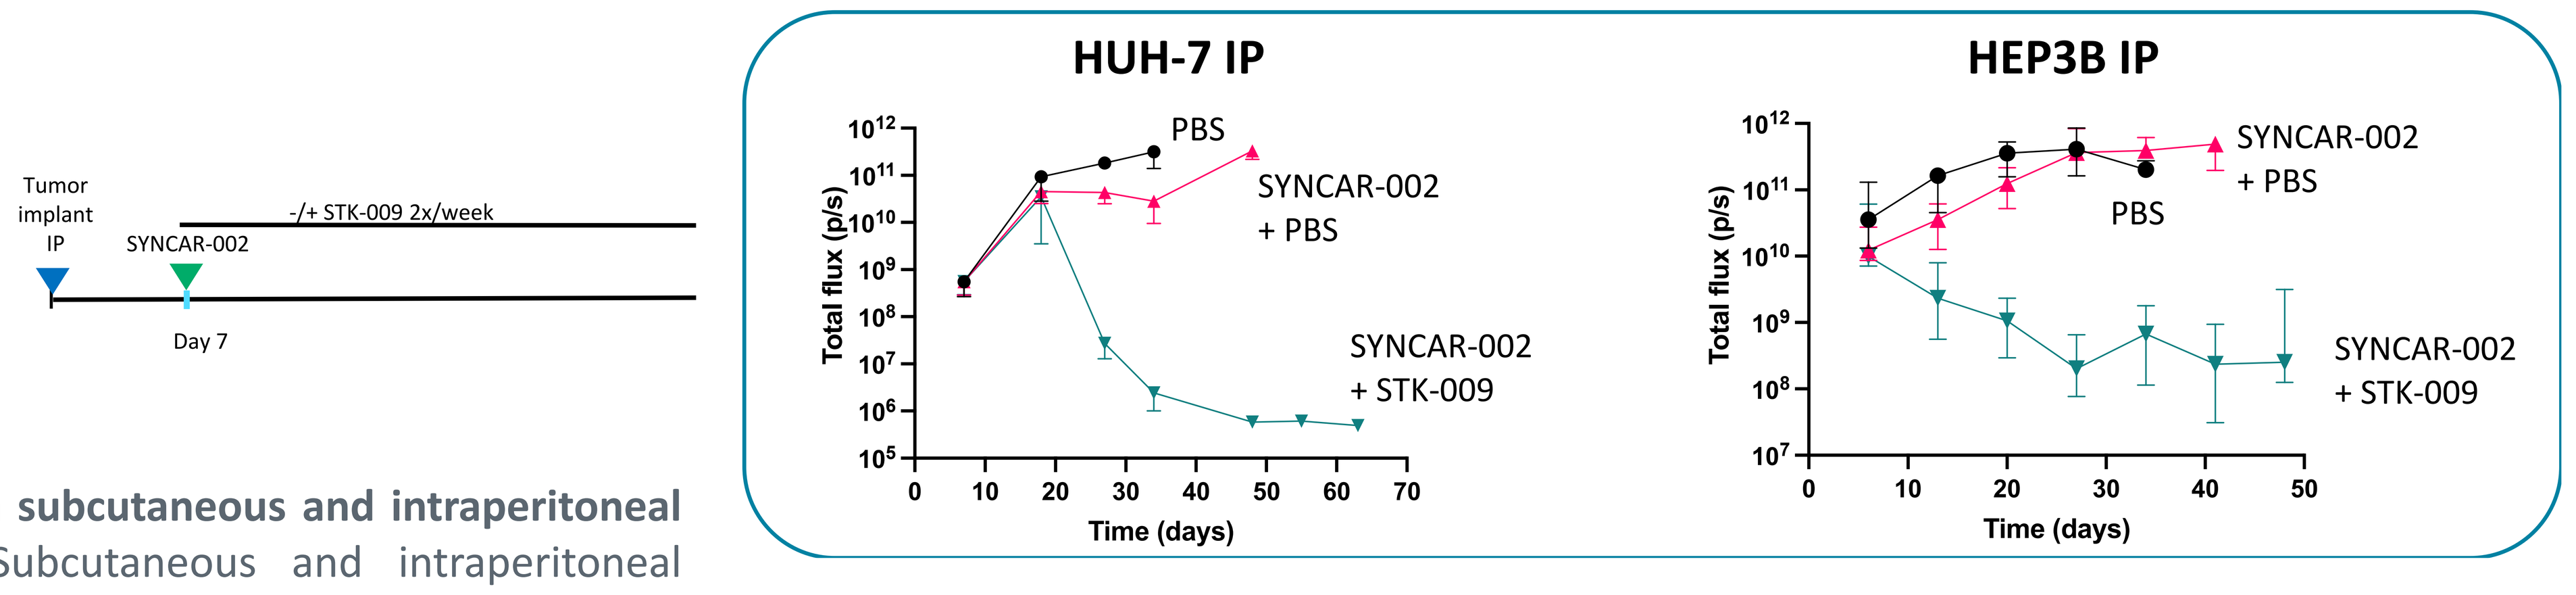
Tumor implant IP
SYNCAR-002
-/+ STK-009 2x/week
Day 7
HUH-7 IP
HEP3B IP
Total flux (p/s)
Time (days)
PBS
SYNCAR-002 + PBS
SYNCAR-002 + STK-009

SYNCAR-002 -/+ STK-009 efficacy in subcutaneous and intraperitoneal HCC xenograft mouse models. Subcutaneous and intraperitoneal models were treated by a suboptimal dose of SYNCAR-002 T cells with or without subcutaneous injection of STK-009 at <100mm³ and on day 7, respectively. Median tumor volume and tumor burden is displayed.

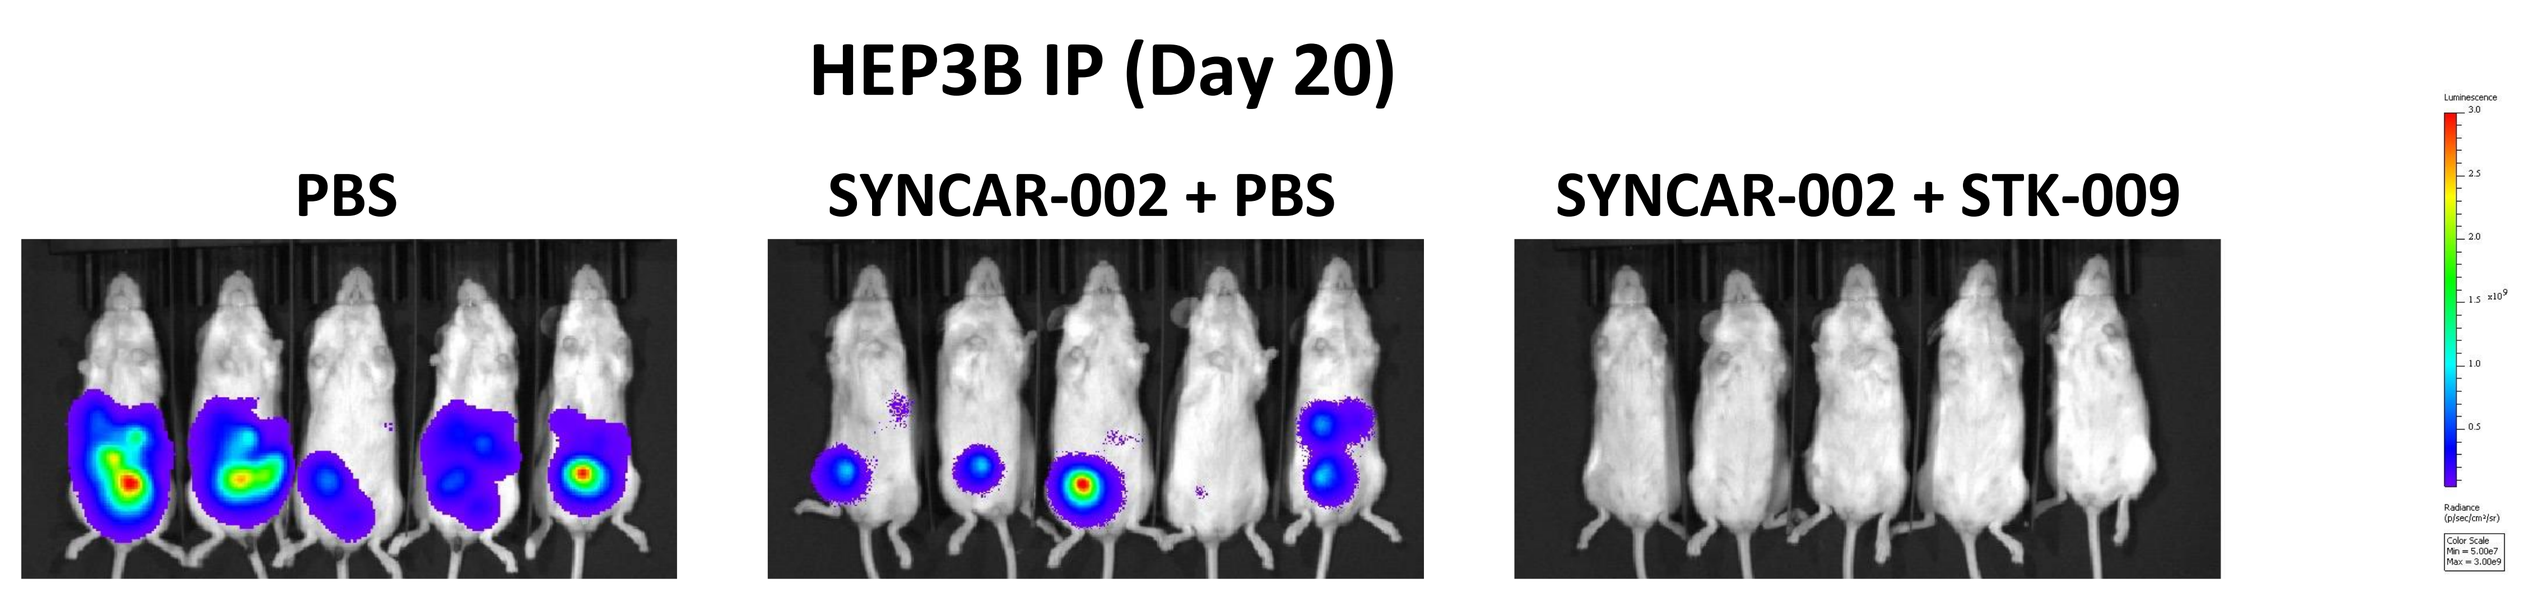
HEP3B IP (Day 20)
PBS
SYNCAR-002 + PBS
SYNCAR-002 + STK-009

STK-009 significantly increases intratumoral SYNCAR-002 T cell levels, proliferation, and effector activity

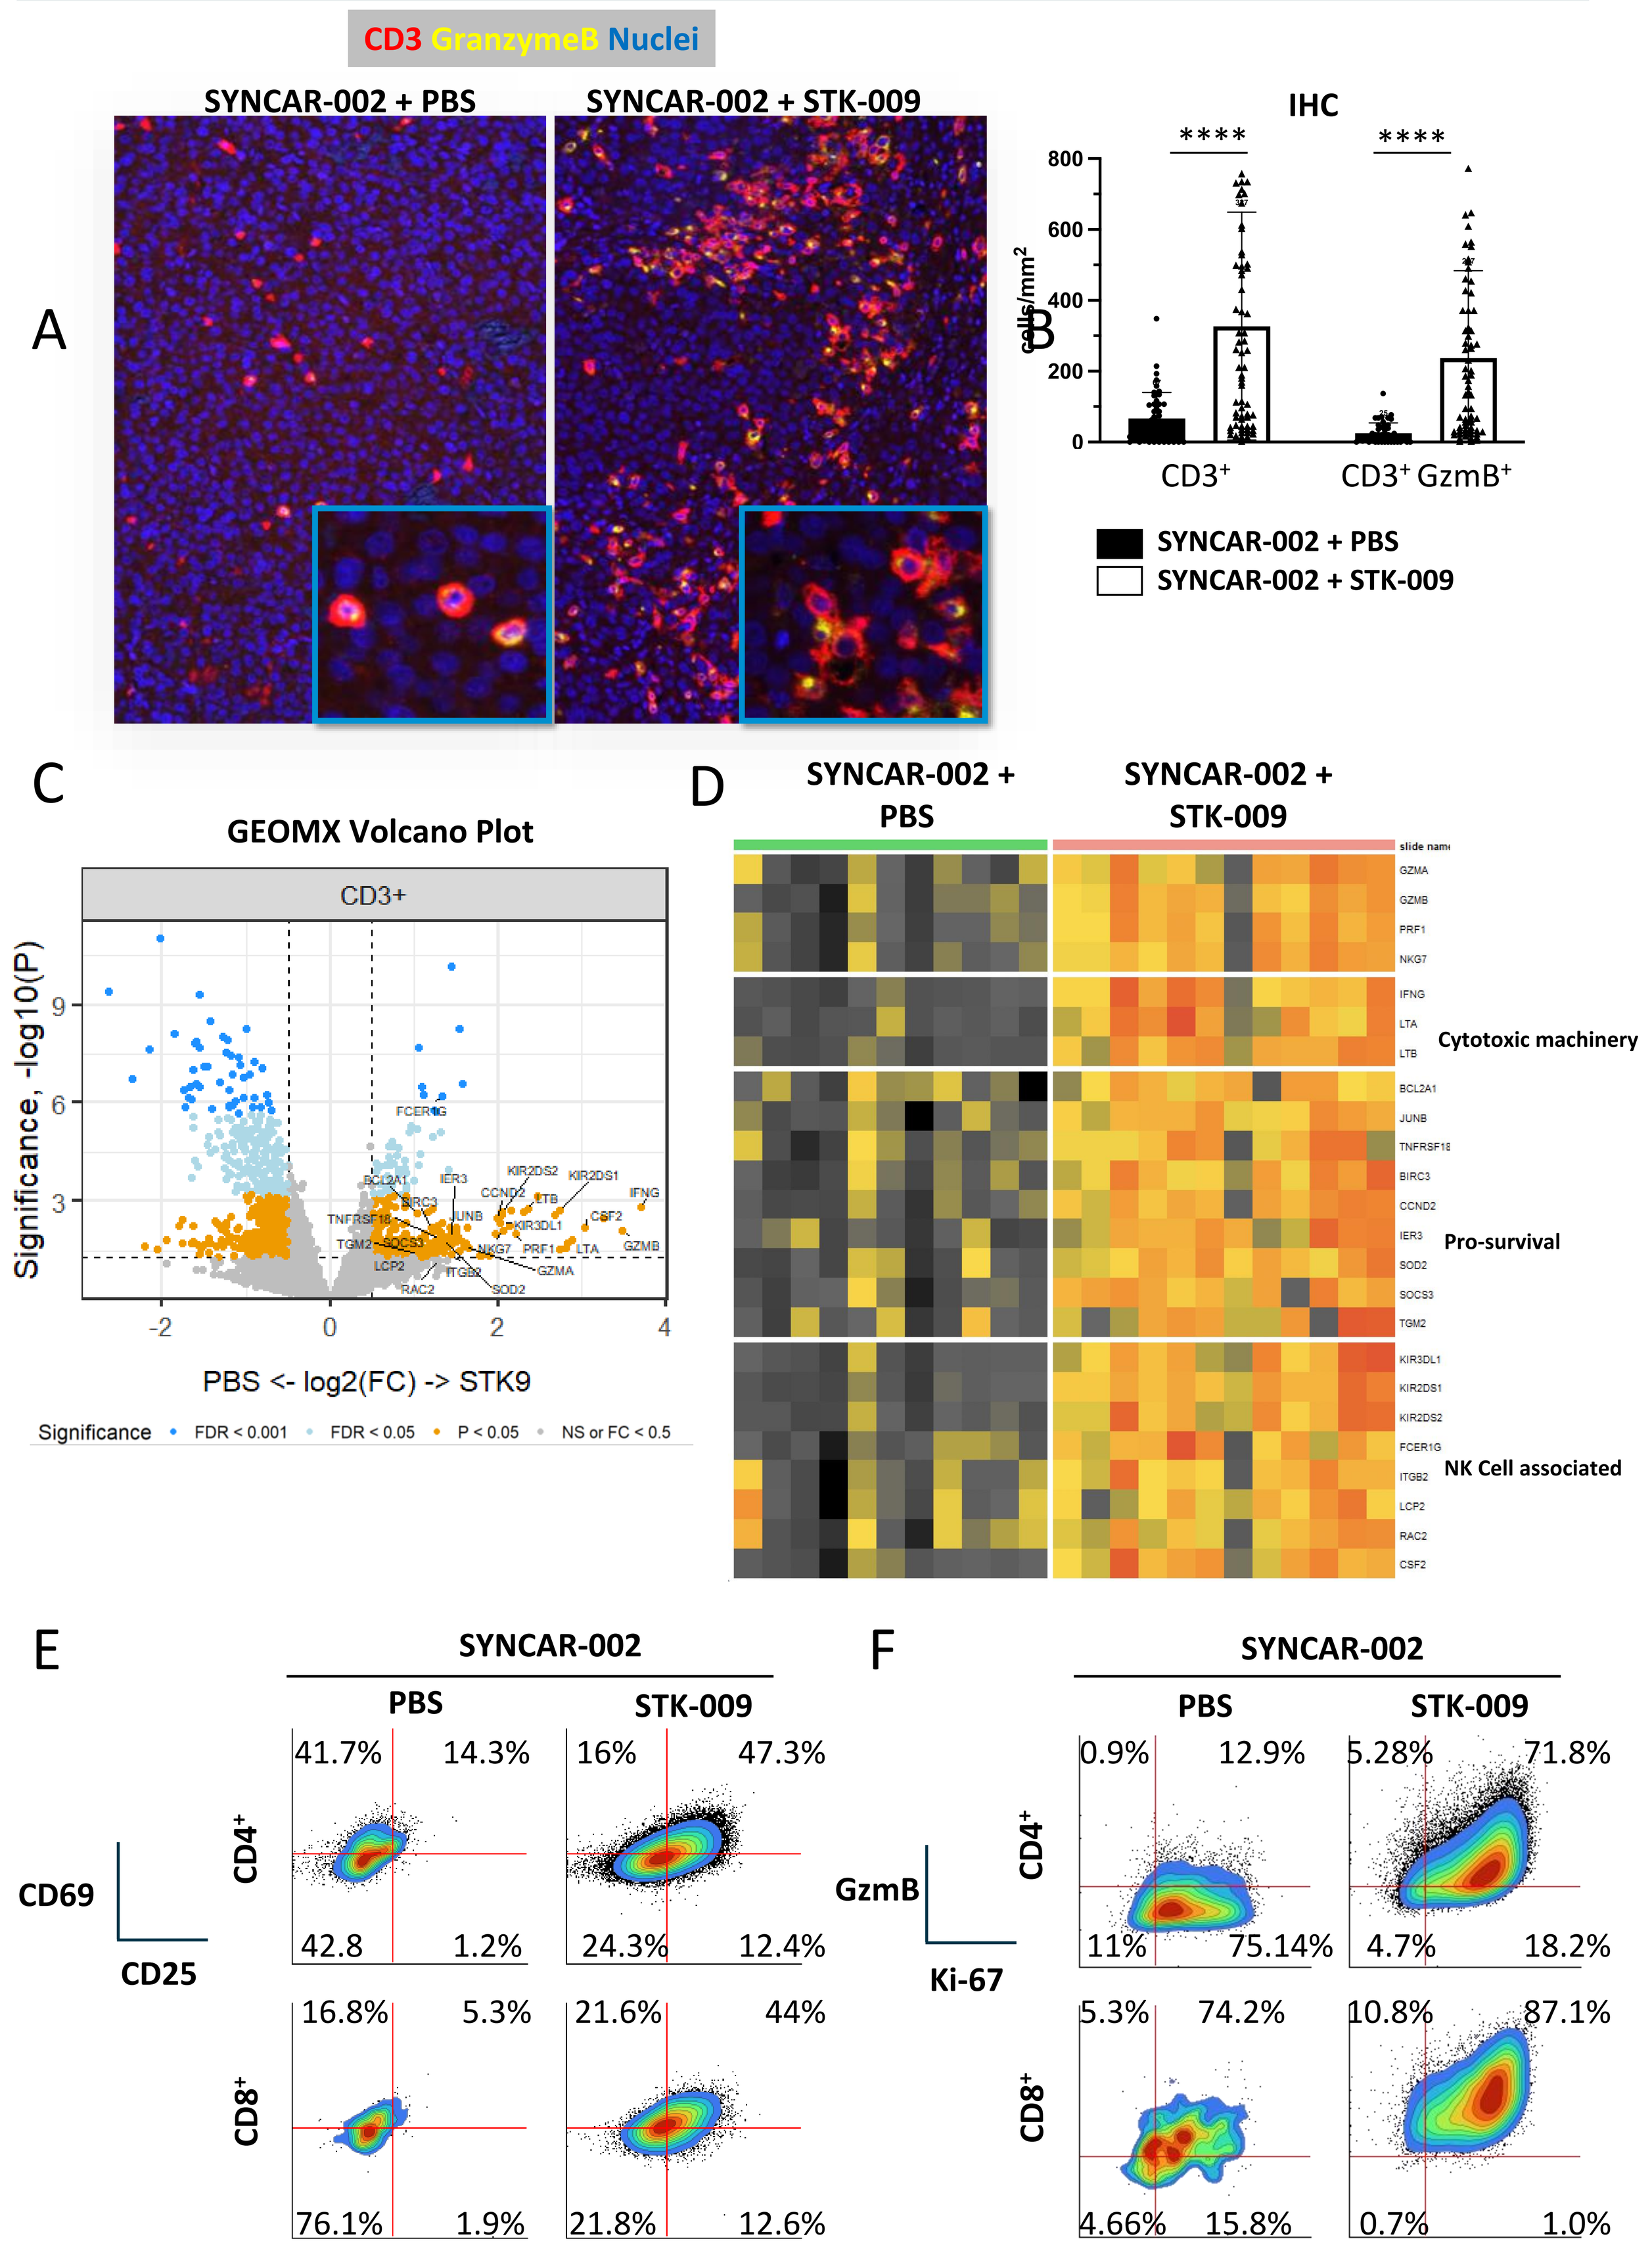
CD3 GranzymeB Nuclei
A
SYNCAR-002 + PBS
SYNCAR-002 + STK-009
B
IHC
cells/mm²

CD3+
CD3+ GzmB+
SYNCAR-002 + PBS
SYNCAR-002 + STK-009
C
GEOMX Volcano Plot
CD3+
Significance, -log10(P)
PBS <- log2(FC) -> STK9
Significance
FDR < 0.001
FDR < 0.05
P < 0.05
NS or FC < 0.5
D
SYNCAR-002 + PBS
SYNCAR-002 + STK-009
Cytotoxic machinery
Pro-survival
NK Cell associated
E
SYNCAR-002
PBS
STK-009
CD69
CD25
CD4+
CD8+
41.7%
14.3%
42.8
1.2%
16%
47.3%
24.3%
12.4%
16.8%
5.3%
76.1%
1.9%
21.6%
44%
21.8%
12.6%
F
GzmB
Ki-67
0.9%
12.9%
11%
75.14%
5.28%
71.8%
4.7%
18.2%
5.3%
74.2%
4.66%
15.8%
10.8%
87.1%
0.7%
1.0%

Assessment of intratumoral SYNCAR-002 T cell levels and activation. For immunofluorescence, HEPG2 tumors were taken down 14 days post-SYNCAR-002 T cell transfer. **(A)** IHC of tumors: hCD3 (red), hGranzyme B (yellow), and nuclei (blue). **(B)** Quantitation of hCD3⁺ and hCD3⁺ hGzmB⁺ cells. ****p-value <0.0001. **(C&D)** GeoMX analysis of hCD3⁺ cells from tumors depicting differentially expressed genes as a volcano plot and specific genes involved in cytotoxicity, survival, and NK cells as a heatmap. **(E&F)** Flow analysis of SYNCAR-002 cells isolated from tumors.

Mice previously cured by SYNCAR-002 + STK-009 withstand HEPG2 tumor rechallenge

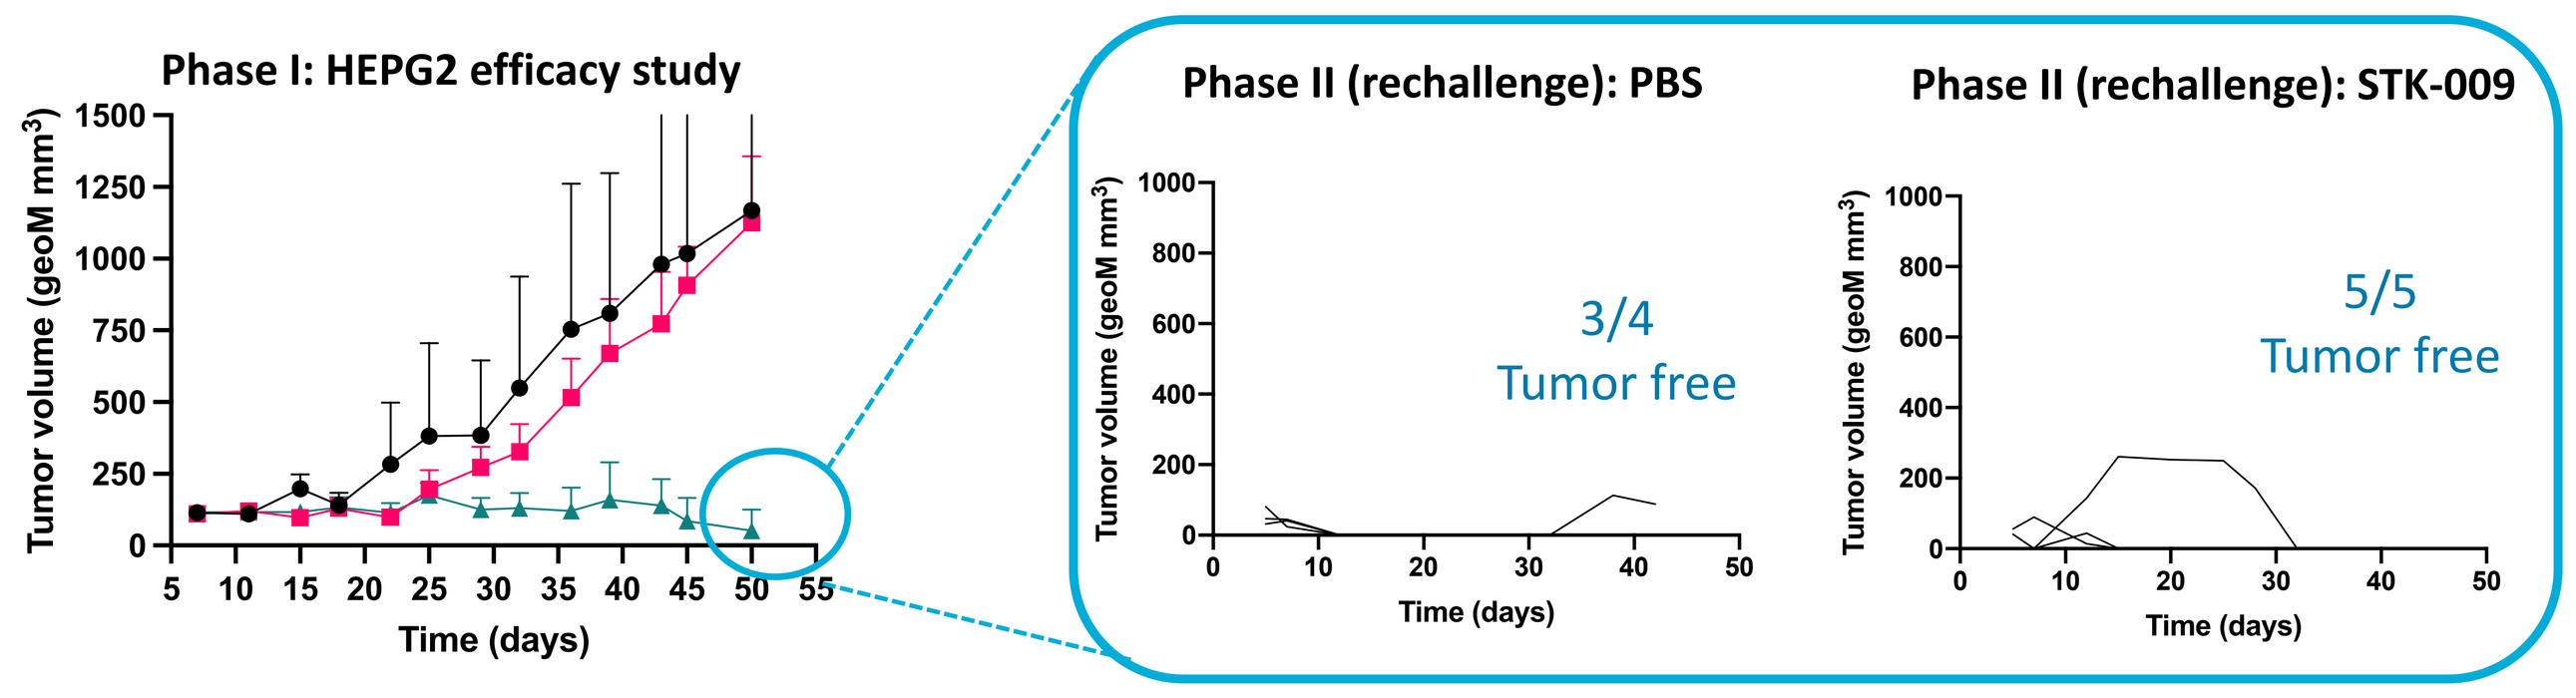
Phase I: HEPG2 efficacy study
Tumor volume (geoM mm³)
Time (days)
Phase II (rechallenge): PBS
3/4
Tumor free
Phase II (rechallenge): STK-009
5/5
Tumor free

HEPG2 rechallenge model. Mice with subcutaneous HEPG2 tumors that received SYNCAR-002 at Day 7 and subsequent doses of STK-009 were cured by Day 50. HEPG2 + Matrigel were implanted into these mice at Day 55 and into naïve mice. Previously cured mice were split into two groups receiving either PBS (n=4) or STK-009 2x/week (n=5).

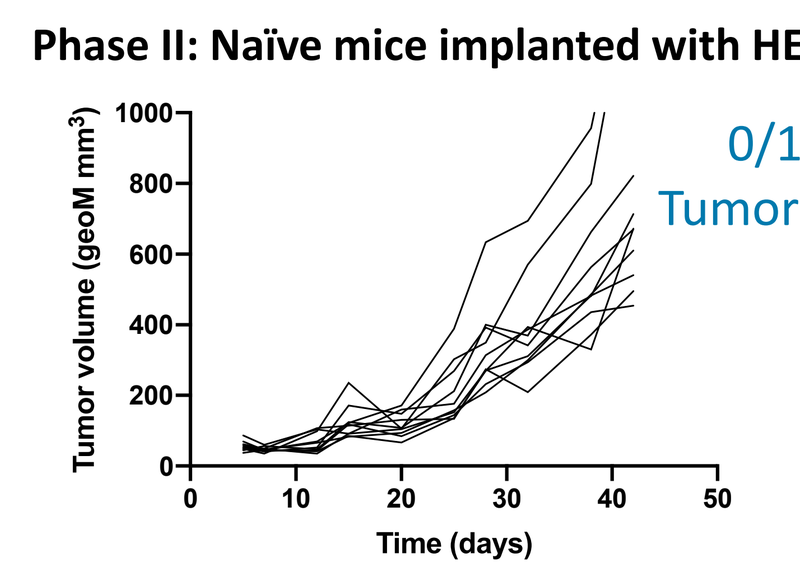
Phase II: Naïve mice implanted with HEPG2
Tumor volume (geoM mm³)
Time (days)
0/10
Tumor free

CONCLUSION

Adoptive T cell therapies, including CAR T cells, are increasingly recognized as important modalities in the treatment of cancer. However, CAR Ts and other ACTs suffer from several challenges that limit their effectiveness and utilization, including poor expansion and short persistence *in vivo*, diminished activity in the tumor microenvironment, and toxicities related to high initial CAR-T doses and uncontrolled expansion. We have designed the STK-009/*ortho*IL2Rβ to address these challenges with CAR-Ts. SYNCAR-002, a GPC3 CAR-T powered by STK-009, demonstrates:

- **Improved anti-tumor efficacy in HCC solid tumor xenograft models**
- **Increased levels and persistence of CAR T cells *in vivo***
- **Increased activation of intratumoral CAR T cells to overcome a hostile tumor microenvironment**

Therefore, the STK-009/SYNCAR platform has the potential to overcome clinically relevant hurdles in cell therapy especially in solid epithelial tumor indications.

[1]Aspuria et al. An Orthogonal IL-2 and IL-2Rb System Drives Persistence and Activation of CAR T cells and Bulky Lymphoma. Science Translational Medicine. 13(625). Dec 2021.