SYNCAR: An Engineered IL-2/IL-2R-system that Selectively Enhances CD19 CAR T cells to Deplete B Cells and Provide Therapeutic Benefit in SLE and RA Mouse Models Without Lymphodepletion

Ethan Jung, Marie Semana, Ivan Cheng, Helena Silva, Sandro Vivona, Somya Singh, Michele Bauer, Mohammed Ali, Henry Rosas, Woei Chang, Alayna Gibbs, Deepti Rokkam, Navneet Ratti, Patrick J. Lupardus, Martin Oft, and Paul-Joseph Aspuria
Synthekine, Inc. Menlo Park, California, USA

Introduction

CD19 CAR-T therapy has shown efficacy in autoimmune (AI) diseases, such as systemic lupus erythematosus (SLE) and lupus nephritis (LN), demonstrating its transformative potential beyond oncology. Additionally, exploring its application in other AI conditions that involve B cells in their pathogenesis, like rheumatoid arthritis (RA), can broaden its therapeutic impact. Lymphodepleting (LD) chemotherapy prior to CAR-T therapy is an essential step to enable T cell engraftment and efficacy by increasing the availability of homeostatic cytokines.

However, LD leads to adverse events and risks including cytokine release syndrome, cytopenia, a greater risk of infection, genotoxic risk and secondary malignancies. To fully capitalize on its therapeutic potential, strategies to minimize LD while maintaining the expansion and efficacy of CD19 CAR-T therapy are desirable.

IL-2, a potent stimulator of T cells, could increase T cell proliferation, but its use is **limited by systemic toxicities** due to widespread immune cell activation. We previously described an orthogonal oIL-2/oIL-2R-system consisting of a pegylated, IL-2 mutein (STK-009) that does not significantly activate the wild type (WT) receptor and a mutated IL-2 Receptor Beta (oRβ) that does not significantly respond to WT IL-2. This system provides a "private IL-2 signal" in engineered oRβ-expressing cells while avoiding signaling in bystander immune cells, reducing toxicities. We have demonstrated STK-009 selectively expands oRβ-expressing CD19 CAR T cells (SYNCAR-001), leading to enhanced anti-tumor control and deep, durable responses in bulky lymphoma mouse models.

SYNCAR: A Lock and Key System Providing a Private IL-2 Signal to Engineered Cells *In Vivo* Obviates the Need for Lymphodepletion

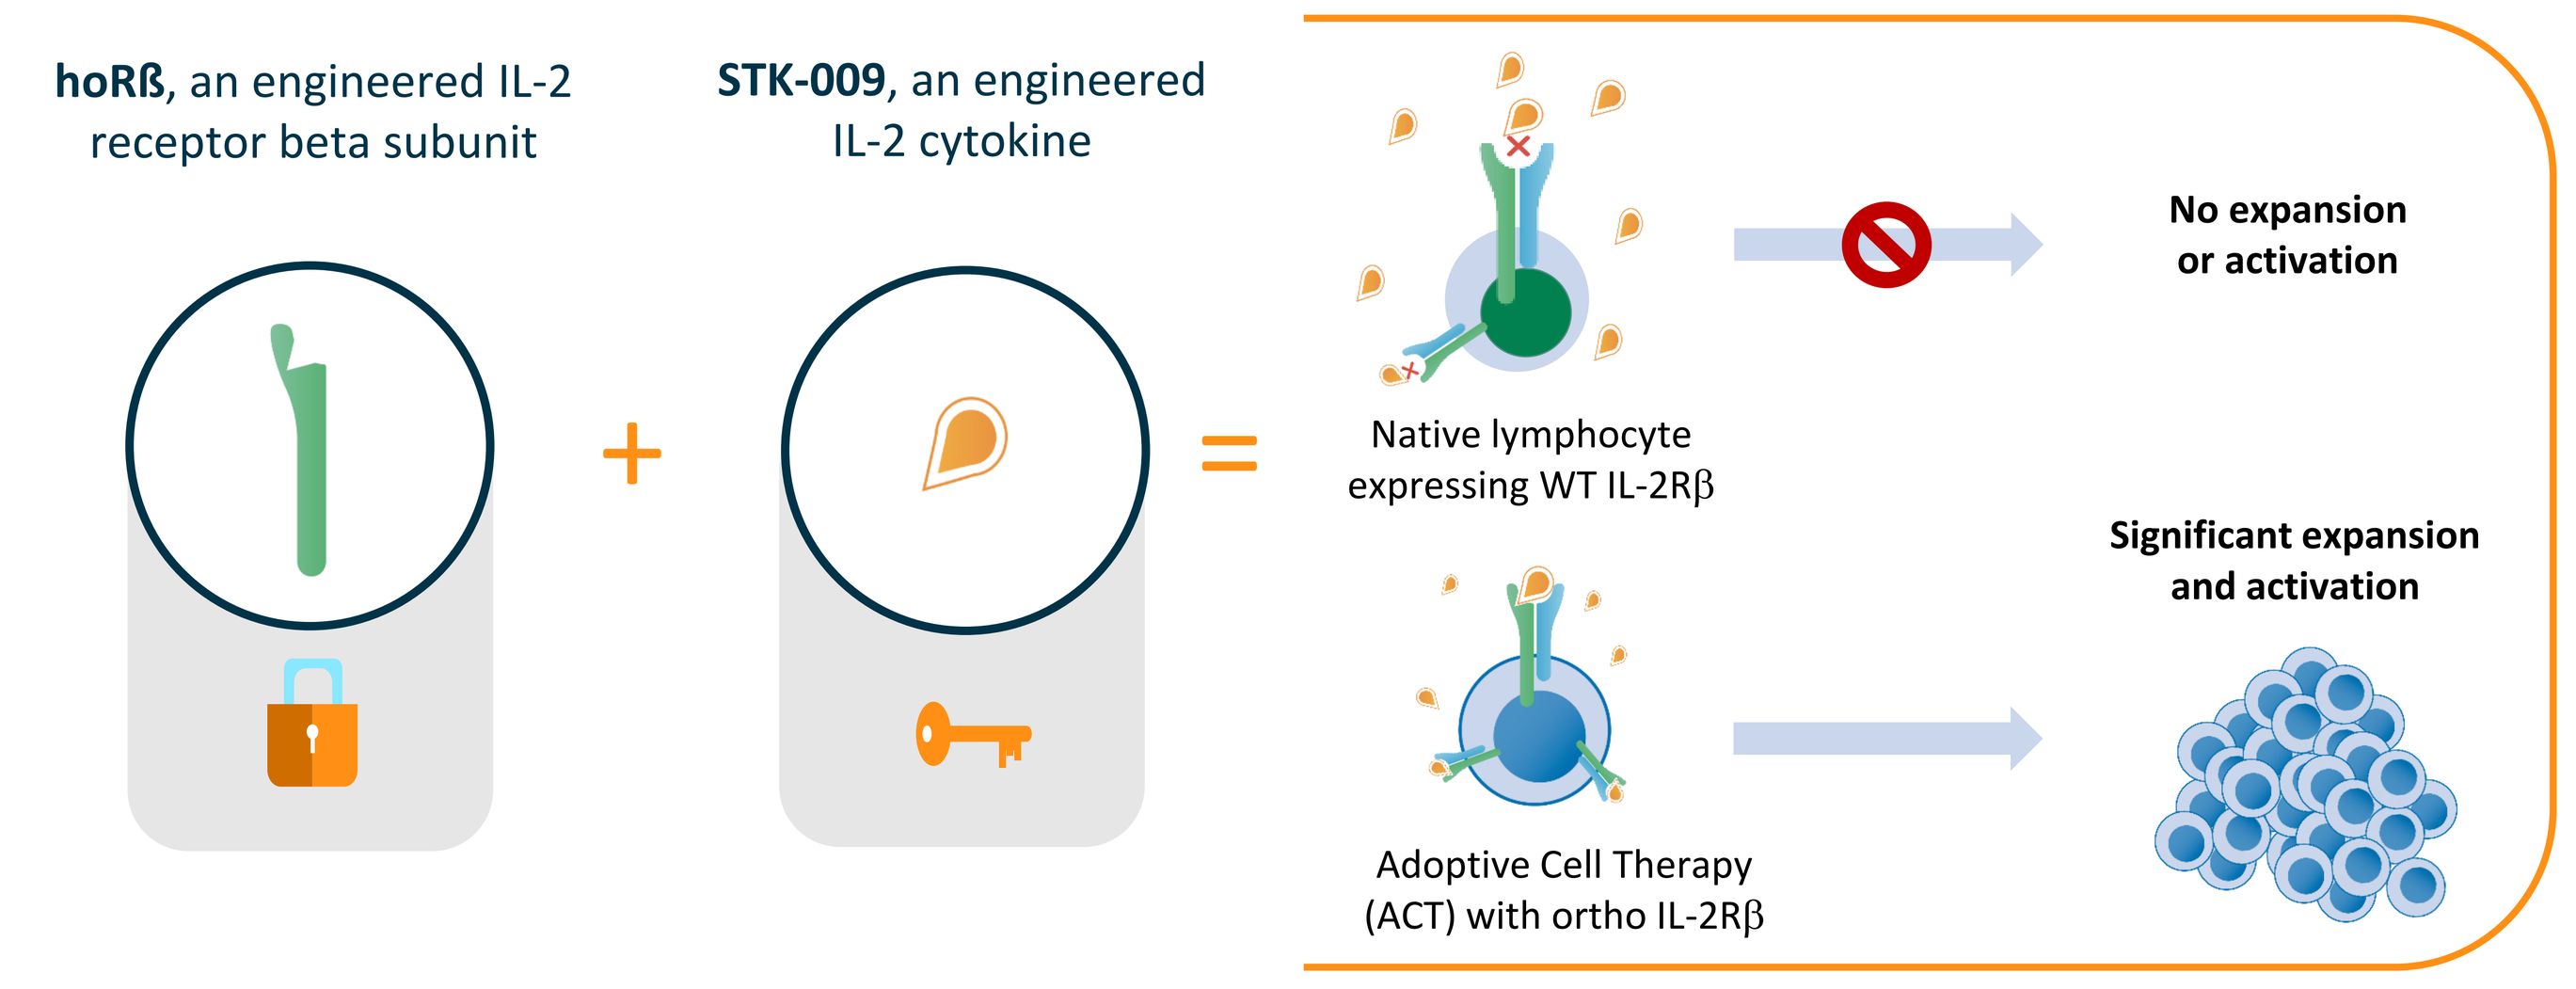
horß, an engineered IL-2 receptor beta subunit
STK-009, an engineered IL-2 cytokine
+
=
Native lymphocyte expressing WT IL-2Rβ
No expansion or activation
Adoptive Cell Therapy (ACT) with ortho IL-2Rβ
Significant expansion and activation

Potential to be incorporated into a wide range of adoptive cell therapies, including CAR-Ts , TCRs, TILs, and Tregs

SYNCAR-001: CD19 CAR T cells Co-Expressing the hoRβ to Target CD19 Expressing Lymphocytes

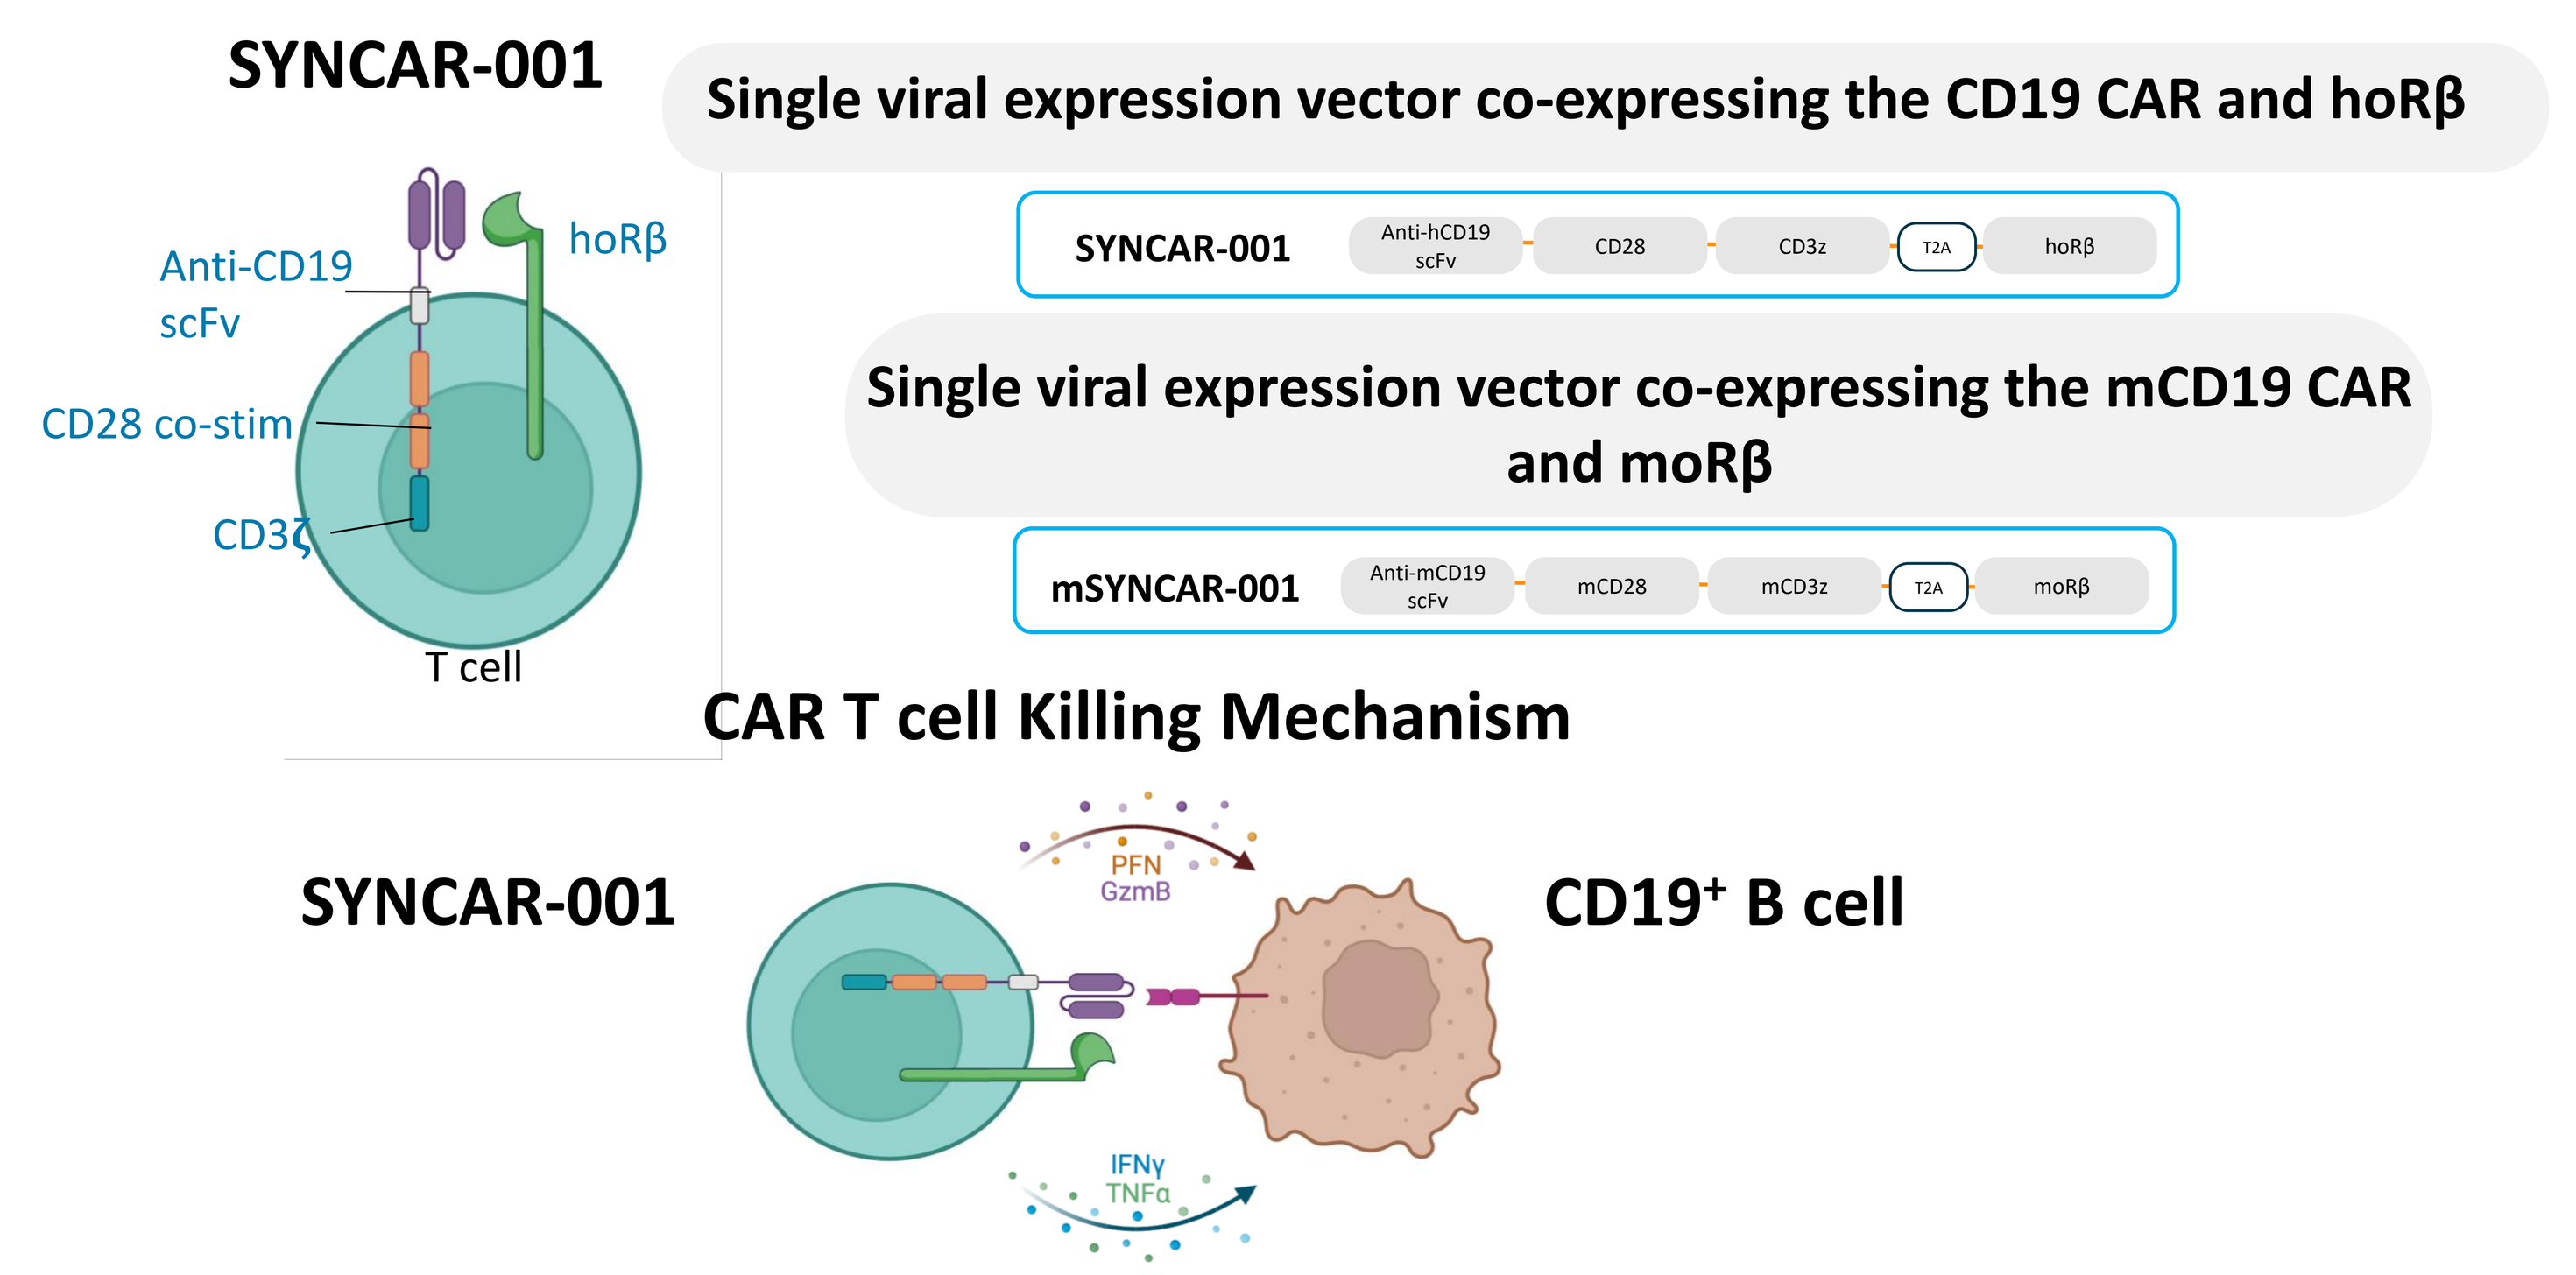
SYNCAR-001
Anti-CD19 scFv
horβ
CD28 co-stim
CD3ζ
T cell
Single viral expression vector co-expressing the CD19 CAR and hoRβ
SYNCAR-001
Anti-hCD19 scFv
CD28
CD3z
T2A
hoRβ
Single viral expression vector co-expressing the mCD19 CAR and moRβ
mSYNCAR-001
Anti-mCD19 scFv
mCD28
mCD3z
T2A
moRβ
CAR T cell Killing Mechanism
SYNCAR-001
PFN
GzmB
CD19⁺ B cell
IFNγ
TNFα

Methods

To investigate the therapeutic potential of the orthogonal IL-2/IL-2R system, we utilized CD19 CAR T cells in autoimmune mouse models without lymphodepletion (LD). Specifically, we administered mSTK-009, the murine (m) surrogate of STK-009, in conjunction with murine T cells co-expressing a mCD19 CAR and moRβ (mSYNCAR-001).

We employed two autoimmune mouse models for systemic lupus erythematosus (SLE) and rheumatoid arthritis (RA). In the SLE model, aged NZB/NZWF1 mice, characterized by elevated anti-dsDNA/nuclear antibodies and kidney dysfunction, were used. For RA, a non-lymphodepleted collagen-induced arthritis model was utilized. In both models, mice received mSYNCAR-001 alone or in combination with mSTK-009. The impact on CD19+ B cell depletion, autoantibody production, kidney function, and arthritis symptoms were assessed in the relevant model.

Murine SYNCAR-001 + STK-009 Induces an "Immune Reset" as Evidenced by Transient B cell Depletion and Sustained Reduction of Lupus Serology in a Therapeutic NZB/W Lupus Model Without Lymphodepletion

Therapeutic treatment model

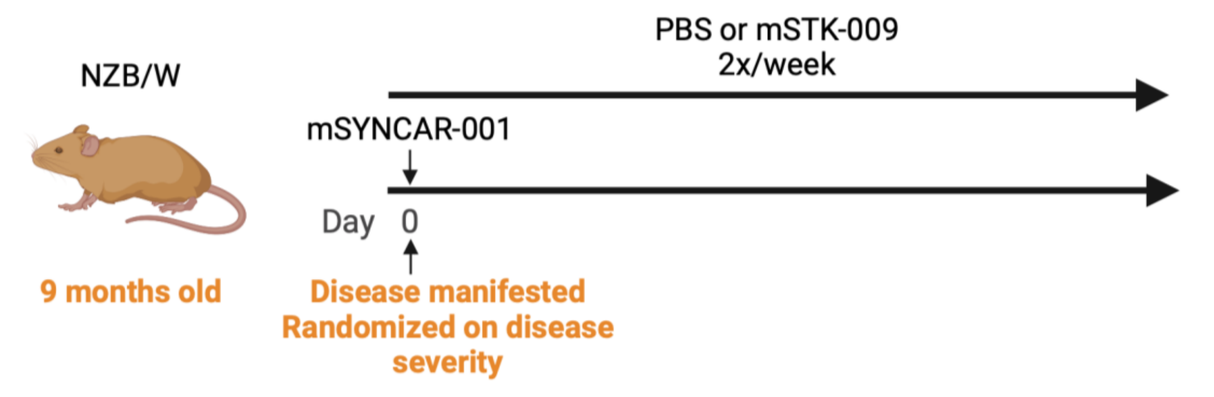
NZB/W
PBS or mSTK-009 2x/week
mSYNCAR-001
Day 0
9 months old
Disease manifested Randomized on disease severity

Peripheral B cell depletion and mSYNCAR-001 expansion with mSTK-009

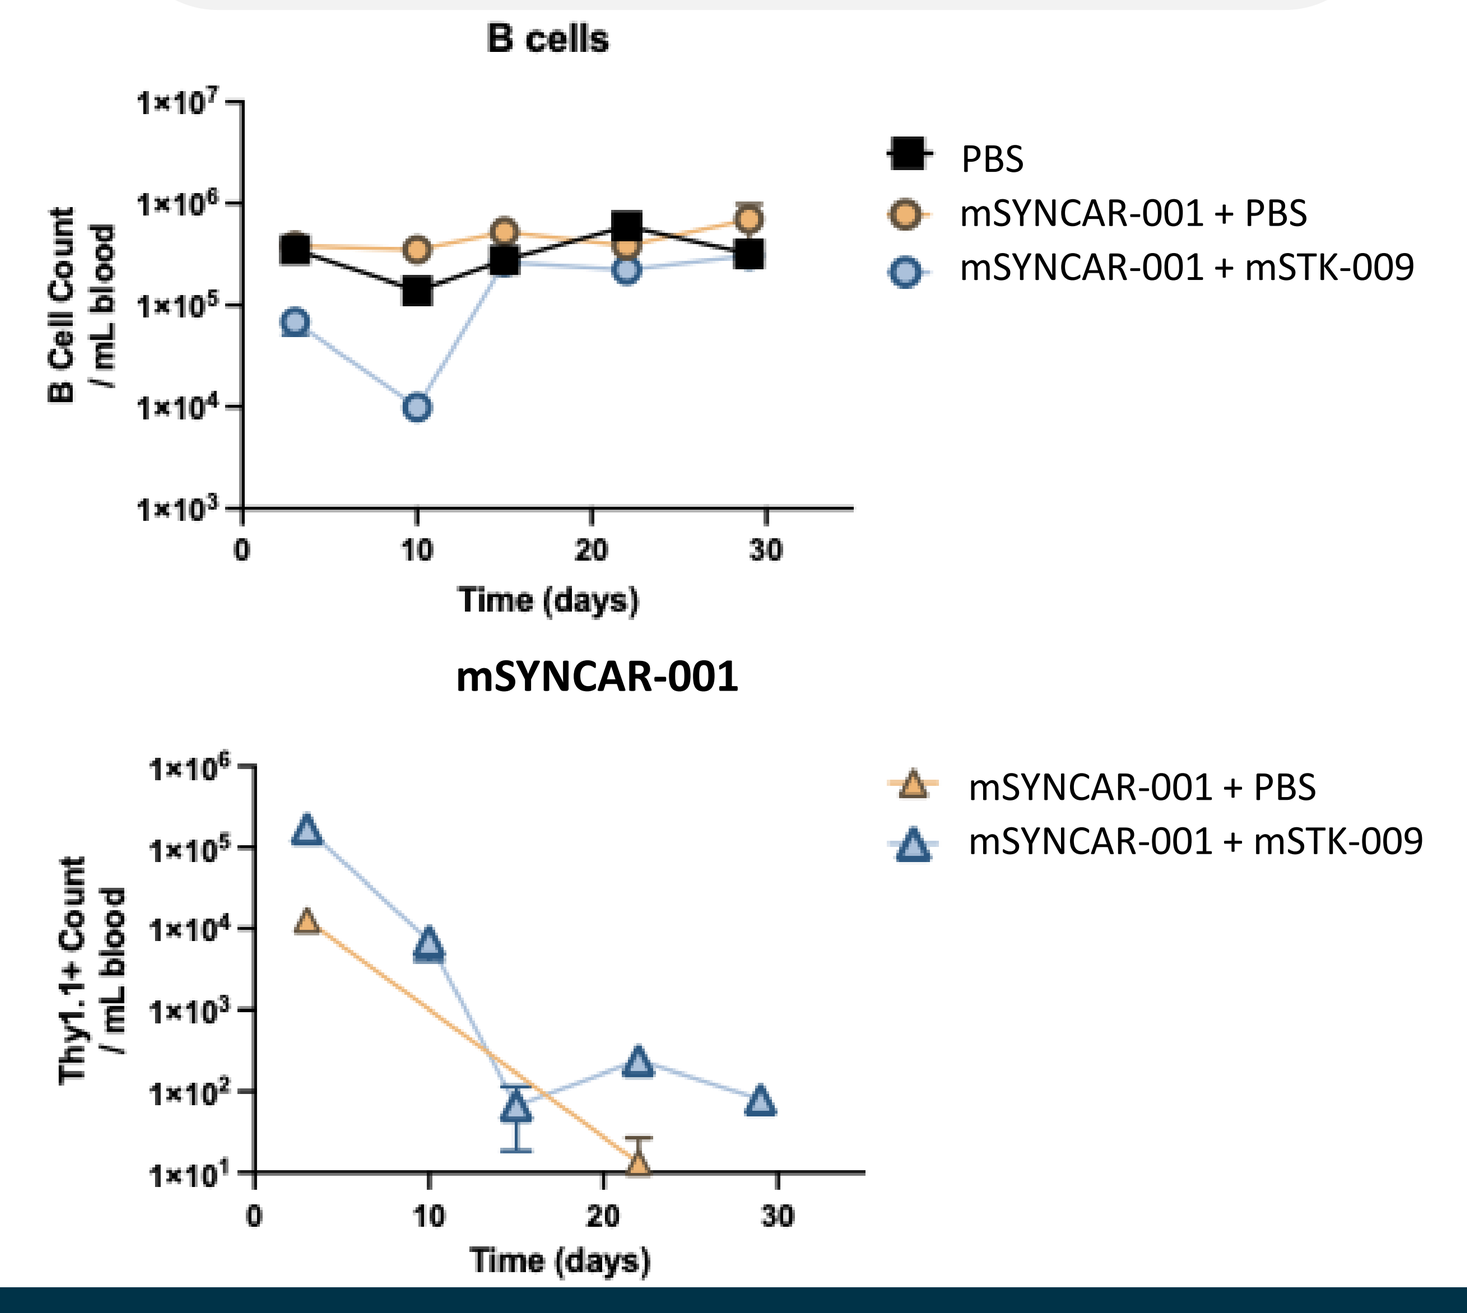
B cells
B Cell Count / mL blood
Time (days)
PBS
mSYNCAR-001 + PBS
mSYNCAR-001 + mSTK-009
mSYNCAR-001
Thy1.1+ Count / mL blood

Deep B cell depletion in peripheral blood and tissues with mSTK-009

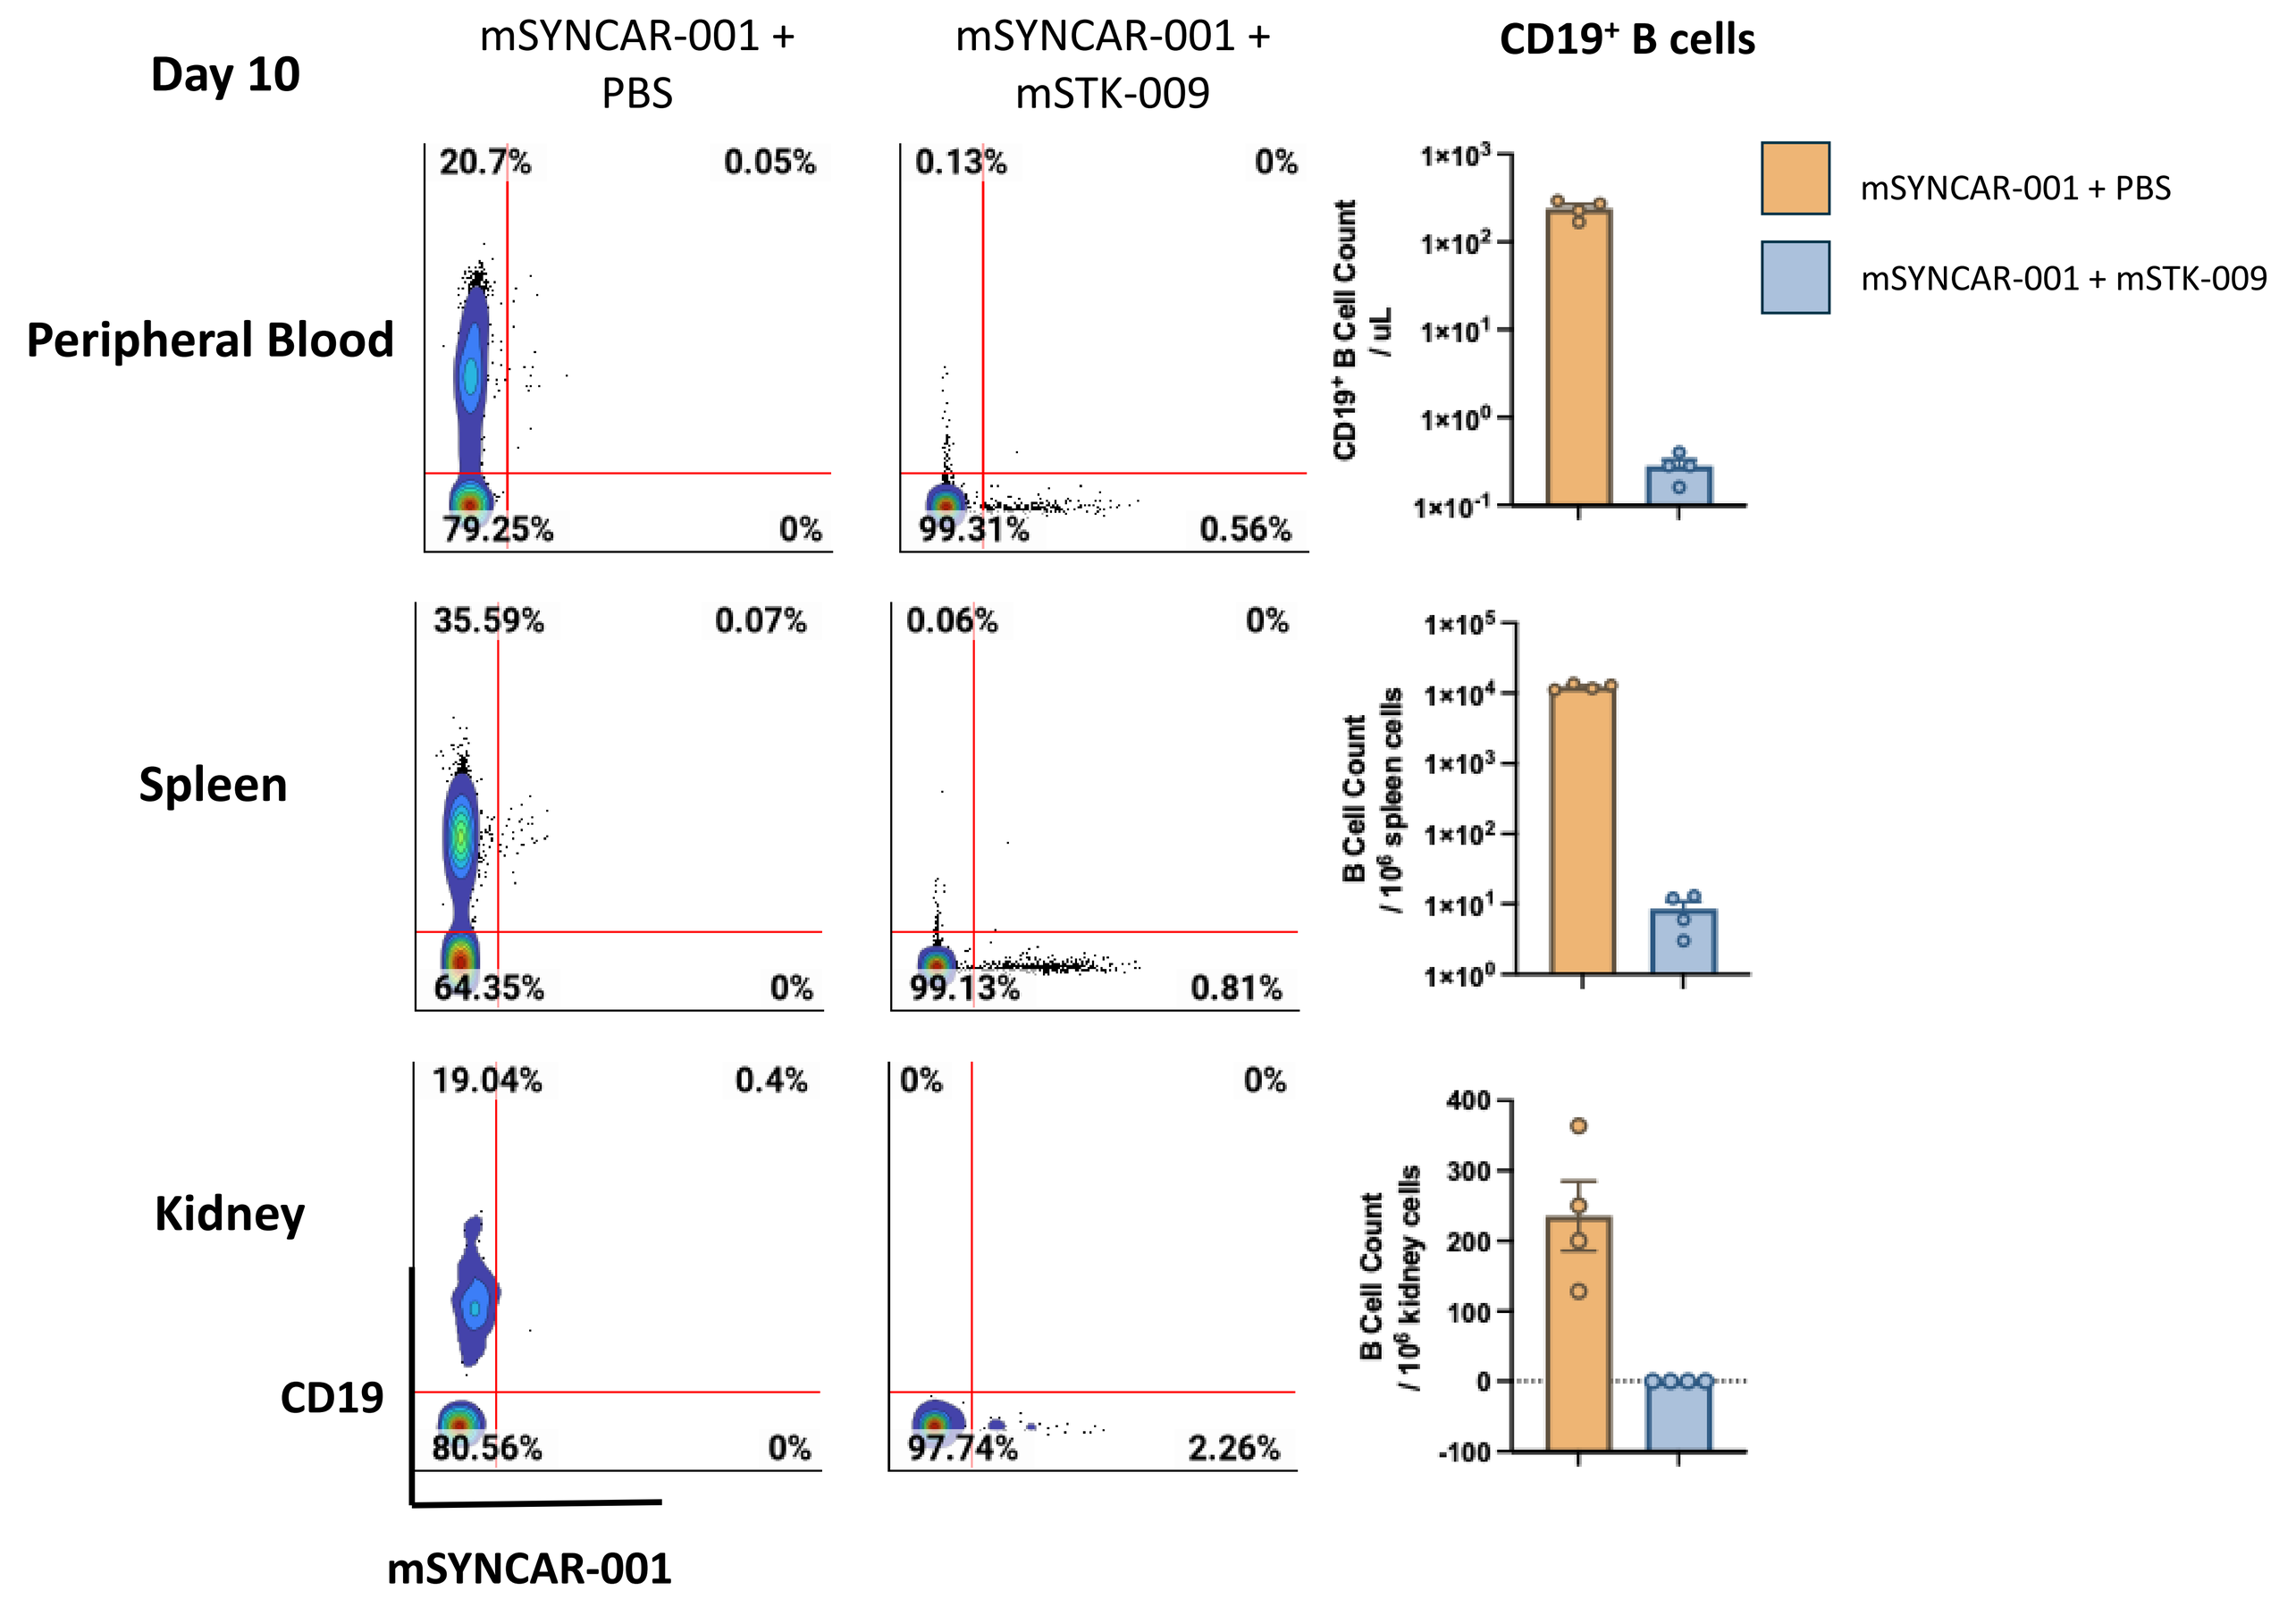
Day 10
mSYNCAR-001 + PBS
mSYNCAR-001 + mSTK-009
CD19⁺ B cells
Peripheral Blood
Spleen
Kidney
CD19
mSYNCAR-001
mSYNCAR-001 + PBS
mSYNCAR-001 + mSTK-009

Low levels of lupus associated antibodies and proteinuria at 6 weeks post-CAR T infusion

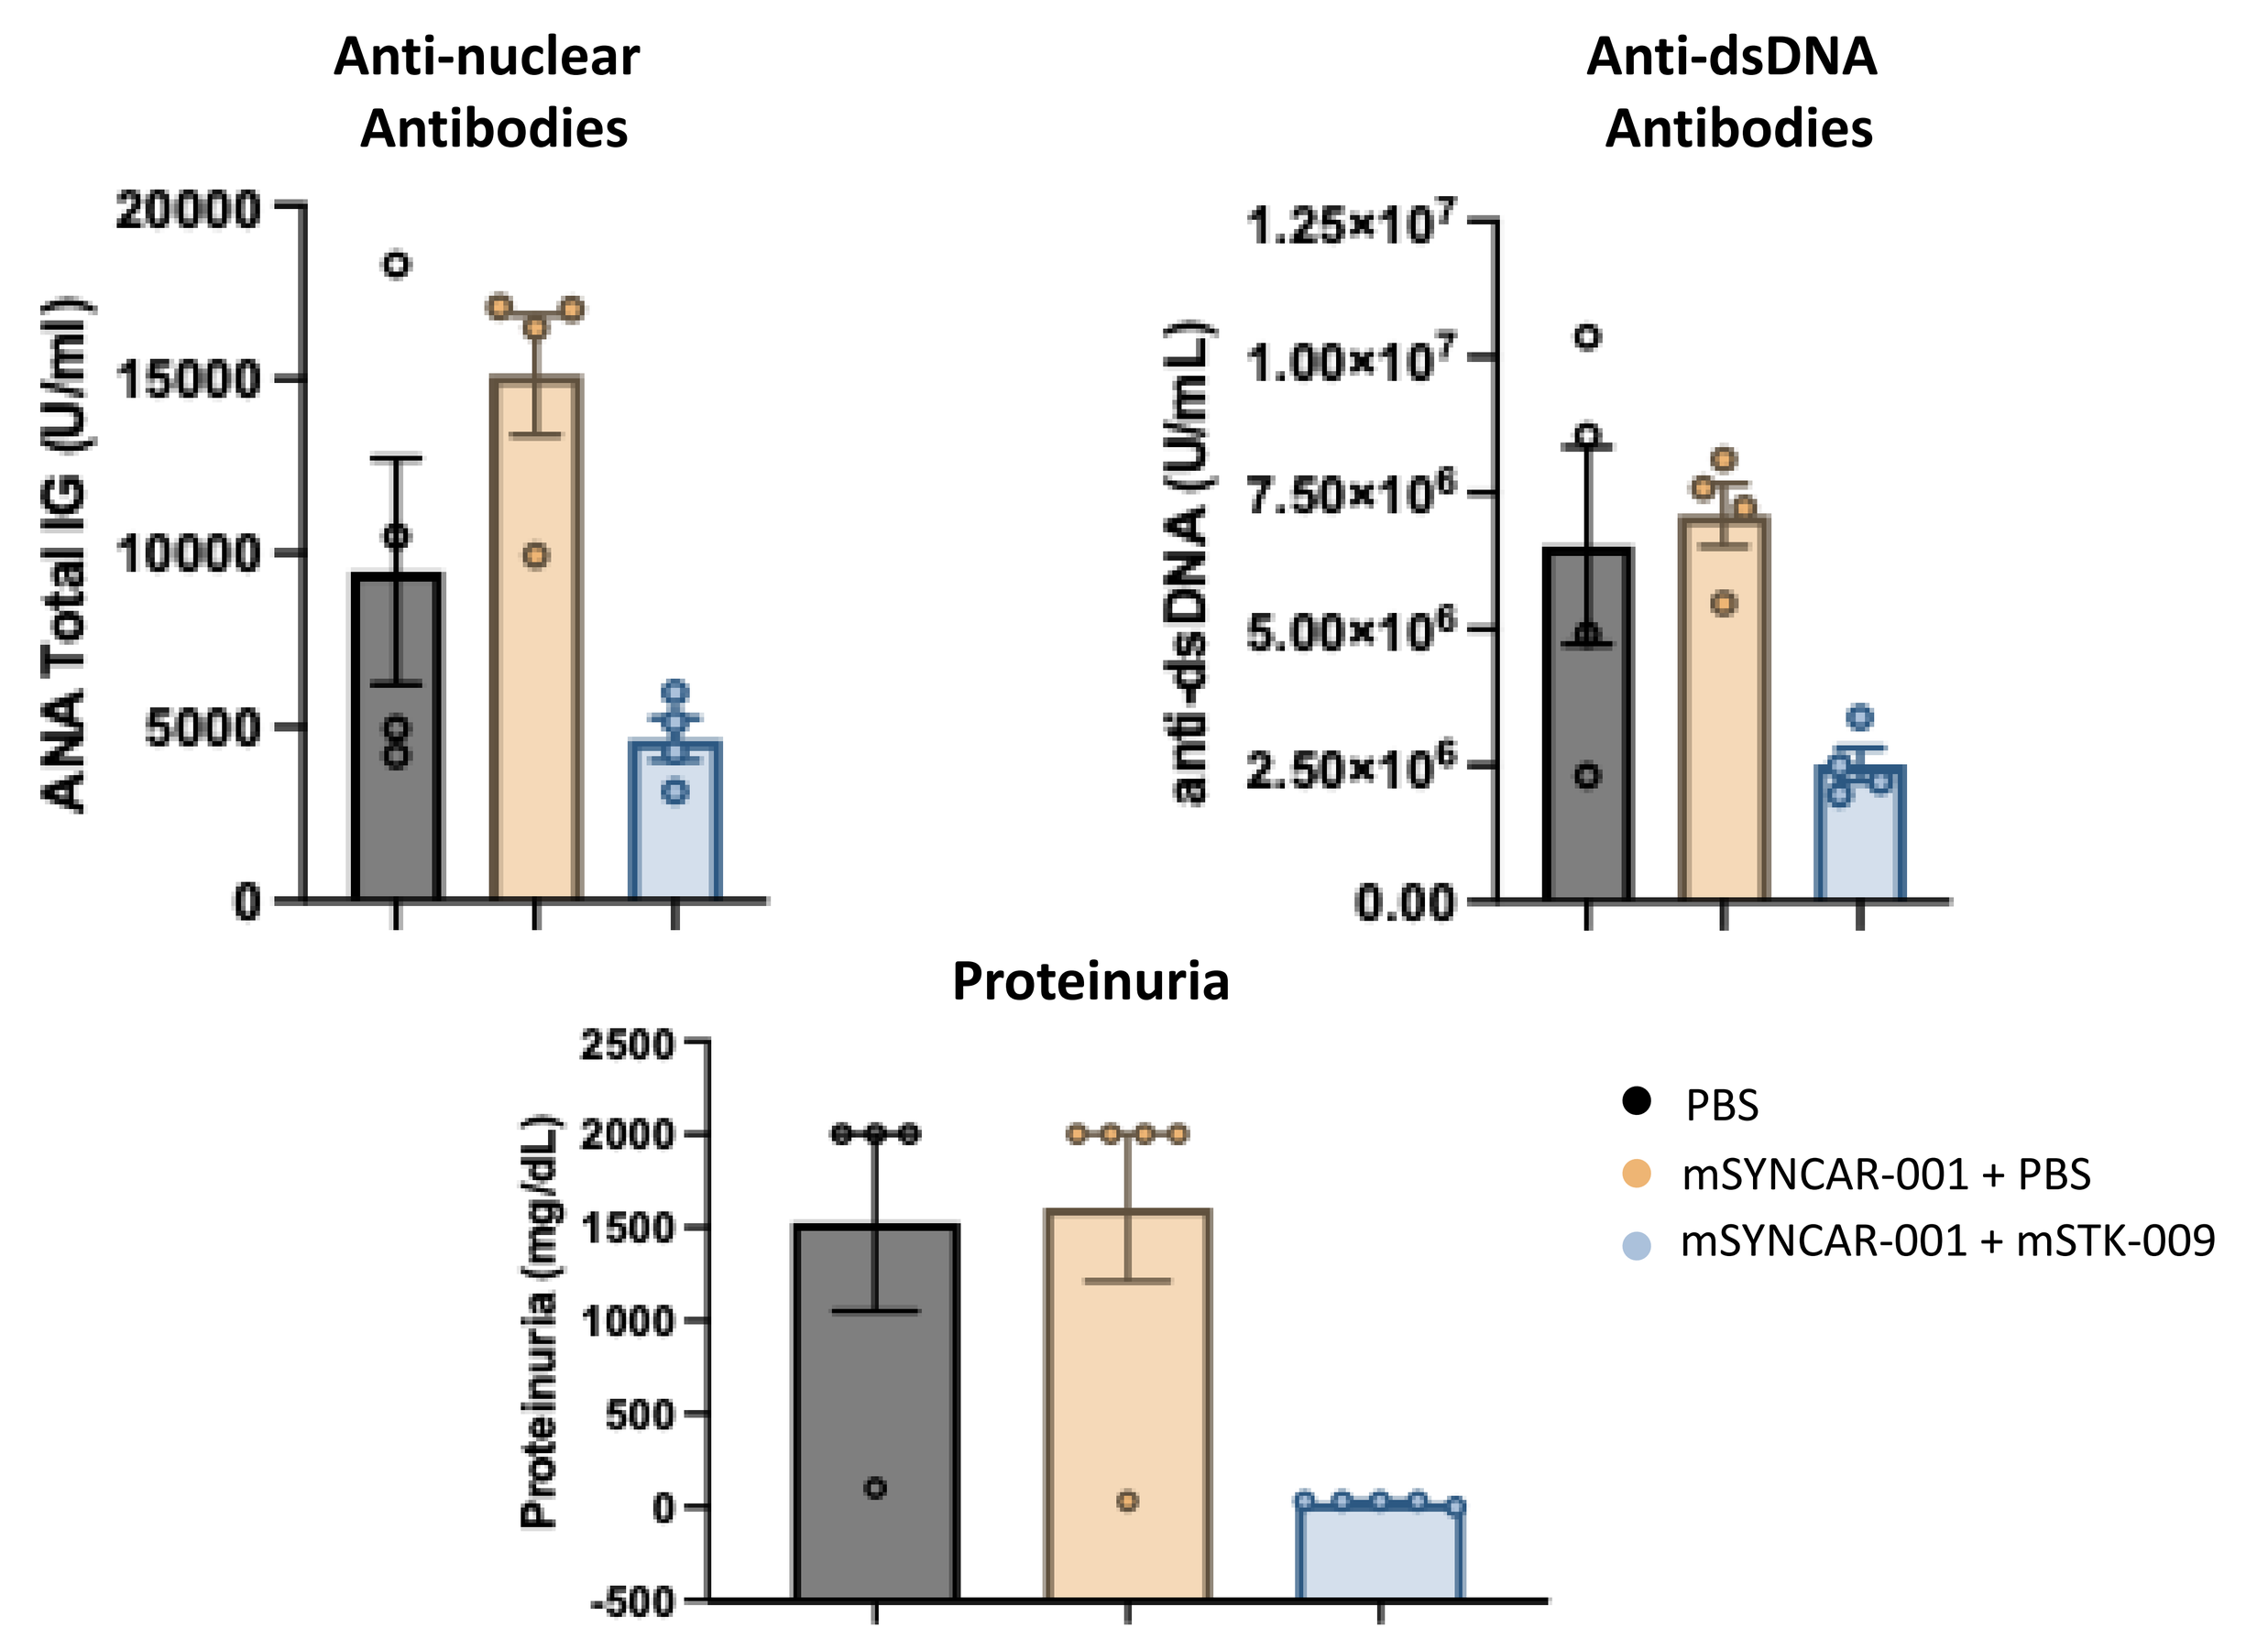
Anti-nuclear Antibodies
ANA Total IG (U/ml)
Anti-dsDNA Antibodies
anti-dsDNA (U/mL)
Proteinuria
Proteinuria (mg/dL)
PBS
mSYNCAR-001 + PBS
mSYNCAR-001 + mSTK-009

Murine SYNCAR-001 + STK-009 Induce B Cell Depletion and Joint Normalization in a Therapeutic Collagen Induced Arthritis (CIA) Model Without Lymphodepletion

Therapeutic treatment model

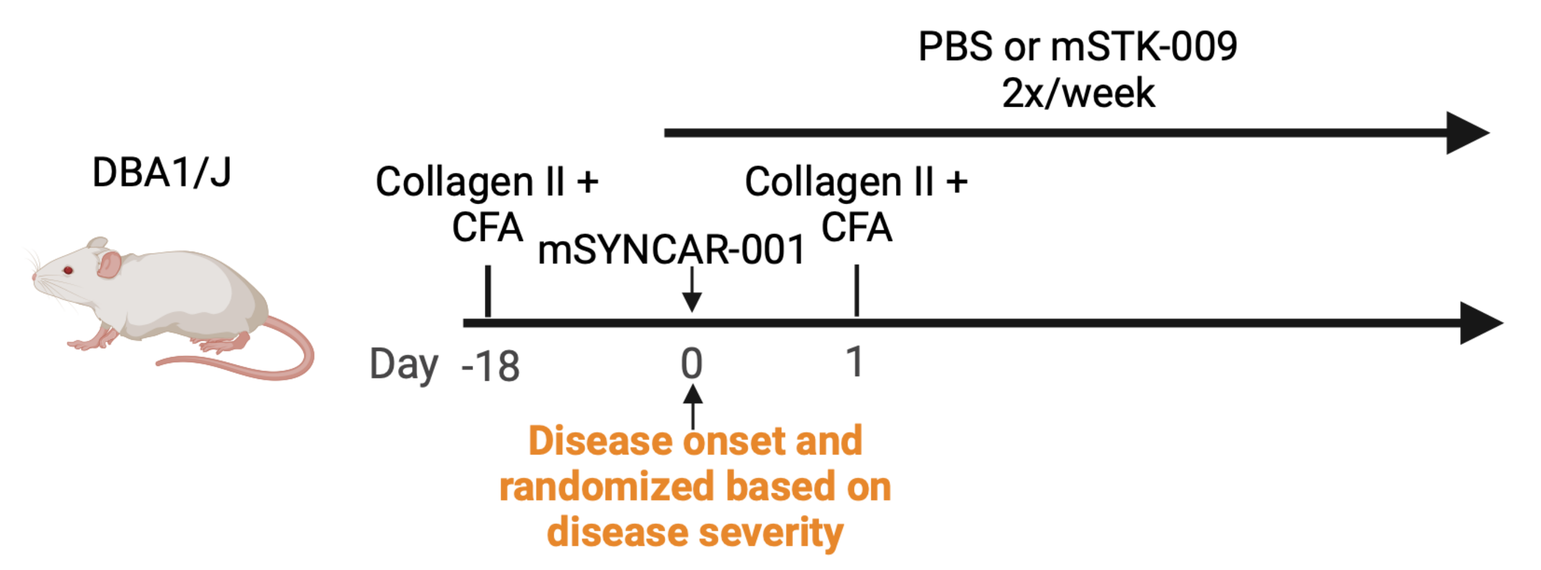
DBA1/J
PBS or mSTK-009 2x/week
Collagen II + CFA
mSYNCAR-001
Collagen II + CFA
Day -18 0 1
Disease onset and randomized based on disease severity

Significant B cell reduction with mSYNCAR-001 + mSTK-009

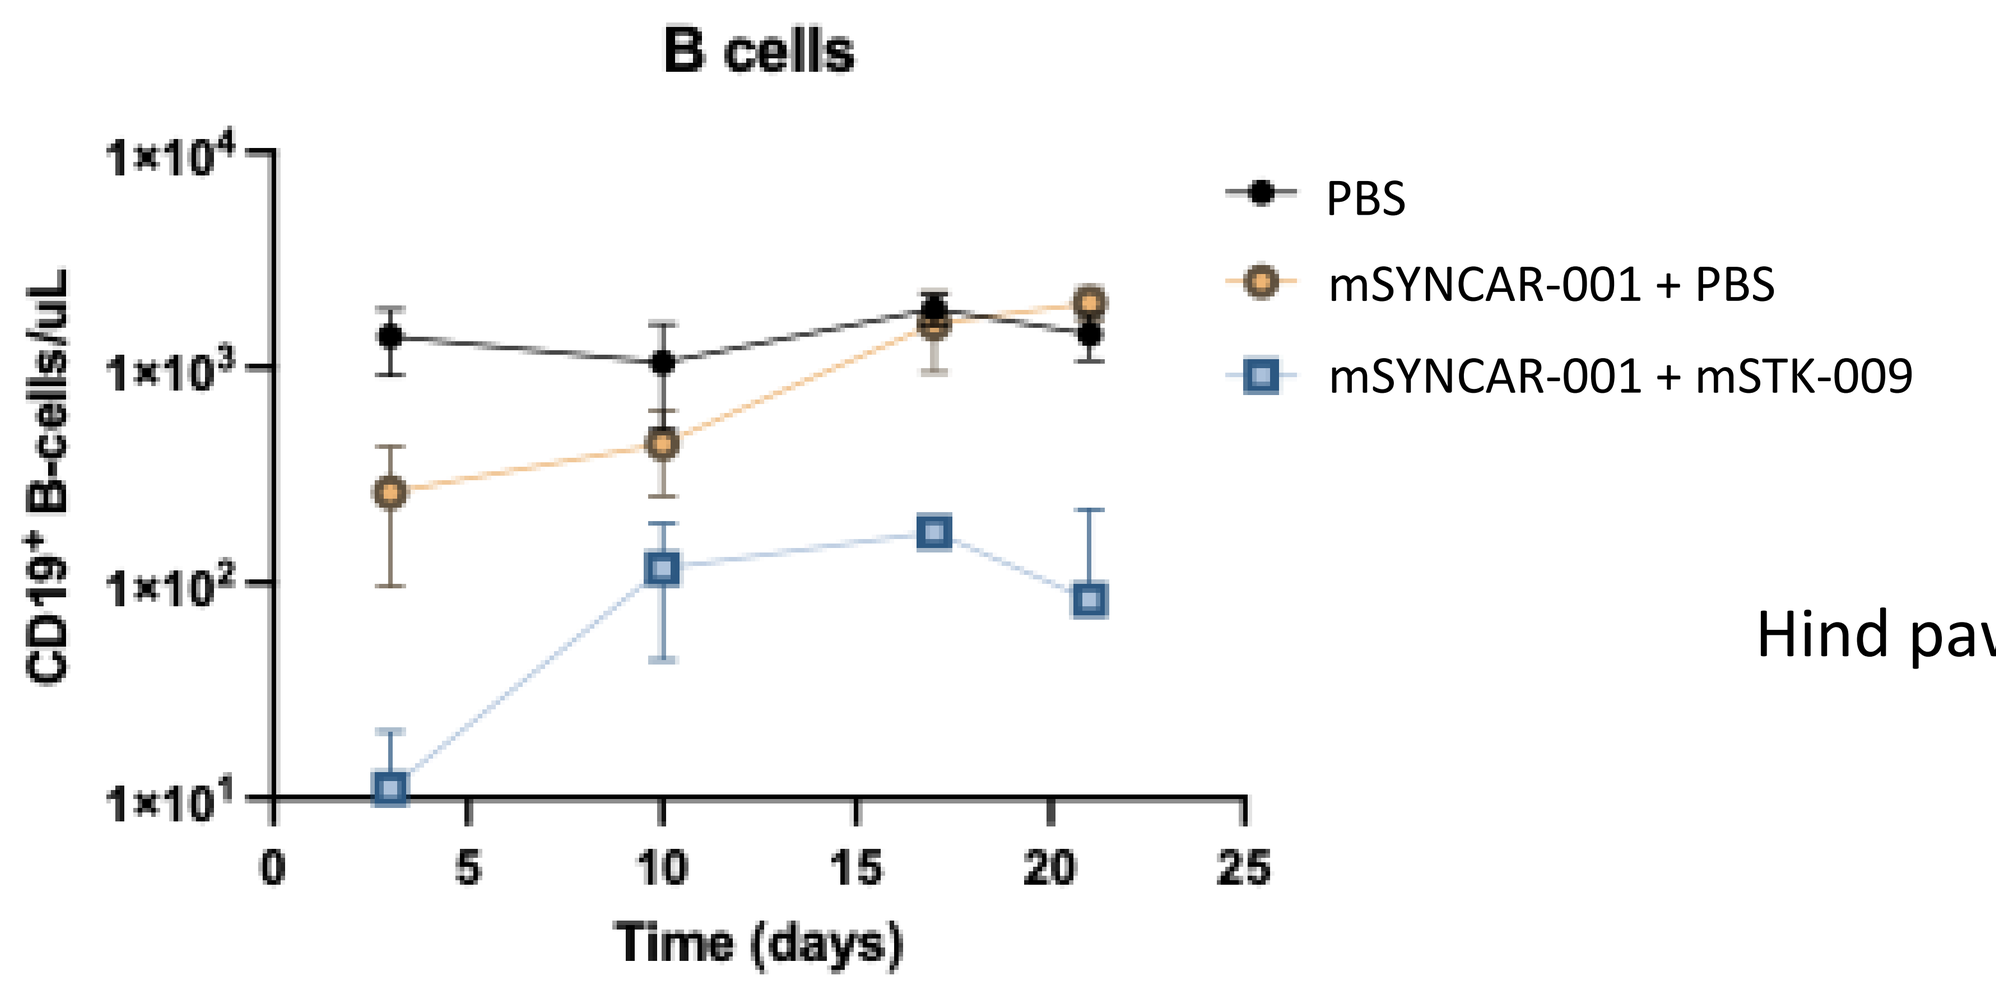
B cells
CD19⁺ B-cells/uL
Time (days)
PBS
mSYNCAR-001 + PBS
mSYNCAR-001 + mSTK-009

Paw swelling is reduced withmSYNCAR-001 + mSTK-009

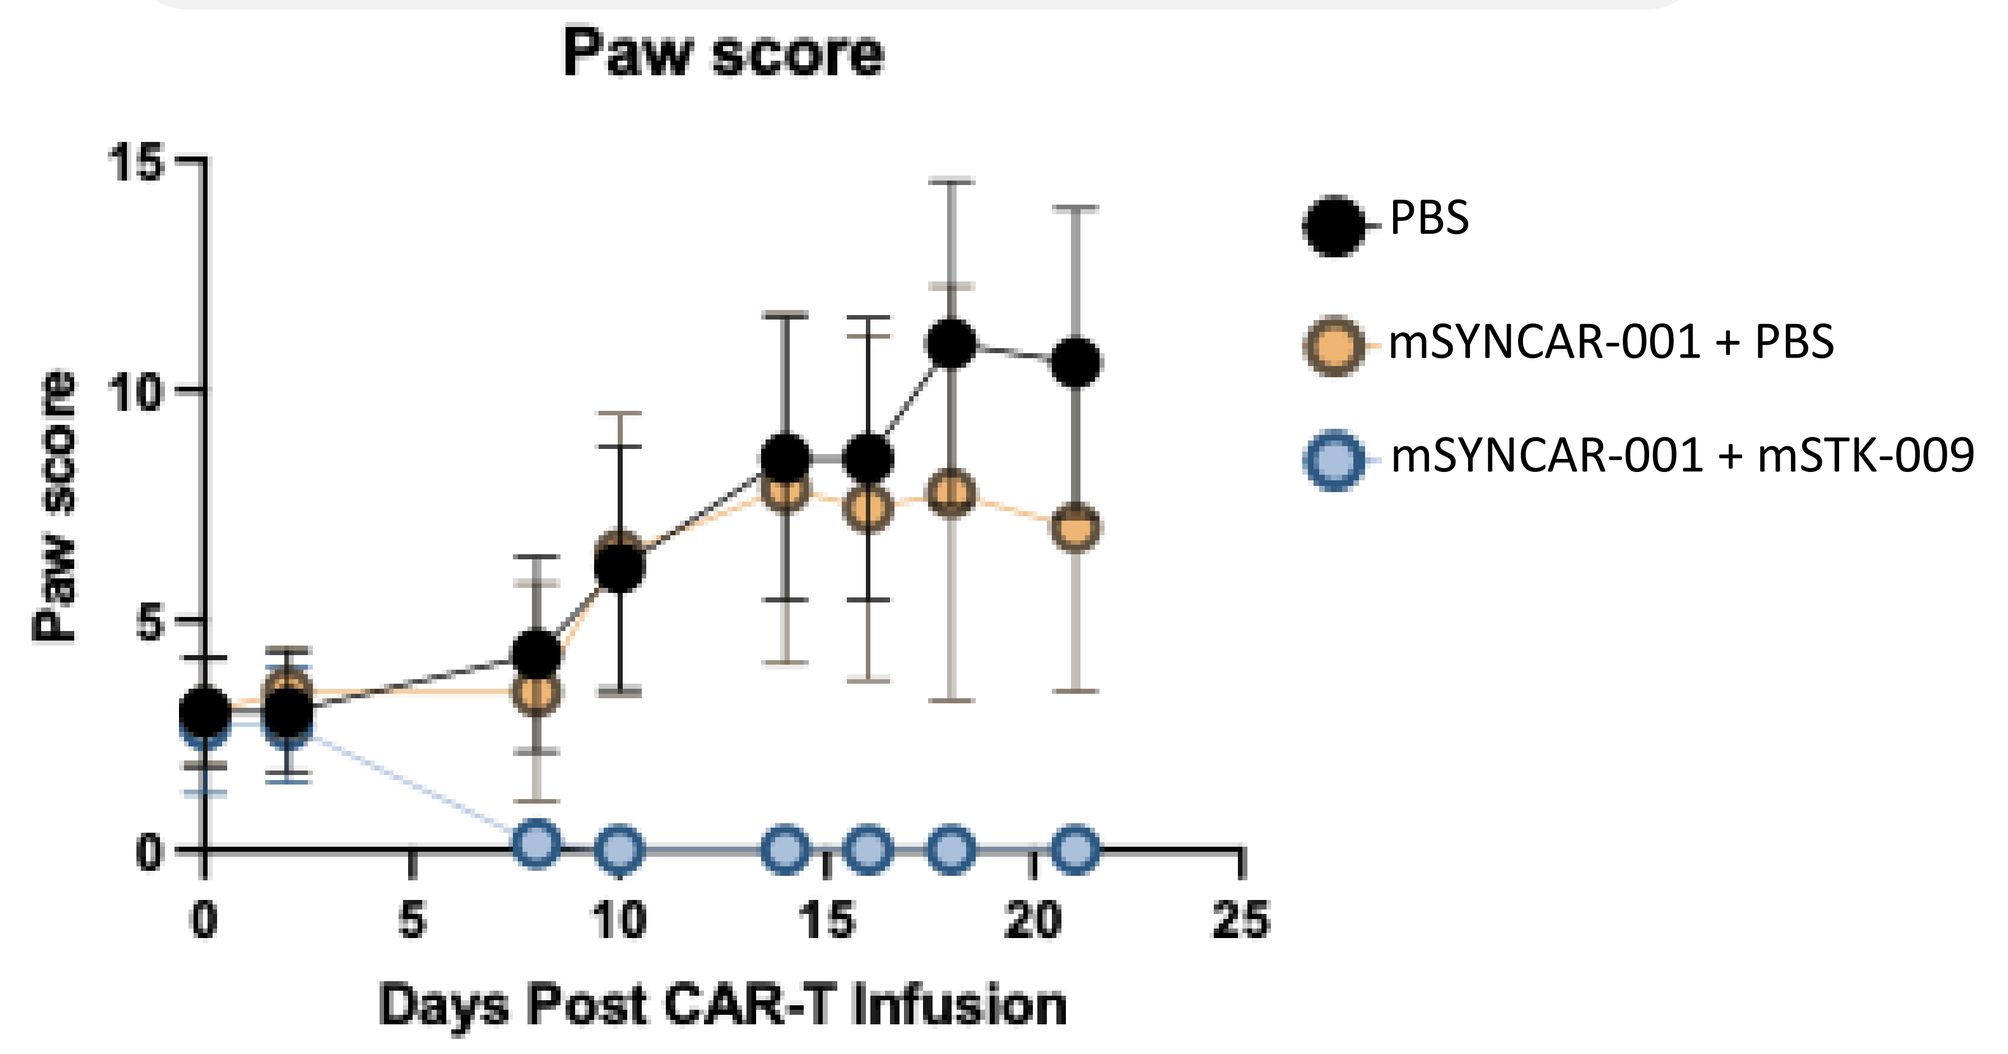
Paw score
Paw score
Days Post CAR-T Infusion
PBS
mSYNCAR-001 + PBS
mSYNCAR-001 + mSTK-009

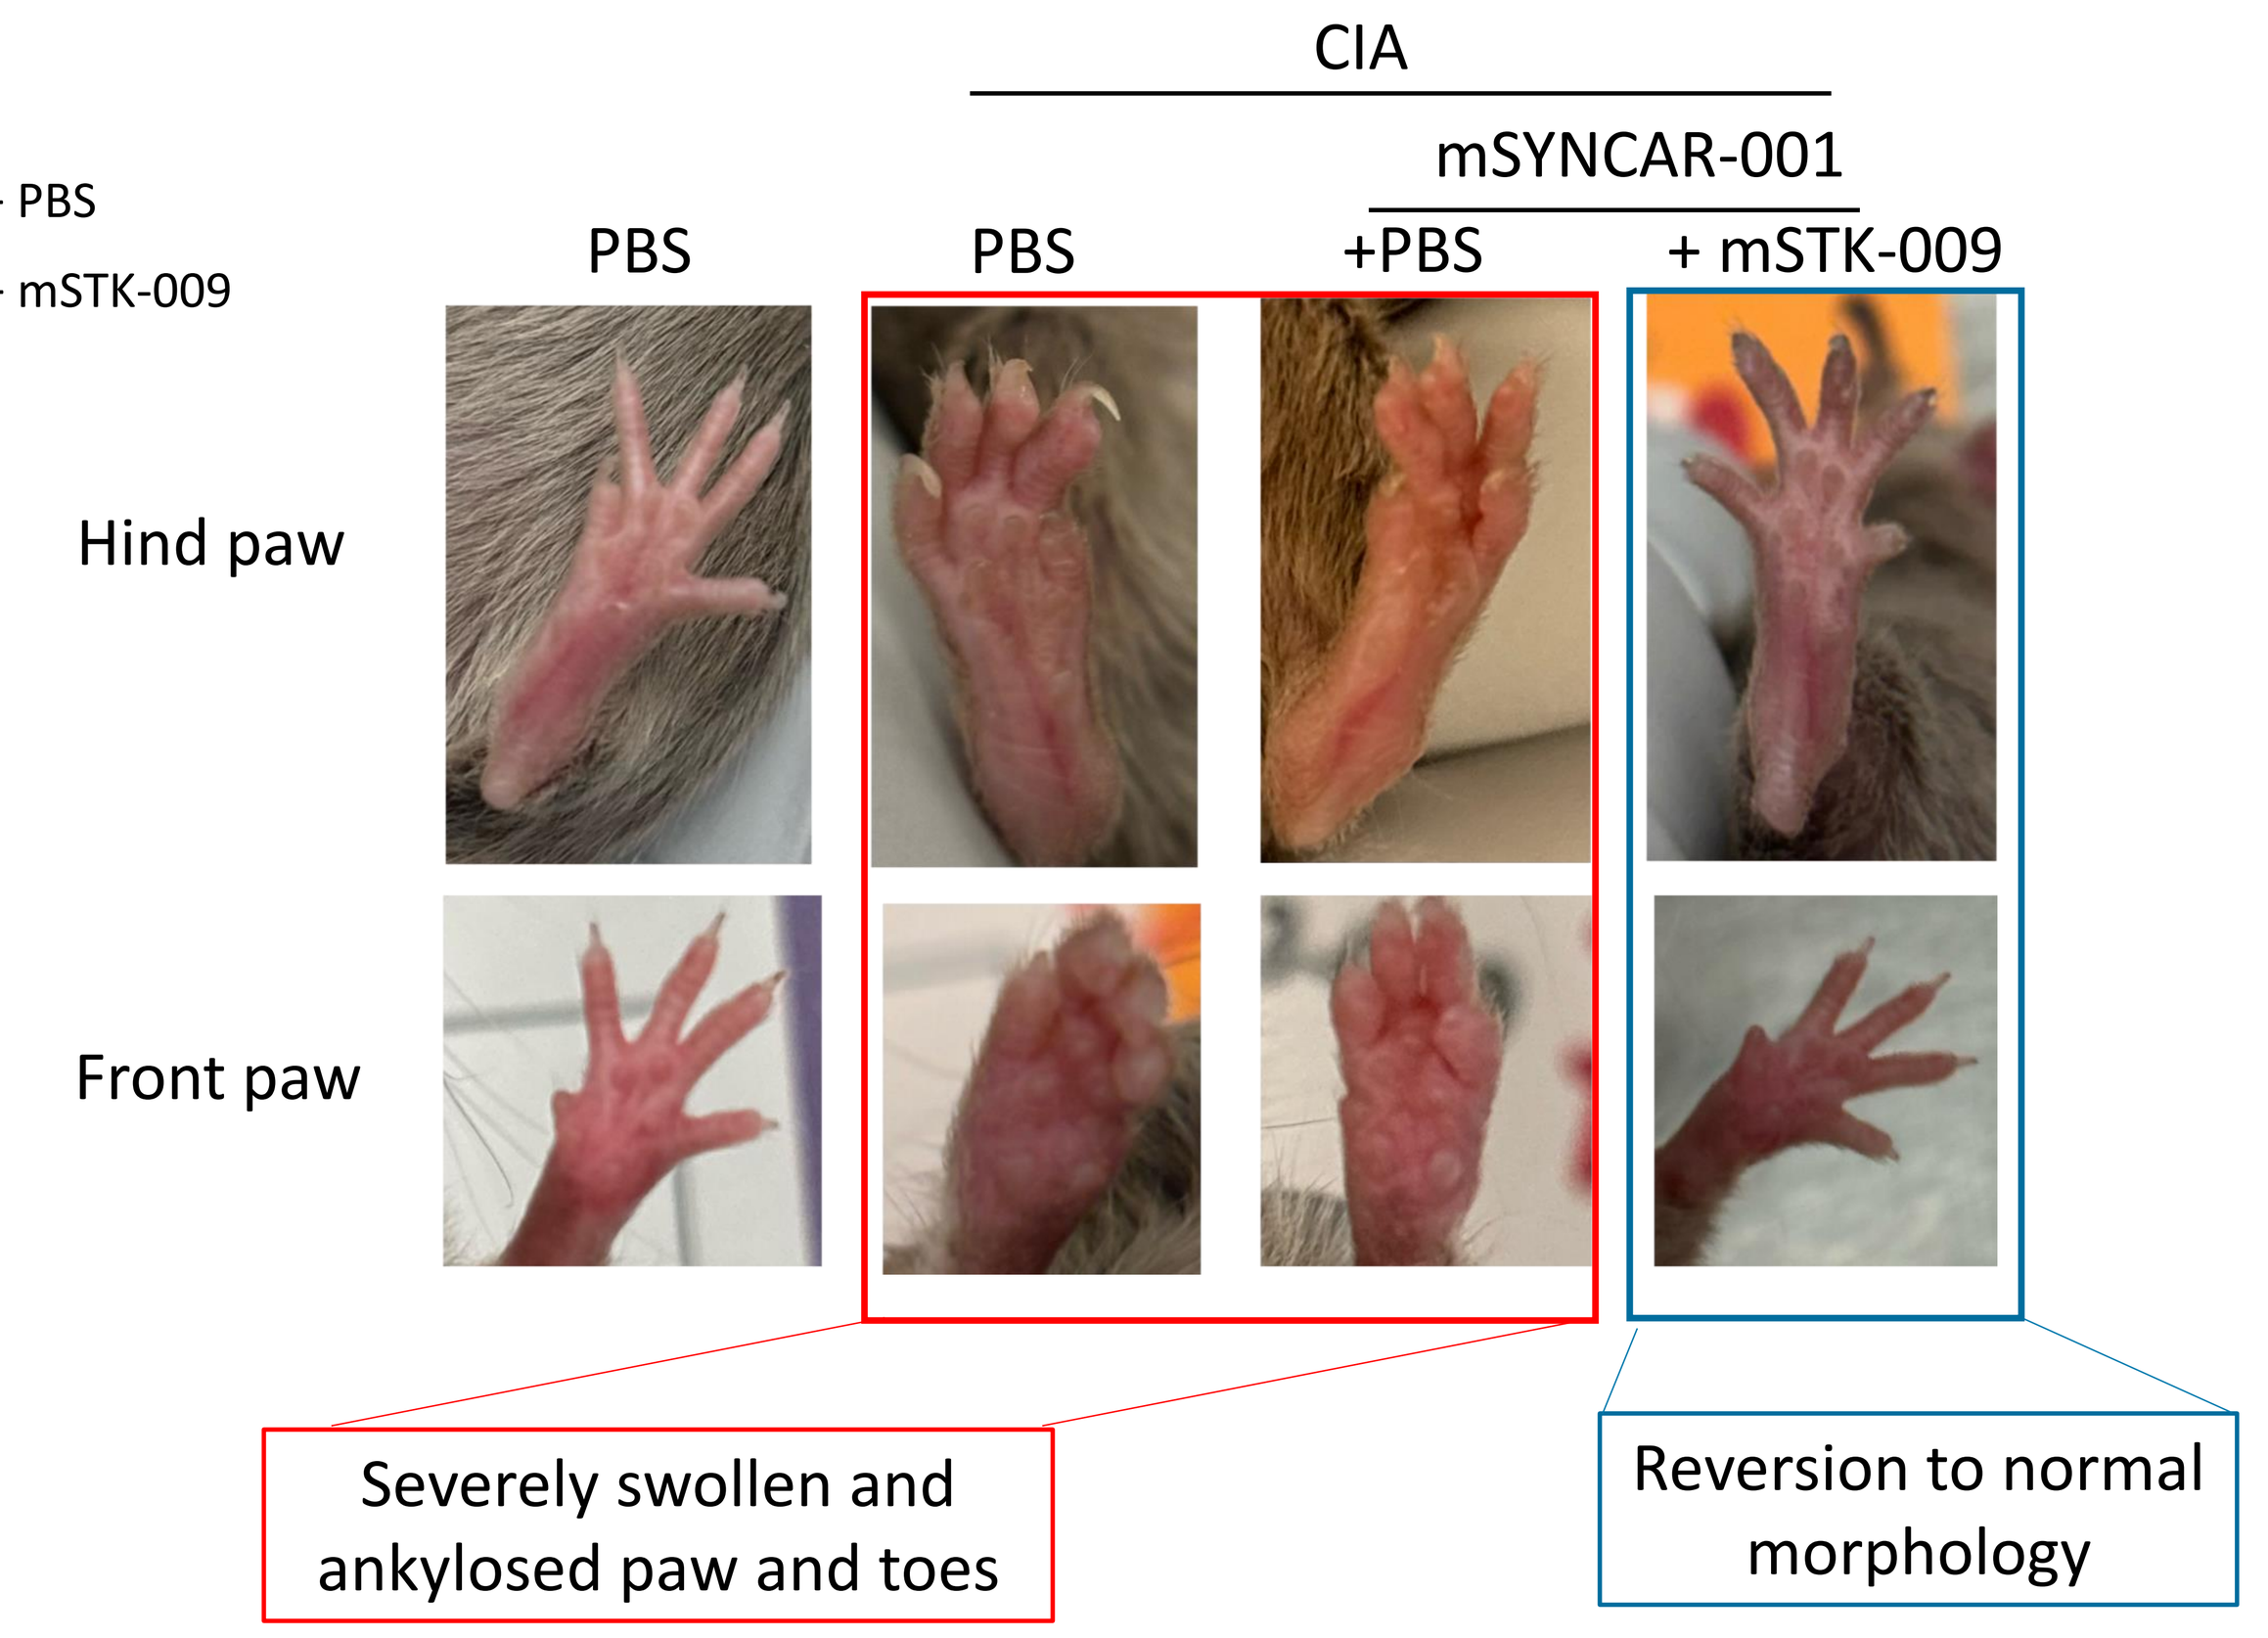
CIA
mSYNCAR-001
PBS
PBS
+PBS
+ mSTK-009
Hind paw
Front paw
Severely swollen and ankylosed paw and toes
Reversion to normal morphology

Complete B cell depletion in the joints and synovia restoration with mSYNCAR-001 + mSTK-009

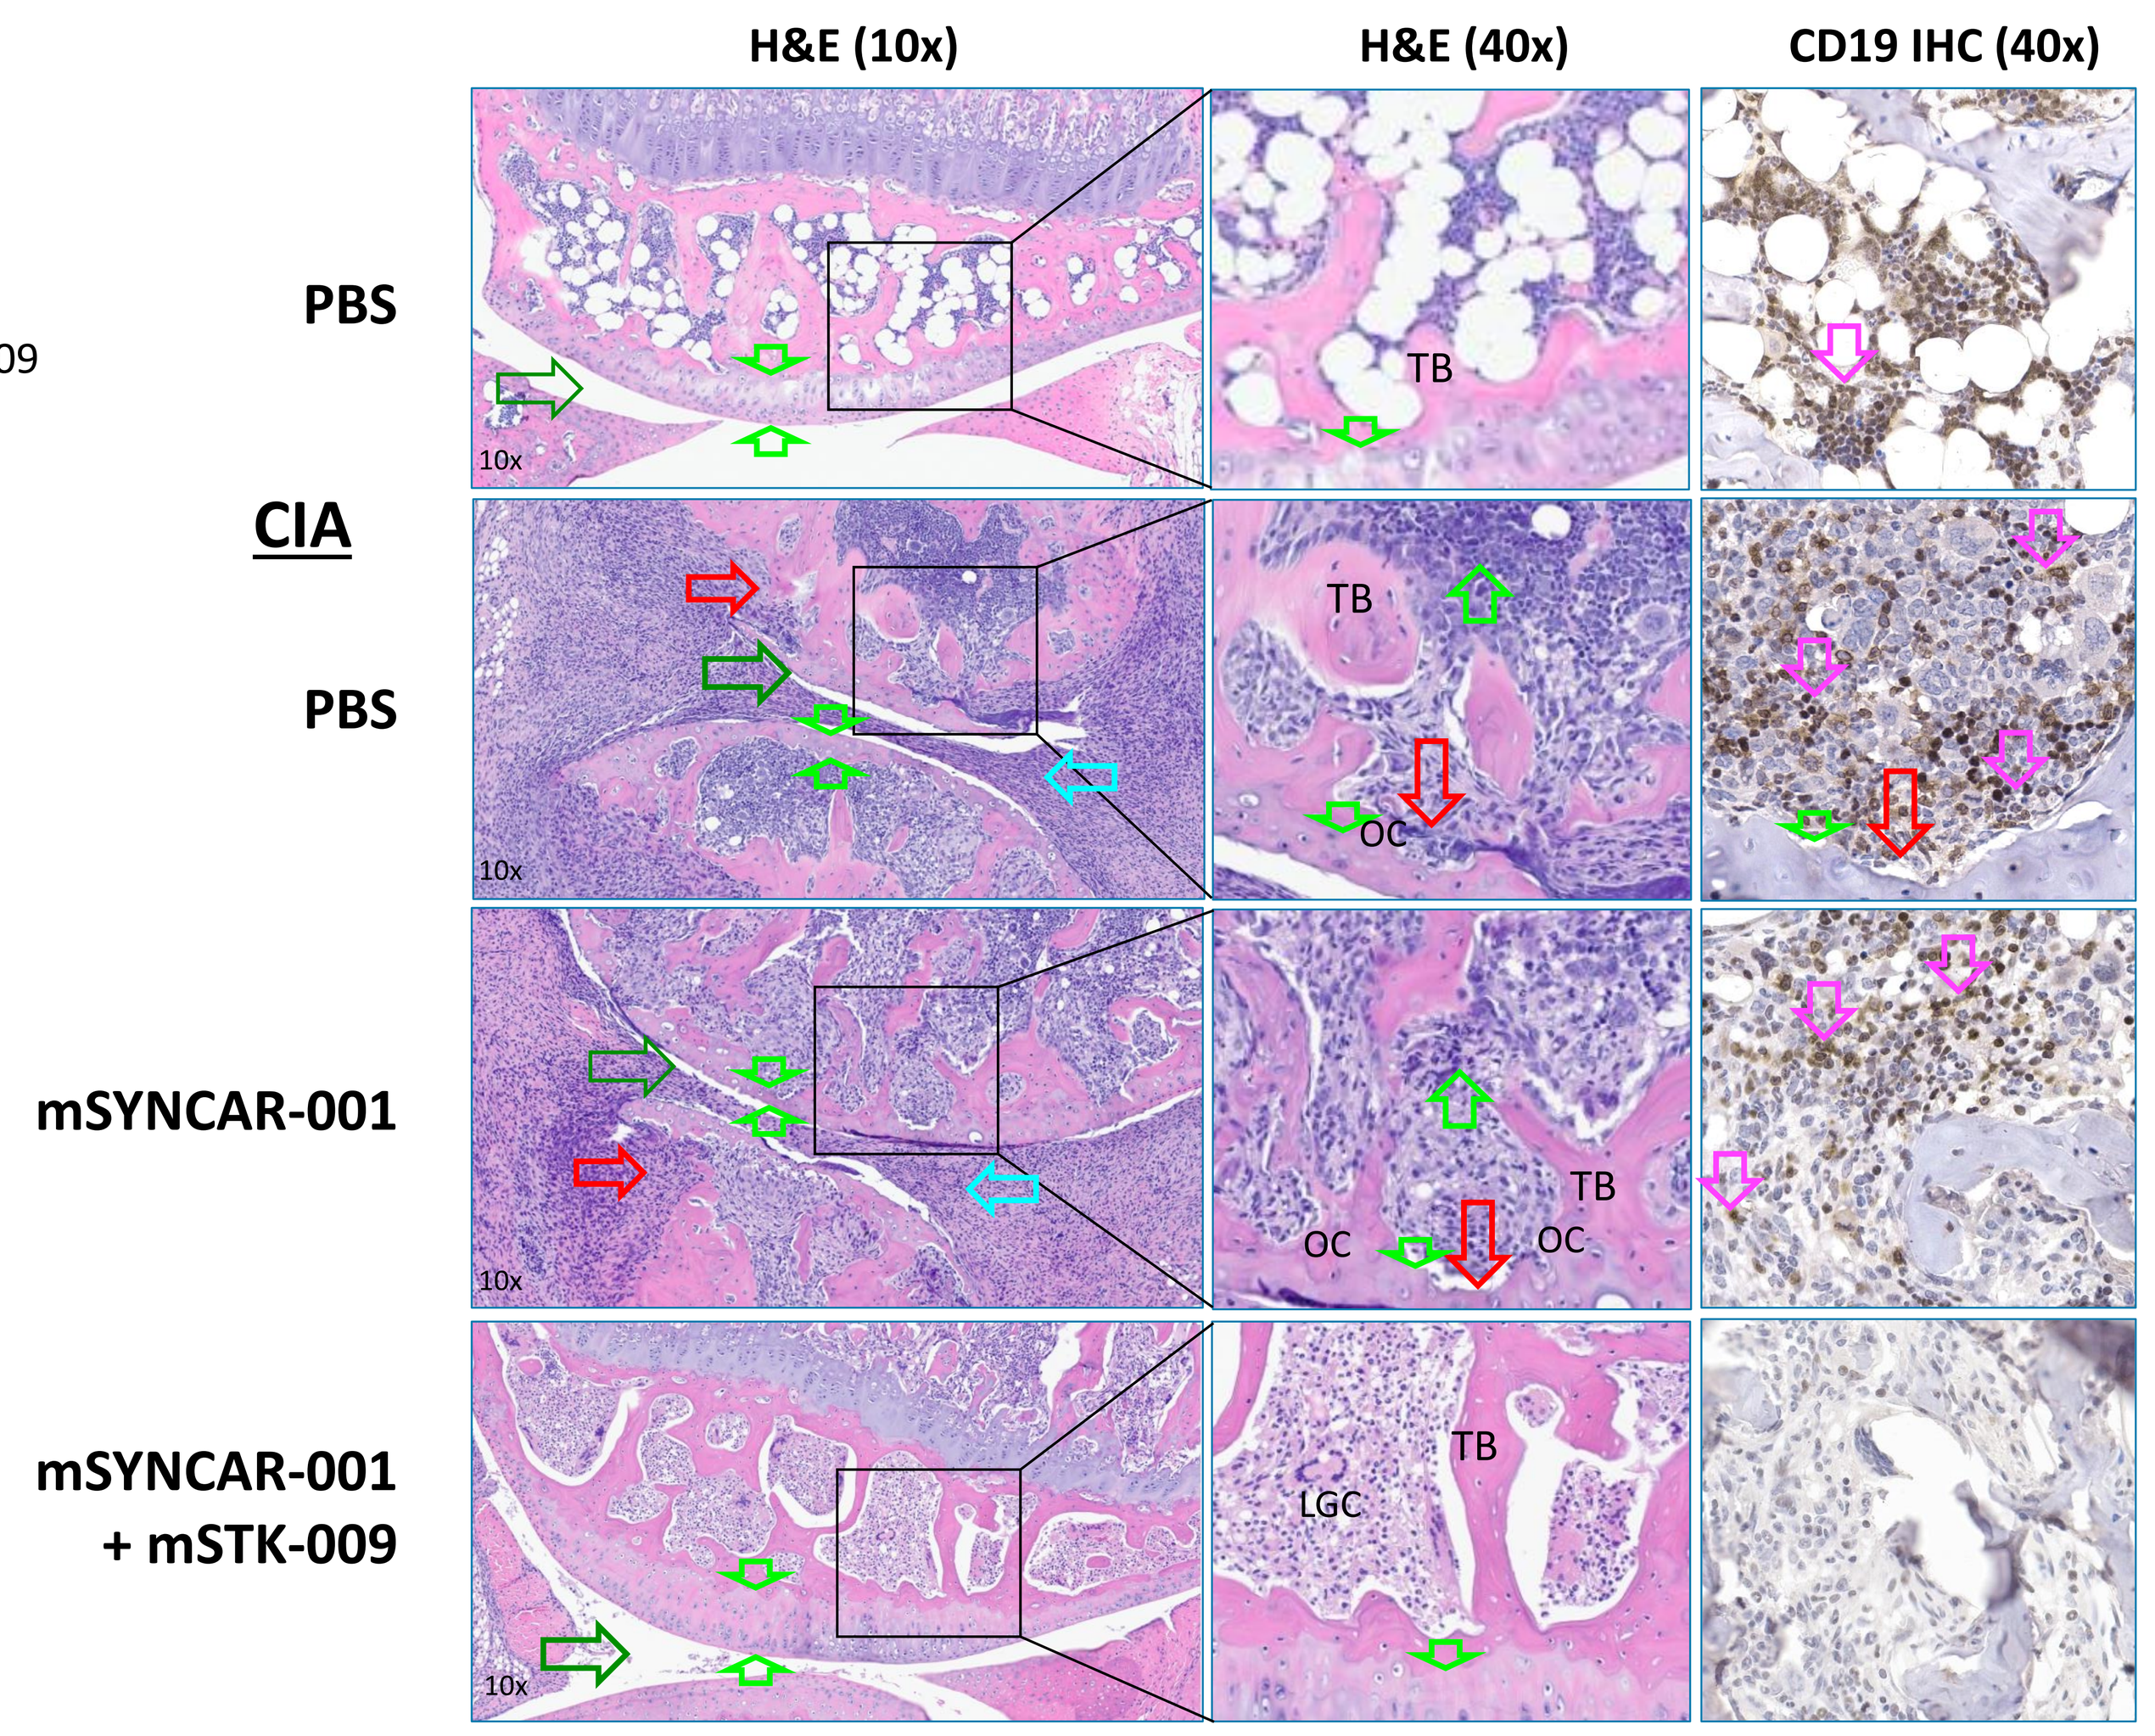
H&E (10x)
H&E (40x)
CD19 IHC (40x)
PBS
CIA
PBS
mSYNCAR-001
mSYNCAR-001 + mSTK-009
TB
OC
LGC
10x

Synovial Cavity	Articular cartilage	B cells	TB Trabecular bone
Invasive pannus	Subchondral bone erosion	Lymphocytes	
Synovitis	LGC Langerhans Giant Cells	OC Osteoclasts	

Histology and immunohistochemistry for CD19 (**brown**) of knee joints at the end of the study.

Collagen induced arthritis led to the accumulation of CD19⁺ B cells **(pink arrows)** and inflammatory lymphocytes, macrophages and osteoclasts in the subchondral bone, leading to the pathologic hallmarks of RA, subchondral bone erosion and the formation of an invasive pannus. mSYNCAR-001 cells alone did not eliminate B cells in the subchondral bone or decrease the inflammatory pannus and bone erosion.
In contrast, mSYNCAR-001 + STK-009 led to the elimination of B cells from the subchondral bone and normalized joint histology.

STK-009 Reinvigorates SYNCAR-001 Cells After an STK-009 Dose Holiday

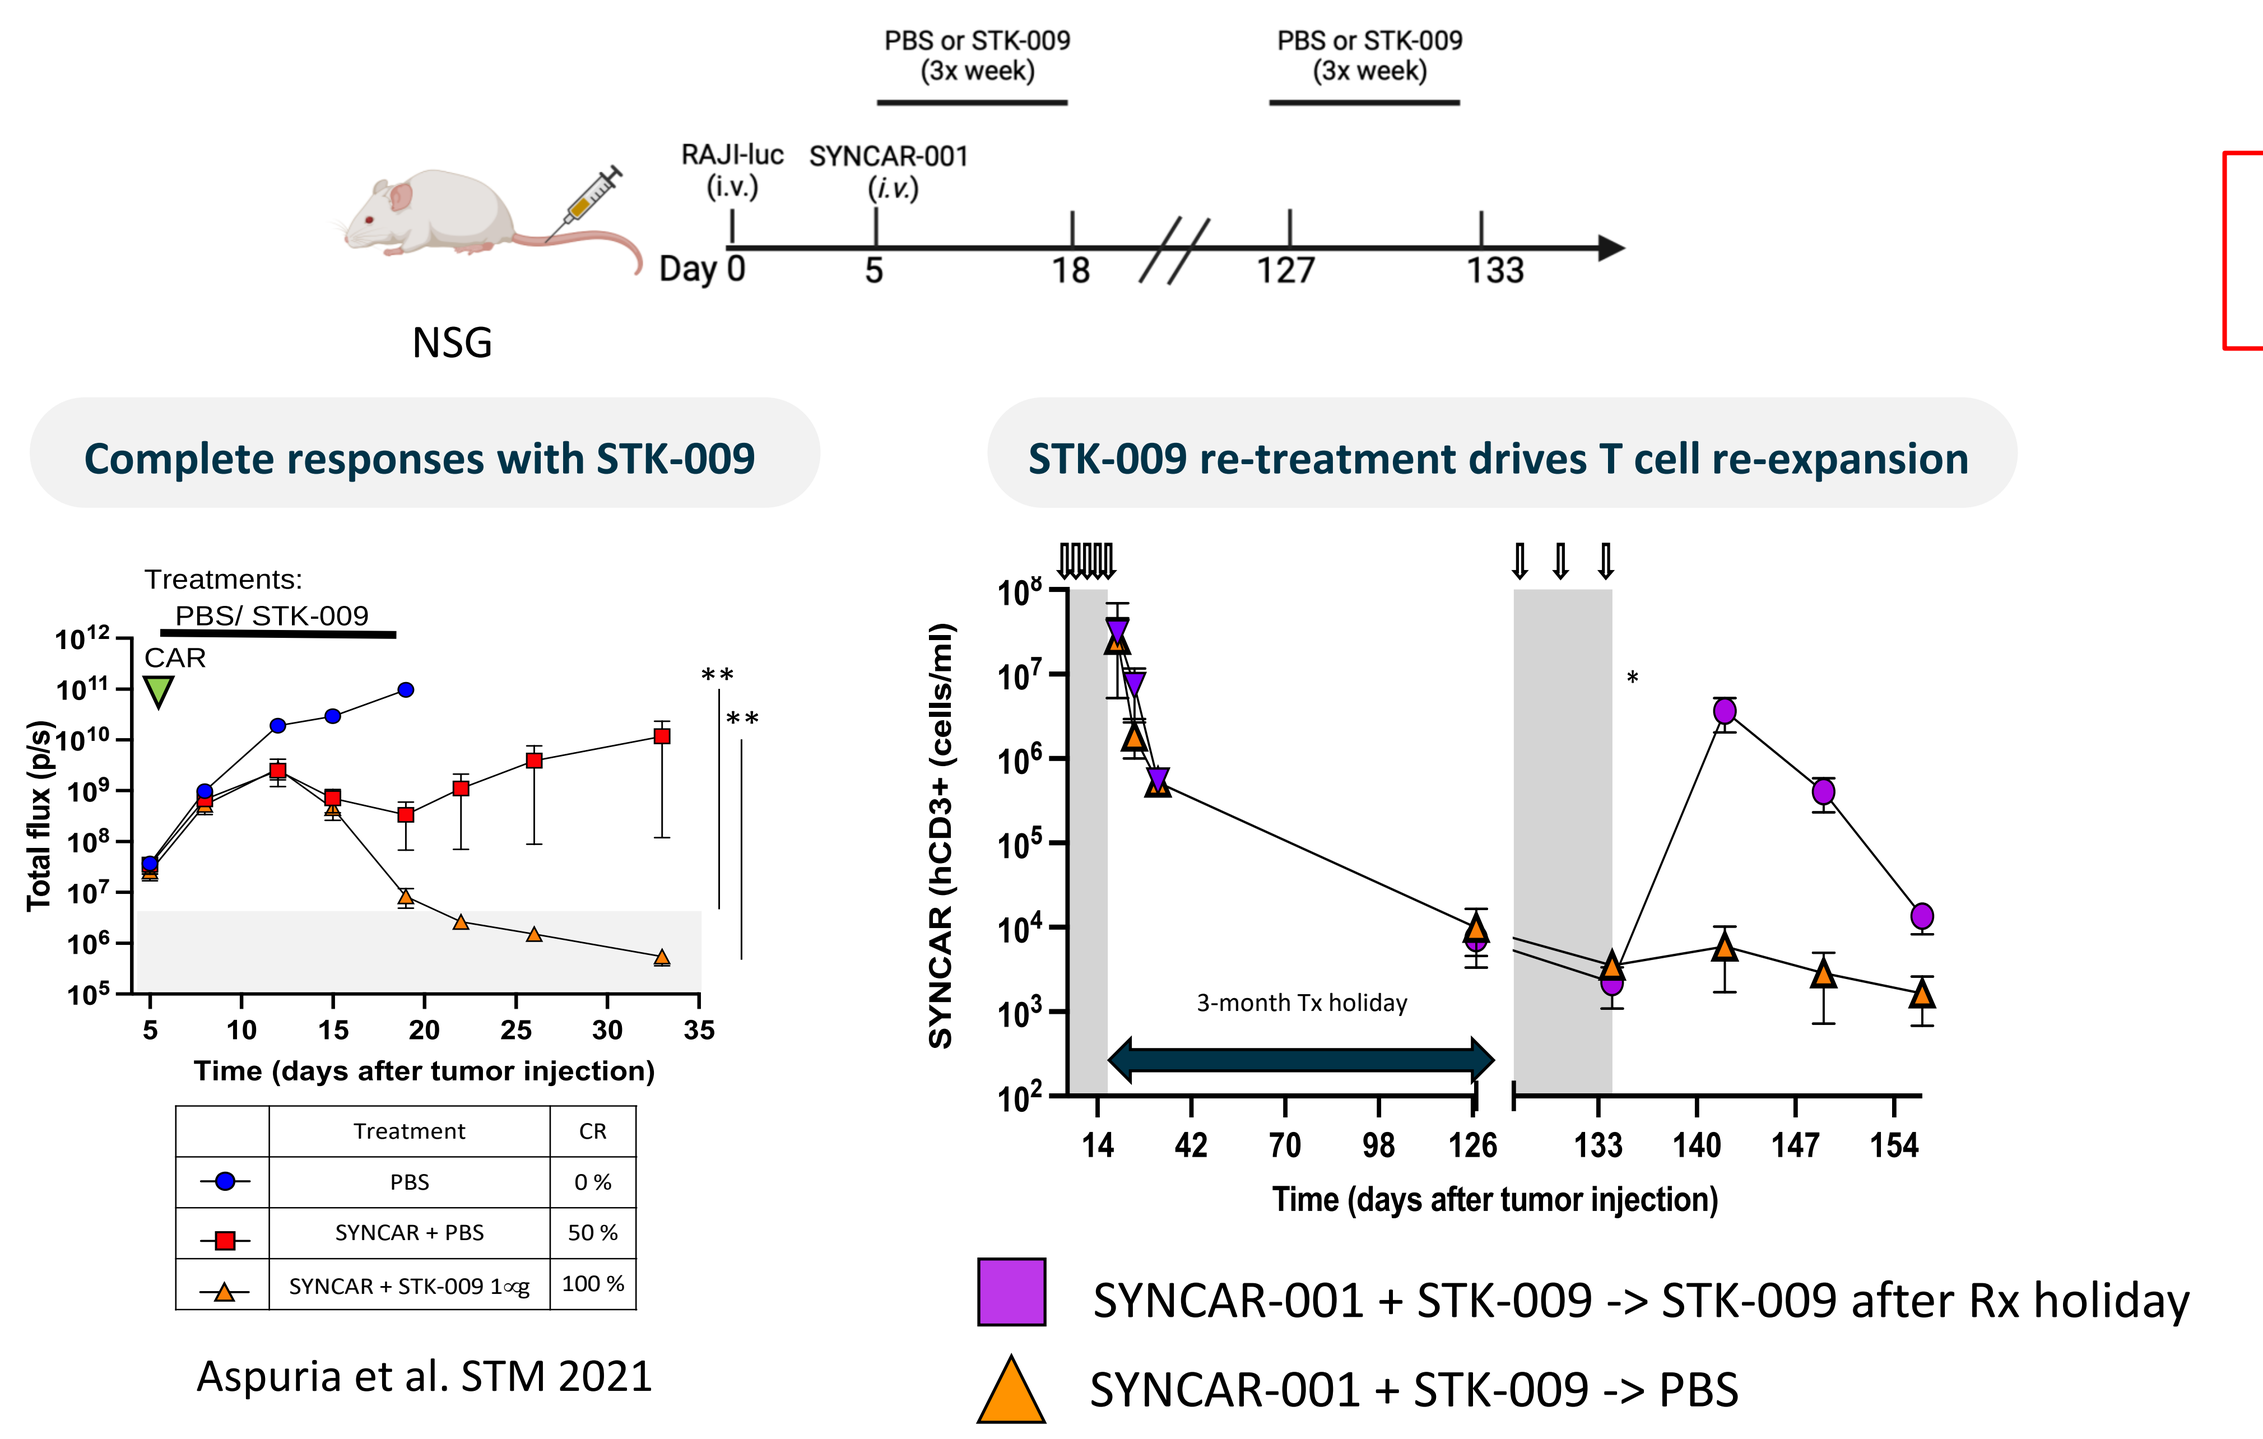
PBS or STK-009 (3x week)
PBS or STK-009 (3x week)
RAJI-luc (i.v.)
SYNCAR-001 (i.v.)
Day 0 5 18 127 133
NSG
Complete responses with STK-009
Treatments:
PBS/ STK-009
CAR
Total flux (p/s)
Time (days after tumor injection)
STK-009 re-treatment drives T cell re-expansion
SYNCAR (hCD3+ (cells/ml)
3-month Tx holiday
Time (days after tumor injection)

	Treatment	CR
●	PBS	0 %
■	SYNCAR + PBS	50 %
▲	SYNCAR + STK-009 1 μg	100 %

Aspuria et al. STM 2021

SYNCAR-001 + STK-009 -> STK-009 after Rx holiday
SYNCAR-001 + STK-009 -> PBS

STK-009 Induced CAR-T Expansion Without Lymphodepletion

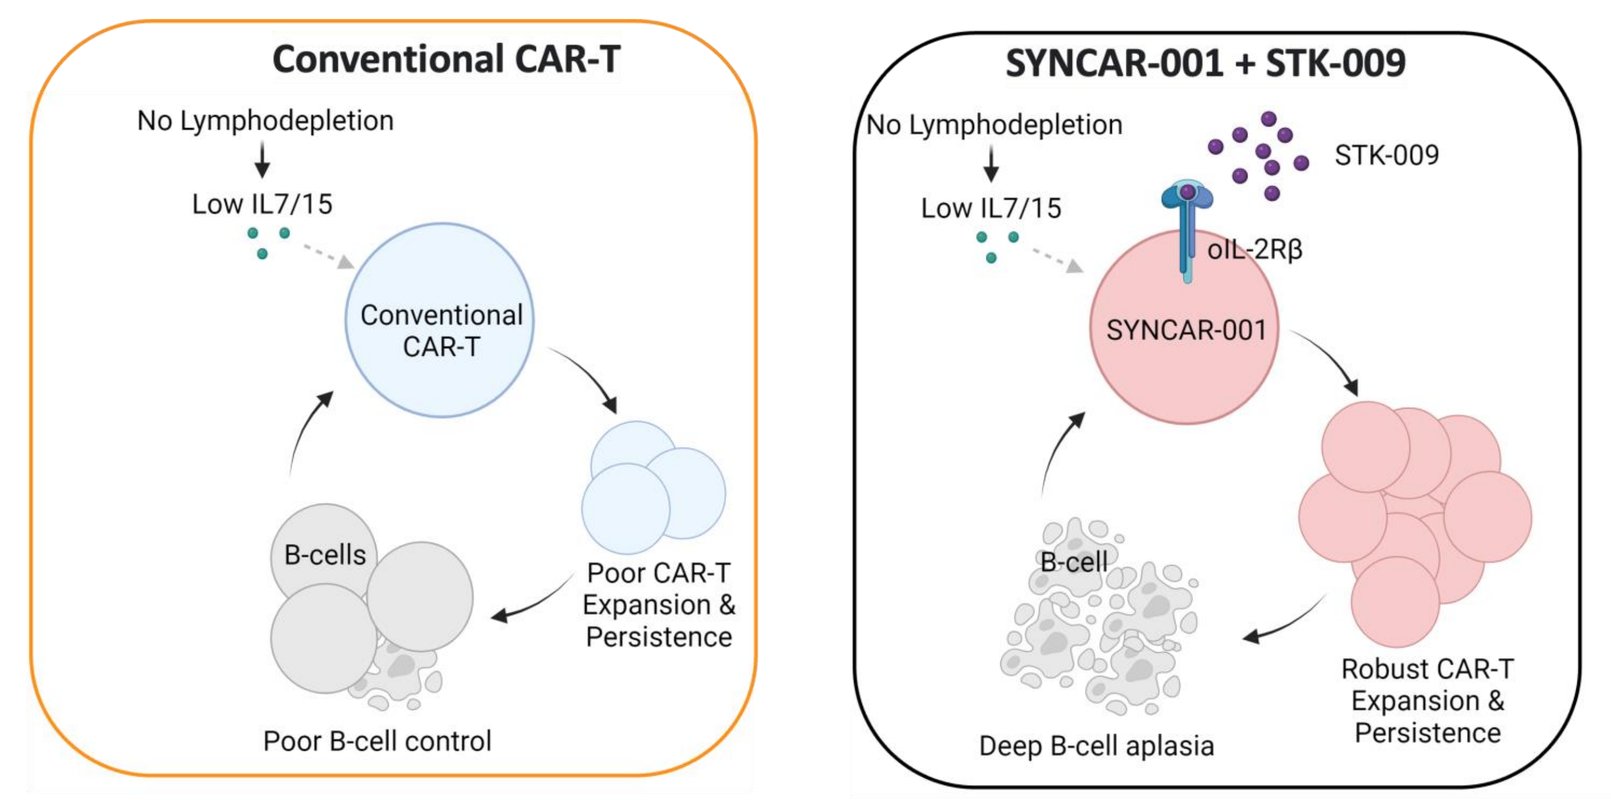
Conventional CAR-T
No Lymphodepletion
Low IL7/15
Conventional CAR-T
Poor CAR-T Expansion & Persistence
B-cells
Poor B-cell control
SYNCAR-001 + STK-009
No Lymphodepletion
STK-009
Low IL7/15
oIL-2Rβ
SYNCAR-001
Robust CAR-T Expansion & Persistence
B-cell
Deep B-cell aplasia

Conclusions

These findings encourage the clinical exploration of SYNCAR-001 + STK-009 in autoimmune diseases including SLE and RA to achieve CD19 CAR T expansion and significant B cell aplasia and depletion of B cells in disease tissue without **genotoxic, lymphodepleting conditioning**. SYNCAR-001 + STK-009 is in Phase I clinical trials in heme malignancies (NCT05665062) and LN and SLE (NCT06544330).