CT183: Initial Results From a Phase 1a/1b Study of STK-012, a First-in-Class α/β IL-2 Receptor Biased Partial Agonist in Advanced Solid Tumors (NCT05098132)

Benjamin Izar[1], Dmitriy Zamarin[2], David R. Spigel[3], Christopher J. Hoimes[4], David F. McDermott[5], Kartik Sehgal[6], Yana G. Najjar[7], Adam J. Schoenfeld[8], Edward B. Garon[9], Ryan J. Sullivan[10], Brian S. Henick[1], Ticiana A. Leal[11], Michael E. Hurwitz[12], Rana R. McKay[13], Alex Azrilevich[14], Naiyer A. Rizvi[14], Martin Oft[14], Anita Mehta Damani[14], Natalie Busby[14], Tony Tran[14], Alexander I. Spira[15]

[1]Columbia University Irving Medical Center, New York, NY, [2]Mount Sinai, New York, NY, [3]Sarah Cannon Research Institute, Nashville, TN, [4]Duke Cancer Institute, Durham, NC, [5]Beth Israel Deaconess Medical Center, Boston, MA, [6]Dana Farber Cancer Institute, Boston, MA, [7]UPMC Hillman Cancer Center, Pittsburgh, PA, [8]Memorial Sloan Kettering Cancer Center, New York, NY, [9]University of California, Los Angeles, Los Angeles, CA, [10]Massachusetts General Hospital, Boston, MA, [11]Emory University, Atlanta, GA, [12]Yale Cancer Center, New Haven, CT, [13]University of California at San Diego, San Diego, CA, [14]Synthekine, Menlo Park, CA, [15]NEXT Oncology Virginia, Fairfax, VA

Synthekine

Background

- High dose intravenous (IV) interleukin-2 (IL-2) induces deep and durable responses in certain cancers, but its use is limited due to toxicities, including severe hypotension and capillary leak syndrome (CLS), and the requirement for inpatient administration.
- Recent approaches to develop IL-2 therapies with an expanded therapeutic index have targeted the dimeric form (β/γ) of the IL-2 receptor, which is predominantly expressed on naïve T cells and NK cells, rather than the high-affinity trimeric form (α/β/γ), which is highly expressed on antigen activated T cells and constitutively expressed at a low level on Tregs.
- Recent findings support that IL-2Rα engagement is essential for optimal antitumor efficacy of IL-2.[1-4]
- STK-012 is a first-in-class α/β-IL-2R biased partial agonist designed to drive antitumor activity by selectively stimulating CD25+ antigen activated T cells, while avoiding hallmark IL-2 toxicities by sparing pleotropic activation of lymphocytes including NK cells.

Table 1. IL-2R Targeting Approaches

	1st Generation[5]	2nd Generation Engineered IL-2[5]	STK-012
IL-2 Construct	High dose IL-2 (Aldesleukin)	"Non-α" IL-2	α/β-biased IL-2
IL-2R Bias	No bias / Binds to high & intermediate affinity IL-2R	Dimeric IL-2 receptor / Binds to intermediate affinity IL-2R	Trimeric IL-2 receptor / Binds to high affinity receptor IL-2R
IL-2R Subunit Sparing	None	Reduced or No binding to IL-2Rα	Reduced binding to IL-2Rγ
Cell Selectivity	No selectivity	NK cells and naïve T cells	Antigen-activated T cells

STK-012-101 Phase 1a/1b Study Design and Status

- STK-012-101 is a Phase 1a/1b first-in-human study of STK-012 administered subcutaneously (SC) in subjects with advanced relapsed/refractory (r/r) solid tumors.
- During the Phase 1a, subjects were enrolled in a 3+3 dose escalation to
 - Part A: STK-012 monotherapy weekly (QW) over 2 dose levels (DL)
 - Part B: STK-012 monotherapy every 3 weeks (Q3W) over 5 DLs
 - Part C: STK-012 Q3W in combination with pembrolizumab 200 mg Q3W over 4 DLs
- During Phase 1b expansions, subjects were enrolled to
 - Part D: STK-012 monotherapy Q3W expansion at the candidate RP2D
- Escalation in Parts A-C has been completed. Data presentation will be restricted to Parts A and B (Ph1a STK-012 monotherapy).

Figure 1. STK-012-101 Study Design

- Part A: STK-012 QW escalation 0.375 mg → 0.75 mg
- Part B: STK-012 Q3W escalation 0.75 mg → 3 mg
- Part C: STK-012 Q3W escalation 0.75 mg → 3 mg + Pembro Q3W
- Part D: STK-012 Ph1b expansions at 2.25 mg Q3W

Candidate RP2D: 2.25 mg Q3W

Enrolling to Ph1b monotherapy expansions and biomarker backfill

Disposition, Demographics, and Baseline Characteristics

Disposition

- As of February 26th, 2024, 47 subjects were treated with STK-012 monotherapy at 7 dose levels across the 2 dosing schedules (QW and Q3W) in the Phase 1a.
- 8 subjects (17%) remained on treatment at time of data extract.

Demographics

- The median age was 63.1 years.
- 44.3% subjects were female.
- 42.6% had an ECOG PS of 0 and 57.4% had ECOG PS of 1.

Table 2. STK-012 Ph1a Monotherapy Baseline Disease Characteristics, N=47

Prior lines in advanced setting	
1	10 (21.3)
2	9 (19.1)
≥3	28 (59.6)
Prior immune checkpoint inhibitor	
0	10 (21.2)
≥1	37 (78.7)
Disease type	
Non-Small Cell Lung Cancer	15 (31.9)
Clear Cell Renal Cell Carcinoma	6 (12.8)
Ovarian Cancer	6 (12.8)
Mucosal Melanoma	4 (8.5)
Other (HNSCC, GEJ, GC, CC, non-ccRCC and MSI-H/dMMR)	13 (27.7)

STK-012 Monotherapy Safety

- Treatment-related adverse events (TRAEs) were reversible with standard management (eg, dose hold/reduction, supportive meds).
- No subjects experienced CLS and very few had associated TRAEs (Table 4)
- No dose-limiting toxicities (DLTs) were observed during the DLT period (Cycle 1 [21 days]).
- The majority of TRAEs were Grade 1 or 2 in severity (Table 3); 1 subject experienced a Grade 4 TRAE and no subjects experienced Grade 5 TRAEs.
 - The Grade 4 TRAE of Anaphylactic reaction occurred at C9D1 (1.5 mg Q3W) and led to treatment discontinuation; the event resolved on the same day with supportive treatment.
- All 3 subjects treated at the maximum dose of 3 mg Q3W experienced Grade 3 GI SAEs (Small bowel obstruction, Enterocolitis, Vomiting) in Cycle 2 (after the DLT period).
- An intermediate dose of 2.25 mg Q3W was then explored and selected as candidate RP2D.

Table 3. Most Common TRAEs (in ≥10% of subjects) on STK-012 monotherapy, N=47

TRAE	All Grade, N (%)	Grade ≥3, N (%)
Rash maculo-papular	19 (40.4)	5 (10.6)
Fatigue	12 (25.5)	1 (2.1)
Injection site reaction	11 (23.4)	0
Nausea	11 (23.4)	1 (2.1)
Diarrhea	9 (19.9)	2 (4.3)
Pruritis	7 (14.9)	0
Vomiting	6 (12.8)	2 (4.3)
Arthralgia	5 (10.6)	1 (2.1)
Total (All TRAEs)	**37 (78.7)**	**15 (31.9)**

Table 4. Capillary Leak associated TRAEs on STK-012 monotherapy, N=47

TRAE	All Grade, N (%)	Grade ≥3, N (%)
Hypotension	2 (4.3)	0
Pyrexia	2 (4.3)	0
Flu-like symptoms	2 (4.3)	0
Peripheral edema	2 (4.3)	0
CRS	1 (2.1)	0
Creatinine ↑	1 (2.1)	1 (2.1)
CLS	0	0
LFT increase	0	0
Lymphopenia	0	0

STK-012 Preliminary PK-PD Data in Part B Cohorts (Q3W)

Fig 2. Serum PK

- Observed Half-life is ~4 days.
- Exposure (AUC) is dose proportional.

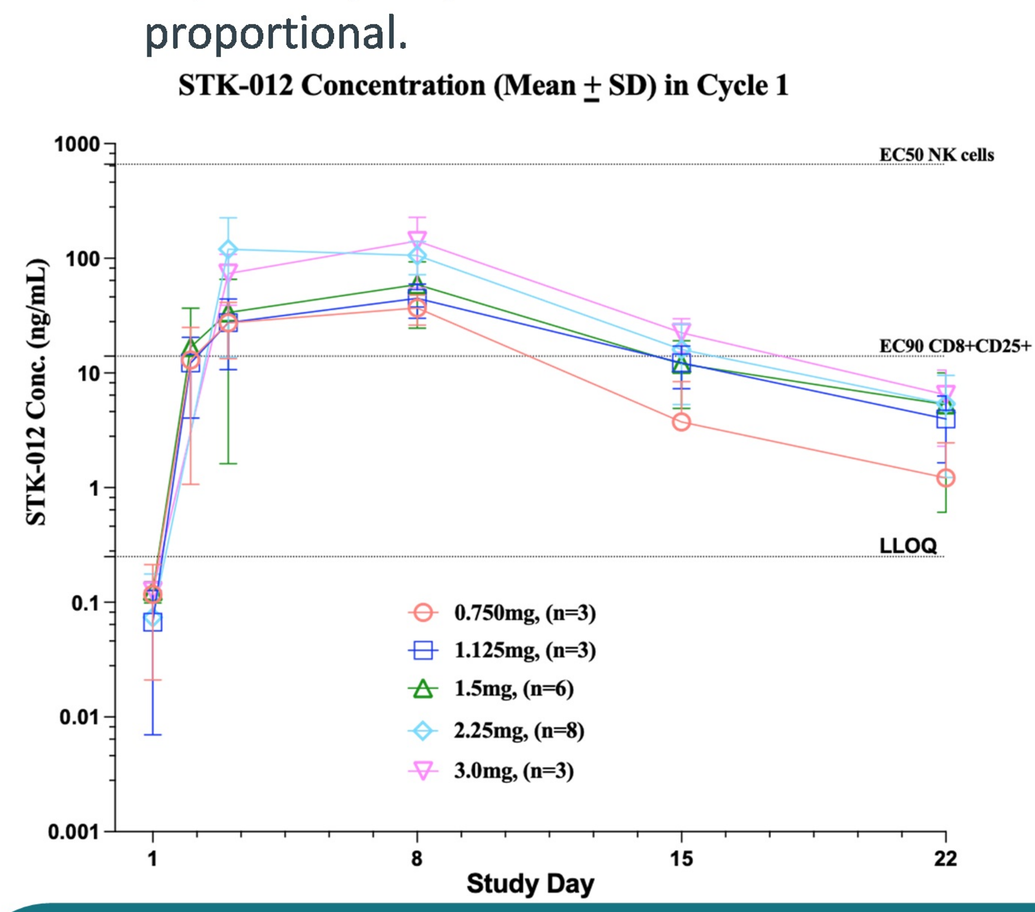
STK-012 Concentration (Mean ± SD) in Cycle 1
EC50 NK cells
EC90 CD8+CD25+
LLOQ
STK-012 Conc. (ng/mL)
Study Day
0.750mg, (n=3)
1.125mg, (n=3)
1.5mg, (n=6)
2.25mg, (n=8)
3.0mg, (n=3)

Fig 3. pSTAT5

- STK-012 demonstrates selectivity for T cells that express CD25 (IL-2Rα)

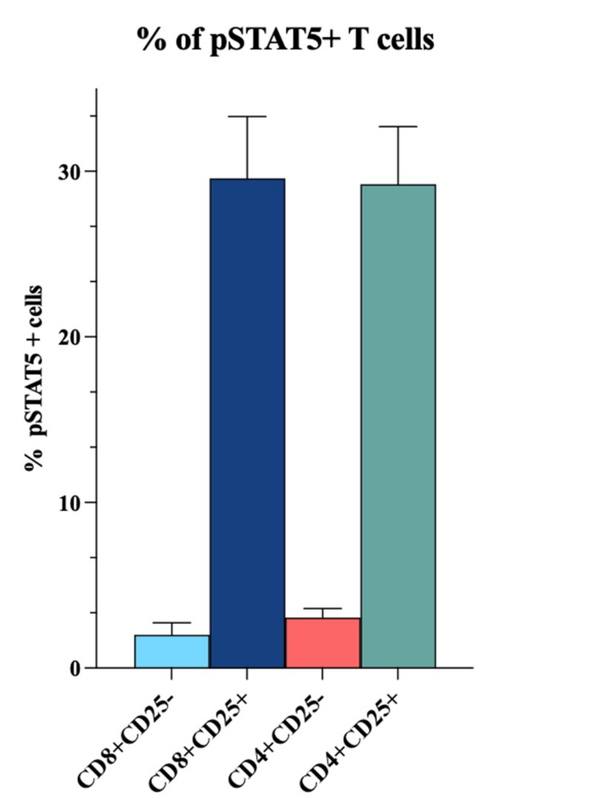
% of pSTAT5+ T cells
% pSTAT5 + cells
CD8+CD25-
CD8+CD25+
CD4+CD25-
CD4+CD25+

Fig 4. Serum IFNγ

- Serum IFNγ increase is dose dependent and peaks approximately at STK-012 Tmax (Day 8)

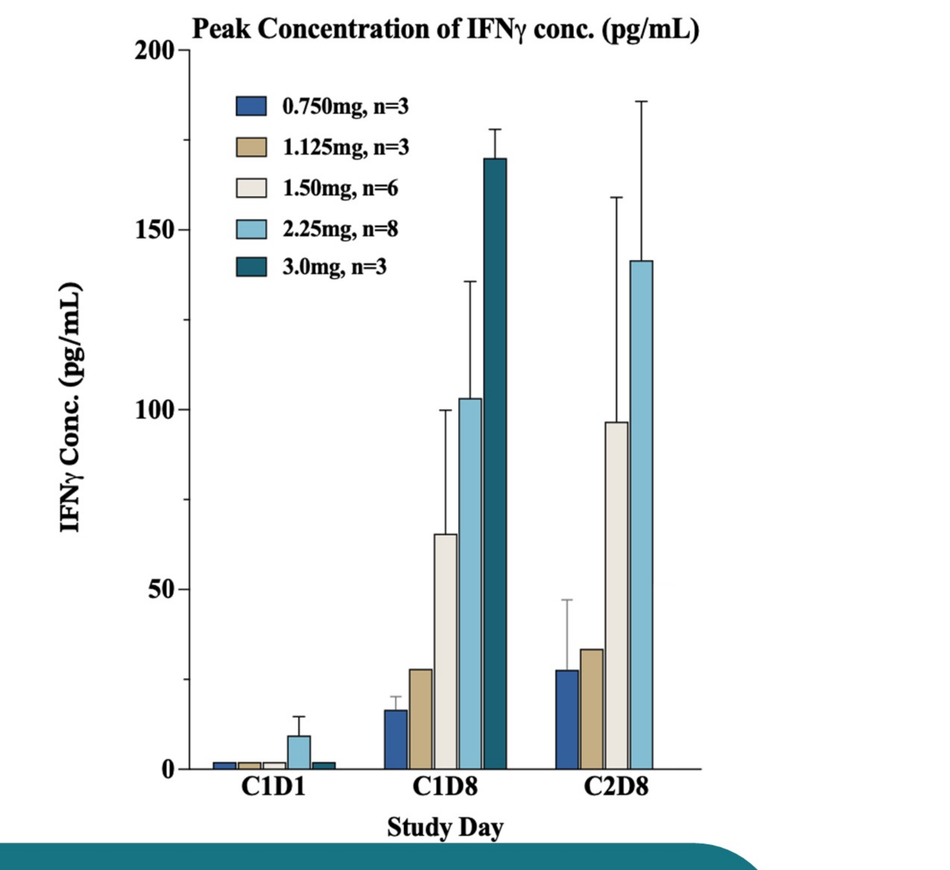
Peak Concentration of IFNγ conc. (pg/mL)
0.750mg, n=3
1.125mg, n=3
1.50mg, n=6
2.25mg, n=8
3.0mg, n=3
IFNγ Conc. (pg/mL)
C1D1
C1D8
C2D8
Study Day

Fig 5a, 5b: Peripheral Flow Cytometry Supports Selectivity for Antigen Activated CD8 T cells

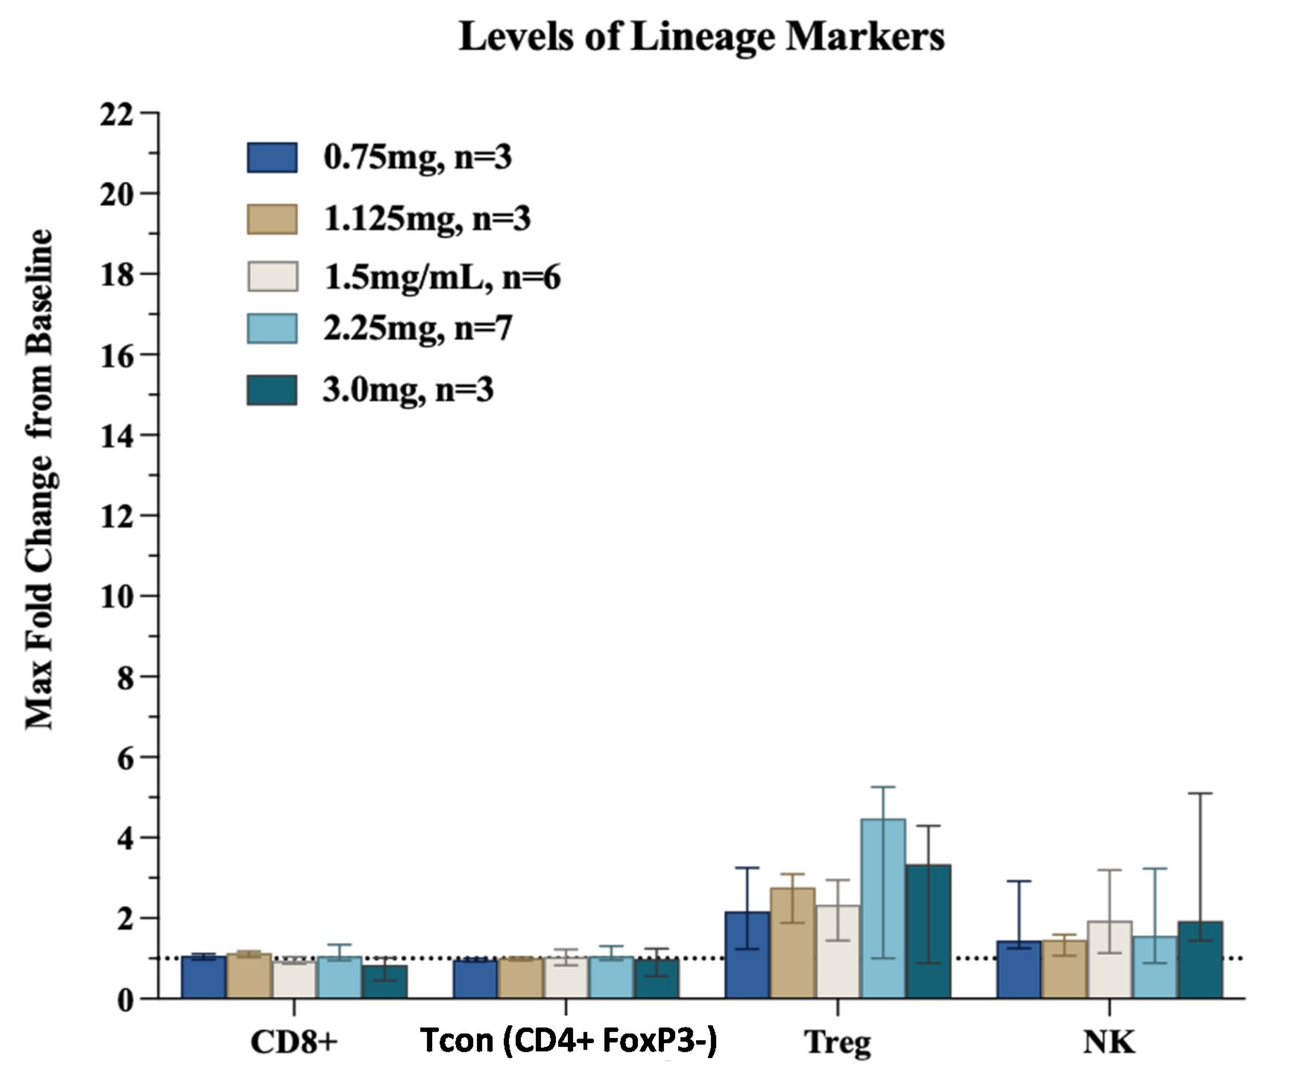
Levels of Lineage Markers
0.75mg, n=3
1.125mg, n=3
1.5mg/mL, n=6
2.25mg, n=7
3.0mg, n=3
Max Fold Change from Baseline
CD8+
Tcon (CD4+ FoxP3-)
Treg
NK

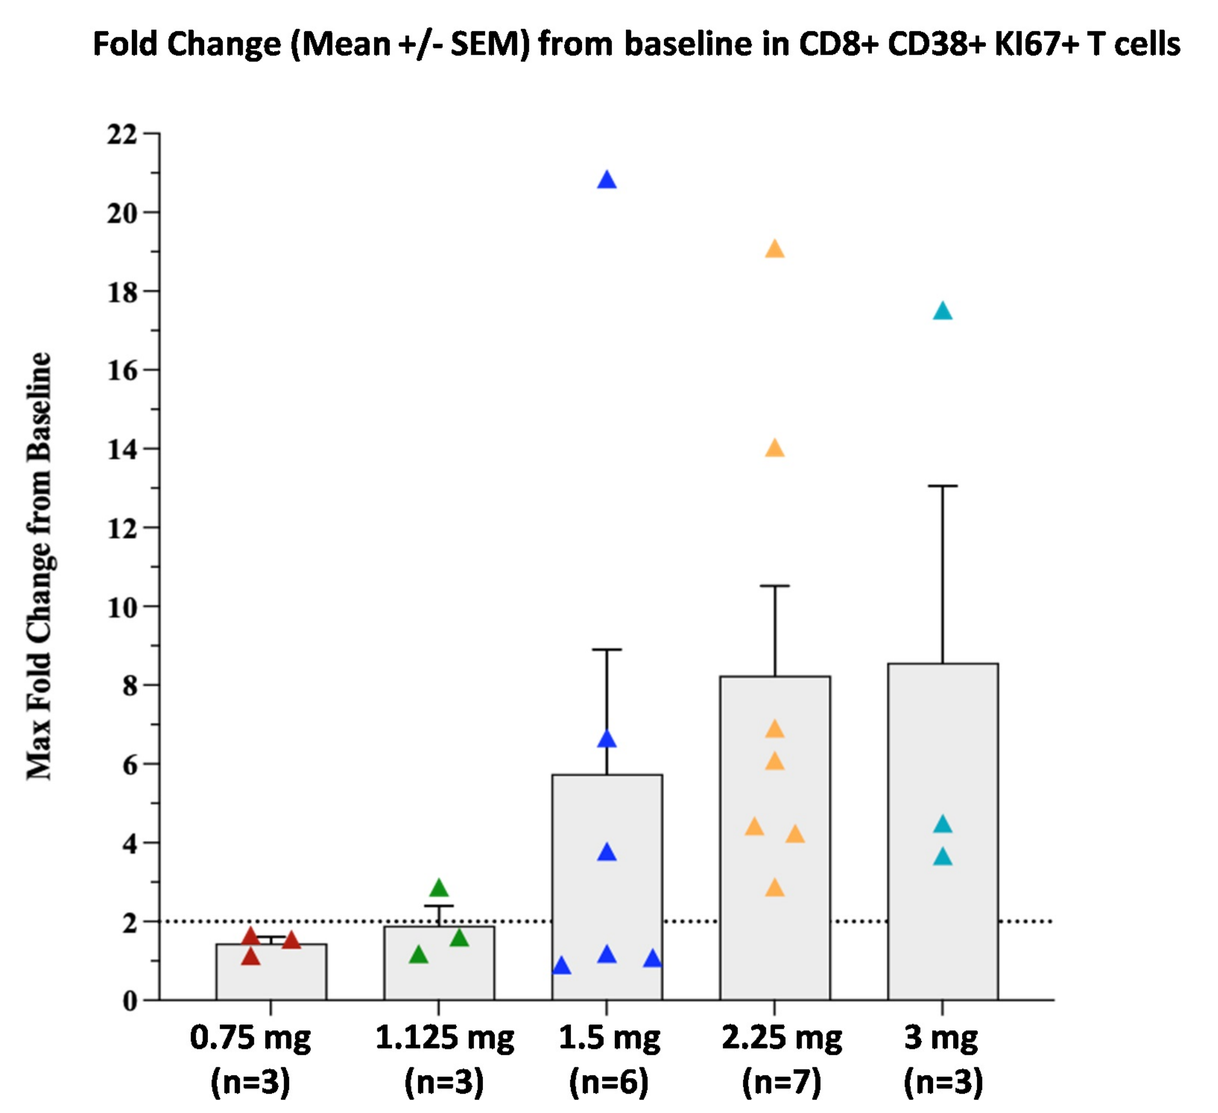
Fold Change (Mean +/- SEM) from baseline in CD8+ CD38+ KI67+ T cells
Max Fold Change from Baseline
0.75 mg (n=3)
1.125 mg (n=3)
1.5 mg (n=6)
2.25 mg (n=7)
3 mg (n=3)

STK-012 Monotherapy Efficacy

- Of 40 efficacy evaluable# subjects, 3 had partial response (PR) and 12 had stable disease (SD) as their best overall response (BOR) by RECIST V1.1.
- The partial responses included 2 confirmed (anti-PD-1 pretreated NSCLC and RCC) and 1 unconfirmed (anti-PD-1 pretreated HNSCC).

Figure 6. Waterfall for Phase 1a Monotherapy

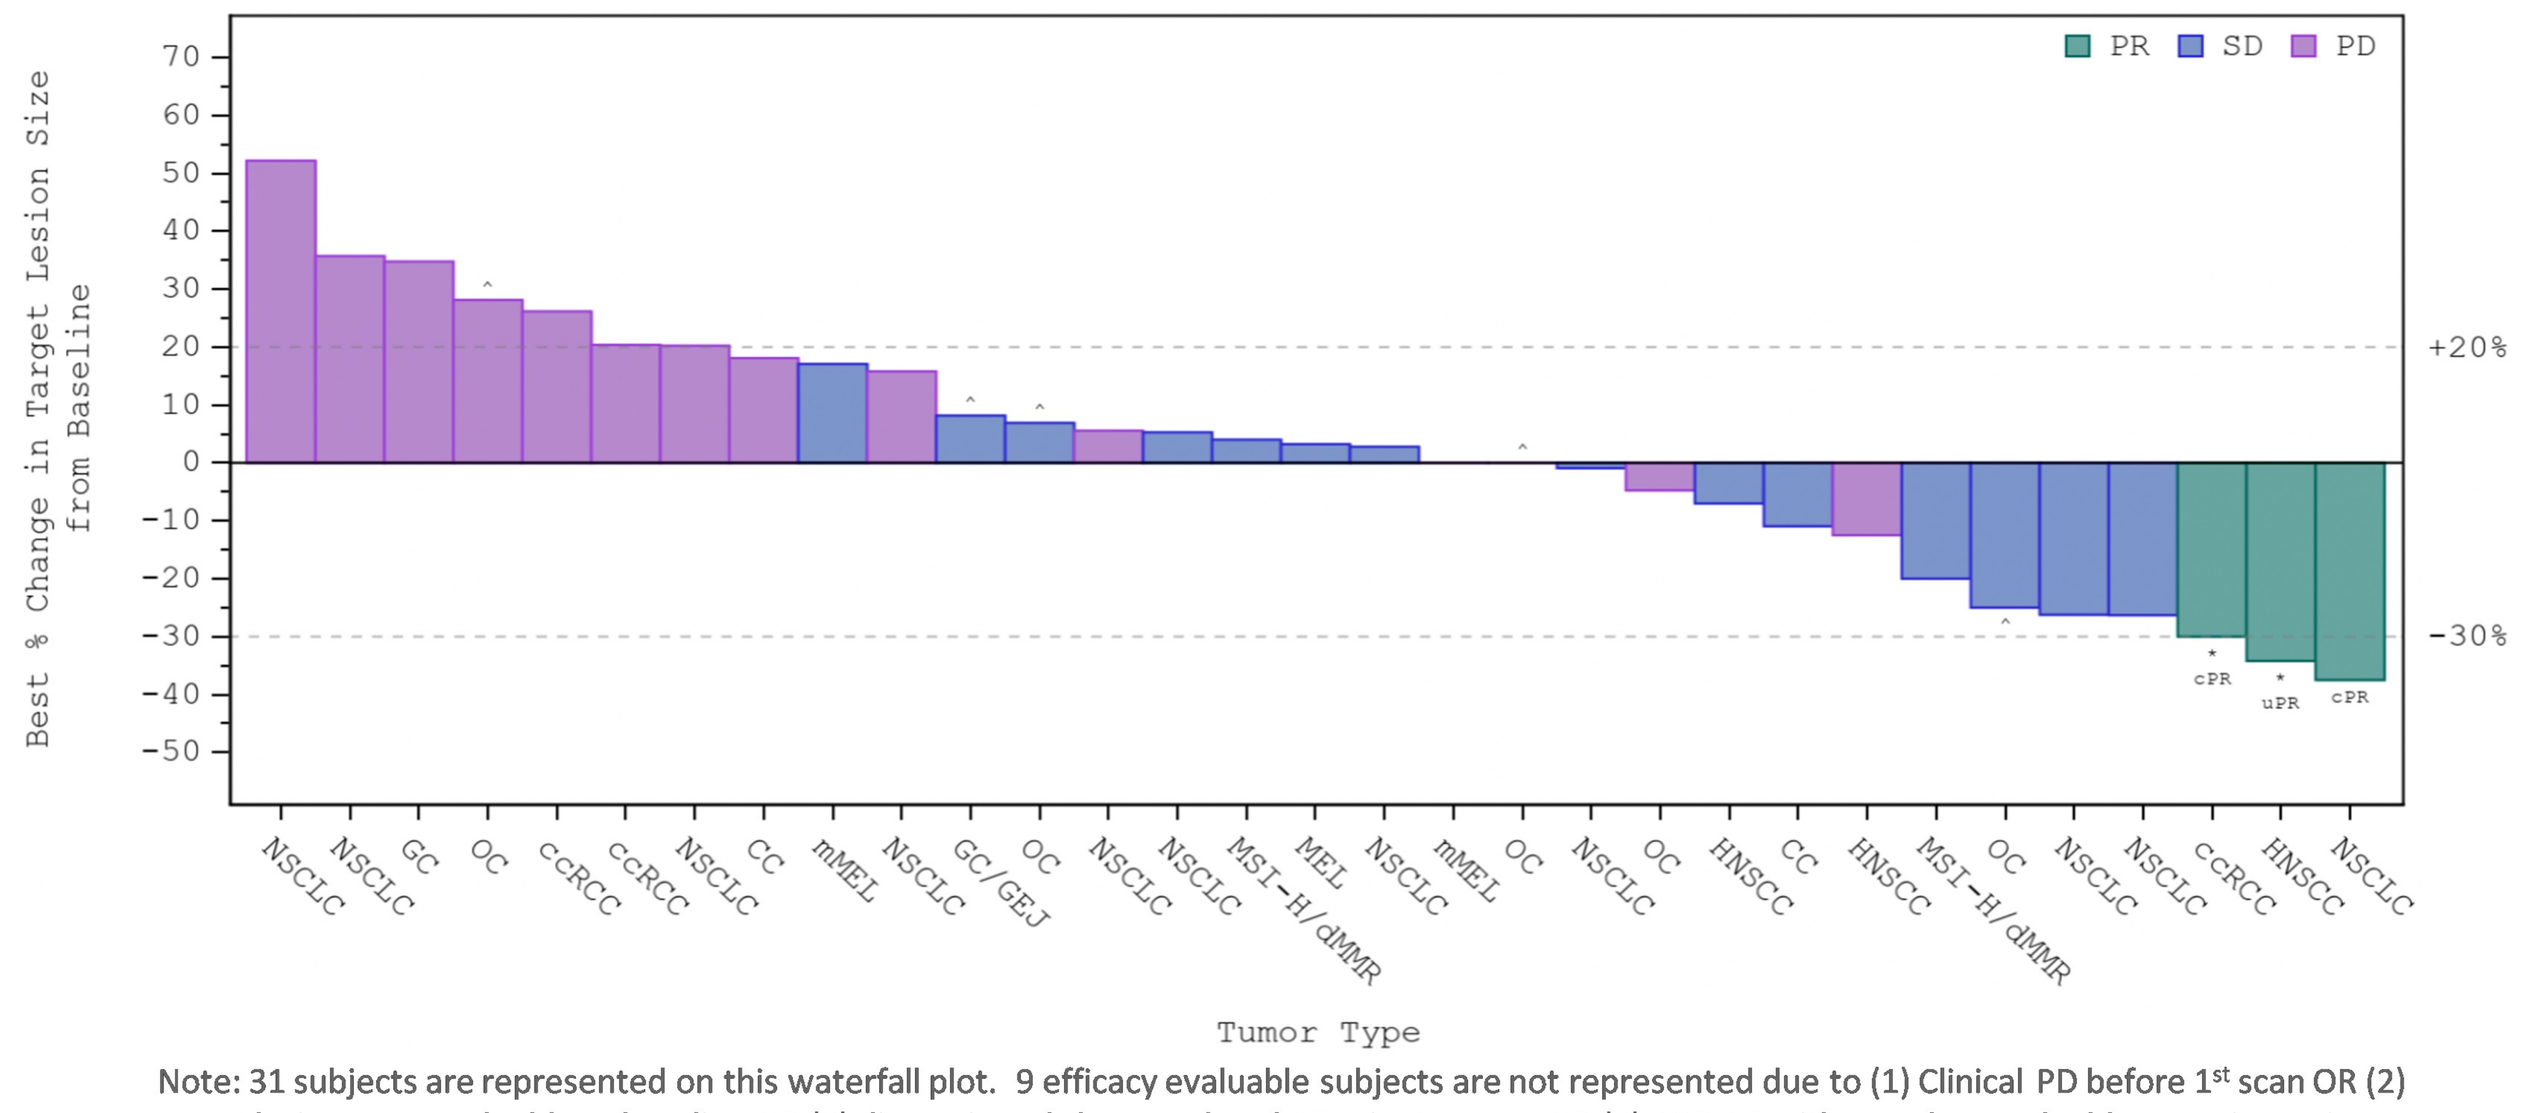
PR
SD
PD
Best % Change in Target Lesion Size from Baseline
+20%
-30%
cPR
uPR
cPR
NSCLC
NSCLC
GC
OC
ccRCC
ccRCC
NSCLC
CC
mMEL
NSCLC
GC/GEJ
OC
NSCLC
NSCLC
MSI-H/dMMR
MEL
NSCLC
mMEL
OC
NSCLC
OC
HNSCC
CC
HNSCC
MSI-H/dMMR
OC
NSCLC
NSCLC
ccRCC
HNSCC
NSCLC
Tumor Type

Note: 31 subjects are represented on this waterfall plot. 9 efficacy evaluable subjects are not represented due to (1) Clinical PD before 1st scan OR (2) target lesions not evaluable at baseline OR (3) discontinued due to related AE prior to scans OR (4) BOR NE with no other evaluable scan timepoints.
- Subjects are censored after the first reporting of radiographic or clinical PD.
* Indicates subjects who are ongoing treatment; ^ indicates subjects who are IO Naive.

Greater Efficacy in IO Pretreated Subjects With 1-2 Prior Lines

- In subjects who progressed on/after prior anti-PD-1 who also had 1-2 prior lines in the advanced setting (N=15 efficacy evaluable), 3 had BOR of PR and 6 BOR of SD
- Durability of effect was observed in multiple subjects with BOR of SD and the ccRCC responder with sustained response for >9 months.

Figure 7. Waterfall for Ph1a Monotherapy (IO Pretreated, 1-2 Prior Lines)

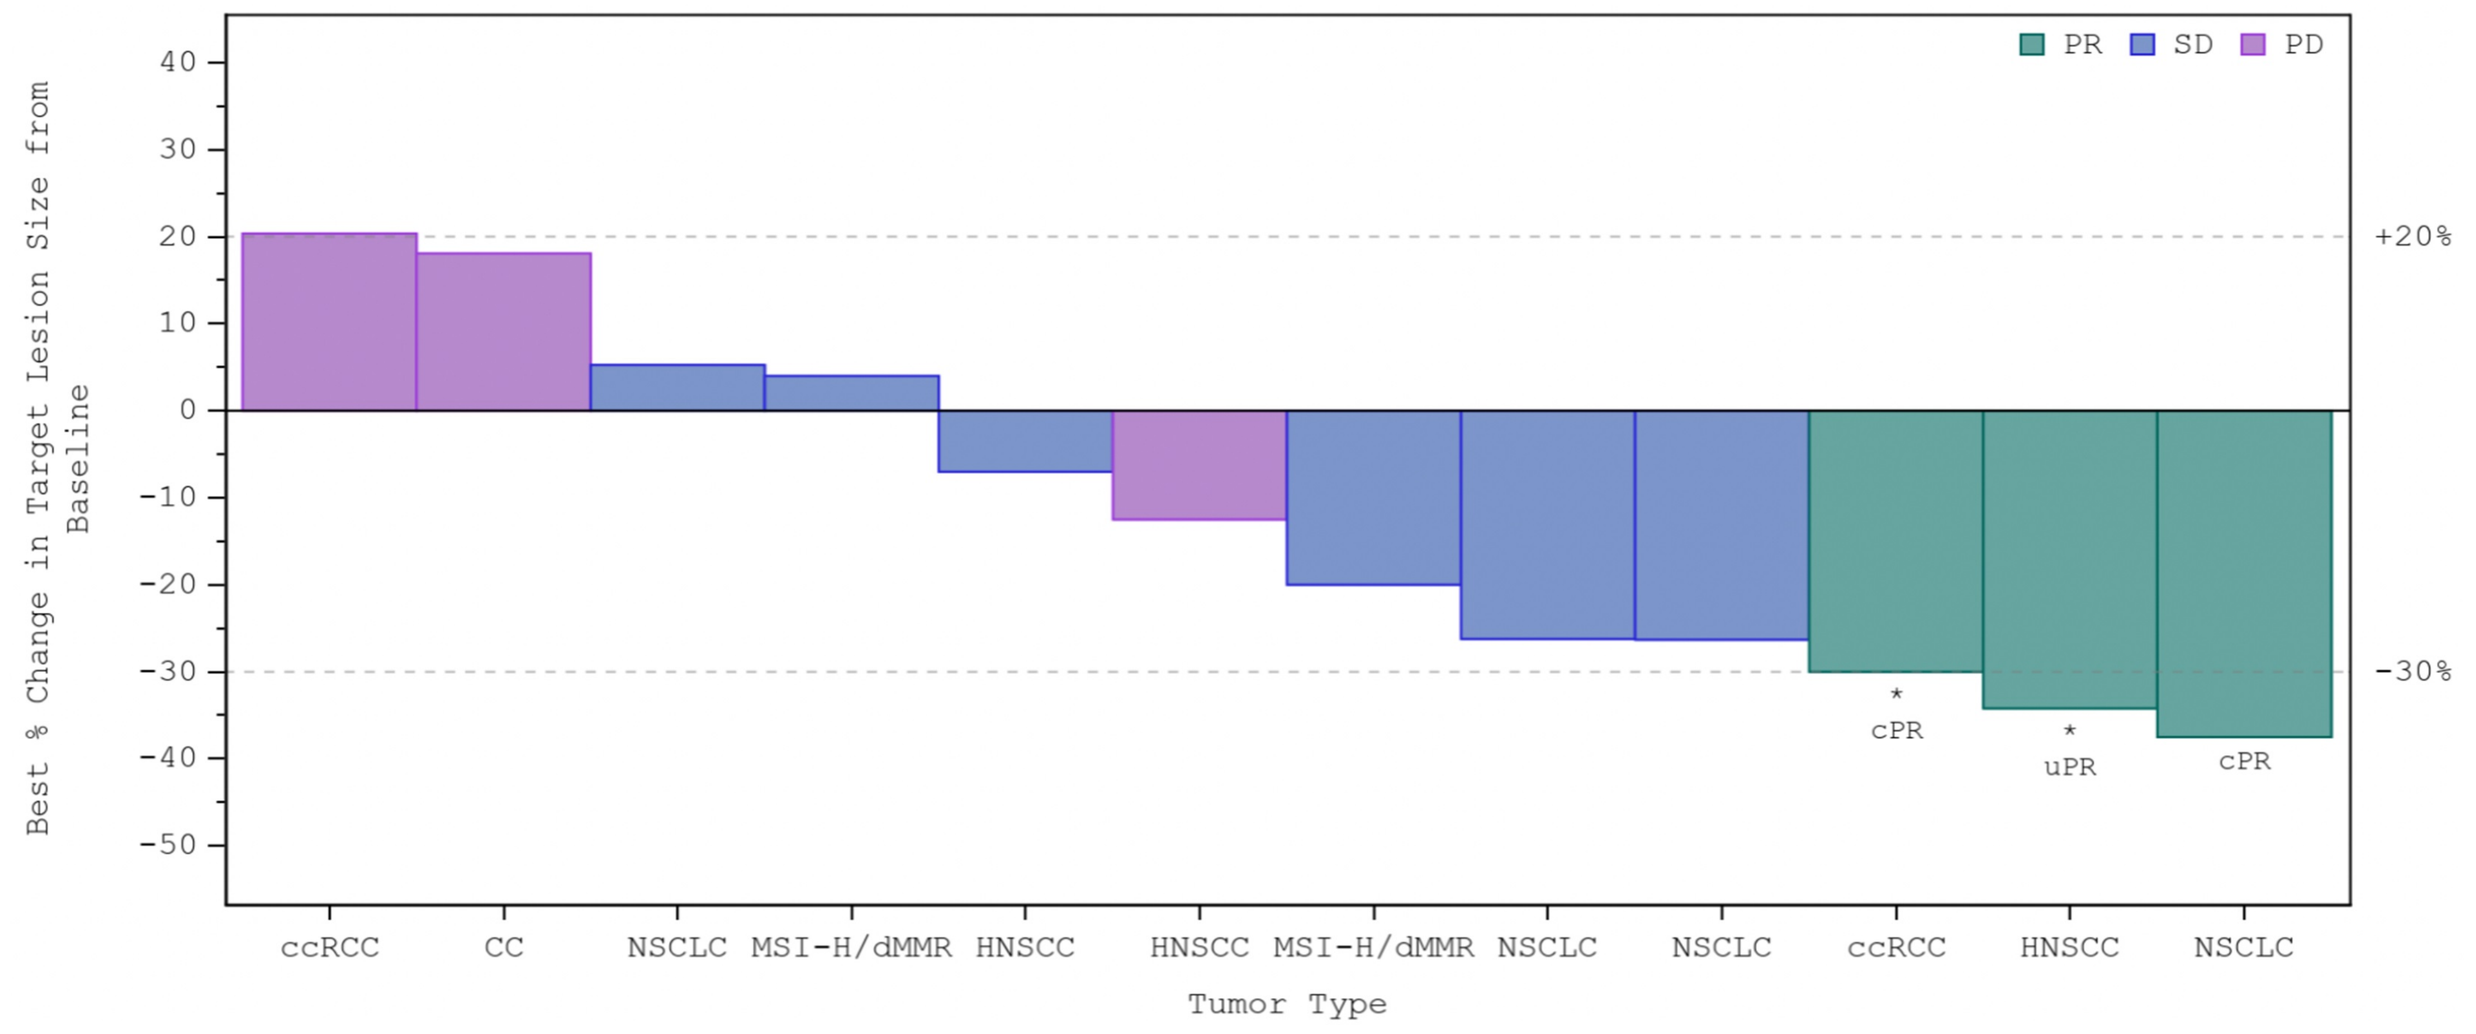
PR
SD
PD
Best % Change in Target Lesion Size from Baseline
+20%
-30%
cPR
uPR
cPR
ccRCC
CC
NSCLC
MSI-H/dMMR
HNSCC
HNSCC
MSI-H/dMMR
NSCLC
NSCLC
ccRCC
HNSCC
NSCLC
Tumor Type

Note: 12 subjects are represented on this waterfall plot. 3 efficacy evaluable subjects are not represented due to (1) Clinical PD before 1st scan.
- Subjects are censored after the first reporting of radiographic or clinical PD.
* Indicates subjects who are ongoing treatment as of the data extract date .

Figure 8. Spider for Ph1a Monotherapy (IO Pretreated with 1-2 Prior Lines)

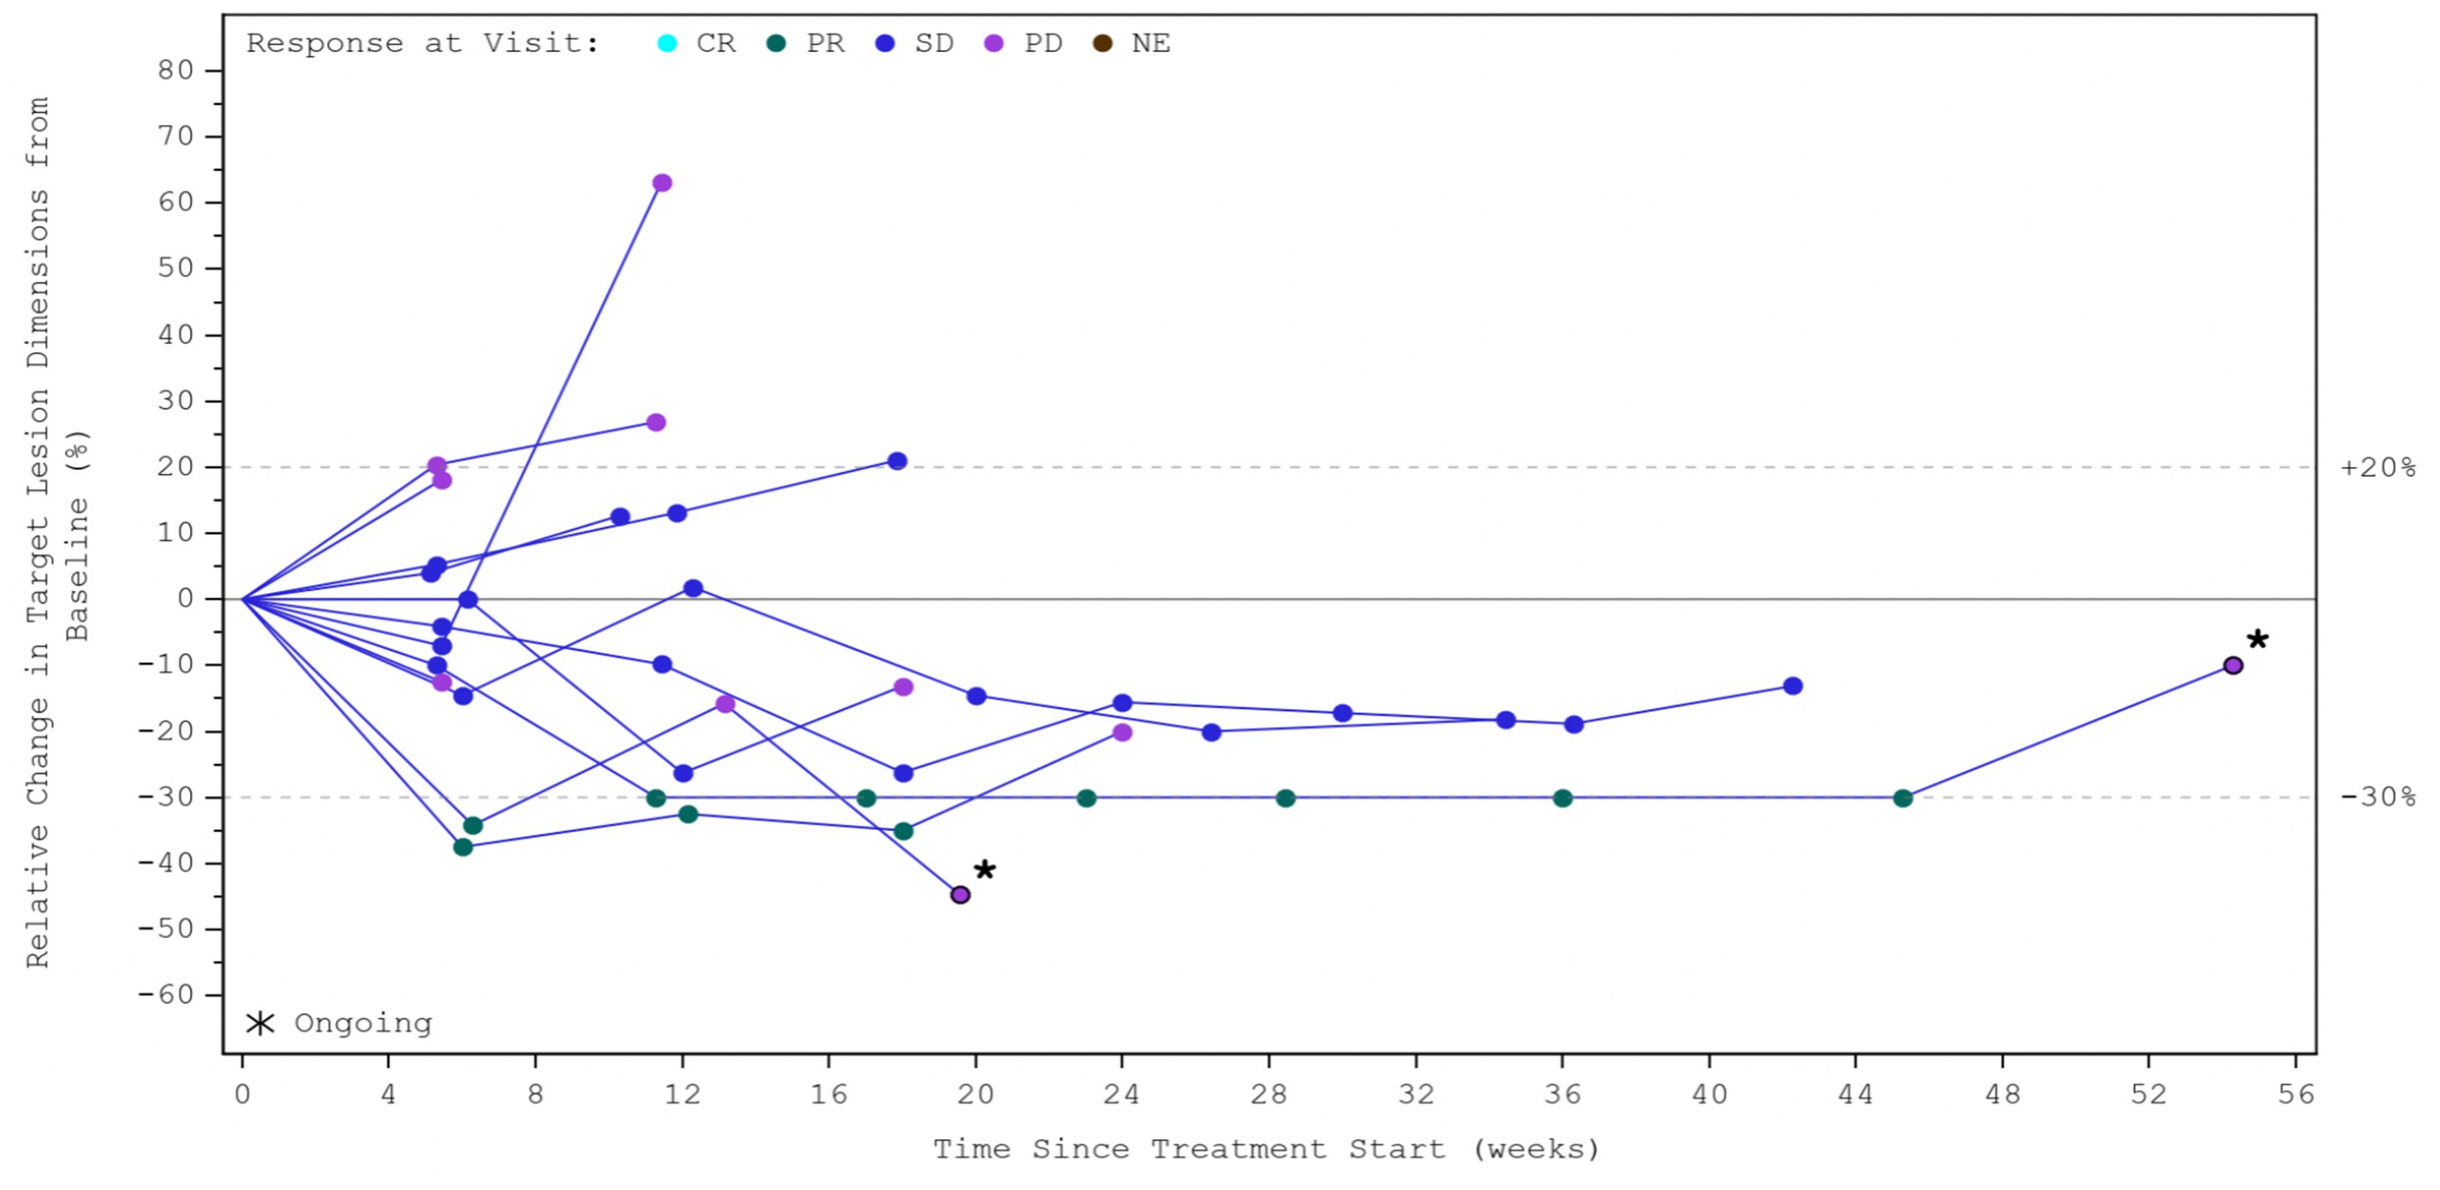
Response at Visit: CR PR SD PD NE
Relative Change in Target Lesion Dimensions from Baseline (%)
+20%
-30%
Ongoing
Time Since Treatment Start (weeks)

Note: 12 subjects are represented on this spider plot. 3 efficacy evaluable subjects are not represented due to clinical PD before 1st scan.
- Subjects are censored after the first reporting of radiographic or clinical PD.
* Indicates subjects who are ongoing treatment as of the data extract date .

Subject With Advanced ccRCC Achieves 85% cPR on Monotherapy STK-012 in Ongoing Phase 1b

Subject History

72-year-old female with Stage IV ccRCC and 2 prior lines of therapy before receiving STK-012 monotherapy

Prior lines:

1. Nivolumab + ipilimumab: Aug 2022 - Mar 2023 (BOR: cPR)
2. Cabozantinib + Nivolumab: Apr 2023 - Aug 2023 (BOR: PD)

Enrollment on STK-012 Ph1b ccRCC monotherapy expansion:

- Treatment Start: Sept 2023
- STK-012 Dose: 2.25mg Q3W Monotherapy

Fig 9. Chest CT

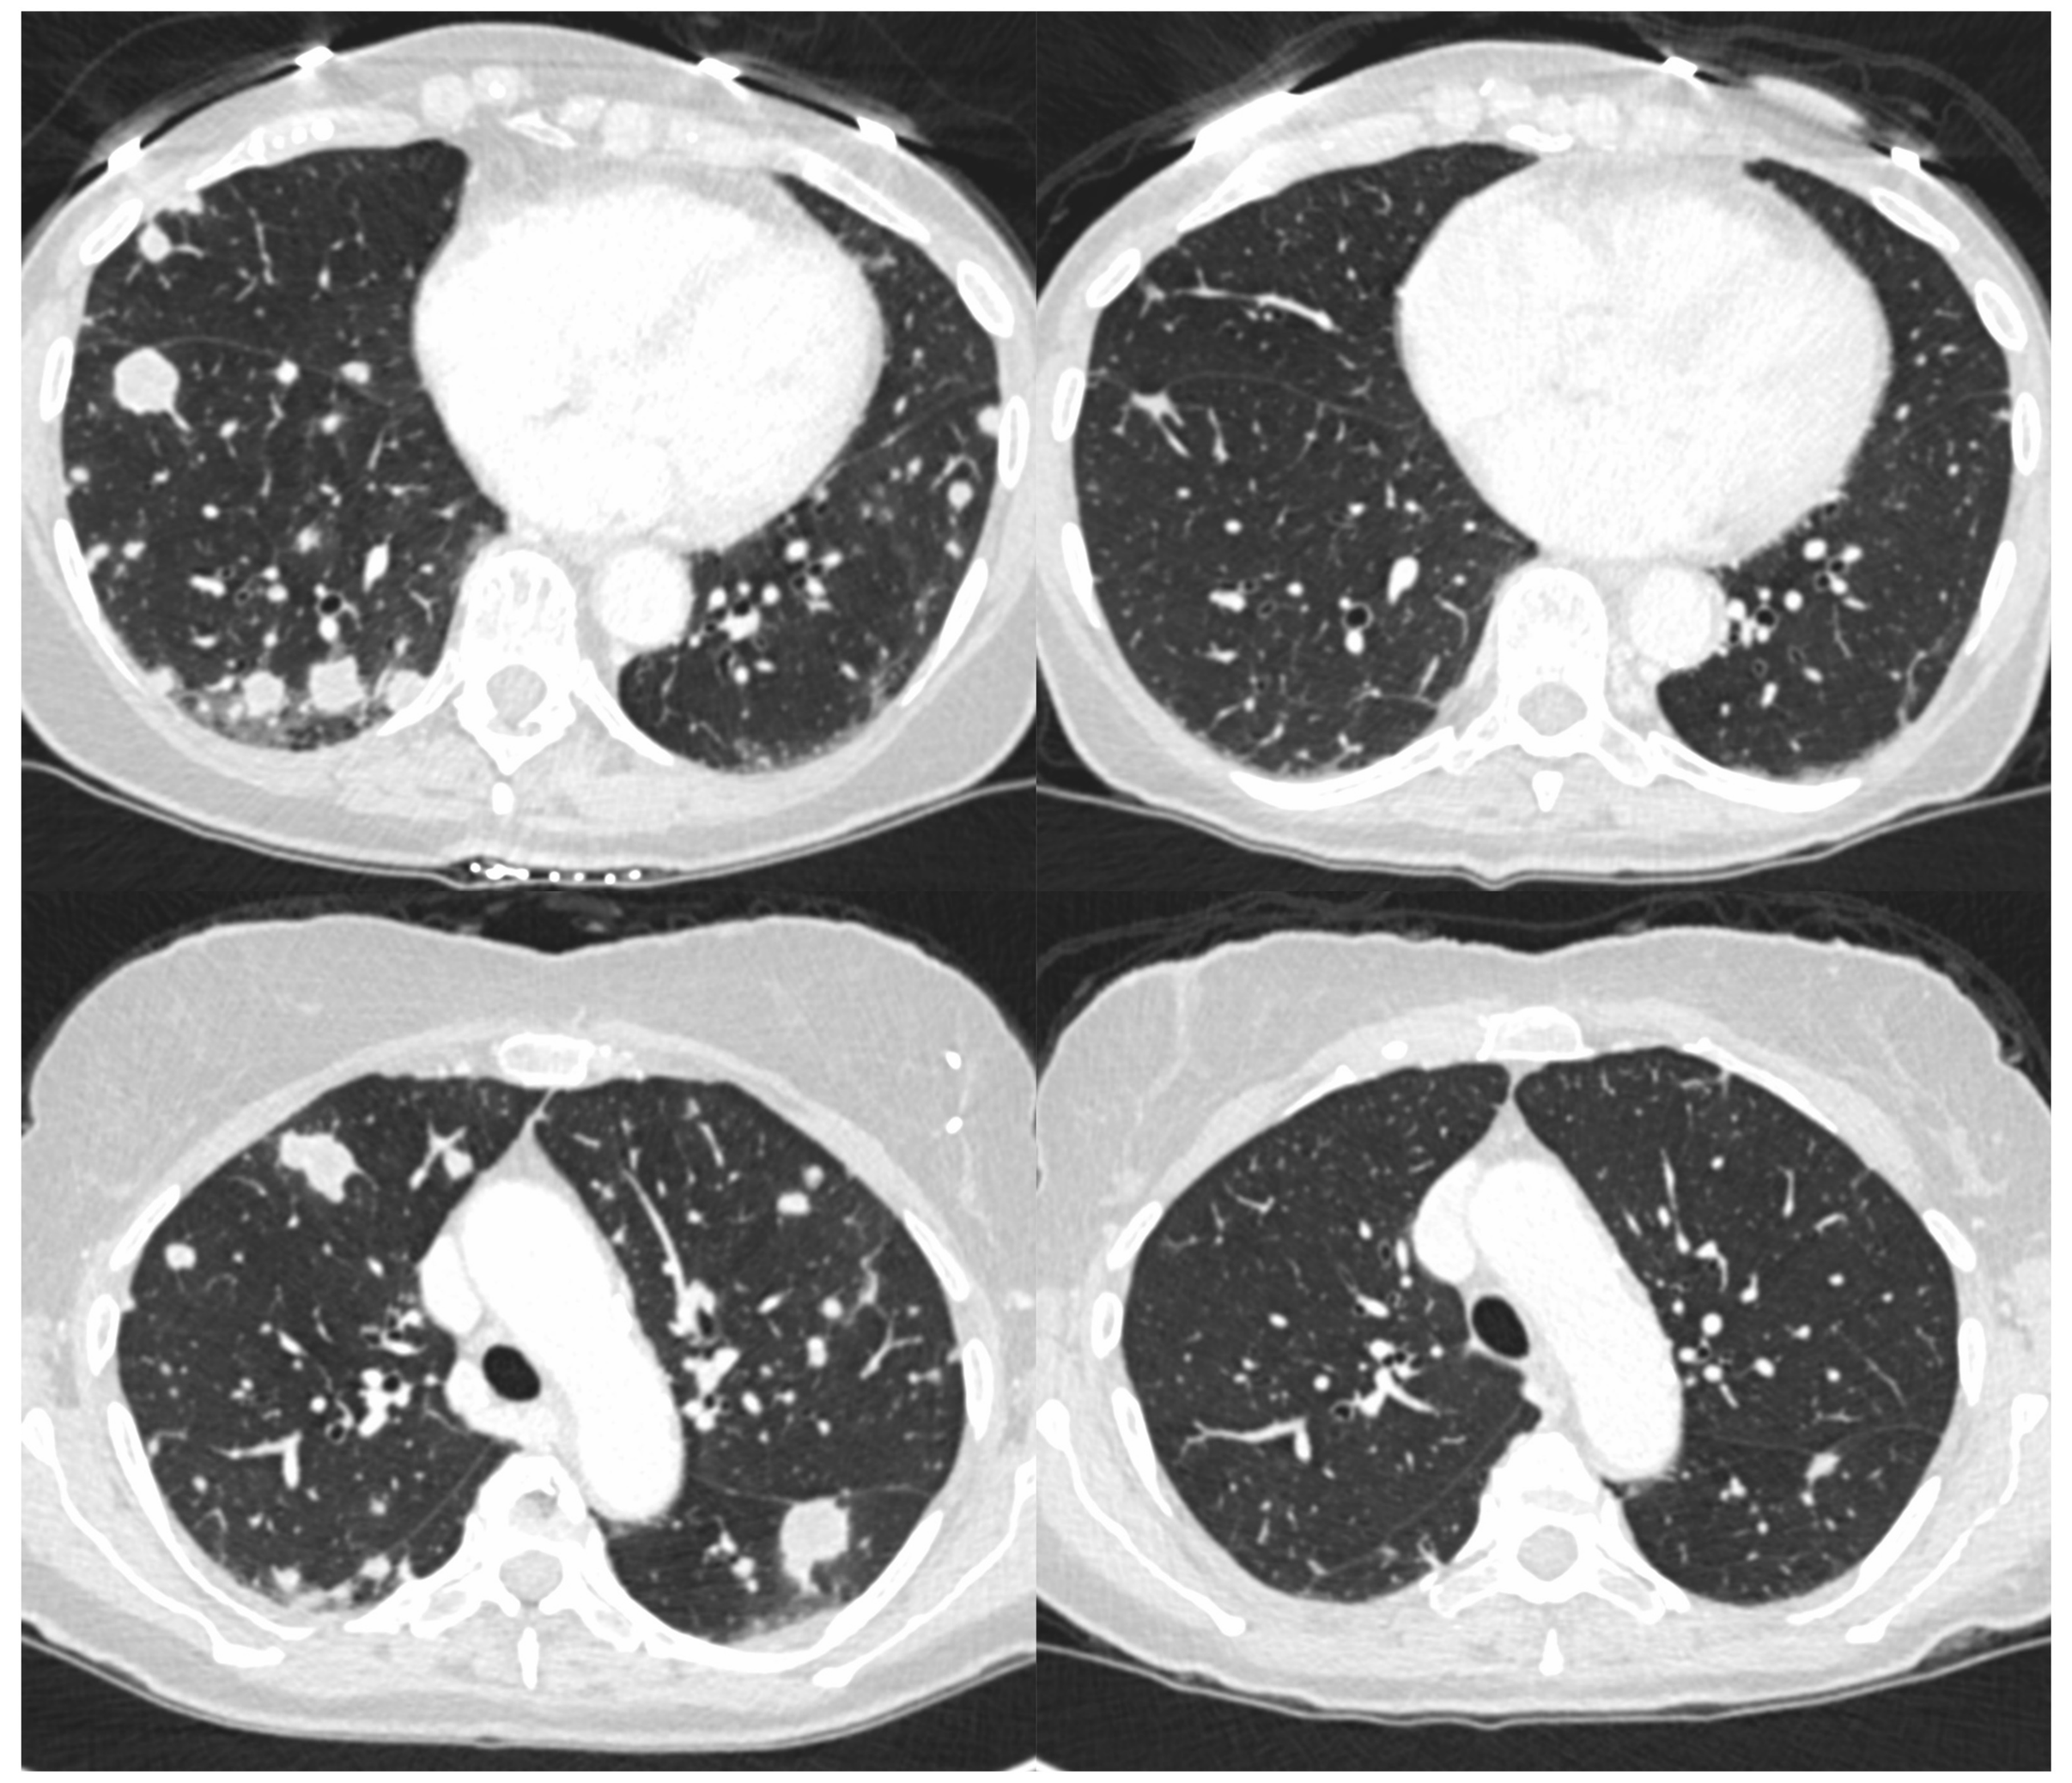
Baseline
Week 12

STK-012 Tumor Response

Baseline: Subject enrolled with "innumerable" lung metastases and a large primary renal mass (non-target).

Wk	Change From Baseline
6	↓50% (PR)
12	↓80% (cPR)
18	↓85% (cPR)
24	↓76% (PD)

STK-012 Tolerability

Dose reduction to 1.5 mg Q3W due to Gr 2 constitutional symptoms.

STK-012 has been well tolerated post dose reduction with no recurrence of the Gr 2 constitutional symptoms.

The subject has not experienced any SAEs.

Conclusions

- In the ongoing STK-012-101 study, monotherapy STK-012 showed favorable safety, PK, and PD profiles with distinct differences from aldesleukin and non-α IL-2 analogues.
- Preliminary PK-PD data support selectivity for IL-2R α/β, selective expansion of activated CD8 T cells, and dose dependent serum IFNγ increase.
- Preliminary efficacy with monotherapy STK-012 (3 partial responses in Phase 1a) was observed in subjects who progressed on/after prior immunotherapy.
- Preliminary evidence of activity was observed in the ongoing Phase 1b.
- Further development is warranted and enrollment in STK-012 monotherapy dose expansions (Phase 1b) in advanced NSCLC and ccRCC is ongoing.

References:
1. Hashimoto, et al. Nature 610, 173–181 (2022).
2. Wu J et al. Cancer Res (2023) 83 (7_Supplement): 4434
3. LaPorte KM et al. JITC 2023;11:e006611.
4. Wu W. et al. Nature Cancer. 2023 Sep;4(9):1309-1325.
5. Raeber ME et al. The Lancet. 2023;90: 104539

#Efficacy Evaluable Definition: Received at least 1 dose of STK-012 and met one of the following criteria: had at least one post baseline tumor assessment OR study discontinuation or death due to disease progression or STK-012 related AE prior to the first postbaseline tumor assessment.