STK-012, an α/β-selective IL-2 activates tumor antigen specific CD25+ CD8 T cells to reject tumors without acute vascular toxicity

Martin Oft, Navneet Ratti, Sandro Vivona, Jan Emmerich, Romina Riener, Ievgen O. Koliesnik, Scott McCauley, Michele Bauer, Marie Semana, Deepti Rokkam, Bhargavi Jayaraman, Rene De Waal Malefyt, Paul-Joseph Aspuria, Michael Totagrande, Anita Mehta-Damani, Patrick J. Lupardus, Rob A. Kastelein. *Synthekine, Menlo Park, CA*

Synthekine
ABSTRACT #1801

ABSTRACT

High-dose Interleukin-2 (IL-2) monotherapy induces complete responses in cancer patients but its use is limited by acute vascular toxicities including capillary leak syndrome and severe hypotension 1,2. IL-2 activates lymphocytes and NK-cells through the intermediate-affinity dimeric IL-2 receptor,IL-2Rβ/γ (CD122/CD132), while antigen activated T-cells and regulatory T-cells (Tregs) have increased sensitivity to IL-2 by expressing the high-affinity trimeric IL-2 receptor, IL-2Rα/β/γ (CD25/CD122/CD132)3. CD25-independent IL-2s ("non-α-IL-2s") aim to increase the therapeutic efficacy of IL2 in cancer patients by avoiding Treg activation through selective binding to IL-2Rβ/γ 4. However, those molecules still are reported to induce fever and hypotension and have limited efficacy as a monotherapy or in combination with anti-PD-1 5,6. Here we show that a novel α/β-IL-2 agonist that was designed to preferentially bind to the IL-2Rα/β/γ receptor which is highly upregulated on antigen activated T-cells can greatly improve on the efficacy of IL-2 while avoiding the vascular toxicity commonly associated with IL-2 treatment. In syngeneic tumor models, this α/β-IL-2 agonist significantly reduced exhaustion of tumor infiltrating T cells compared to WT-IL-2 or a non-α-IL-2 leading to improved expansion of tumor antigen specific CD25+PD-1+CD8+ T cells systemically and in the tumor microenvironment. This resulted in complete responses and tumor immune memory with α/β-IL-2 monotherapy as well as improved outcomes in combination with anti-PD-1 therapy in PD-1 refractory syngeneic tumors. In contrast,WT-IL-2 reduced T cell exhaustion and drove antigen specific T cell responses to a lesser degree, resulting in reduced combinatorial efficacy withanti-PD-1, while the non-α-IL-2 failed to do either. Furthermore, the α/β-IL-2 agonist reduced intratumoral Tregs compared to treatment with WT-IL-2 or PBS improving the intratumoral CD8 to Treg ratio. In non-human primates and mice, WT-IL-2 and a non-α-IL-2 led to broad extravasation of lymphocytes and NK cells and activation of intra-pulmonal T cells resulting in systemic tissue inflammation and NK cell-mediated lethal capillary leak syndrome whereas the α/β-IL-2 agonist, which avoids binding the dimeric IL-2Rβ/γ expressed on NK cells, avoided systemic lymphocyte activation which facilitated continuous treatment without acute vascular toxicities. Overall, through selective engagement of CD25+ T cells, this α/β-IL-2 agonist demonstrated improved efficacy and tolerability of IL-2 in preclinical tumor models. Clinical trials with STK-012, a human α/β-IL-2 agonist, are in progress (NCT05098132).

1 Atkins, et al.; JCO 1999, 2 Dutcher, et al.; JITC 2014, 3 Liao, et al.; Immunity 2013, 4 Levin, et al.; Nature 2012, 5 Janku, et al.; Cancer Research 2021; 6 Diab, et al.; Cancer Disc. 2020

Distribution and Function of the Trimeric and Dimeric IL-2 Receptor

High Affinity Receptor

Expressed on ***antigen activated CD8s*** *and Tregs*

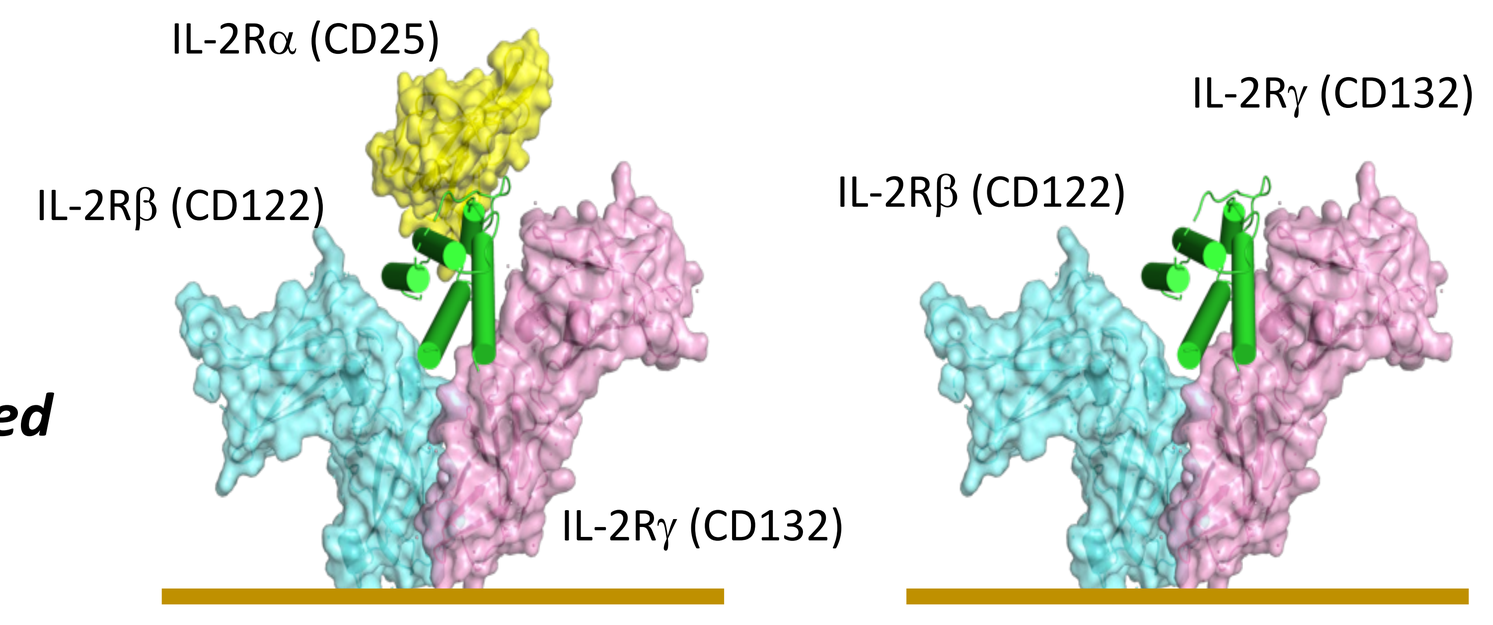
IL-2Rα (CD25)
IL-2Rβ (CD122)
IL-2Rγ (CD132)
IL-2Rβ (CD122)
IL-2Rγ (CD132)

Wang et al (Garcia)., Science 2005

Intermediate Affinity Receptor

Receptor complex most commonly expressed: ***NK cells*** *and naïve (and* ***not activated*** *T cells)*

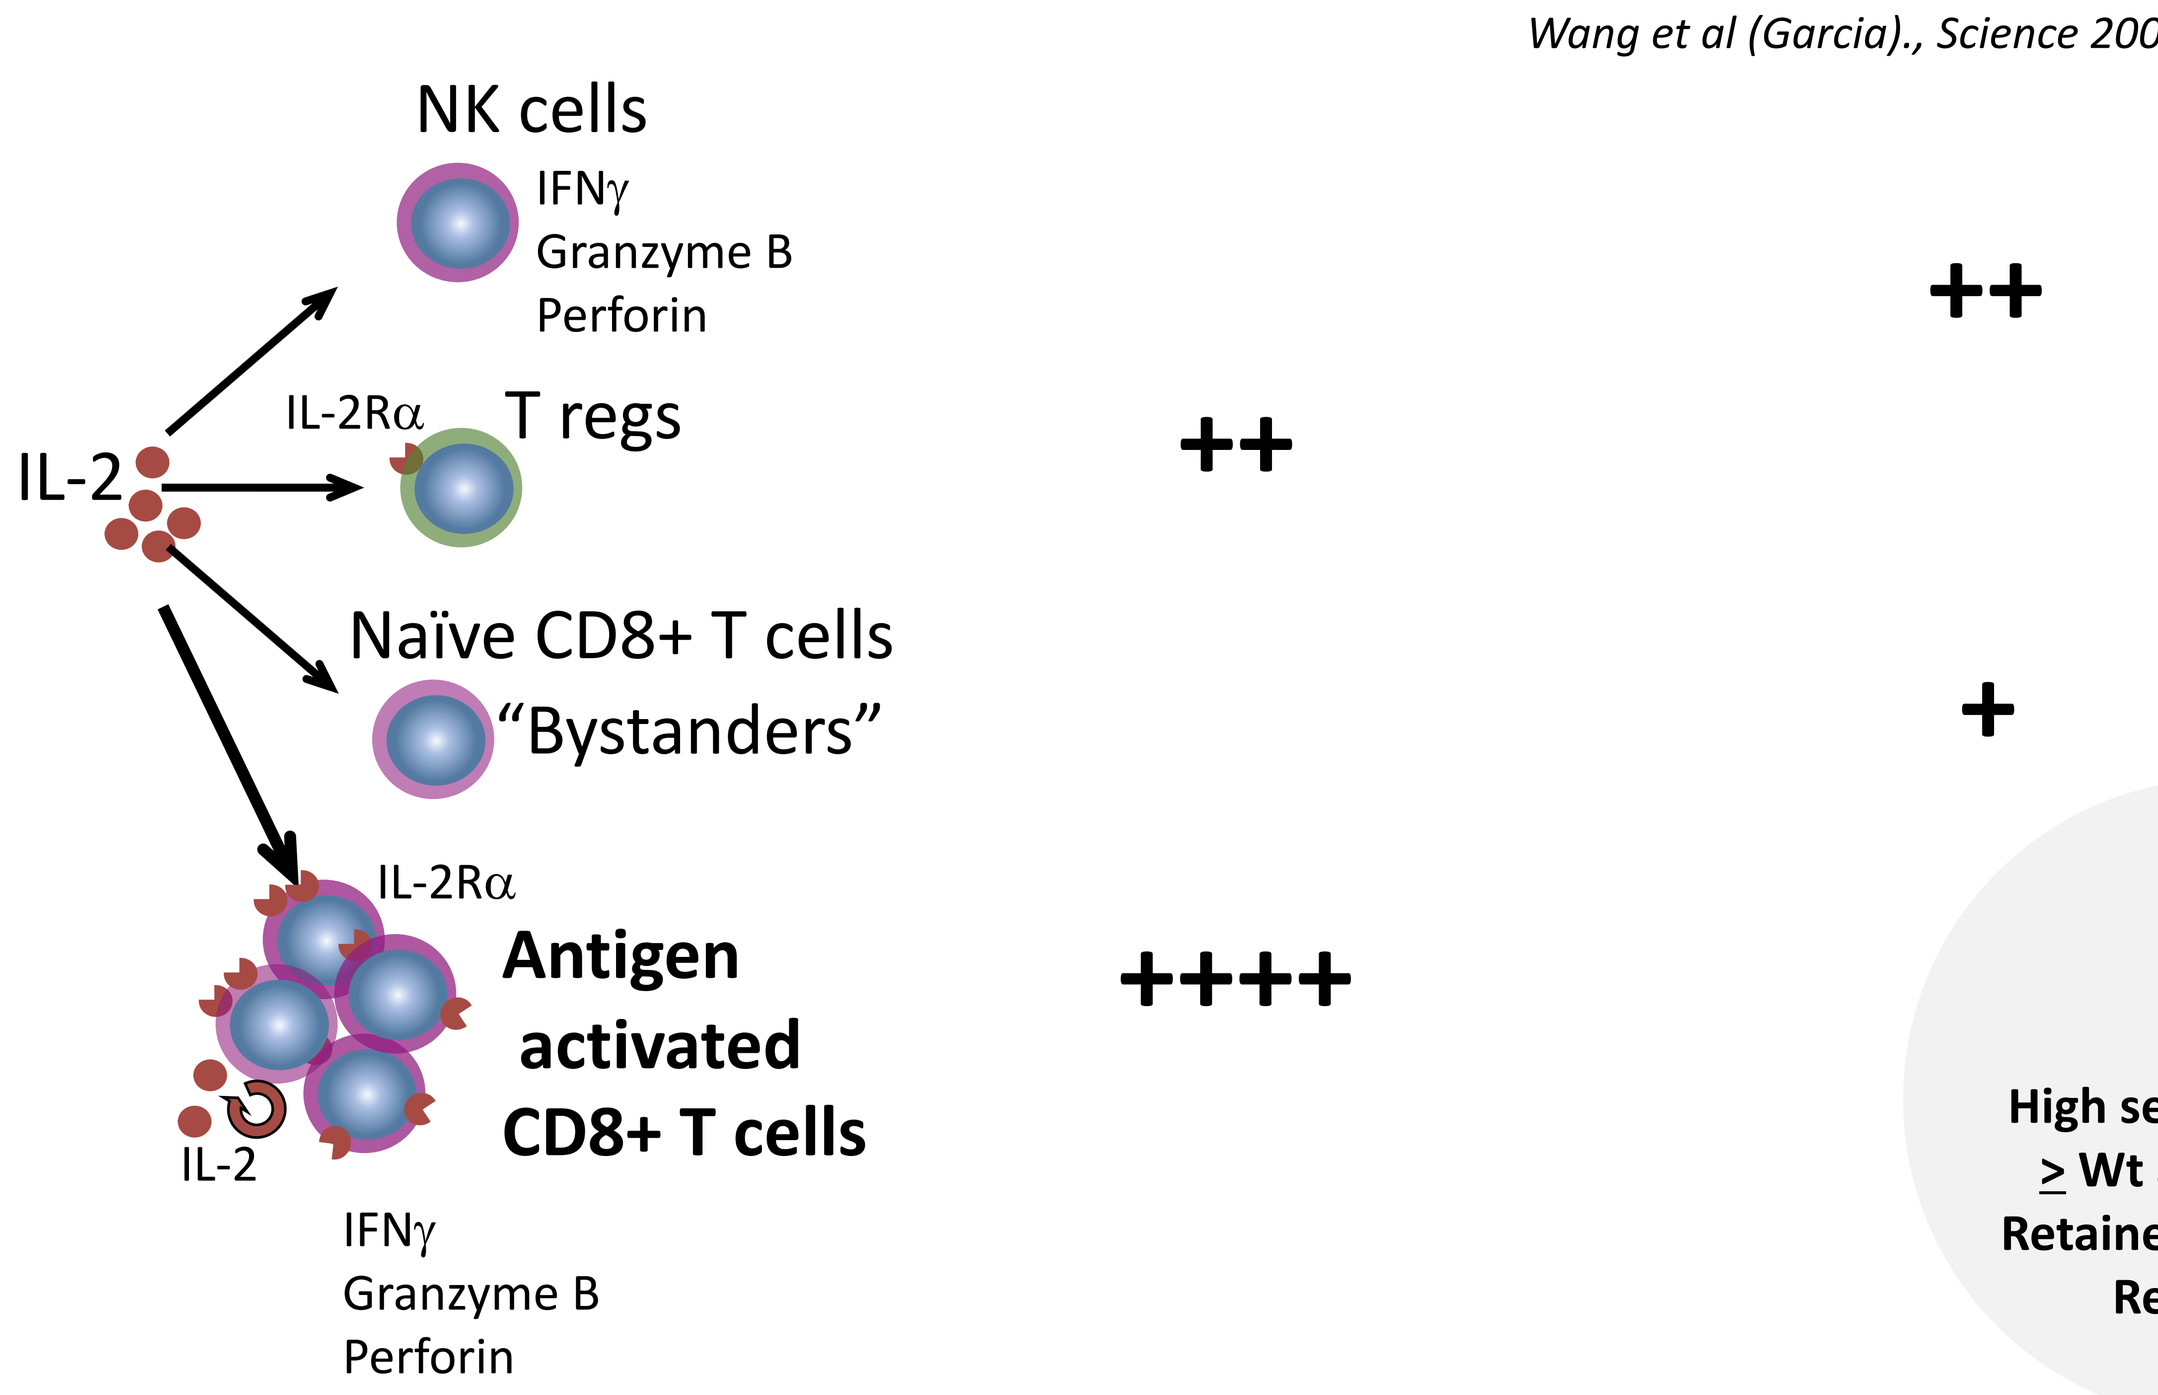
NK cells
IFNγ
Granzyme B
Perforin
IL-2
IL-2Rα
T regs
Naïve CD8+ T cells
"Bystanders"
IL-2Rα
Antigen activated CD8+ T cells
IL-2
IFNγ
Granzyme B
Perforin
++
++
+
++++

Design Criteria

High selectivity for CD25+++ cells
≥ Wt activity on CD25+++ cells
Retained IL-2Rβ binding (efficacy)
Reduced IL-2Rγ binding

Induction of the Trimeric IL-2 Receptor upon T cell receptor Activation

CD25 marks antigen activated PD-1+ T cells (T_{PEX})

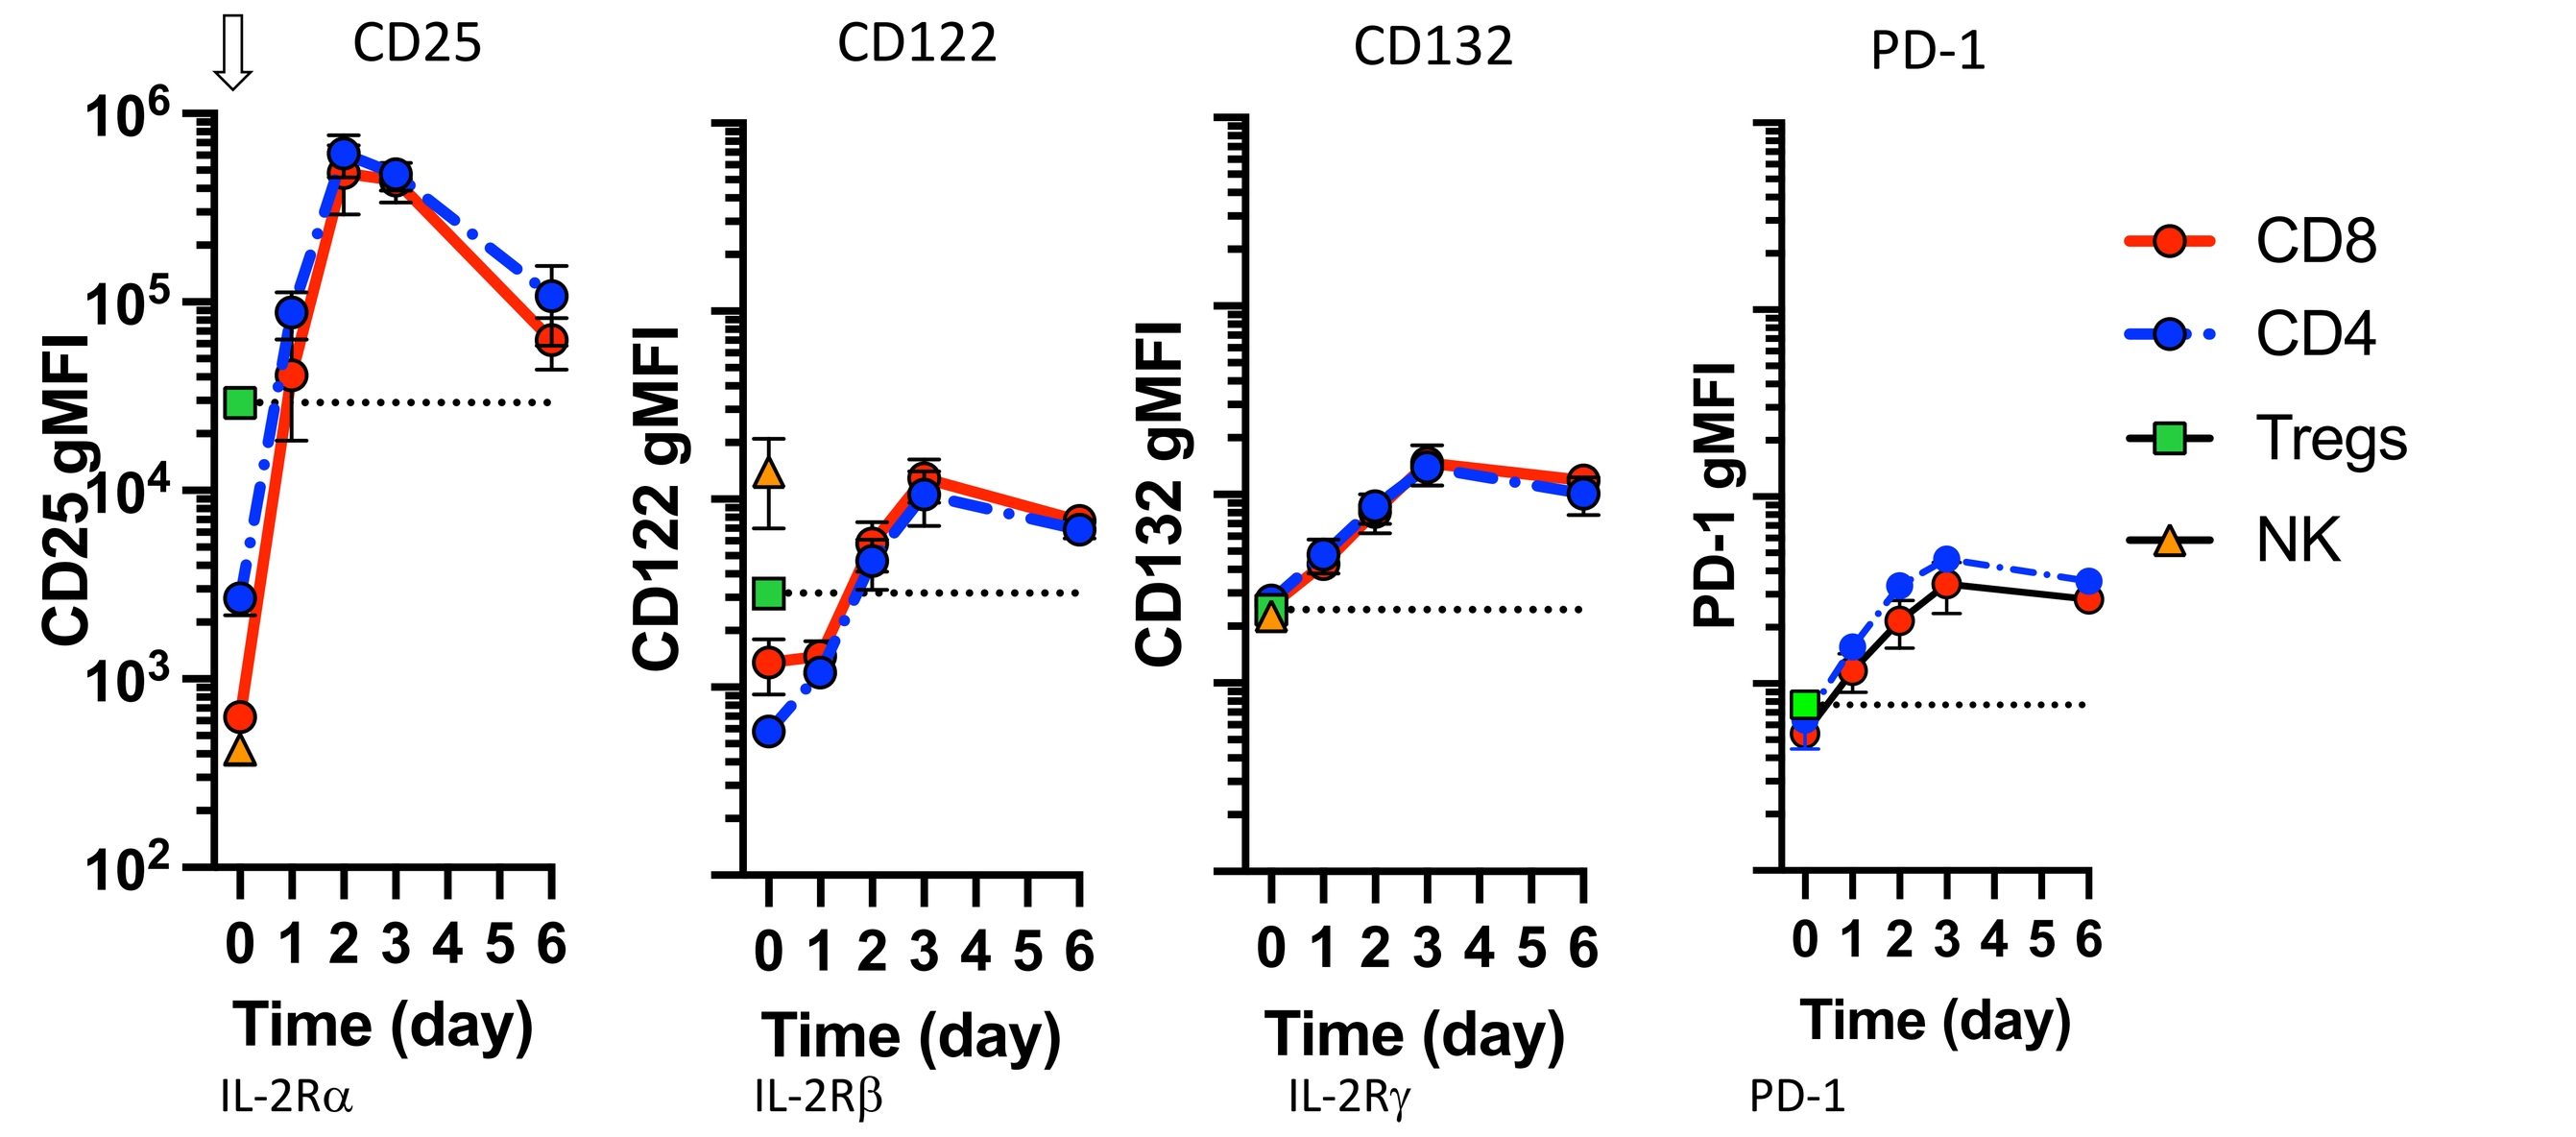
Anti-CD3/CD28
CD25
CD122
CD132
PD-1
CD25 gMFI
CD122 gMFI
CD132 gMFI
PD-1 gMFI
Time (day)
IL-2Rα
IL-2Rβ
IL-2Rγ
PD-1
CD8
CD4
Tregs
NK

Fig. 2. IL-2 receptor upregulation by T cell receptor stimulation CD4+ and CD8+ T cells were stimulated with CD3/CD28 beads; IL-2 receptors and PD-1 were quantified on FACS for one week

STK-012 has increased selectivity for CD25+ T cells vs NK cells

Synthekine Approach
- STK-012
 - Synthekine's IL-2 clinical candidate (pegylated)
 - Selective for Rα+ / Rβ+ T cells
 - Modulated Rγ binding
- mSTK-012
 - Mouse surrogate for STK-012 (pegylated)

Currently Used in Cancer
- IL-2
 - approved in melanoma and RCC (Proleukin®)
 - mIL-2 was pegylated

1st Generation "improved" IL-2 in Cancer
- non-α-IL-2
 - Engineered for reduced IL2Rα binding, IL-2 maintains binding selective for Rβ/γ
 - Proxy for NKTR-214, THOR-707, NL-201, etc.
 - (pegylated)

Current Approach in AI
- non-β-IL-2 (Treg proliferation)
 - Engineered for reduced/no Rβ binding IL-2 maintains binding selective for Rα
 - Proxy for N88D, N88R, V91K, D20T, etc.

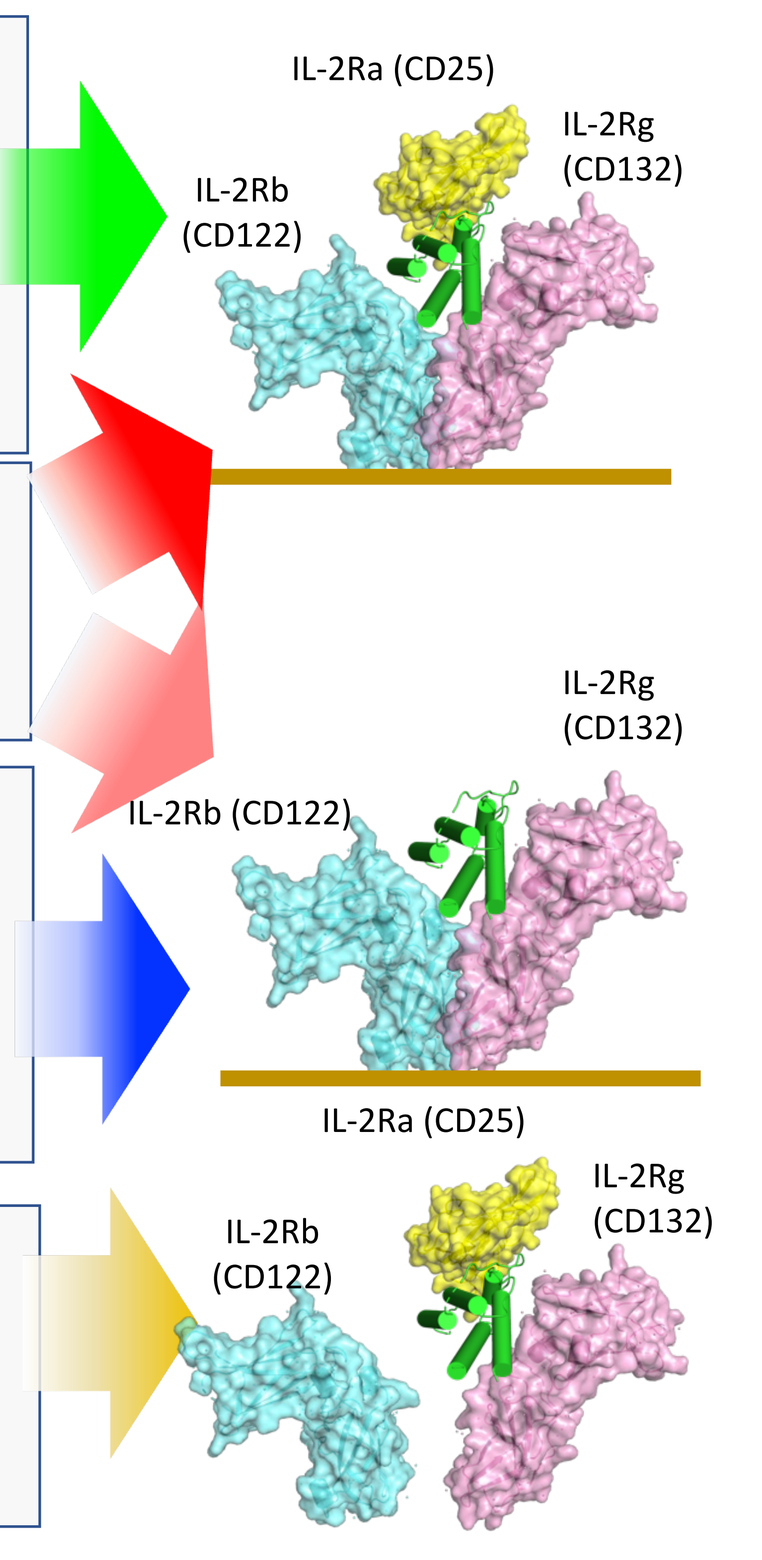
IL-2Ra (CD25)
IL-2Rb (CD122)
IL-2Rg (CD132)

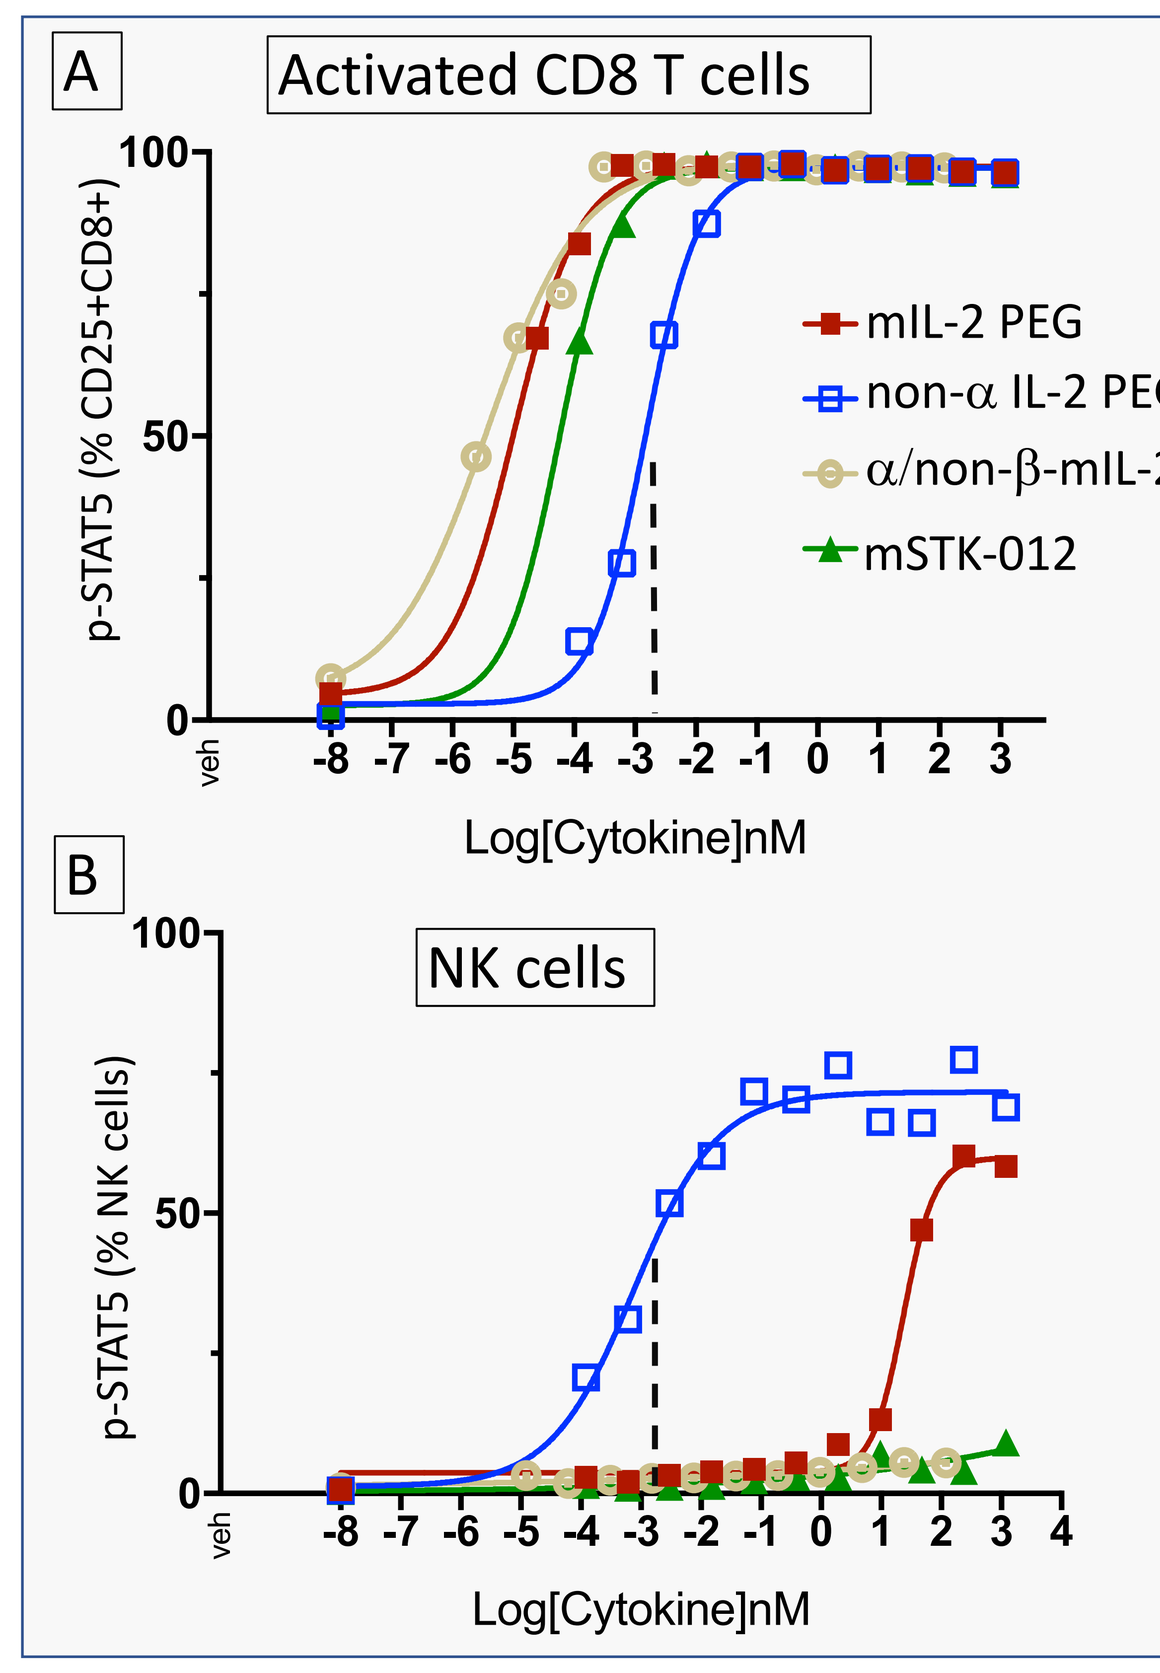
A
Activated CD8 T cells
p-STAT5 (% CD25+CD8+)
mIL-2 PEG
non-α IL-2 PEG
α/non-β-mIL-2 PEG
mSTK-012
Log[Cytokine]nM
B
NK cells
p-STAT5 (% NK cells)
Log[Cytokine]nM

Fig.3 Selectivity of wt-IL-2 and STK-012 but not non-a-IL-2-PEG for TCR activated T cells (**A.B**) STAT5 phosphorylation in response to IL-2 variants, (**A**) CD8 T cells, activated for 3 days with CD3/28 or (**B**) NK cells were stimulated for 20min with IL-2. Note: IL-2 has high selectivity for activated T cells vs. NK cells, but non-α-IL-2 has equal EC50 on both cell types. mSTK-012 does not significantly stimulate NK cells

Mouse IL-2 and non-α-IL-2 induces capillary leak

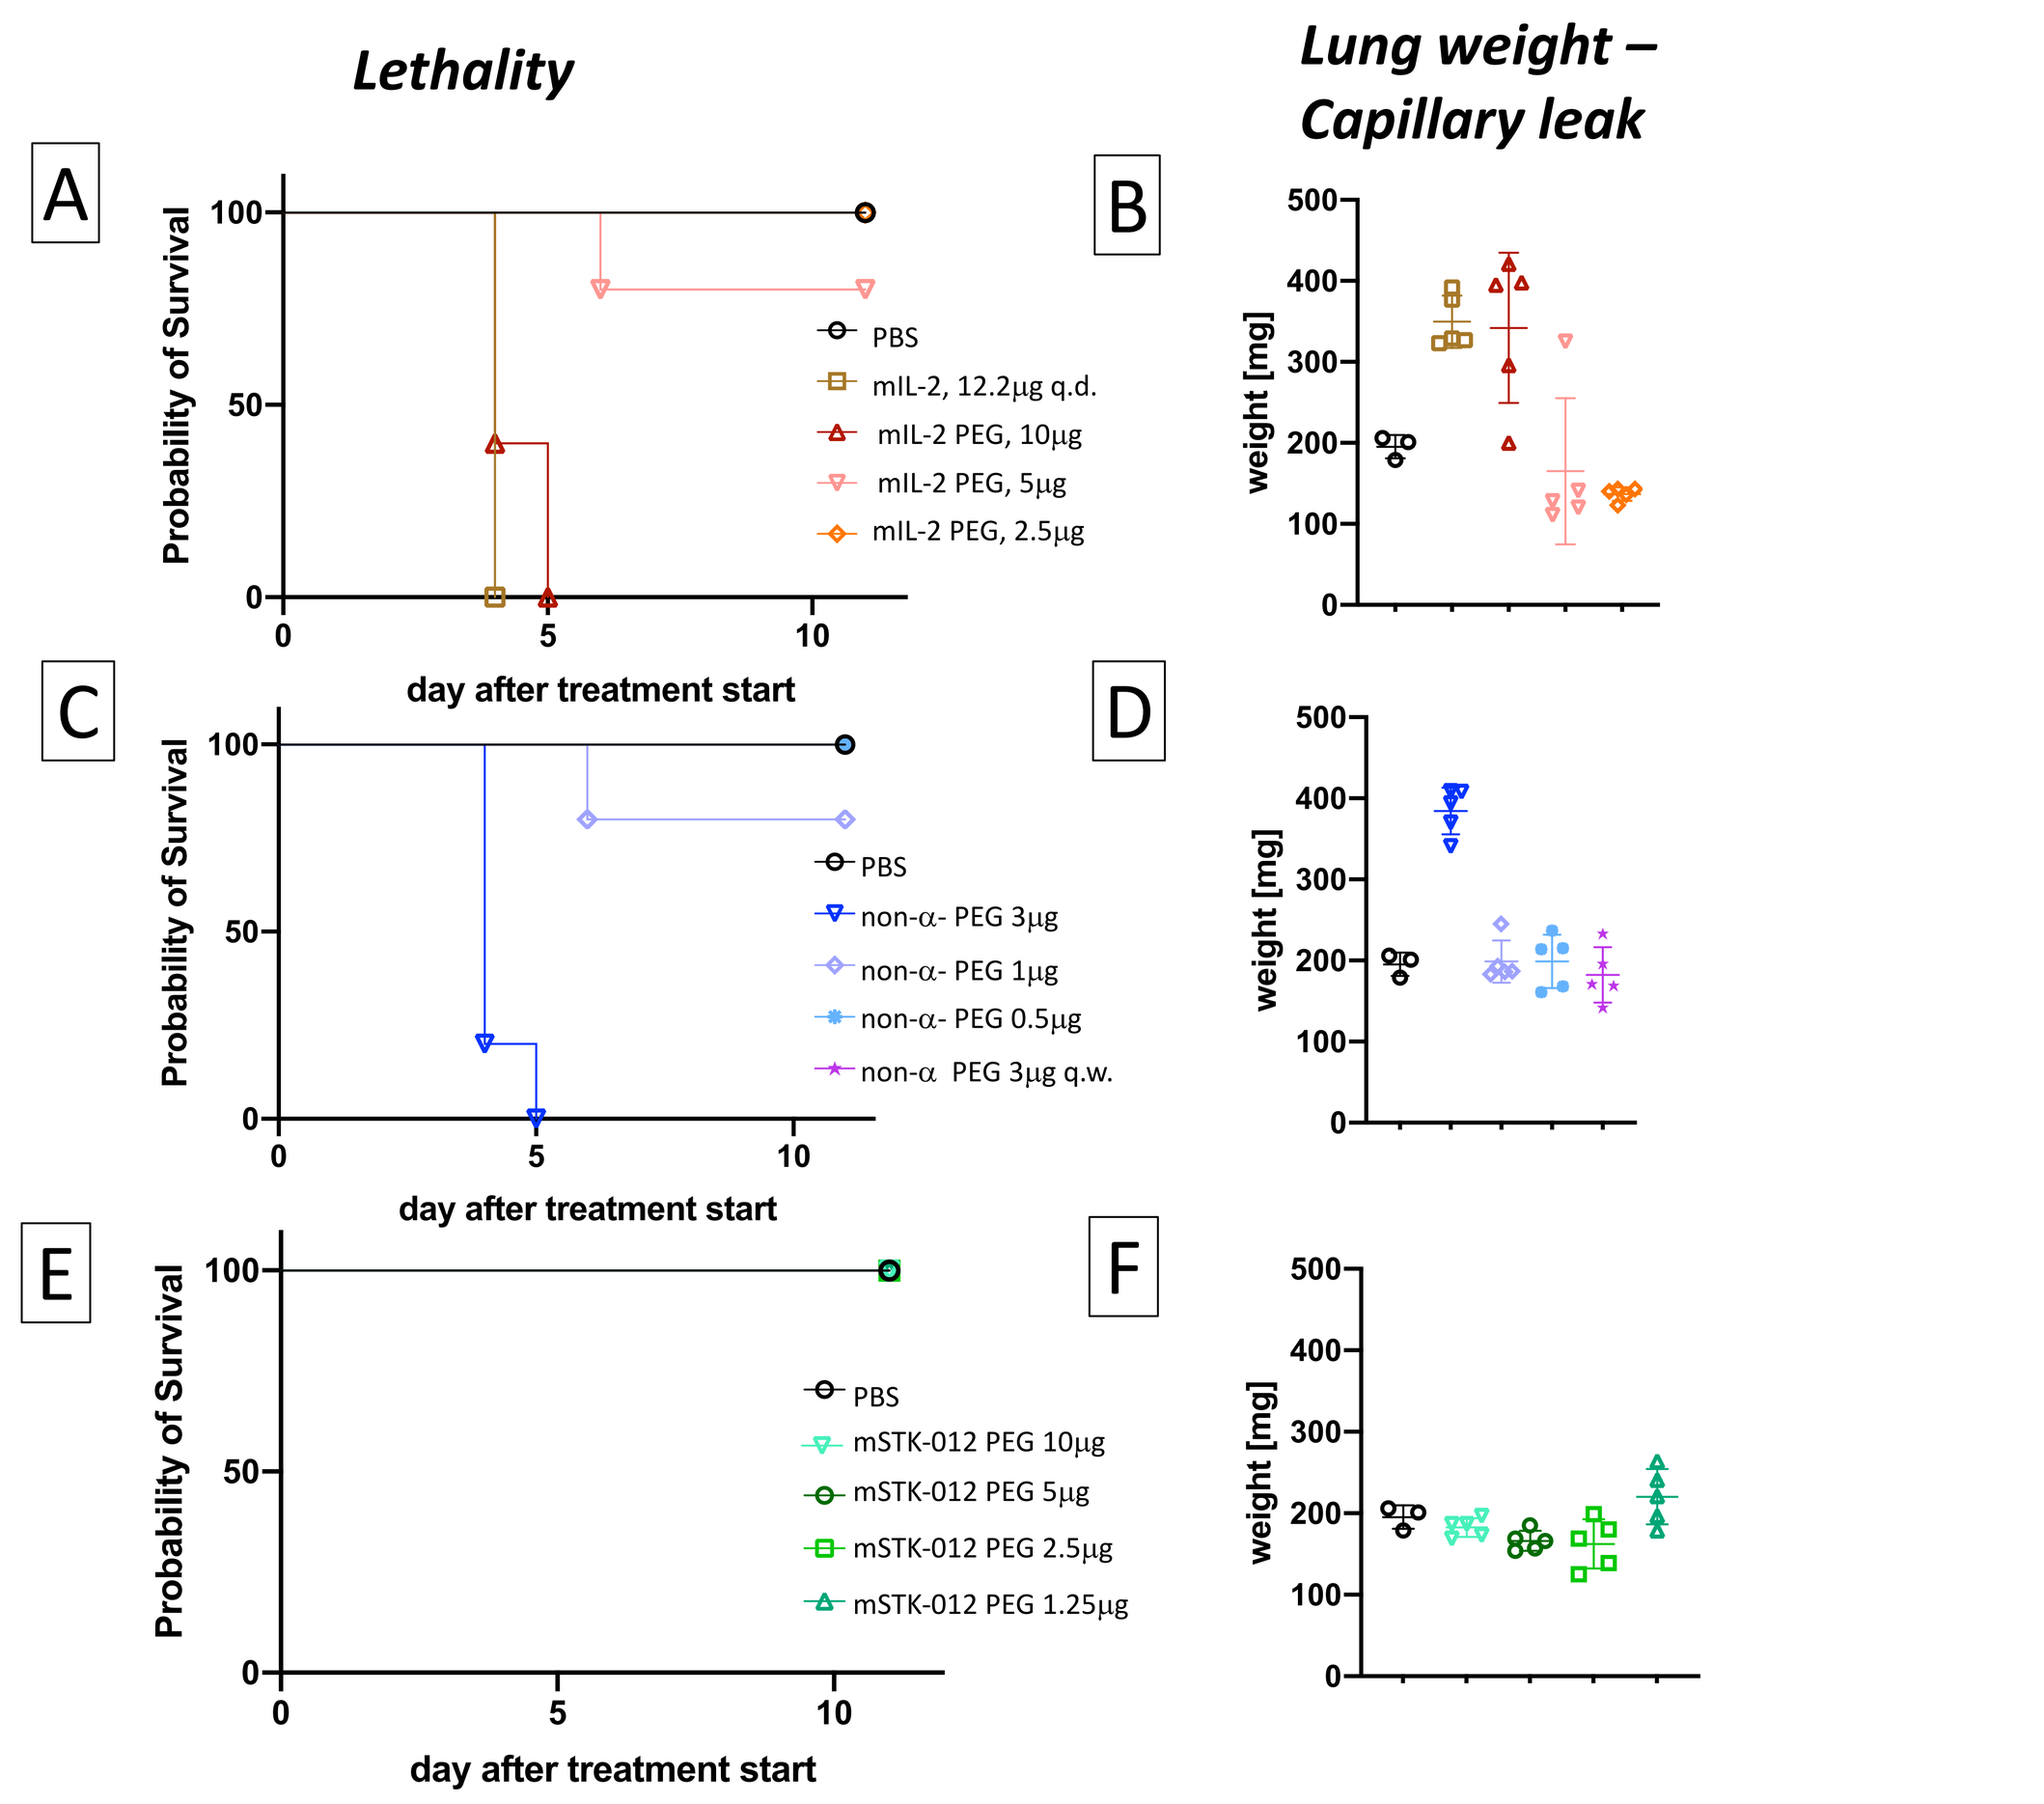
Lethality
Lung weight – Capillary leak
A
B
C
D
E
F
Probability of Survival
day after treatment start
weight [mg]
PBS
mIL-2, 12.2µg q.d.
mIL-2 PEG, 10µg
mIL-2 PEG, 5µg
mIL-2 PEG, 2.5µg
non-α- PEG 3µg
non-α- PEG 1µg
non-α- PEG 0.5µg
non-α PEG 3µg q.w.
mSTK-012 PEG 10µg
mSTK-012 PEG 5µg
mSTK-012 PEG 2.5µg
mSTK-012 PEG 1.25µg

Fig. 4. Acute toxicity, lethality and vascular leak syndrome in wt-mIL-2 and non-α-IL-2-PEG but not with mSTK-012 treated mice (A,C,E) Survival and wet lung weight (**B,D,F**) of mice treated every second day (unless indicated) with the indicated doses of wt-mIL-2 (**A,B**) non-α- (**C,D**) α/β-mIL-2 (**E,F**). (q.d. daily dosing, q.w. weekly dosing). Lungs were harvested at the time of death or at the end of the study. Wet lung weights were determined by subtracting the lung wet after desiccation from the fresh lung weight. All IL-2 PEG molecules were covalently conjugated with a 40kD polyethylene glycol (PEG) except mIL-2 control in (A).

"Tolerated" Doses of mIL-2 / non-a-IL-2 induce Capillary Leak

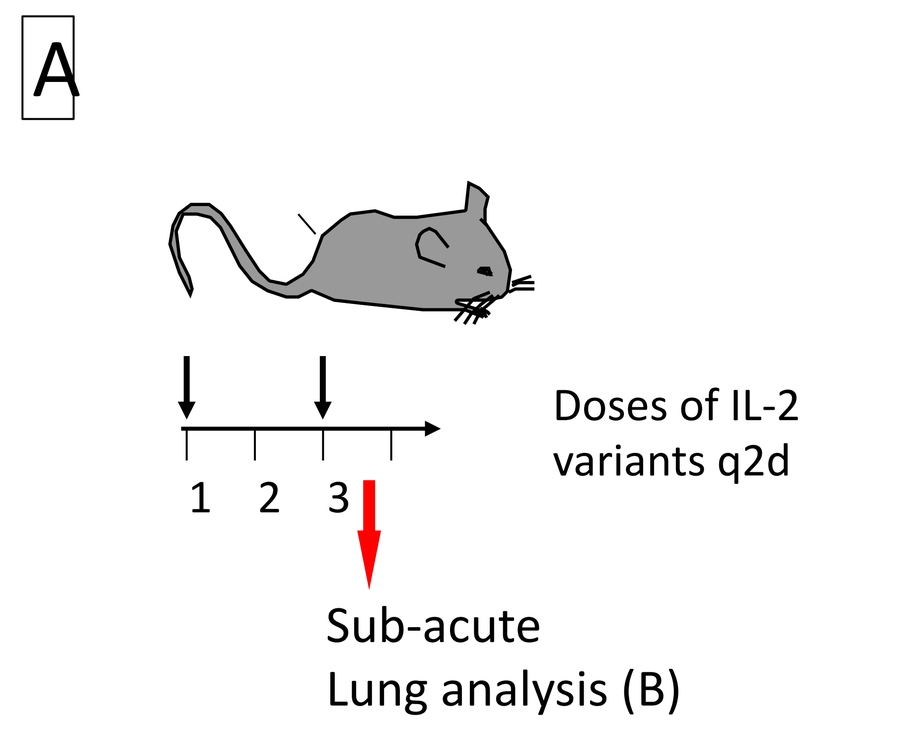
A
Doses of IL-2 variants q2d
Sub-acute Lung analysis (B)

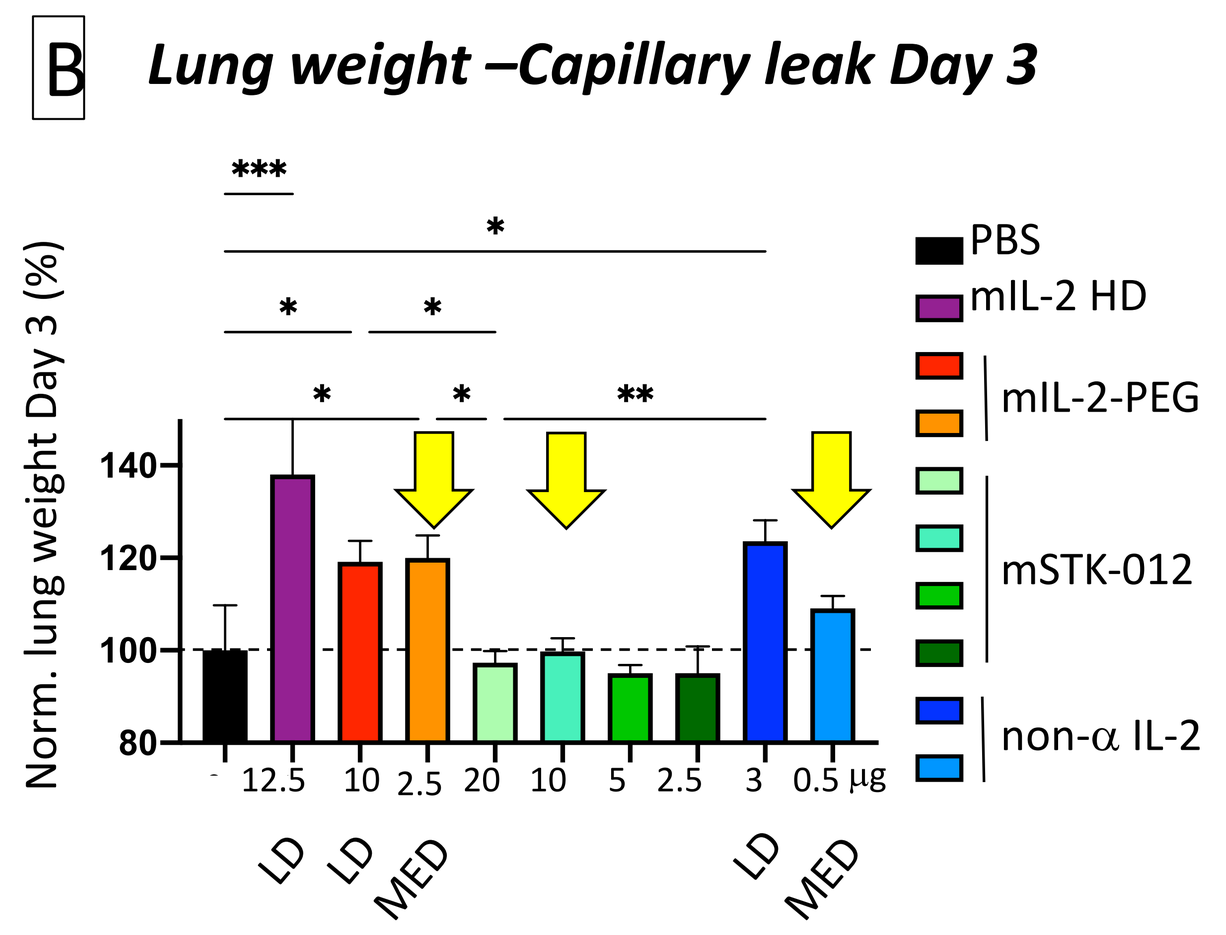
B
Lung weight –Capillary leak Day 3
Norm. lung weight Day 3 (%)
PBS
mIL-2 HD
mIL-2-PEG
mSTK-012
non-α IL-2
LD
LD
MED
LD
MED

Fig.5 Acute Immune toxicity to wt-mIL-2, or non-α-IL-2-PEG but not mSTK-012 Clinically aldesleukin and non-α-IL-2s are dosed for 3 days or less / cycle. Therefore, we investigated the toxicity on day 3 of IL-2 variants. (**A**) Treatment schedule, lung weights (**B**) of mice in IL-2 induced acute toxicity model. (**B**) Subacute lung weights of mice on day 3 of treatment with IL-2 variants (µg)indicating vascular leak syndrome (VLS) in wt-IL-2 and non-α-IL-2 cohorts. LD: lethal dose; MED: Maximal efficacious and non-lethal dose used in efficacy experiments

IL-2 and Non-a-IL-2 but not STK-012 avoids toxicity by sparing systemic NK cell activation and margination

A Acute capillary leak syndrome in cynomolgus monkeys

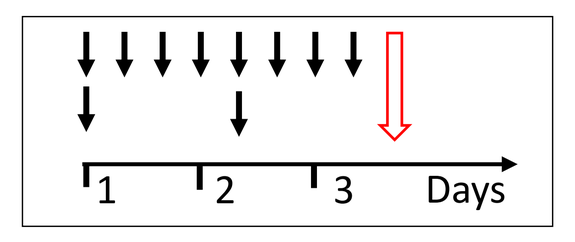
Days

8x Proleukin® (37µg/kg/ 600.00IU/kg, 3x/day IV)
2x non-α-IL-2 / STK-012
⇩Analysis for acute capillary leak

IL-2 (Proleukin) and Non-a-IL-2 caused Lymphocyte extravasation

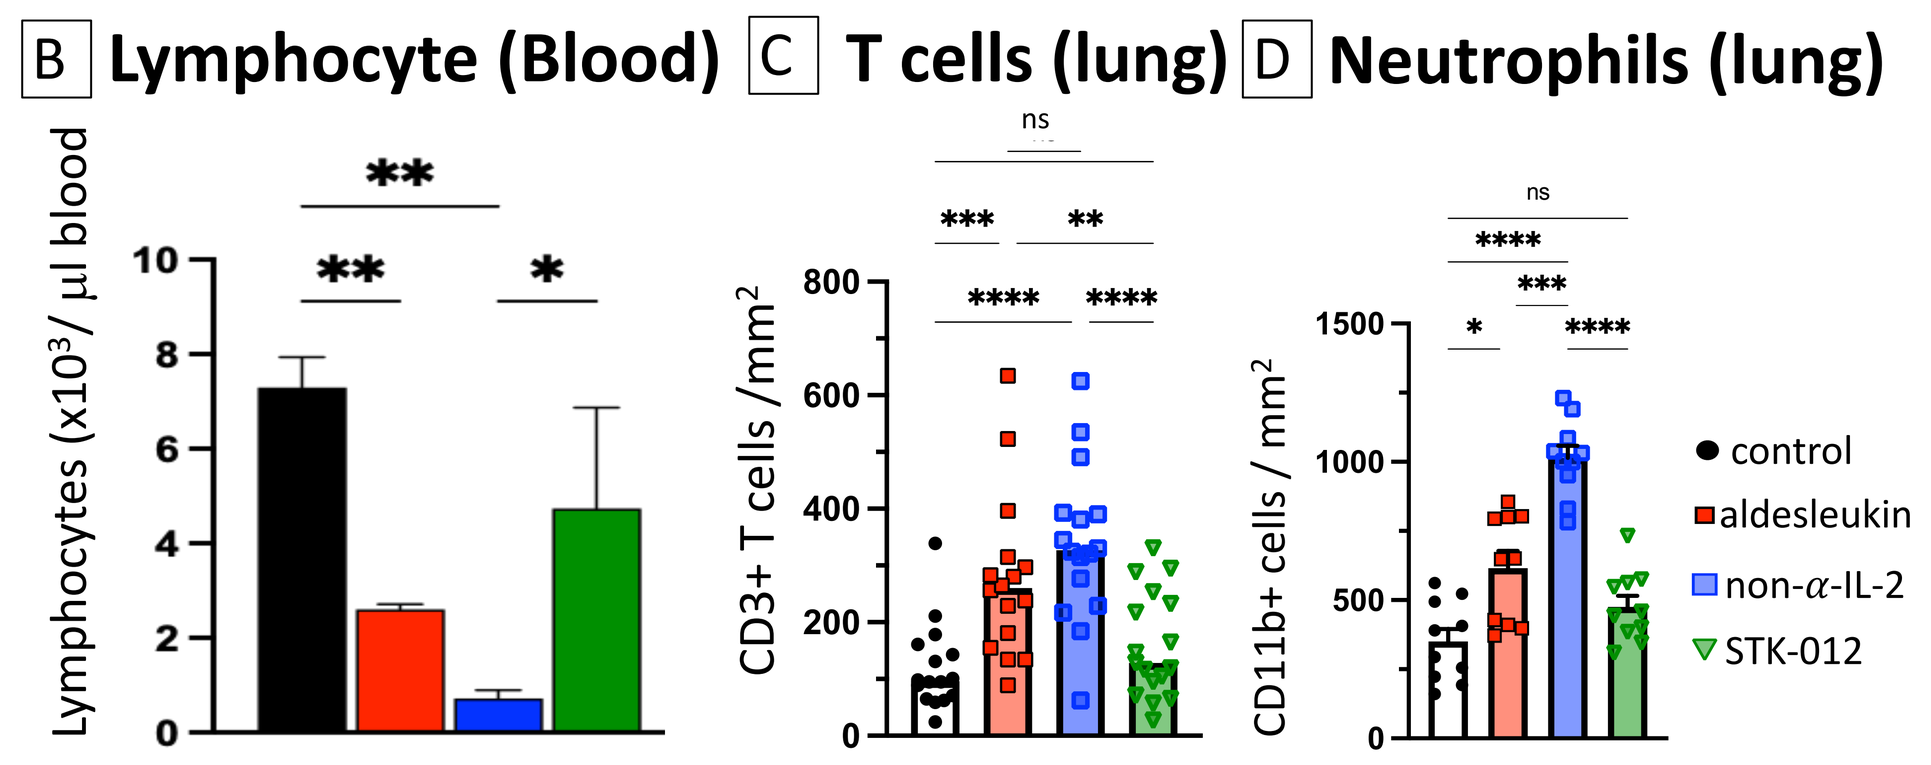
B Lymphocyte (Blood)
C T cells (lung)
D Neutrophils (lung)
Lymphocytes (x10³/ µl blood)
CD3+ T cells /mm²
CD11b+ cells / mm²
control
aldesleukin
non-α-IL-2
STK-012

Fig.6 Acute Immune toxicity to HD-IL-2, or non-α-IL-2 (PEG) but not to STK-012 (PEG) (A) Treatment schedule of hIL-2 (Proleukin®, every 8h), STK-012 or non-α-IL-2-PEG (2x) in NHP; (**B**) absolute lymphocyte count in the blood at day3 (**C**) Quantitation of T cells in the lung; (**D-H**) immunofluorescence (IF) for CD11b (green) and Foxp3+ Treg (red) in NHP acute lung injury model on day 3, (**D**) quantitation of CD11b+ cells (NK cells, macrophage, Granulocytes), (**E**) control lung, (**F**) Proleukin®, (**G**), non-α-IL-2 PEG, and (**H**) STK-012. Yellow arrow: subendothelial CD11b+ infiltrate; red arrow: Foxp3+ Treg.

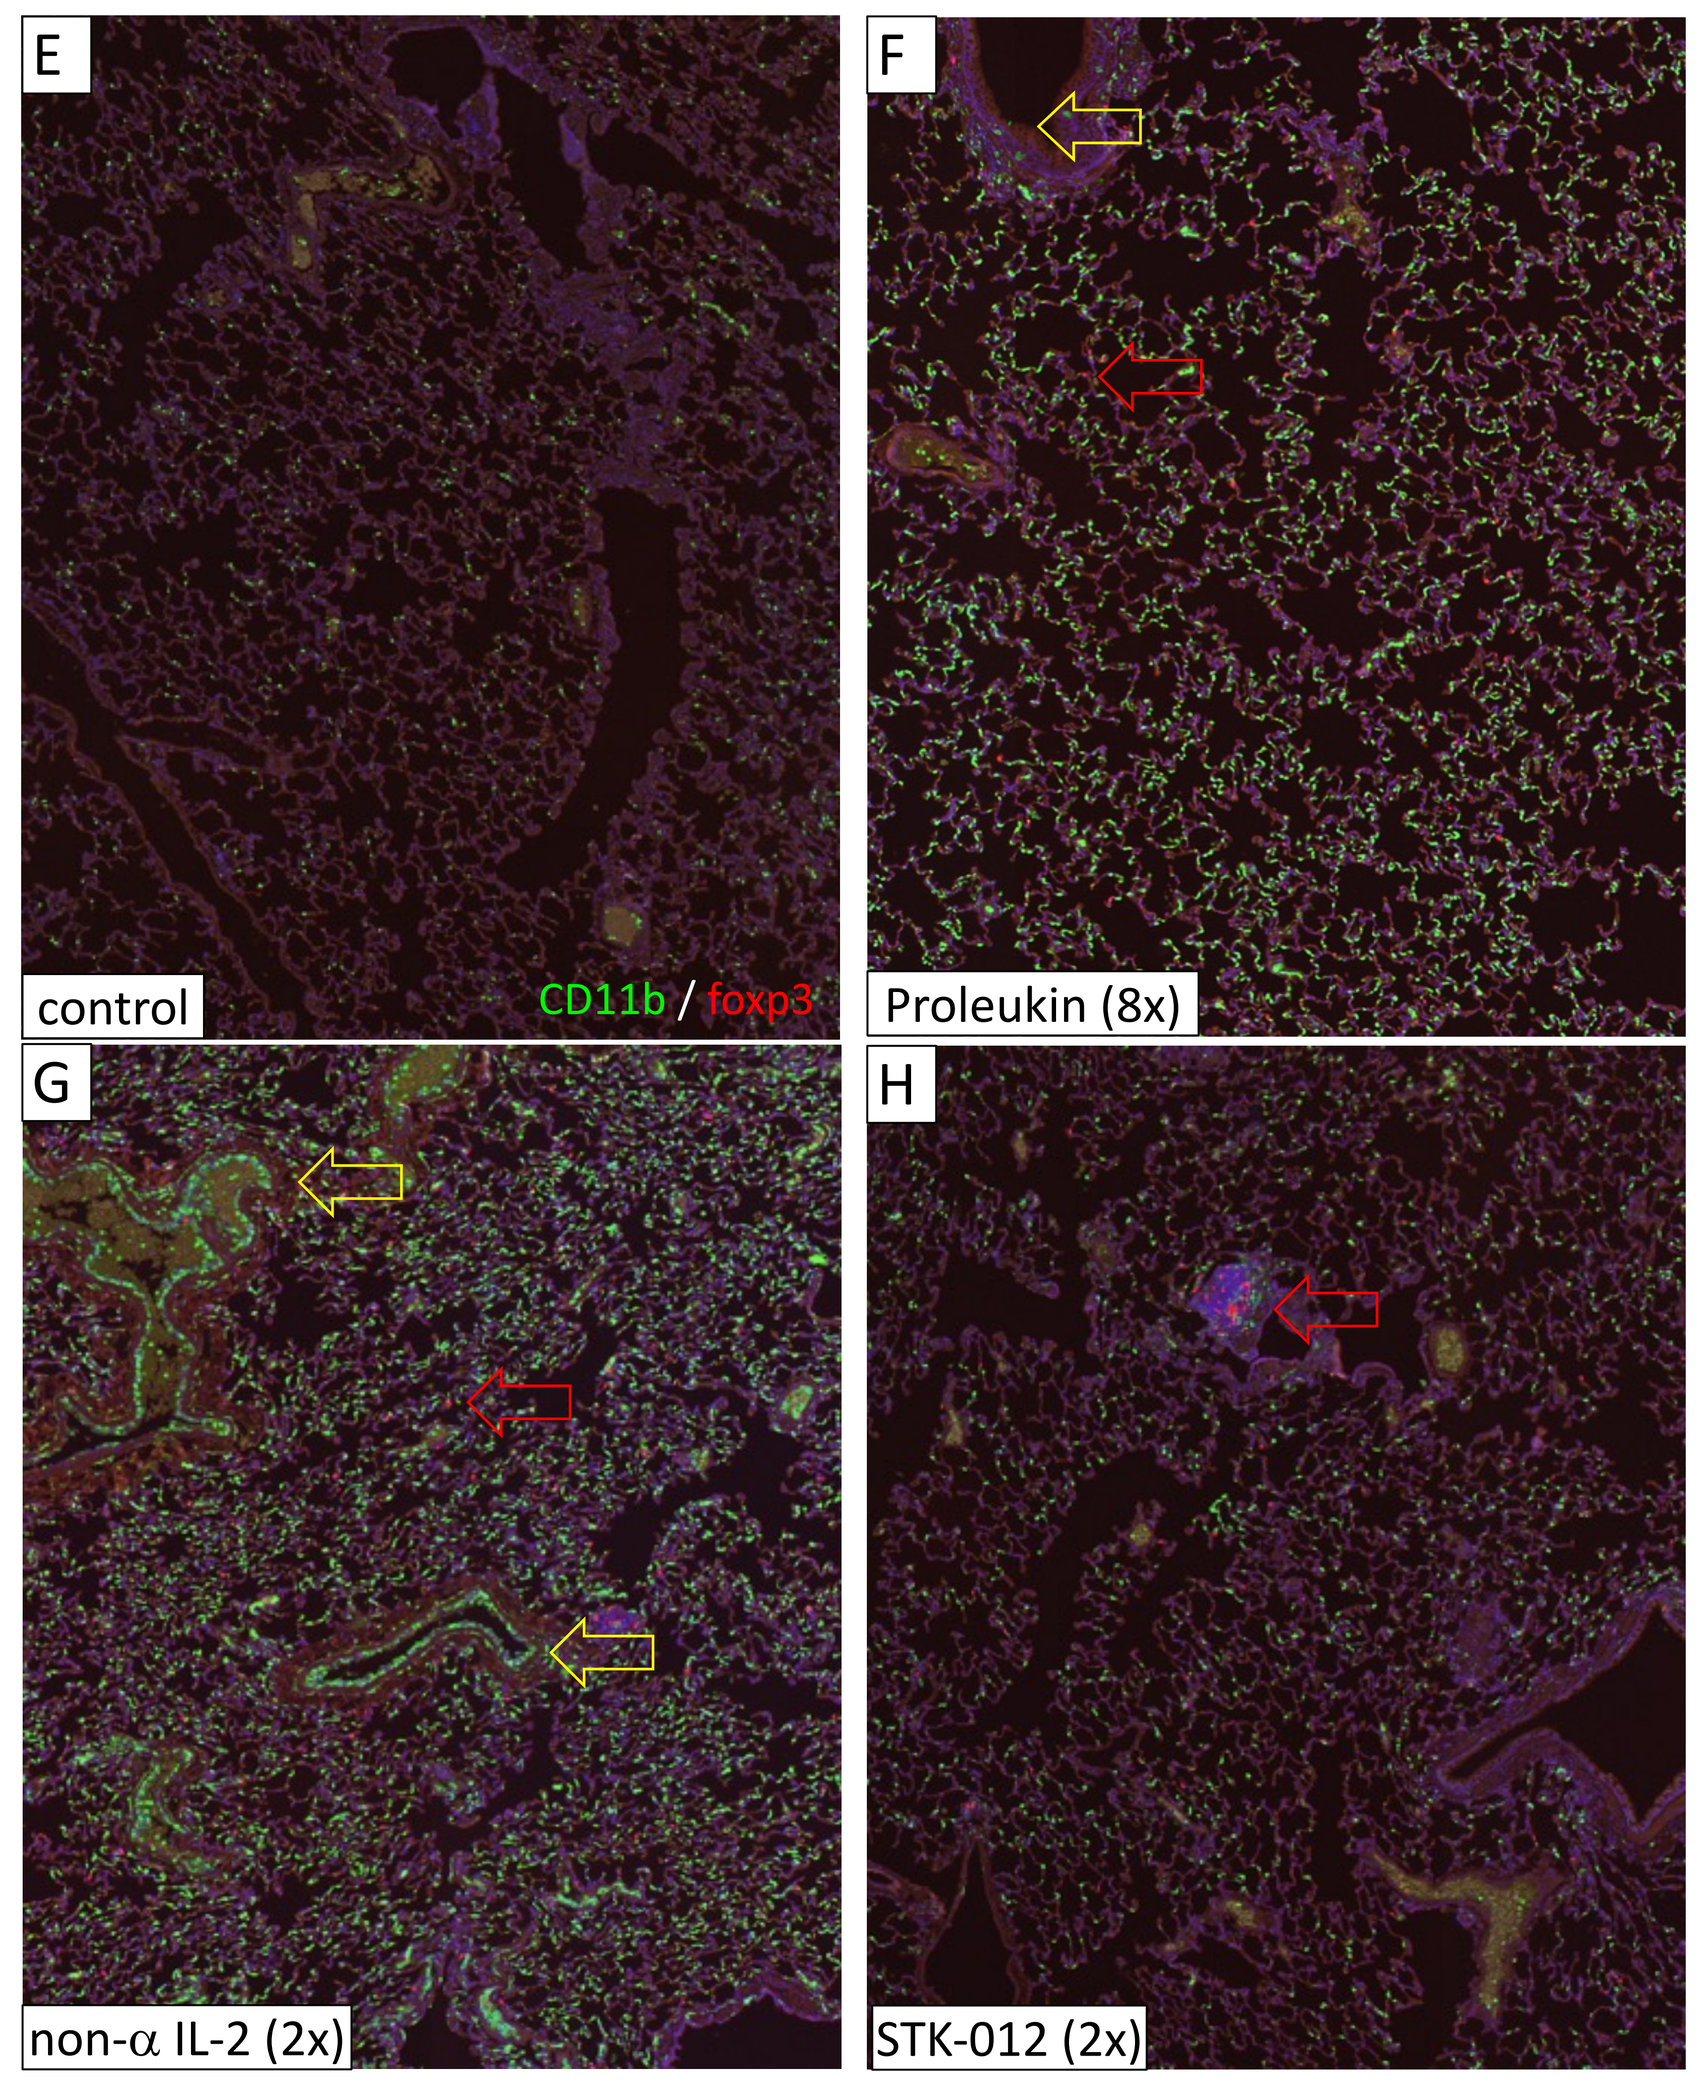
E
F
G
H
control
CD11b / foxp3
Proleukin (8x)
non-α IL-2 (2x)
STK-012 (2x)

mSTK-012 has improved anti-tumor efficacy and increases Tumor infiltrating CD8 and CD25+ CD8 T cells

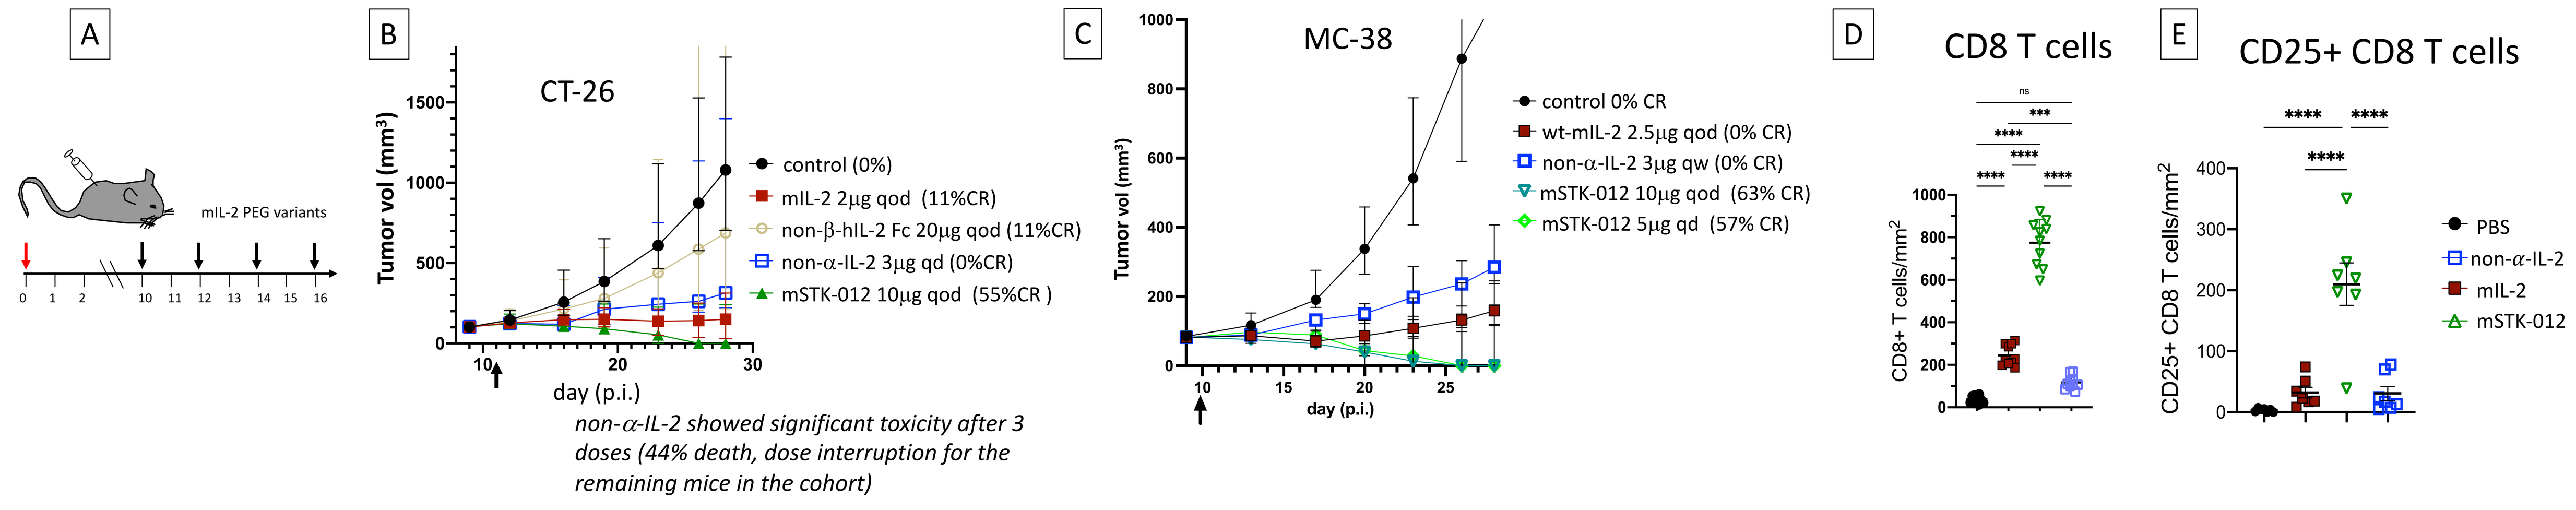
A
mIL-2 PEG variants
B
CT-26
Tumor vol (mm³)
day (p.i.)
control (0%)
mIL-2 2µg qod (11%CR)
non-β-hIL-2 Fc 20µg qod (11%CR)
non-α-IL-2 3µg qd (0%CR)
mSTK-012 10µg qod (55%CR)
non-α-IL-2 showed significant toxicity after 3 doses (44% death, dose interruption for the remaining mice in the cohort)
C
MC-38
control 0% CR
wt-mIL-2 2.5µg qod (0% CR)
non-α-IL-2 3µg qw (0% CR)
mSTK-012 10µg qod (63% CR)
mSTK-012 5µg qd (57% CR)
D
CD8 T cells
CD8+ T cells/mm²
E
CD25+ CD8 T cells
CD25+ CD8 T cells/mm²
PBS
non-α-IL-2
mIL-2
mSTK-012

Fig.7 mSTK-012 Enhances Tumor and CD8+ T cell response (A) treatment schema, (**B**) tumor growth of CT-26 syngeneic colon cancer, (**C**) Tumor growth of MC38 tumors treated with IL-2 variants (2.5µg mIL-2 every other day (MTD), 3µg non-α-IL-2 weekly (MTD) or 10µg mSTK-012 every other day).
wt-mIL-2 PEG, non-α-IL-2 and non-α-IL-2 PEG were dosed at MTD (Fig. 4); ⬇tumor injection;⬇dosing start; tumor volumes plotted post tumor inoculation; qd: daily; qod: every other day; qw: weekly.

mSTK-012 but not non-α-IL-2 Expands Tumor Antigen Specific PD-1+ CD8+ T cells

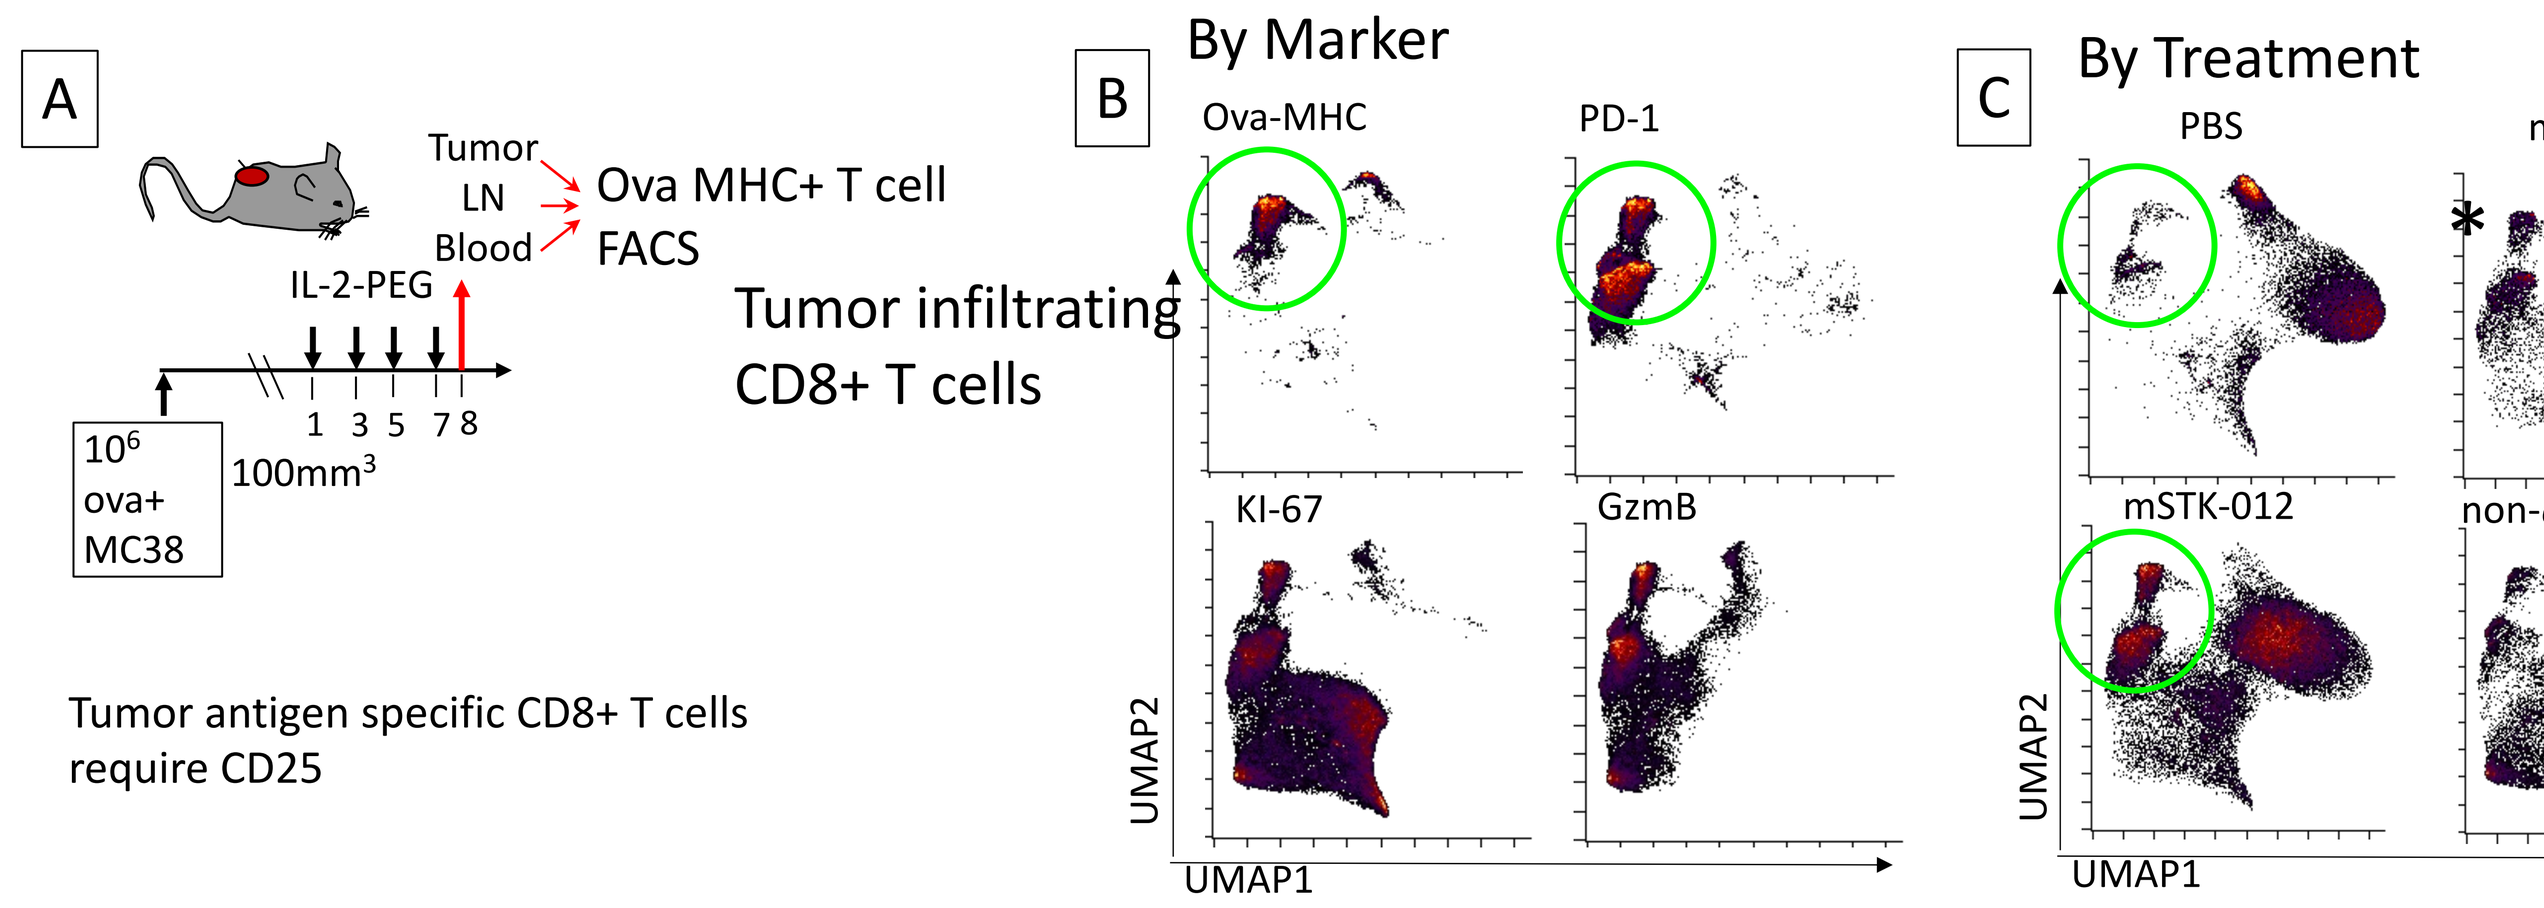
A
Tumor
LN
Blood
Ova MHC+ T cell FACS
IL-2-PEG
10⁶ ova+ MC38
100mm³
Tumor infiltrating CD8+ T cells
Tumor antigen specific CD8+ T cells require CD25
B
By Marker
Ova-MHC
PD-1
KI-67
GzmB
C
By Treatment
PBS
mIL-2
mSTK-012
non-α-IL-2
UMAP1
UMAP2

Figure 8. mSTK-012 Increases PD1+ GzmB+ KI-67+ tumor antigen specific CD8 T cells.
A. Model Setup and treatment schema of C57BL/6 mice implanted with MC38-Ova cells and treated qod with mIL-2, non-α-IL-2 or mSTK-012. (**B,C**) IL-2 and STK-012 treatment increased the antigen specific Tumor infiltrating T cells(b)

Combination of anti-PD-1 with mSTK-012 has increased anti-tumor efficacy

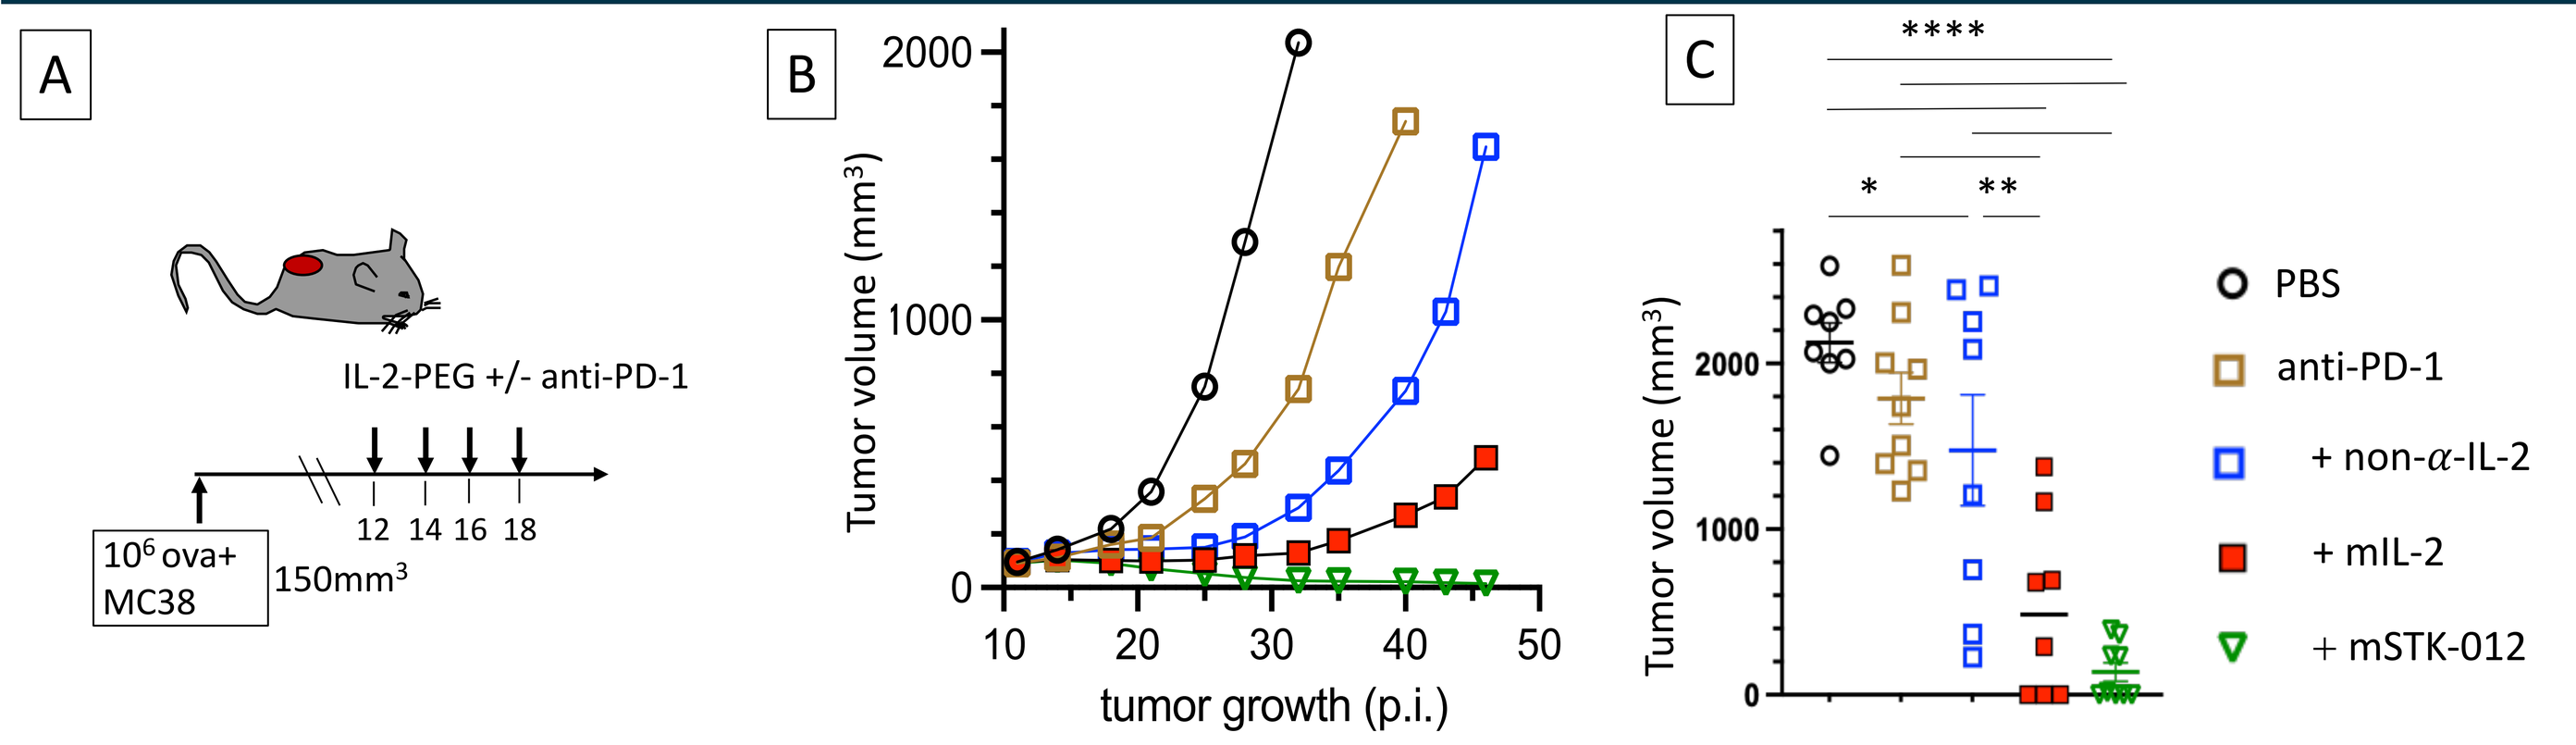
A
IL-2-PEG +/- anti-PD-1
10⁶ ova+ MC38
150mm³
B
Tumor volume (mm³)
tumor growth (p.i.)
C
Tumor volume (mm³)
PBS
anti-PD-1
+ non-α-IL-2
+ mIL-2
+ mSTK-012

Figure 9. mSTK-012 has increased combination efficacy with anti-PD1
A. Model and treatment schema of C57BL/6 mice implanted with MC38 cells and treated qod with mIL-2, non-α-IL-2 or mSTK-012. (**B,C**) mSTK-012 treatment increased the anti-tumor efficacy T cells of anti-PD-1

Conclusions

- STK-012 is highly selective for antigen activated T cells
 - Limited activity in NK cells and resting, bystander T cells
- No IL-2 related acute toxicity and capillary leak induced by mSTK-012 and STK-012
 - Acute NK cell dependent lung and liver toxicity observed with wt-IL-2 and non-α-IL-2
- mSTK-012 has strongly increased anti-tumor activity in syngeneic tumor models compared to wt-mIL-2, non-α-IL-2 or non-β-IL-2
 - Complete responses as monotherapy (in large tumors)
 - Efficacy correlates with strongly increased intratumoral $CD8^+$ T cells and $CD25^+CD8^+$ T cells
 - Strongest increase of intratumoral CD8 / Treg ratio compared to other IL-2s
 - Significantly stronger expansion of antigen specific T_{PEX} cells with mSTK-012
 - Improved combination anti-tumor efficacy with anti-PD-1

- In summary, STK-012 avoids IL-2 mediated acute toxicity which may enable specific expansion of antigen activated memory T cells in cancer patients, enabling a more durable anti-tumor response.
- Clinical trials with STK-012, a human STK-012, are in progress (NCT05098132).