STK-012, a First-in-Class α/β IL-2 Receptor Biased Partial Agonist for the Treatment of Advanced Solid Tumors

PJ Aspuria*1, , Benjamin Izar2, Dmitriy Zamarin3, David R. Spigel4, Christopher J. Hoimes5, David F. McDermott6, Kartik Sehgal7, Yana G. Najjar8, Adam J. Schoenfeld9, Edward B. Garon10, Ryan J. Sullivan11, Brian S. Henick2, Ticiana A. Leal12, Michael E. Hurwitz13, Rana R. McKay14, Alexander I. Spira15, Jan Emmerich1, Ievgen Koliesnik1, Sandro Vivona1, Michele Bauer1, Alize Marangoz Stager1, Xue Gong1, Sushama Rokade1, Patrick Lupardus1, Alex Azrilevich1, Naiyer A. Rizvi1, Anita Mehta Damani1, Martin Oft1

1 Synthekine, 2 Irving Medical Center, Columbia University, 3 Mount Sinai, MSSN, 4 Sara Cannon Research Institute, Sara Cannon Research Institute, 5 Duke Cancer Institute, Duke University, 6 Hem Onc, Beth Israel Deaconess Medical Center, 7 Dana Farber Cancer Institute, Harvard University, 8 Hillman Cancer Center, UPMC, 9 MSKCC, Memorial Sloan Kettering Cancer Center, 10 Thoracic Oncology, University of California, Los Angeles, 11 Massachusetts General Hospital, Havard University, 12 Emory University, 13 Yale Cancer Center, Yale University, 14 Med Oncology, University of California at San Diego, 15 NEXT Oncology Virginia, Fairfax, VA ; United States

Synthekine
1505 O'Brien Dr, Menlo Park, CA, 94025, USA

Overview

STK-012 is a first-in-class IL-2Rα/β-biased partial agonist designed to drive antitumor activity by selectively stimulating CD25+ antigen-activated T-cells, avoiding toxicities by sparing pleotropic activation of lymphocytes including NK-cells.

Preclinically, mouse STK-012 significantly reduced exhaustion of tumor infiltrating T-cells, improved expansion of tumor antigen specific CD25+PD-1+CD8+ T-cells systemically and intratumorally and reduced intratumoral Tregs. mSTK-012 had improved anti-tumor responses over IL-2. WT-IL-2 and non-α-IL-2 but not STK-012 induced widespread extravasation of lymphocytes and NK-cells resulting in lung and systemic tissue inflammation in non-human primates and mice. This WT-IL-2 induced lethal capillary leak syndrome (CLS) was NK-cell-dependent in mice.

In Phase1a/b dose escalation in advanced, relapsed/refractory (r/r) solid tumors, 45 subjects were treated at 7 dose levels across 2 schedules (QW-Q3W, 0.375mg-3mg) with STK-012 monotherapy. 2.25mg Q3W was advanced into Phase1b. No DLTs were observed. The most common treatment related AEs (TRAEs) were maculo-papular rash (38%), injection site reactions (28.9%), fatigue (28.9%), and nausea (24.4%). Grade3 TRAEs occurred in 12 subjects (26.6%) with maculo-papular rash (4), vomiting (2). TRAEs were reversible, no Grade5 TRAEs occurred. No subjects had CLS and <5% experienced CLS related TRAEs such as hypotension, AST/ALT increase or edema. Of 12 evaluable subjects with 1 or 2 prior treatments, 3 had a partial response (PR), 8 had stable disease, including 2 confirmed PR (anti-PD-1r/r NSCLC and RCC) and 1 unconfirmed (anti-PD-1r/r SCCHN). After data-extract-date, a subject with anti-PD-1r/r RCC achieved a cPR with 80% reduction in target lesions.

STK-012 has a half-life of 4 days, induced STAT5 phosphorylation in CD25+_T-cells and dose dependent proliferation of activated CD38+CD8+_T-cells and increase of IFNγ, but very limited expansion of NK-cells and Tregs.

Pegylated α/β-biased IL-2: STK-012:

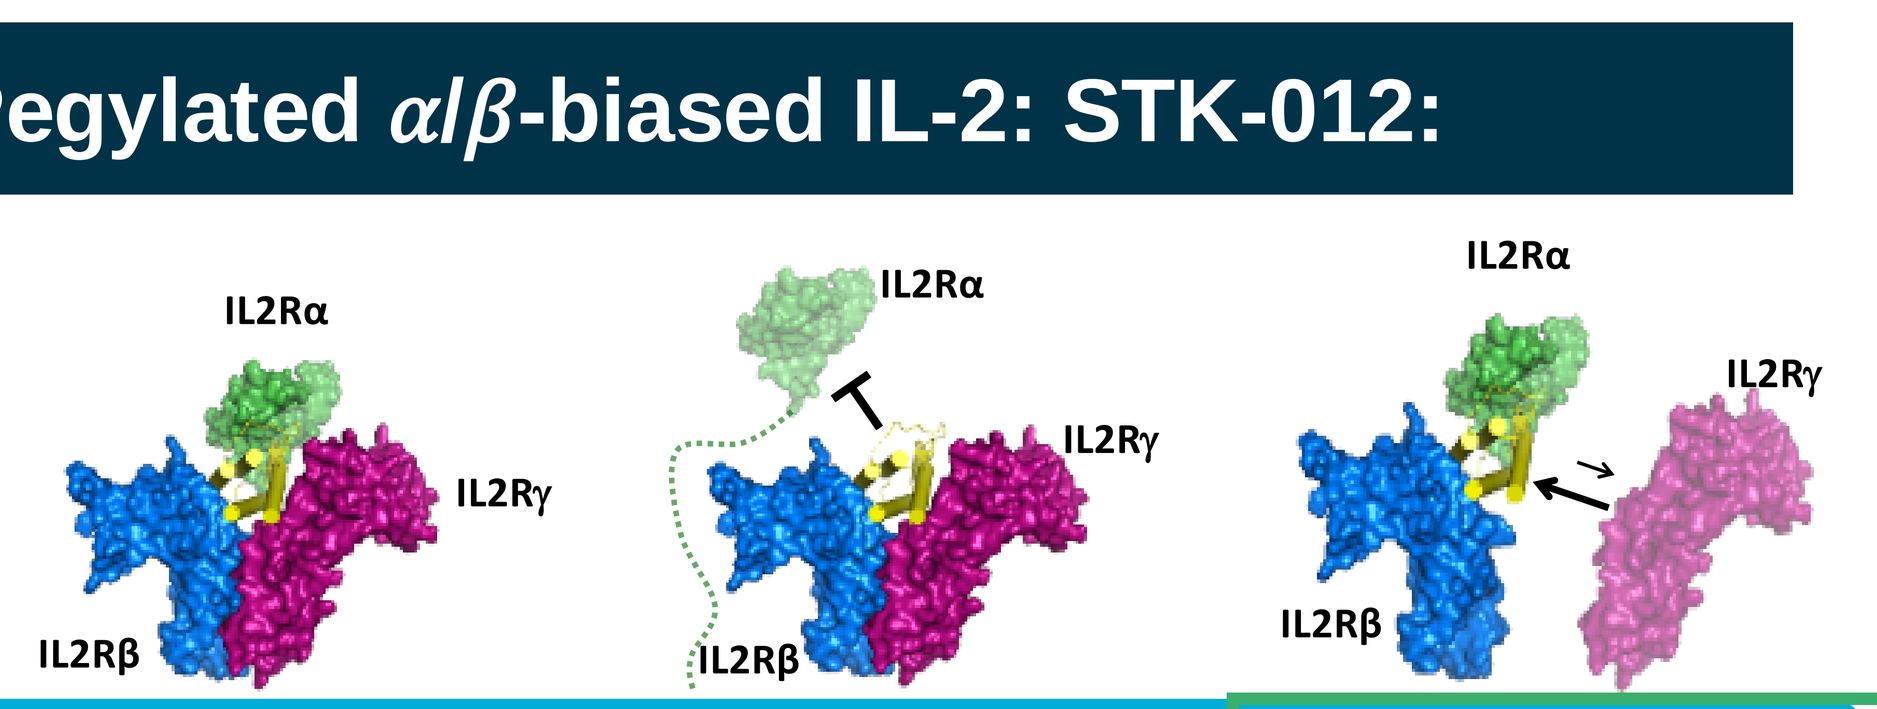
IL2Rα
IL2Rγ
IL2Rβ

	1st Generation	2nd Generation Engineered IL-2	STK-012
	High Dose IL-2 (Aldesleukin)	"Non-α" IL-2	α/β- IL-2
IL-2R Bias	Trimeric >> dimeric IL-2R	Dimeric IL-2 receptor	Trimeric IL-2 receptor
IL-2R subunit sparing	None	Reduced binding IL-2Rα	Reduced binding to IL-2Rγ
Cell Selectivity	Act T cells > Tregs > NK > resting lymphocytes	Act T cells = Tregs = NK = resting lymphocytes	Act T cells > Tregs >>> NK cells and resting lymphocytes
	Deep, durable responses in certain patients but limited use given toxicity	*Avoids Tregs for enhanced anti-tumor function Avoids eosinophils / endothelial cells?*	*Avoids NK cells (toxicity) Targets Activated T cells / Tregs*

Preclinical Surrogates	IL-2	non-α-IL-2-PEG	STK-012 (α/β-hIL-2)
	approved in melanoma and RCC (Proleukin®)	Proxy for Super 2, NKTR-214, THOR-707, NL-201, etc.	Selective for Rα+ / Rβ+ T cells Modulated Rγ binding
	mIL-2-PEG	NL-201-PEG	α/β-mIL-2-PEG

α/β-mIL-2 has Retained Efficacy Compared to mIL-2

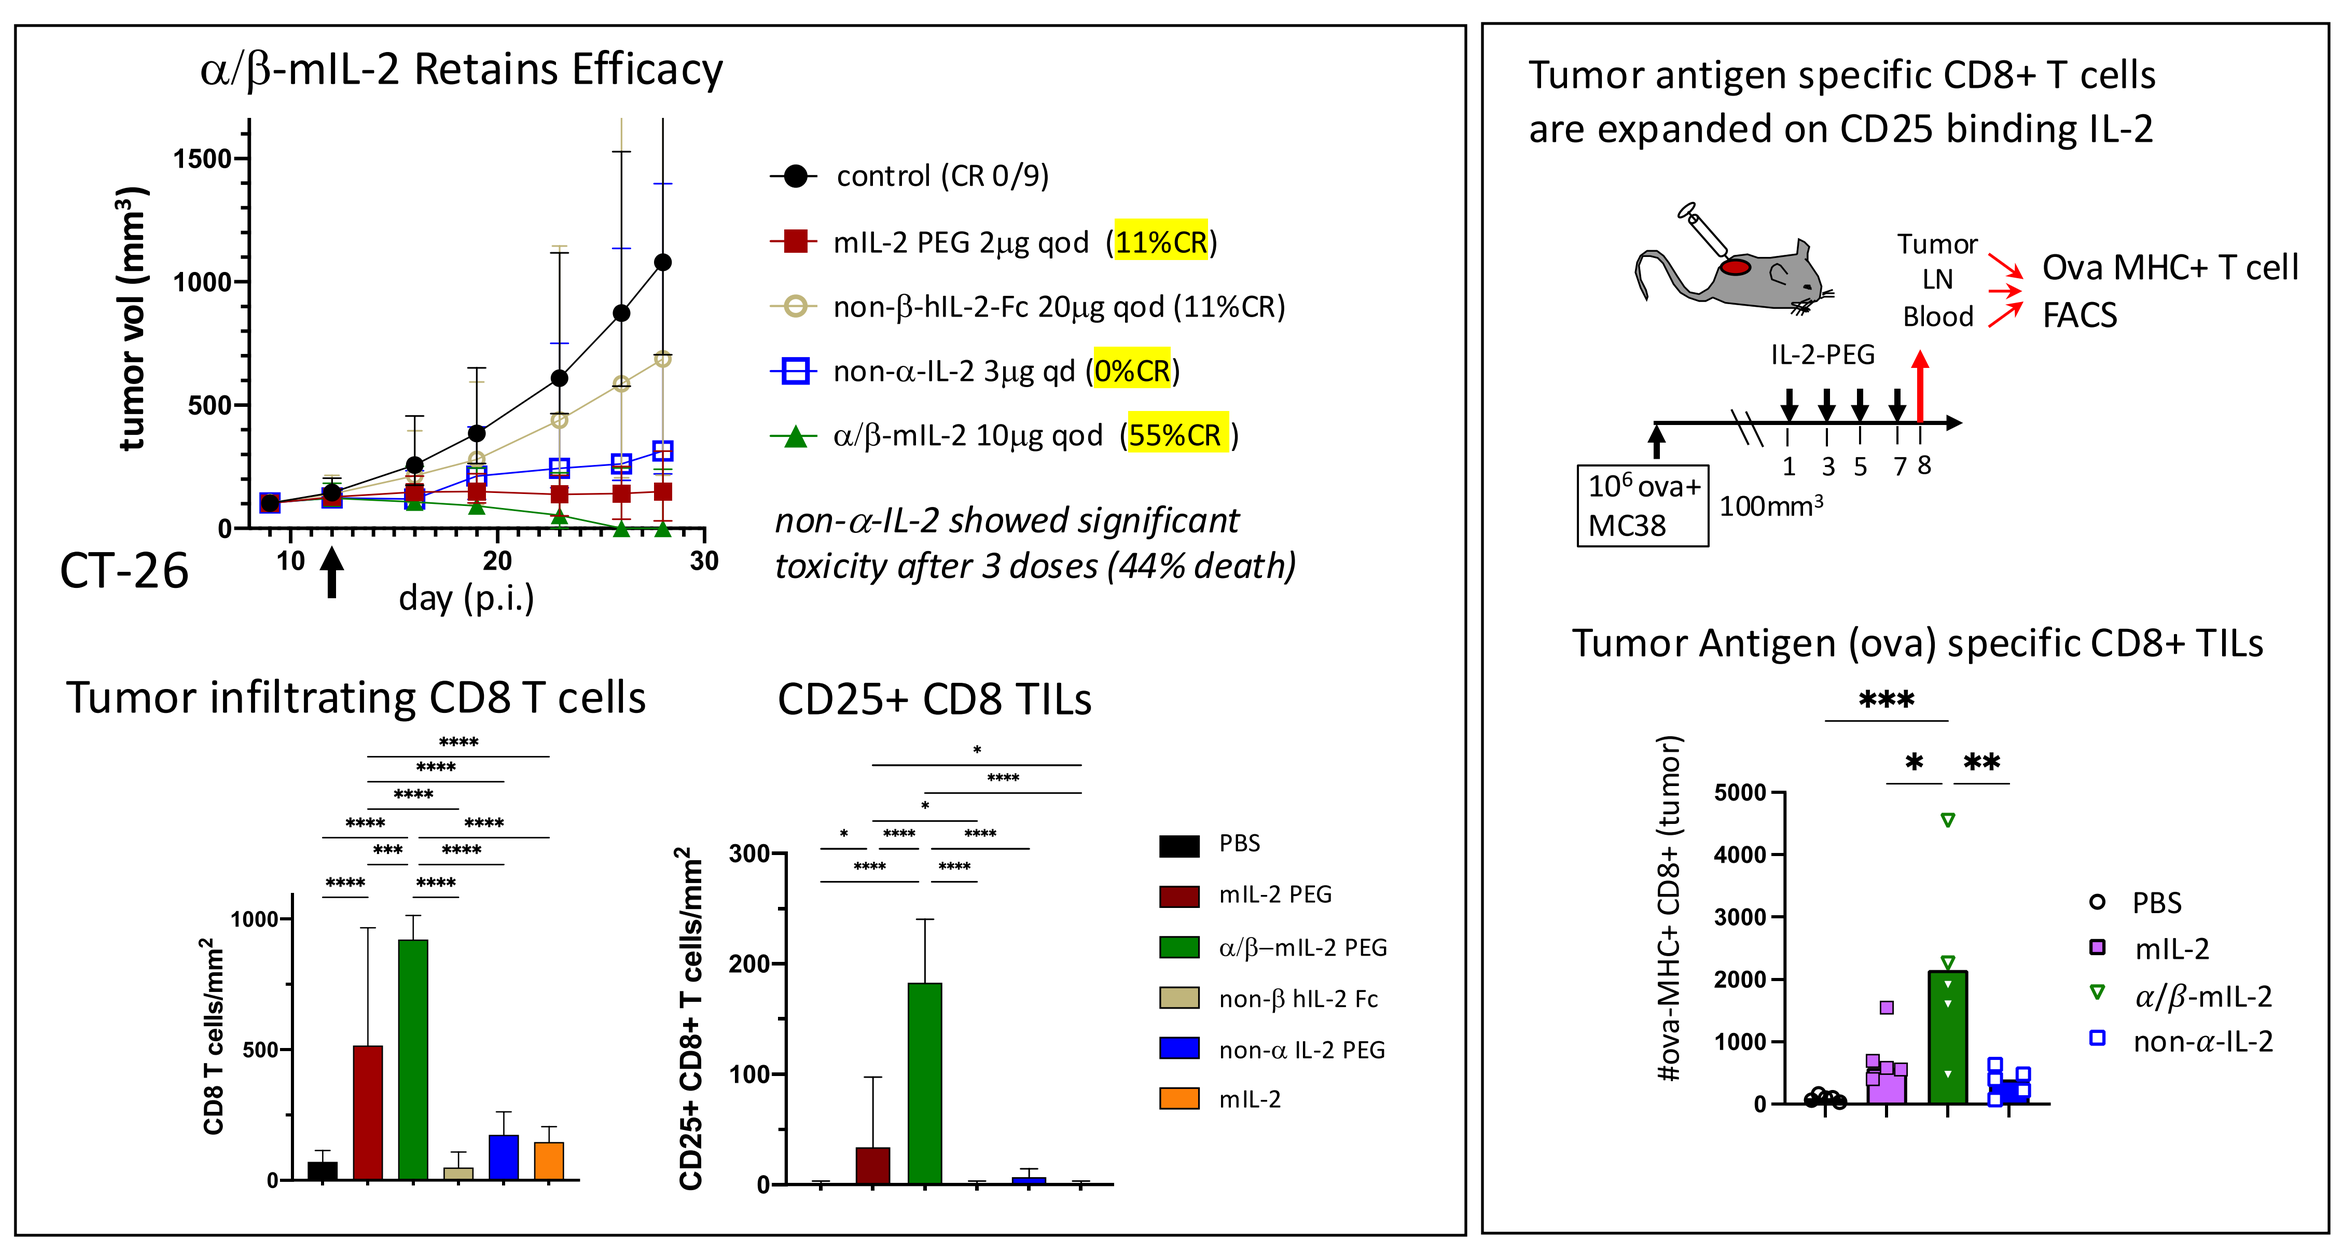
α/β-mIL-2 Retains Efficacy
tumor vol (mm³)
control (CR 0/9)
mIL-2 PEG 2μg qod (11%CR)
non-β-hIL-2-Fc 20μg qod (11%CR)
non-α-IL-2 3μg qd (0%CR)
α/β-mIL-2 10μg qod (55%CR)
non-α-IL-2 showed significant toxicity after 3 doses (44% death)
CT-26
day (p.i.)
Tumor infiltrating CD8 T cells
CD25+ CD8 TILs
PBS
mIL-2 PEG
α/β–mIL-2 PEG
non-β hIL-2 Fc
non-α IL-2 PEG
mIL-2
Tumor antigen specific CD8+ T cells are expanded on CD25 binding IL-2
Tumor LN Blood
Ova MHC+ T cell FACS
IL-2-PEG
10⁶ ova+ MC38
100mm³
Tumor Antigen (ova) specific CD8+ TILs
#ova-MHC+ CD8+ (tumor)
PBS
mIL-2
α/β-mIL-2
non-α-IL-2

WT IL-2 and non-α-IL-2 induce Capillary Leak Syndrome (CLS)

- IL-2 and non-α-IL-2 increase NK cells, T cells and myeloid cells in the lung
- IL-2 and non-α-IL-2 induce CLS
- In mice – NK cells depletion (anti-NK1.1) avoids CLS

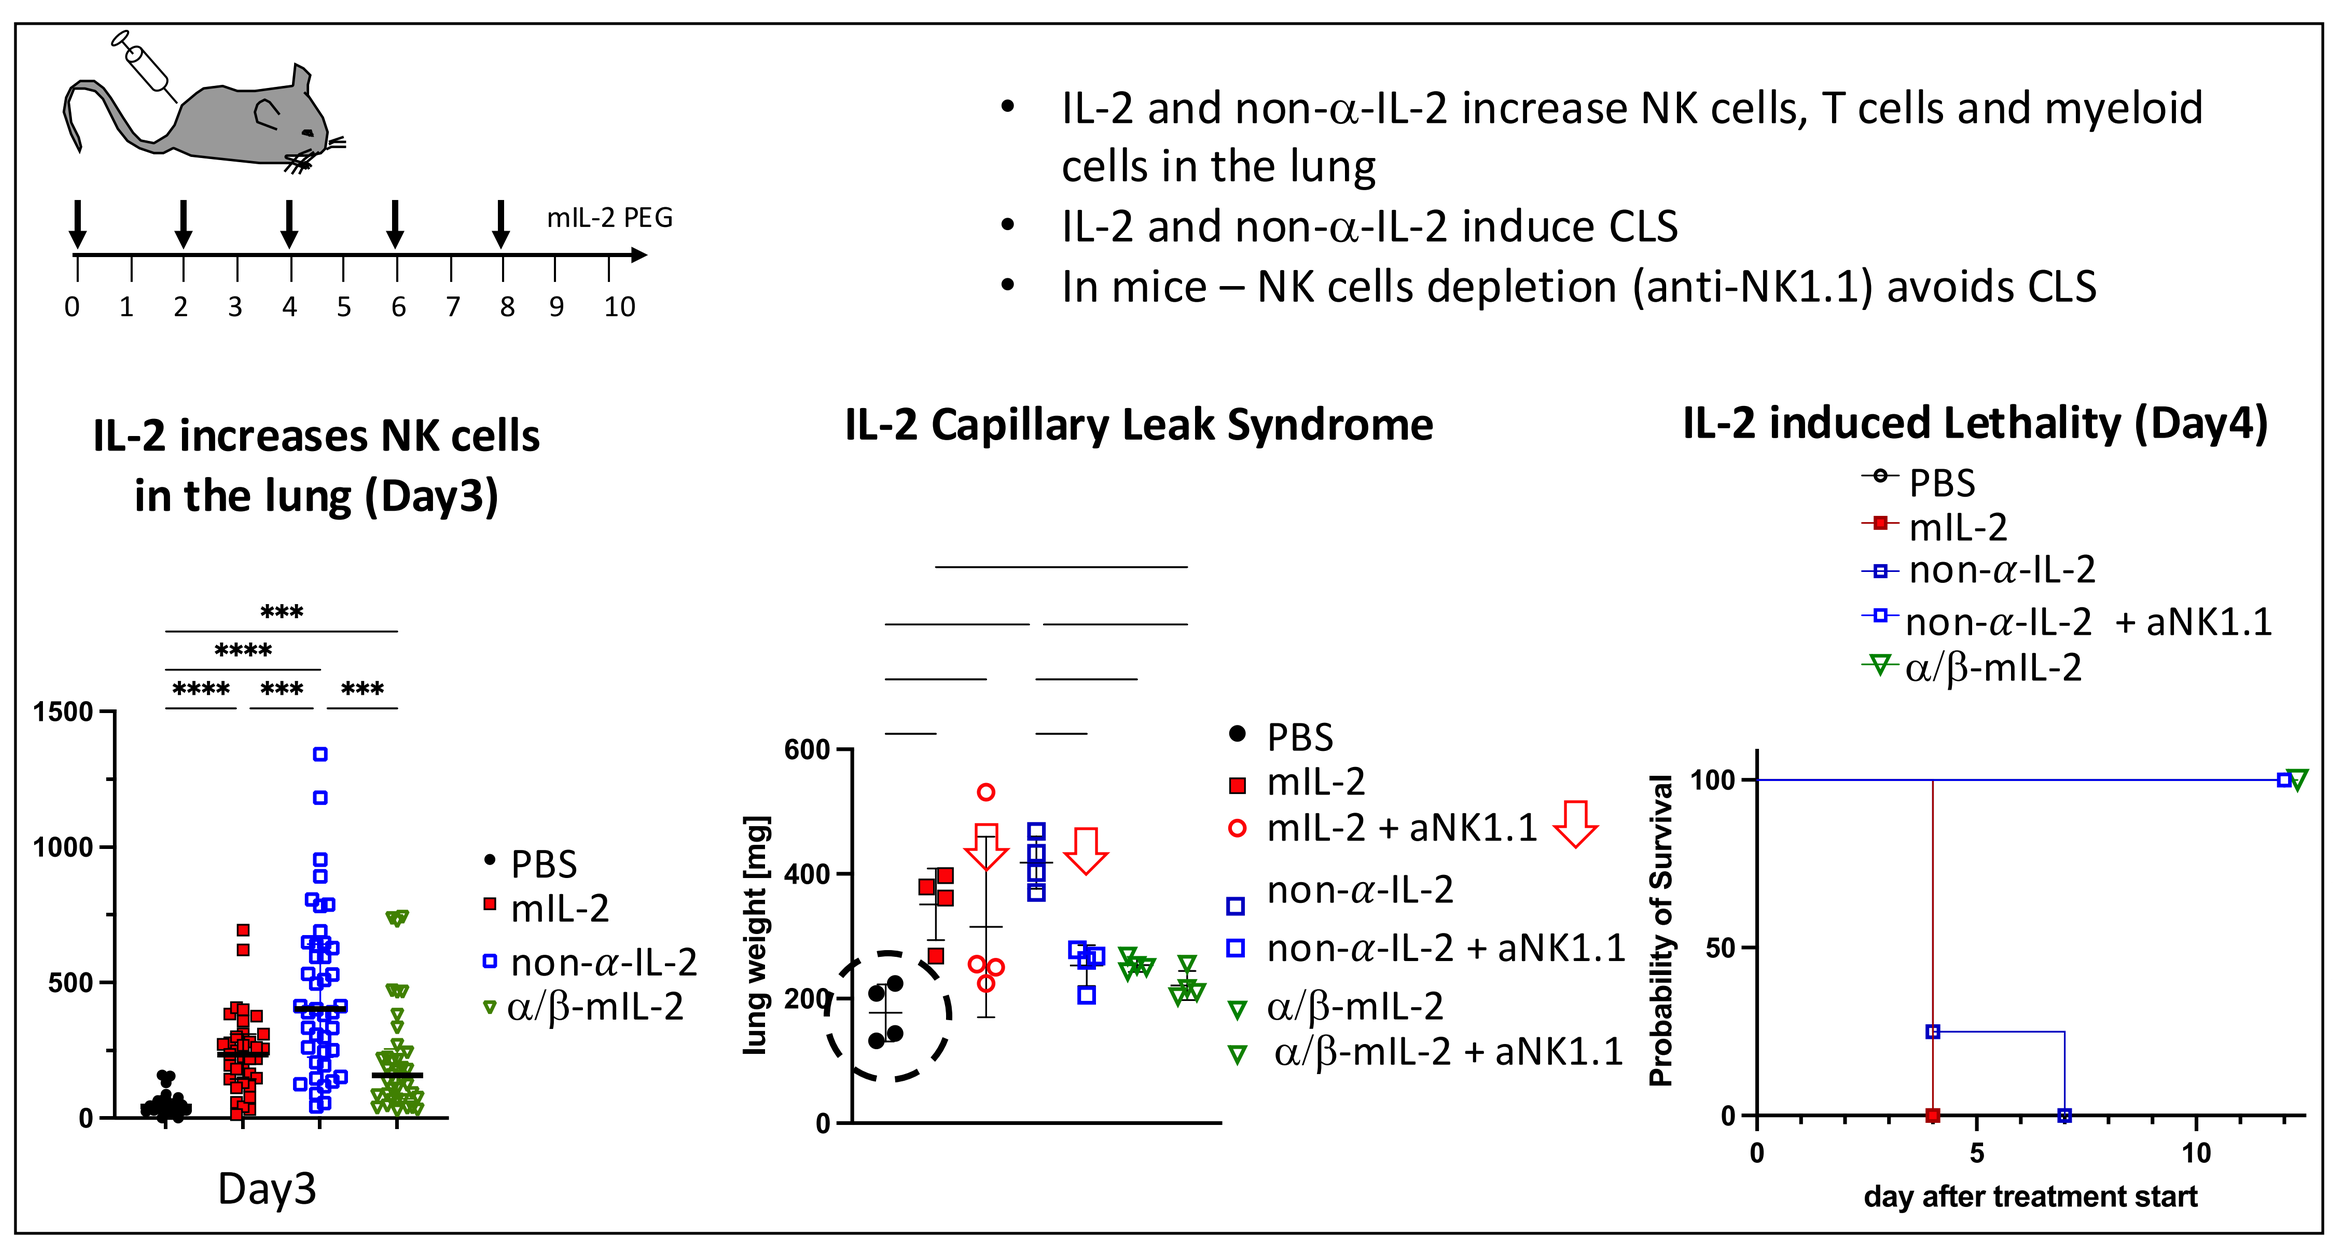
mIL-2 PEG
IL-2 increases NK cells in the lung (Day3)
PBS
mIL-2
non-α-IL-2
α/β-mIL-2
Day3
IL-2 Capillary Leak Syndrome
lung weight [mg]
mIL-2 + aNK1.1
non-α-IL-2 + aNK1.1
α/β-mIL-2 + aNK1.1
IL-2 induced Lethality (Day4)
Probability of Survival
day after treatment start

Clinically, IL-2 induces capillary leak within 3 days, forcing dose interruption and vasopressor treatment. In mice, 2 doses of IL-2 or non-α-IL-2 lead to CLS on day 3 and lethality on day 4. mSTK-012 did not induce CLS or lethality. Treatment with anti-NK1.1 avoided IL-2 induced CLS.

STK-012 Avoids IL-2 Induced Capillary Leak Syndrome in Cynomolgus Monkeys

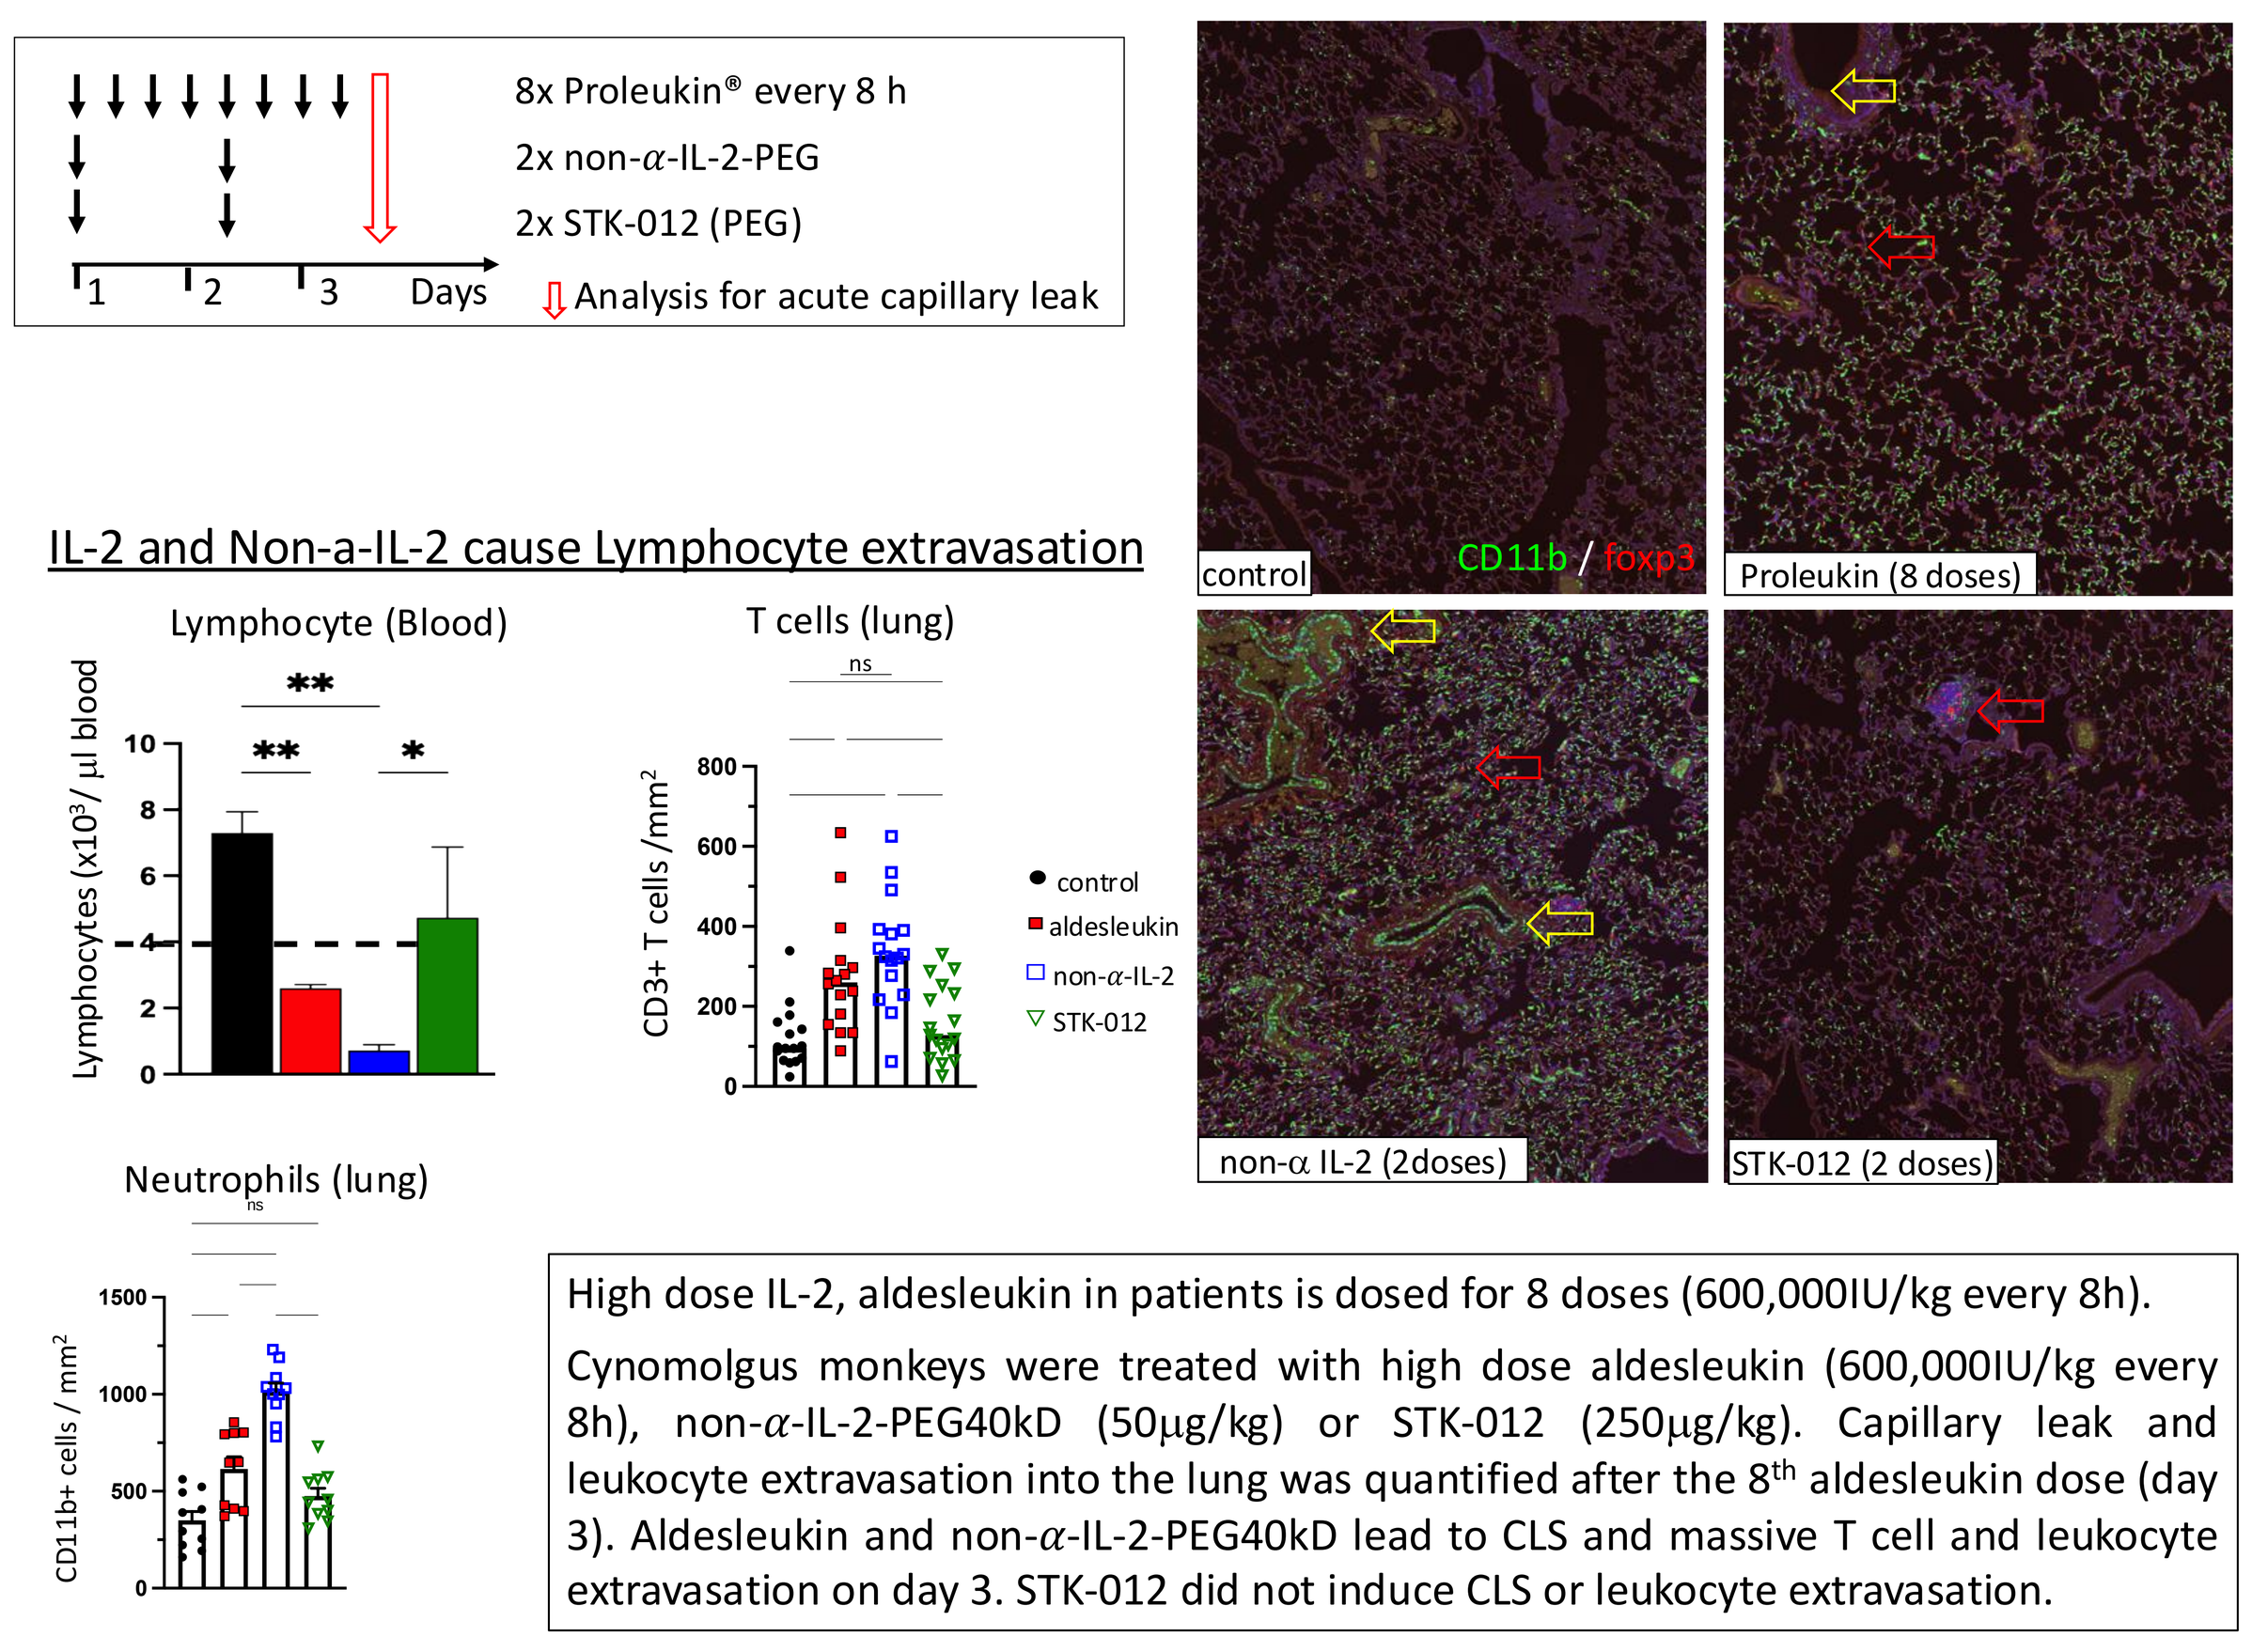
8x Proleukin® every 8 h
2x non-α-IL-2-PEG
2x STK-012 (PEG)
Days
Analysis for acute capillary leak
IL-2 and Non-a-IL-2 cause Lymphocyte extravasation
Lymphocyte (Blood)
Lymphocytes (x10³/ μl blood
T cells (lung)
CD3+ T cells /mm²
control
aldesleukin
non-α-IL-2
STK-012
Neutrophils (lung)
CD11b+ cells / mm²
control
CD11b / foxp3
Proleukin (8 doses)
non-α IL-2 (2doses)
STK-012 (2 doses)

High dose IL-2, aldesleukin in patients is dosed for 8 doses (600,000IU/kg every 8h).

Cynomolgus monkeys were treated with high dose aldesleukin (600,000IU/kg every 8h), non-α-IL-2-PEG40kD (50μg/kg) or STK-012 (250μg/kg). Capillary leak and leukocyte extravasation into the lung was quantified after the 8th aldesleukin dose (day 3). Aldesleukin and non-α-IL-2-PEG40kD lead to CLS and massive T cell and leukocyte extravasation on day 3. STK-012 did not induce CLS or leukocyte extravasation.

Phase 1 Design and Patients in Monotherapy Escalation

- STK-012-101 is a Phase 1a/1b first-in-human study of STK-012 administered subcutaneously (SC) in subjects with advanced relapsed/refractory (r/r) solid tumors.
- During the Phase 1a, subjects were enrolled in a 3+3 dose escalation to
 - Part A: STK-012 monotherapy weekly (QW) over 2 dose levels (DL)
 - Part B: STK-012 monotherapy every 3 weeks (Q3W) over 5 DLs
 - Part C: STK-012 Q3W in combination with pembrolizumab 200 mg Q3W over 4 DLs
- During Phase 1b expansions, subjects were enrolled to
 - Part D: STK-012 monotherapy Q3W expansion at the candidate RP2D
- Escalation in Parts A-C has been completed. Data presentation will be restricted to Parts A and B (Ph1a STK-012 monotherapy).

Part A: STK-012 QW escalation 0.375 mg → 0.75 mg (N=19)

Part B: STK-012 Q3W escalation 0.75 mg → 3 mg (N=28)

Part C: STK-012 Q3W escalation 0.75 mg → 3 mg + Pembro Q3W

Part D: STK-012 Ph1b expansions at 2.25 mg Q3W

Candidate RP2D

2.25 mg Q3W

Baseline Disease Characteristics & Demographics , N=47	
Prior lines in advanced setting	
1	10 (21.3)
2	9 (19.1)
≥3	28 (59.6)
Prior immune checkpoint inhibitor	
0	10 (21.2)
≥1	37 (78.7)
Disease type	
Non-Small Cell Lung Cancer	15 (31.9)
Clear Cell Renal Cell Carcinoma	6 (12.8)
Ovarian Cancer	6 (12.8)
Mucosal Melanoma	4 (8.5)
Other (HNSCC, GEJ, GC, CC, non-ccRCC, MSI-H/dMMR CRC and HNSCC)	13 (27.7)
Age (median)	63.1y
female	44.3%
ECOG 1/0	42.6 / 57.4

STK-012 Monotherapy Safety

- Treatment-related adverse events (TRAEs) were reversible with standard management (e.g. dose hold/reduction, supportive meds).
- No subjects experienced CLS and very few had associated TRAEs (Table 4)
- No dose-limiting toxicities (DLTs) were observed during the DLT period (Cycle 1 [21 days]).
- The majority of TRAEs were Grade 1 or 2 in severity (Table 3); 1 subject experienced a Grade 4 TRAE and no subjects experienced Grade 5 TRAEs.
 - The Grade 4 TRAE of Anaphylactic reaction occurred at C9D1 (1.5 mg Q3W) and led to treatment discontinuation; the event resolved on the same day with supportive treatment.
- All 3 subjects treated at the maximum dose of 3 mg Q3W experienced Grade 3 GI SAEs (Small bowel obstruction, Enterocolitis, Vomiting) in Cycle 2 (after the DLT period).
- An intermediate dose of 2.25 mg Q3W was then explored and selected as candidate RP2D.

Most Common TRAEs (≥10% of subjects) STK-012 monotherapy, N=47

TRAE	All Grade, N (%)	Grade ≥3, N (%)
Rash maculo-papular	19 (40.4)	5 (10.6)
Fatigue	12 (25.5)	1 (2.1)
Injection site reaction	11 (23.4)	0
Nausea	11 (23.4)	1 (2.1)
Diarrhea	9 (19.9)	2 (4.3)
Pruritis	7 (14.9)	0
Vomiting	6 (12.8)	2 (4.3)
Arthralgia	5 (10.6)	1 (2.1)
Total (TRAEs)	**37 (78.7)**	**15 (31.9)**

Capillary Leak associated TRAEs STK-012 monotherapy, N=47

TRAE	All Grade, N (%)	Grade ≥3, N (%)
Hypotension	2 (4.3)	0
Pyrexia	2 (4.3)	0
Flu-like symptoms	2 (4.3)	0
Peripheral edema	2 (4.3)	0
CRS	1 (2.1)	0
Creatinine Increase	1(2.1)	1(2.1)
CLS	0	0
LFT increase	0	0
Lymphopenia	0	0

STK-012 Monotherapy

- Of 40 efficacy evaluable# subjects, 3 had partial response (PR) and 12 had stable disease (SD) as their best overall response (BOR) by RECIST V1.1.
- The partial responses included 2 confirmed (anti-PD-1 pretreated NSCLC and RCC) and 1 unconfirmed (anti-PD-1 pretreated HNSCC).

Waterfall for Phase 1a Monotherapy

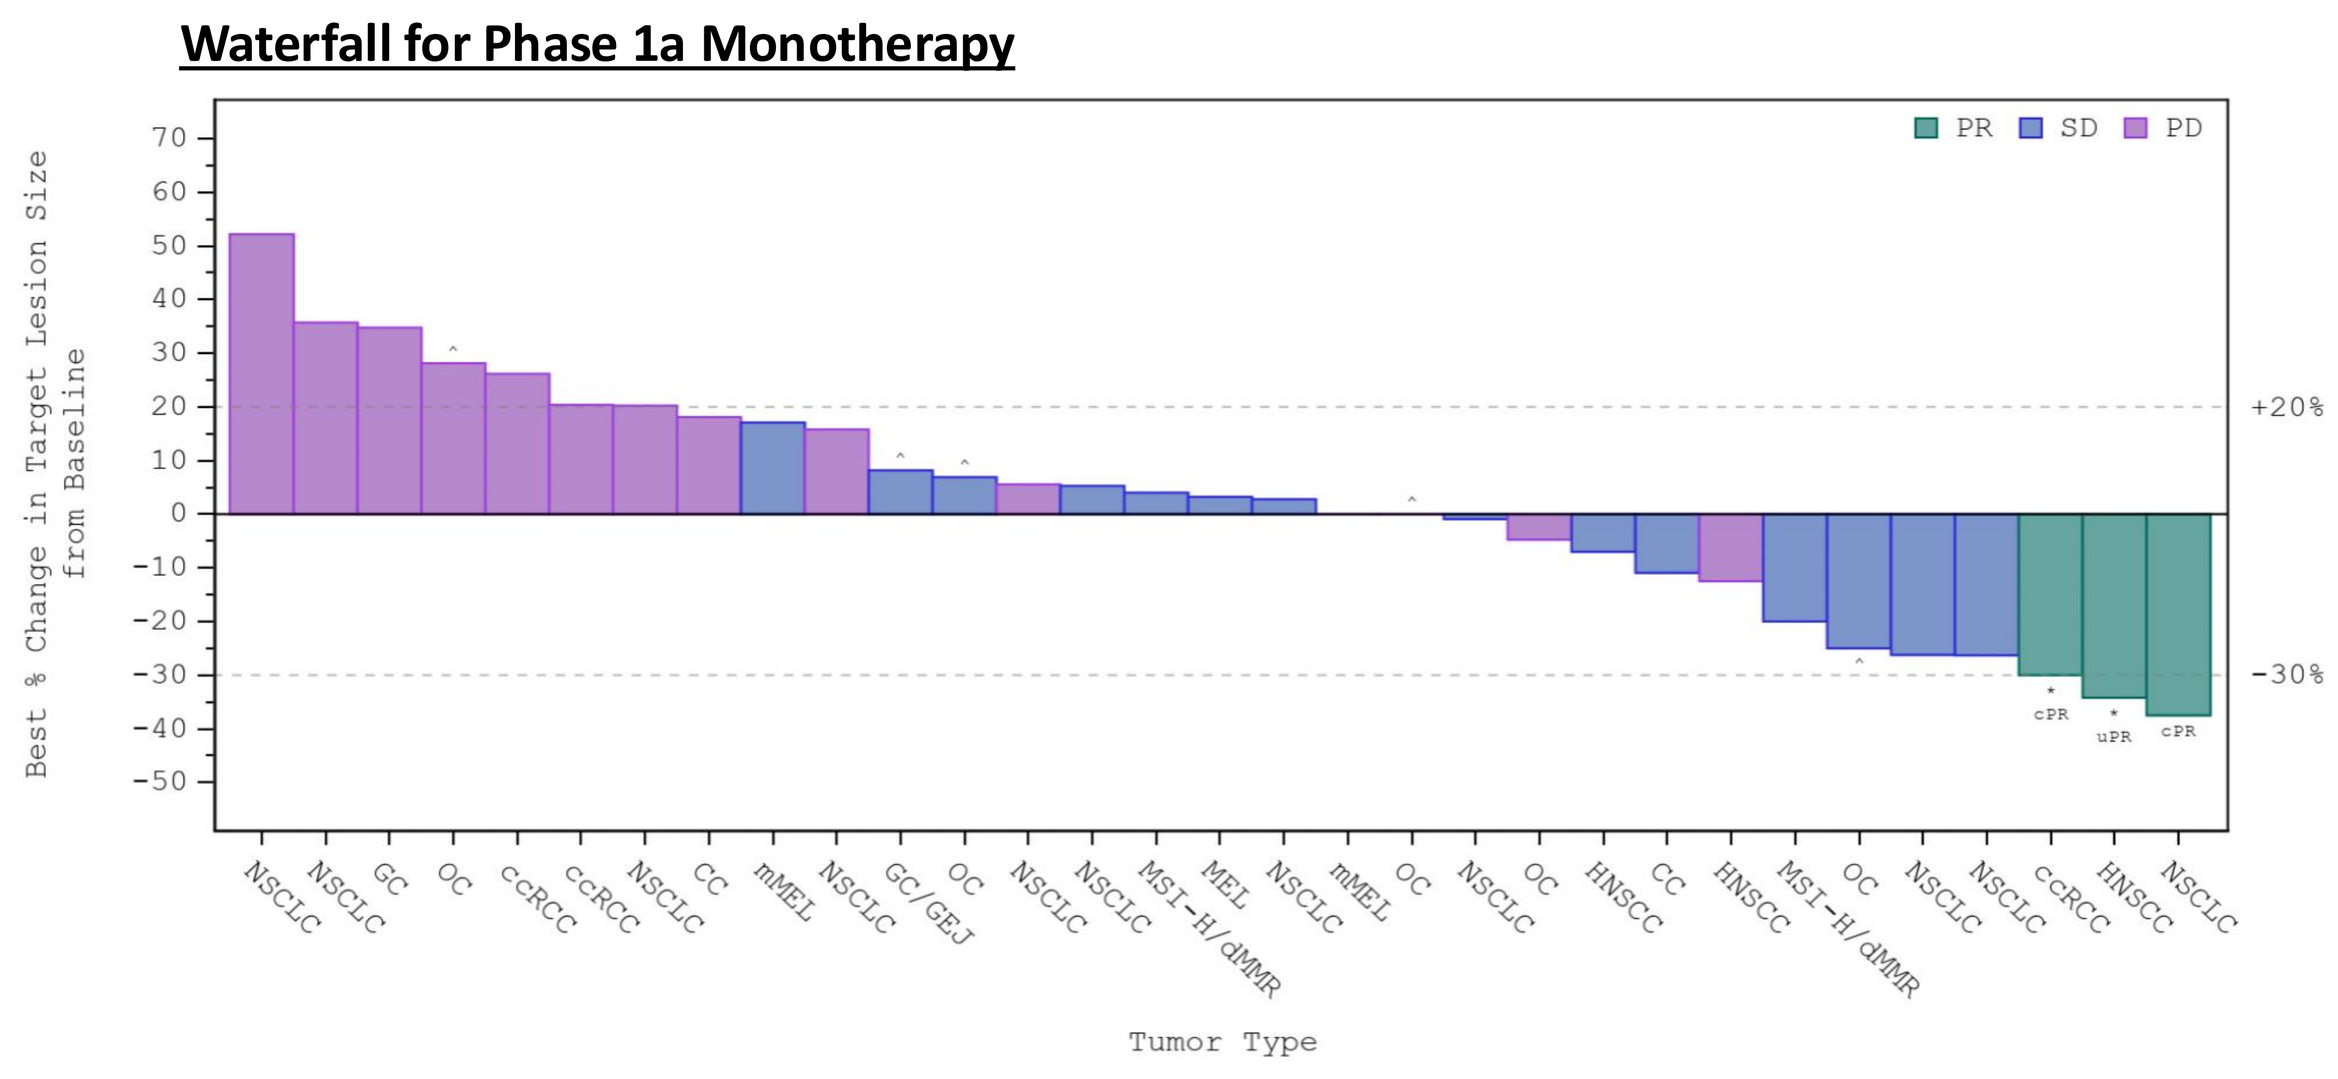
PR
SD
PD
Best % Change in Target Lesion Size from Baseline
+20%
-30%
cPR
uPR
cPR
NSCLC
NSCLC
GC
OC
ccRCC
ccRCC
NSCLC
CC
mMEL
NSCLC
GC/GEJ
OC
NSCLC
NSCLC
MSI-H/dMMR
MEL
NSCLC
mMEL
OC
NSCLC
OC
HNSCC
CC
HNSCC
MSI-H/dMMR
OC
NSCLC
NSCLC
ccRCC
HNSCC
NSCLC
Tumor Type

Note: 31 subjects are represented on this waterfall plot. 9 efficacy evaluable subjects are not represented due to (1) Clinical PD before 1st scan OR (2) target lesions not evaluable at baseline OR (3) discontinued due to related AE prior to scans OR (4) BOR NE with no other evaluable scan timepoints. - Subjects are censored after the first reporting of radiographic or clinical PD.
* Indicates subjects who are ongoing treatment; ^ indicates subjects who are IO Naive.

Greater Efficacy in IO Pretreated Subjects With 1-2 Prior Lines

- In subjects who progressed on/after prior anti-PD-1 who also had only 1-2 prior lines in the advanced setting (N=15 efficacy evaluable), 3 had BOR of PR and 6 BOR of SD
- Durability of effect was observed in multiple subjects with BOR of SD and the ccRCC responder with sustained response for >9 months.

Waterfall for Ph1a Monotherapy (IO Pretreated, 1-2 Prior Lines)

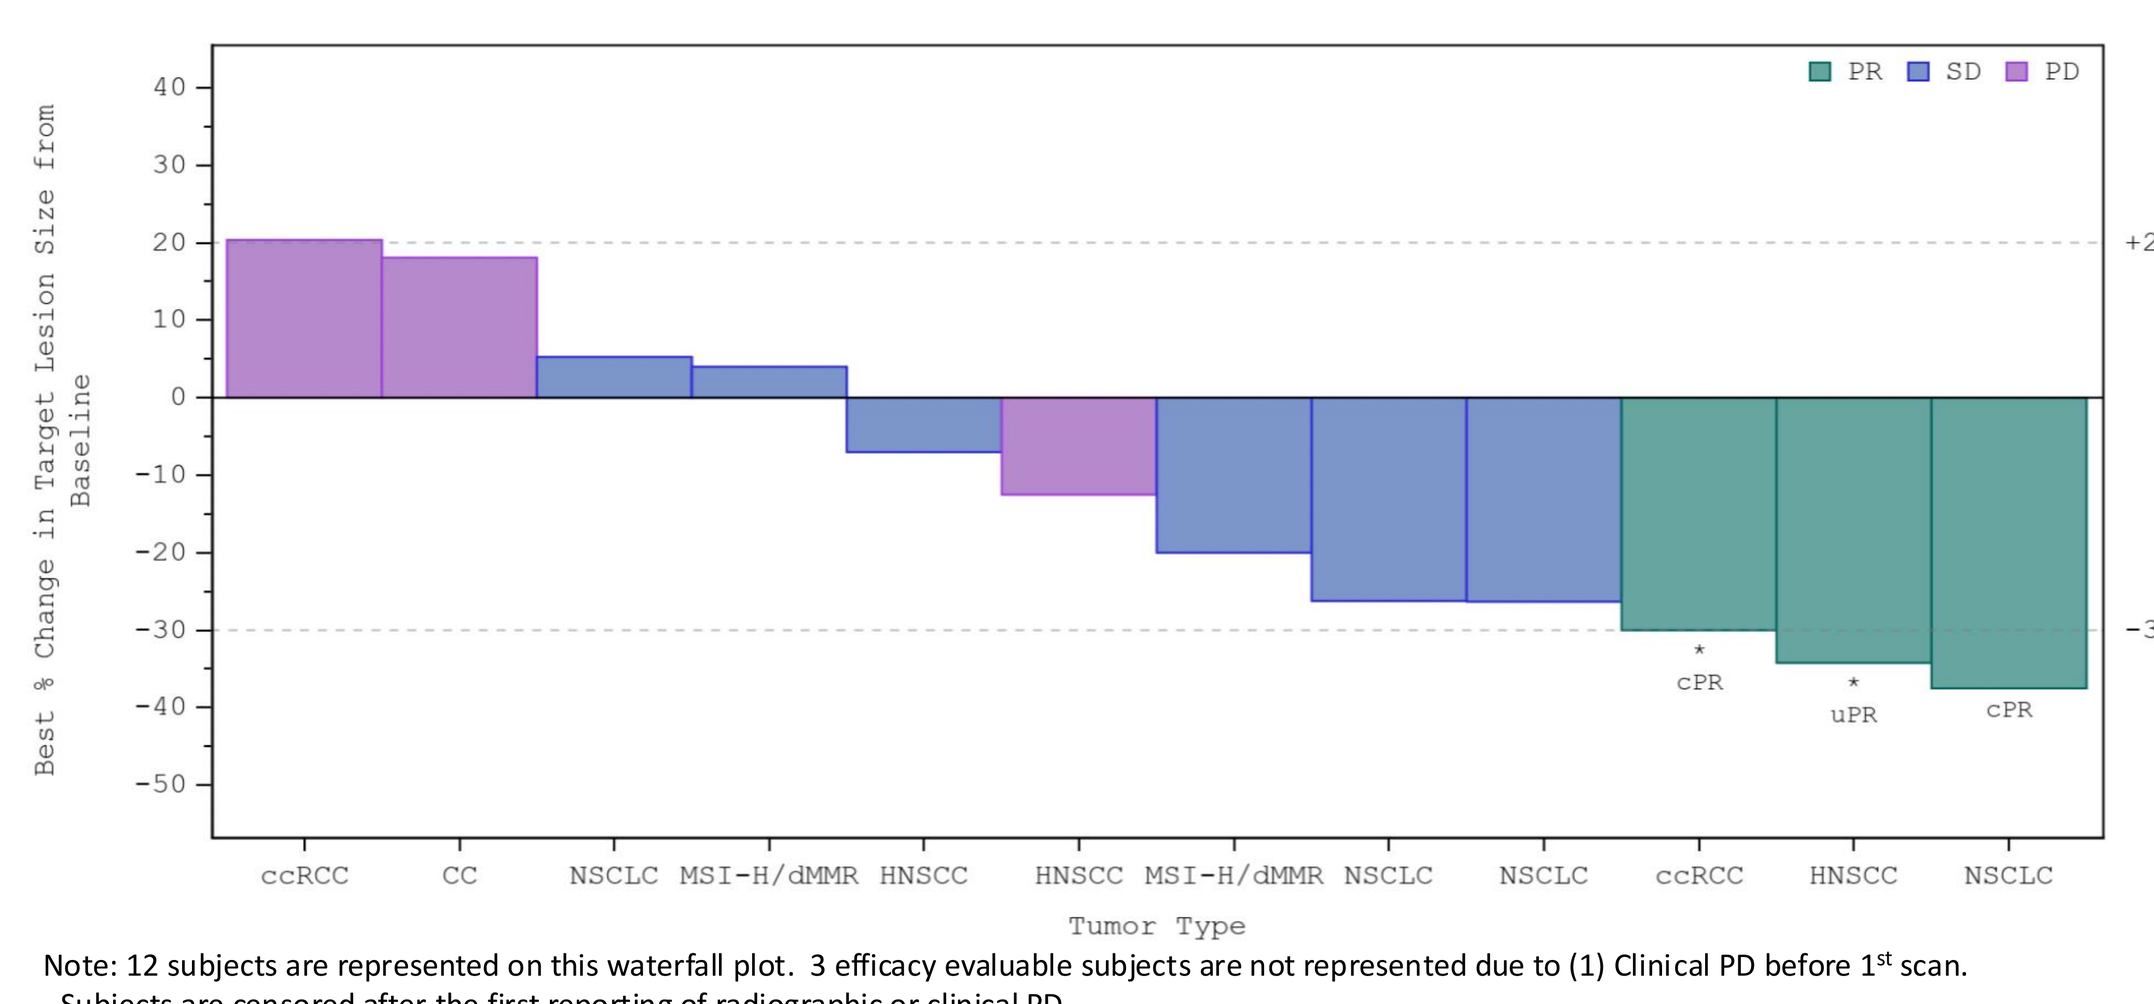
PR
SD
PD
Best % Change in Target Lesion Size from Baseline
+20%
-30%
cPR
uPR
cPR
ccRCC
CC
NSCLC
MSI-H/dMMR
HNSCC
HNSCC
MSI-H/dMMR
NSCLC
NSCLC
ccRCC
HNSCC
NSCLC
Tumor Type

Note: 12 subjects are represented on this waterfall plot. 3 efficacy evaluable subjects are not represented due to (1) Clinical PD before 1st scan.
- Subjects are censored after the first reporting of radiographic or clinical PD.
* Indicates subjects who are ongoing treatment as of the data extract date .

Spider for Ph1a Monotherapy (IO Pretreated with 1-2 Prior Lines)

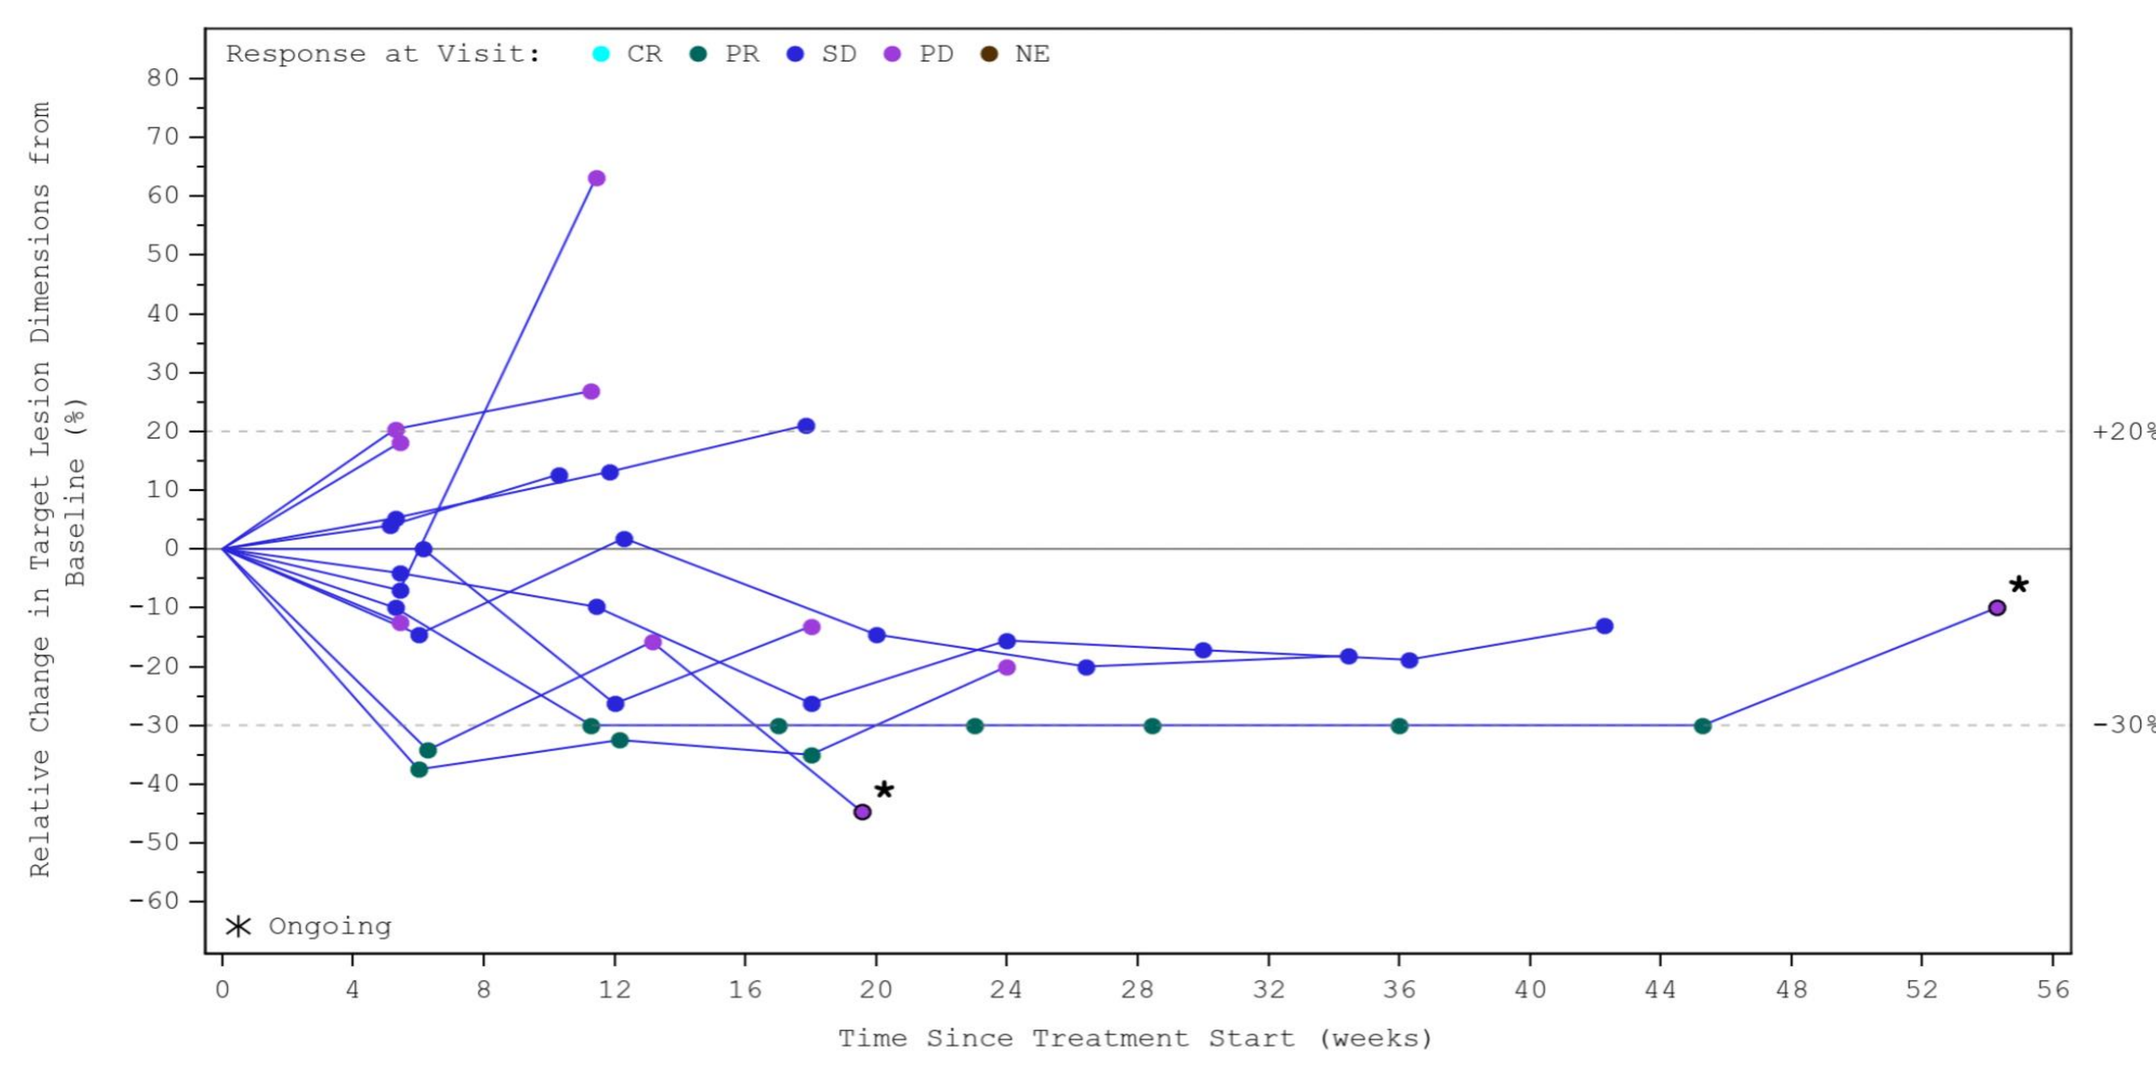
Response at Visit:
CR
PR
SD
PD
NE
Relative Change in Target Lesion Dimensions from Baseline (%)
+20%
-30%
Ongoing
Time Since Treatment Start (weeks)

Note: 12 subjects are represented on this spider plot. 3 efficacy evaluable subjects are not represented due to clinical PD before 1st scan.
- Subjects are censored after the first reporting of radiographic or clinical PD.
* Indicates subjects who are ongoing treatment as of the data extract date .

STK-012 Targets CD25+ activated T cells

STK-012 PK

- Observed Half-life is ~4 days.
- STK-012 remains above EC50 for 3 weeks
- Exposure (AUC) is dose proportional.

CD25 selective p-STAT5

STK-012 demonstrates selectivity for CD25 (IL-2Rα) expressing T cells

IFNγ Response

Serum IFNγ increase is dose dependent and peaks at STK-012 Tmax (Day 8)

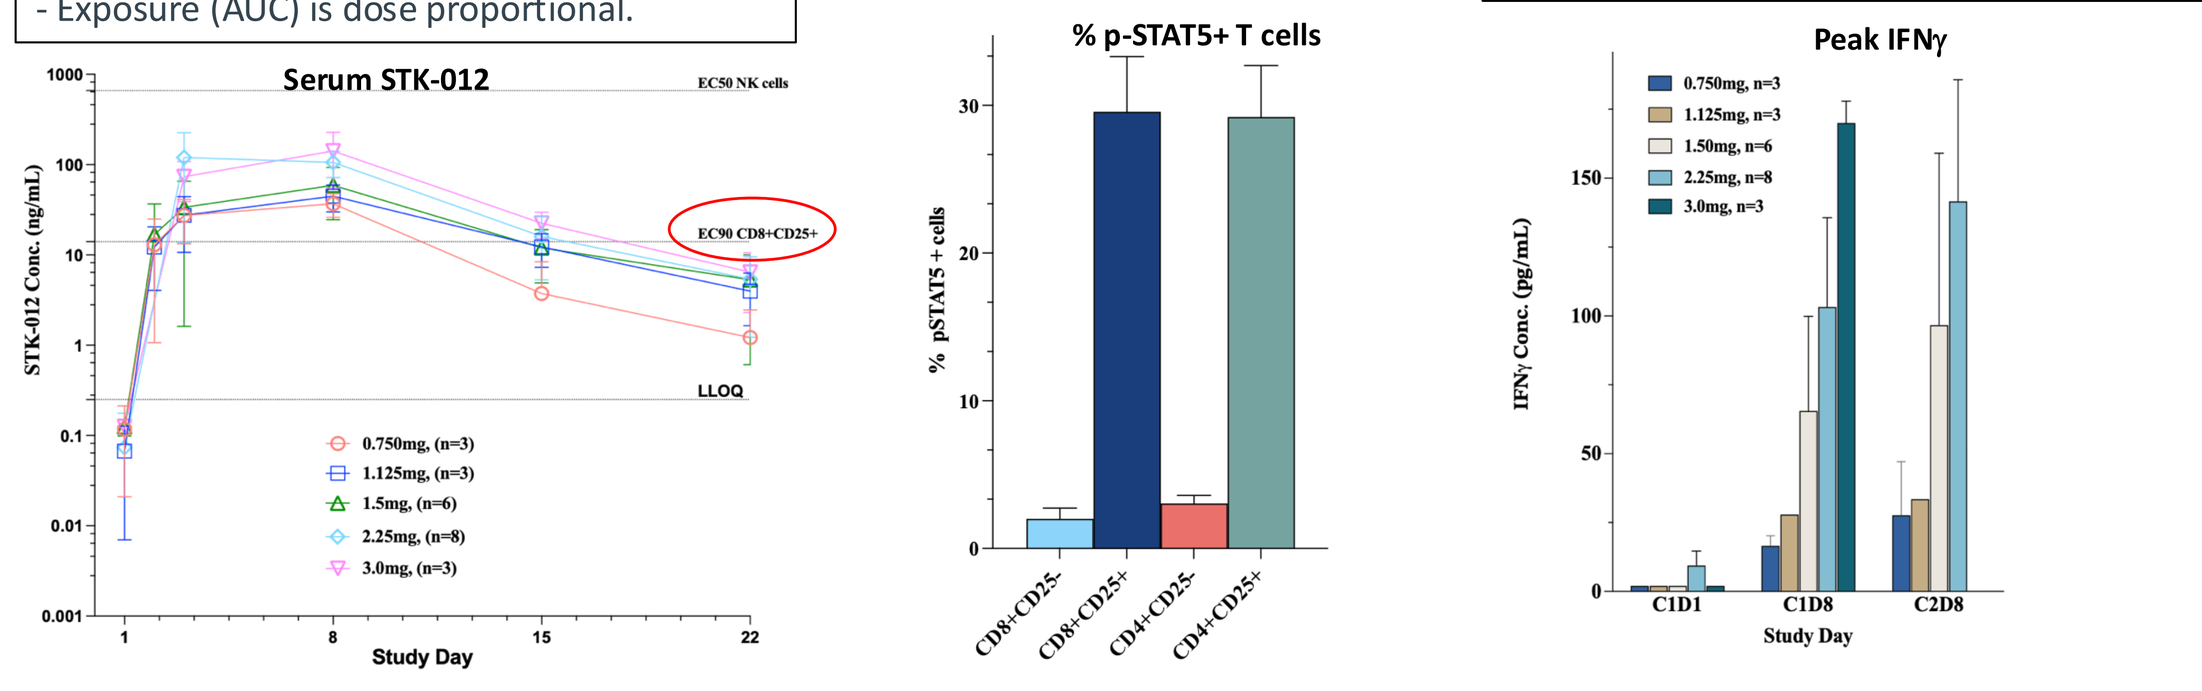
Serum STK-012
EC50 NK cells
EC90 CD8+CD25+
LLOQ
STK-012 Conc. (ng/mL)
0.750mg, (n=3)
1.125mg, (n=3)
1.5mg, (n=6)
2.25mg, (n=8)
3.0mg, (n=3)
Study Day
% p-STAT5+ T cells
% pSTAT5 + cells
CD8+CD25-
CD8+CD25+
CD4+CD25-
CD4+CD25+
Peak IFNγ
IFNγ Conc. (pg/mL)
0.750mg, n=3
1.125mg, n=3
1.50mg, n=6
2.25mg, n=8
3.0mg, n=3
C1D1
C1D8
C2D8
Study Day

Peripheral Flow Cytometry Supports Selectivity for Antigen Activated CD8 T cells

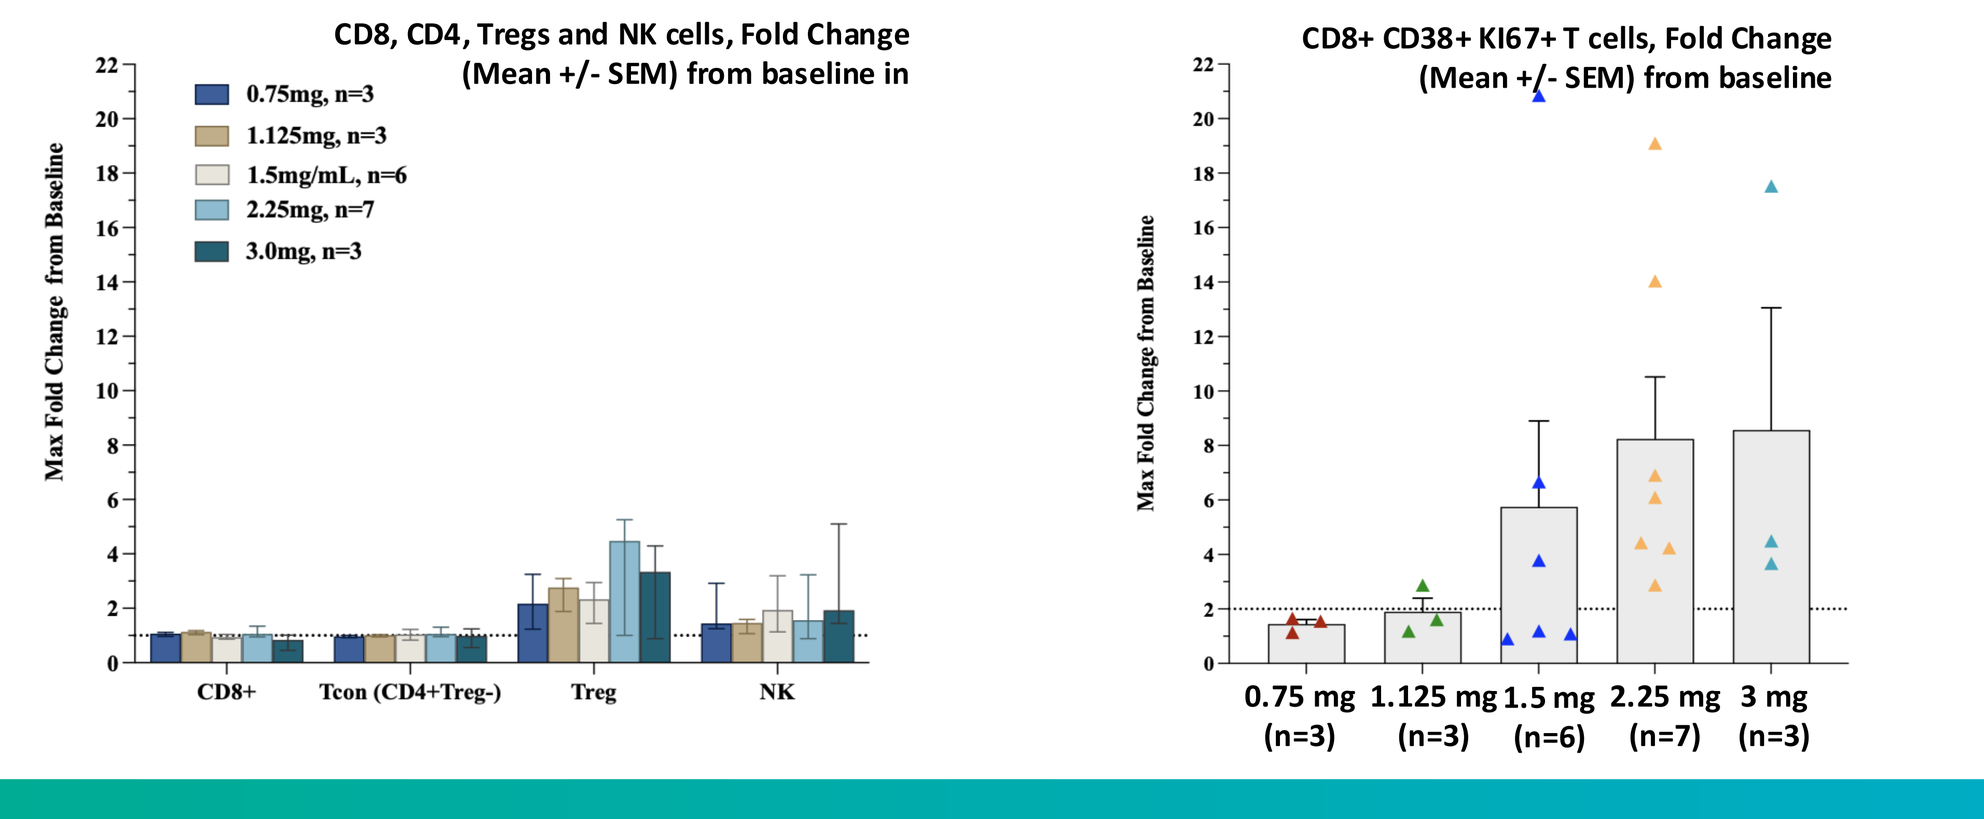
CD8, CD4, Tregs and NK cells, Fold Change (Mean +/- SEM) from baseline in
0.75mg, n=3
1.125mg, n=3
1.5mg/mL, n=6
2.25mg, n=7
3.0mg, n=3
Max Fold Change from Baseline
CD8+
Tcon (CD4+Treg-)
Treg
NK
CD8+ CD38+ KI67+ T cells, Fold Change (Mean +/- SEM) from baseline
0.75 mg (n=3)
1.125 mg (n=3)
1.5 mg (n=6)
2.25 mg (n=7)
3 mg (n=3)

Case Studies

Subject With Advanced ccRCC - 85% cPR on STK-012

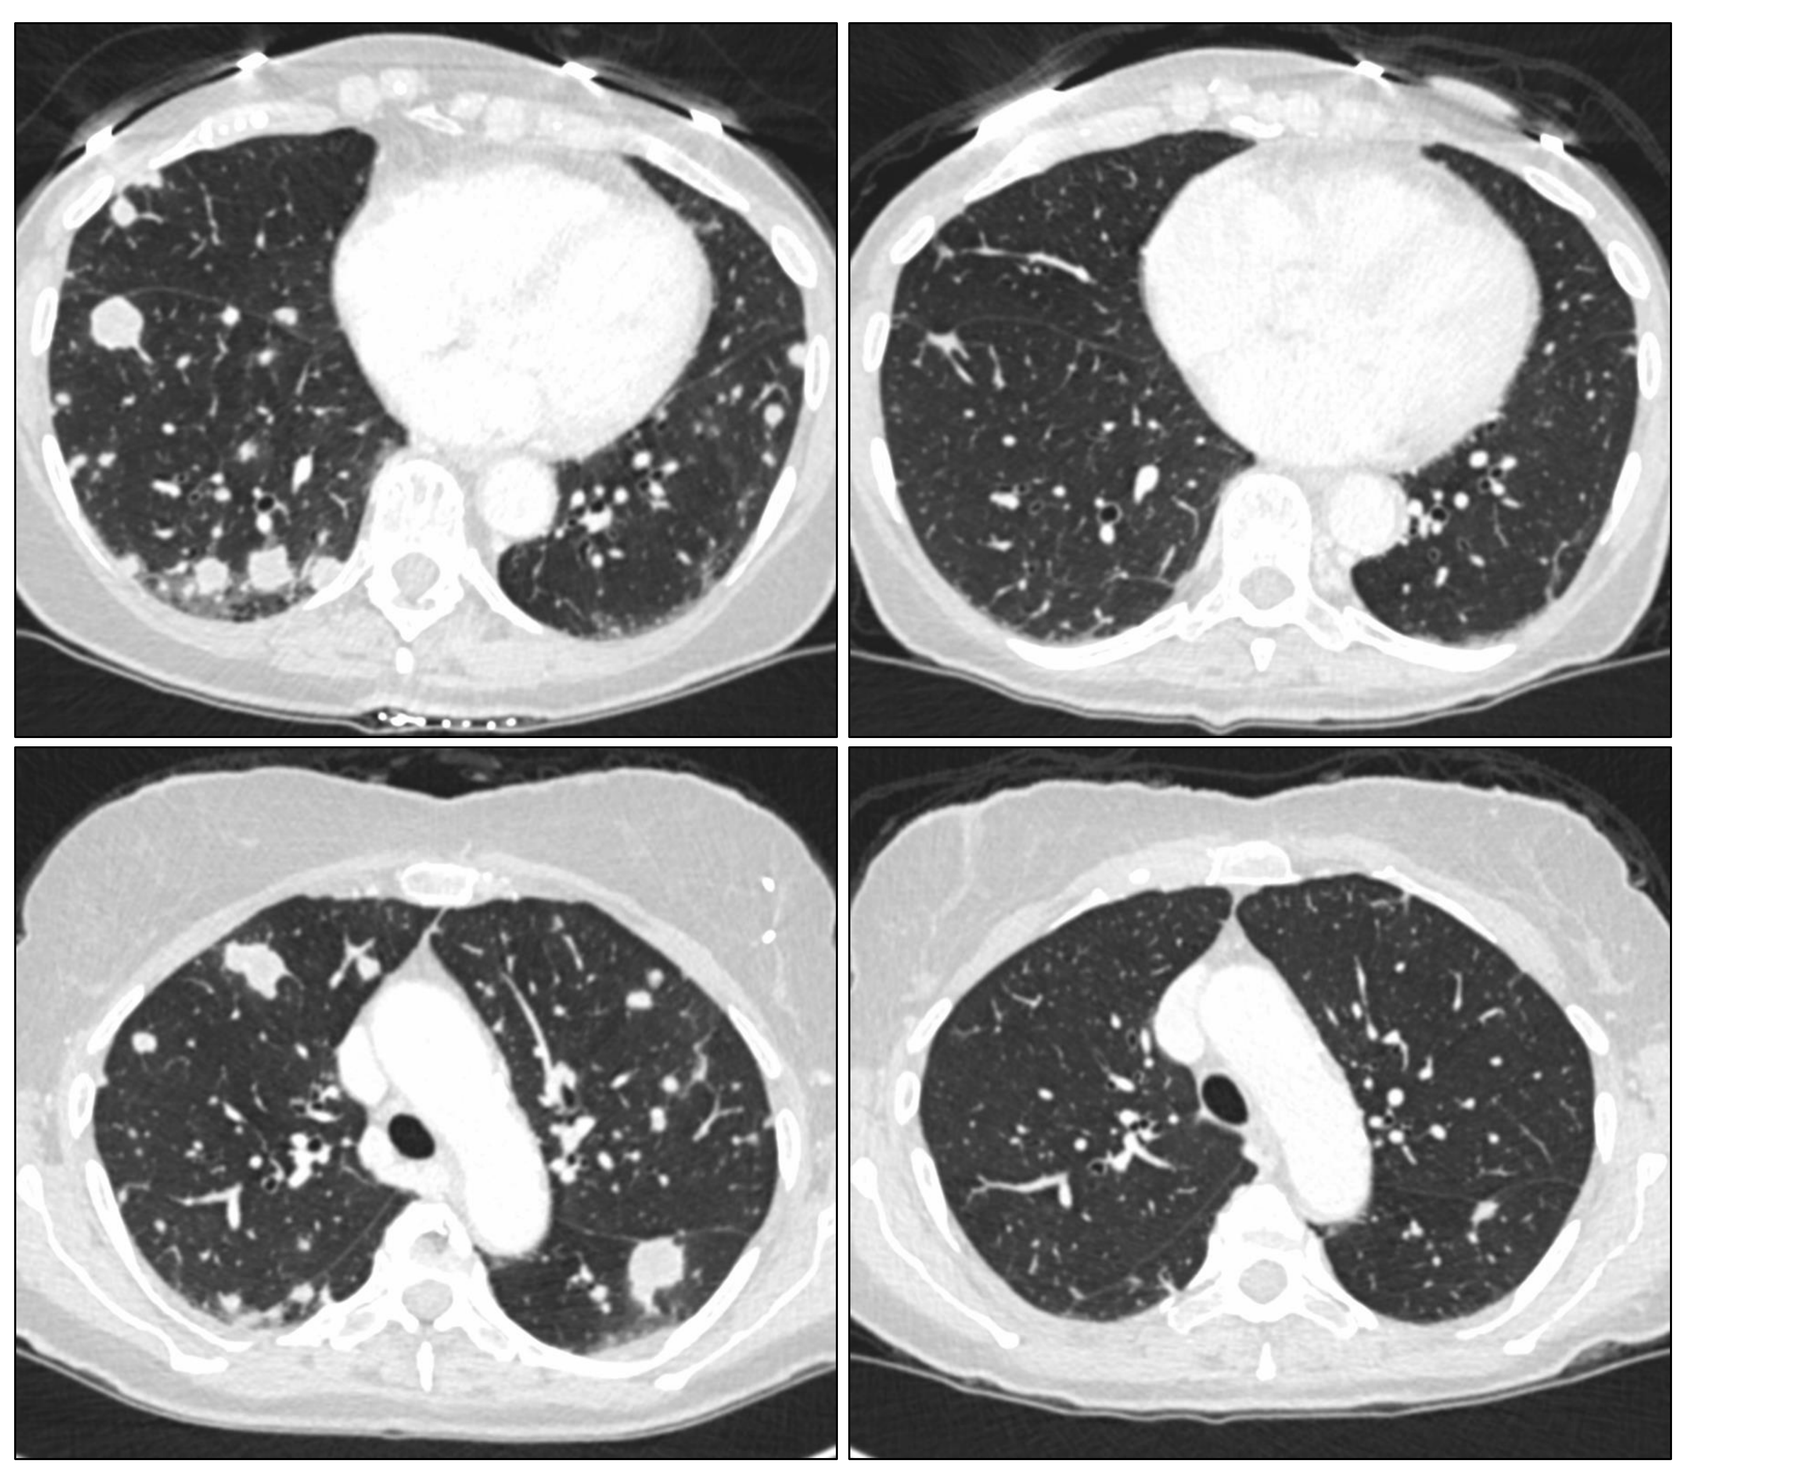
Chest CT
Baseline
Week 12

Subject History

72-year-old female with Stage IV ccRCC, 2 prior lines of therapy before STK-012 monotherapy

Prior lines:

1. Nivolumab + ipilimumab: Aug 2022 - Mar 2023 (BOR: cPR)
2. Cabozantinib + Nivolumab: Apr 2023 - Aug 2023 (BOR: PD)

Response on STK-012 Ph1b ccRCC monotherapy in expansion:

- Baseline: "innumerable" lung metastases, large primary renal mass (non-target).
- STK-012 Dose: 2.25mg Q3W Monotherapy
- 1.5 mg Q3W due to Gr 2 constitutional symptoms

Wk	Change From Baseline
6	↓50% (PR)
12	↓80% (cPR)
18	↓85% (cPR)
24	↓76% (PD)

Subject With Advanced NSCLC 28% ↓ Tumor at 6 weeks

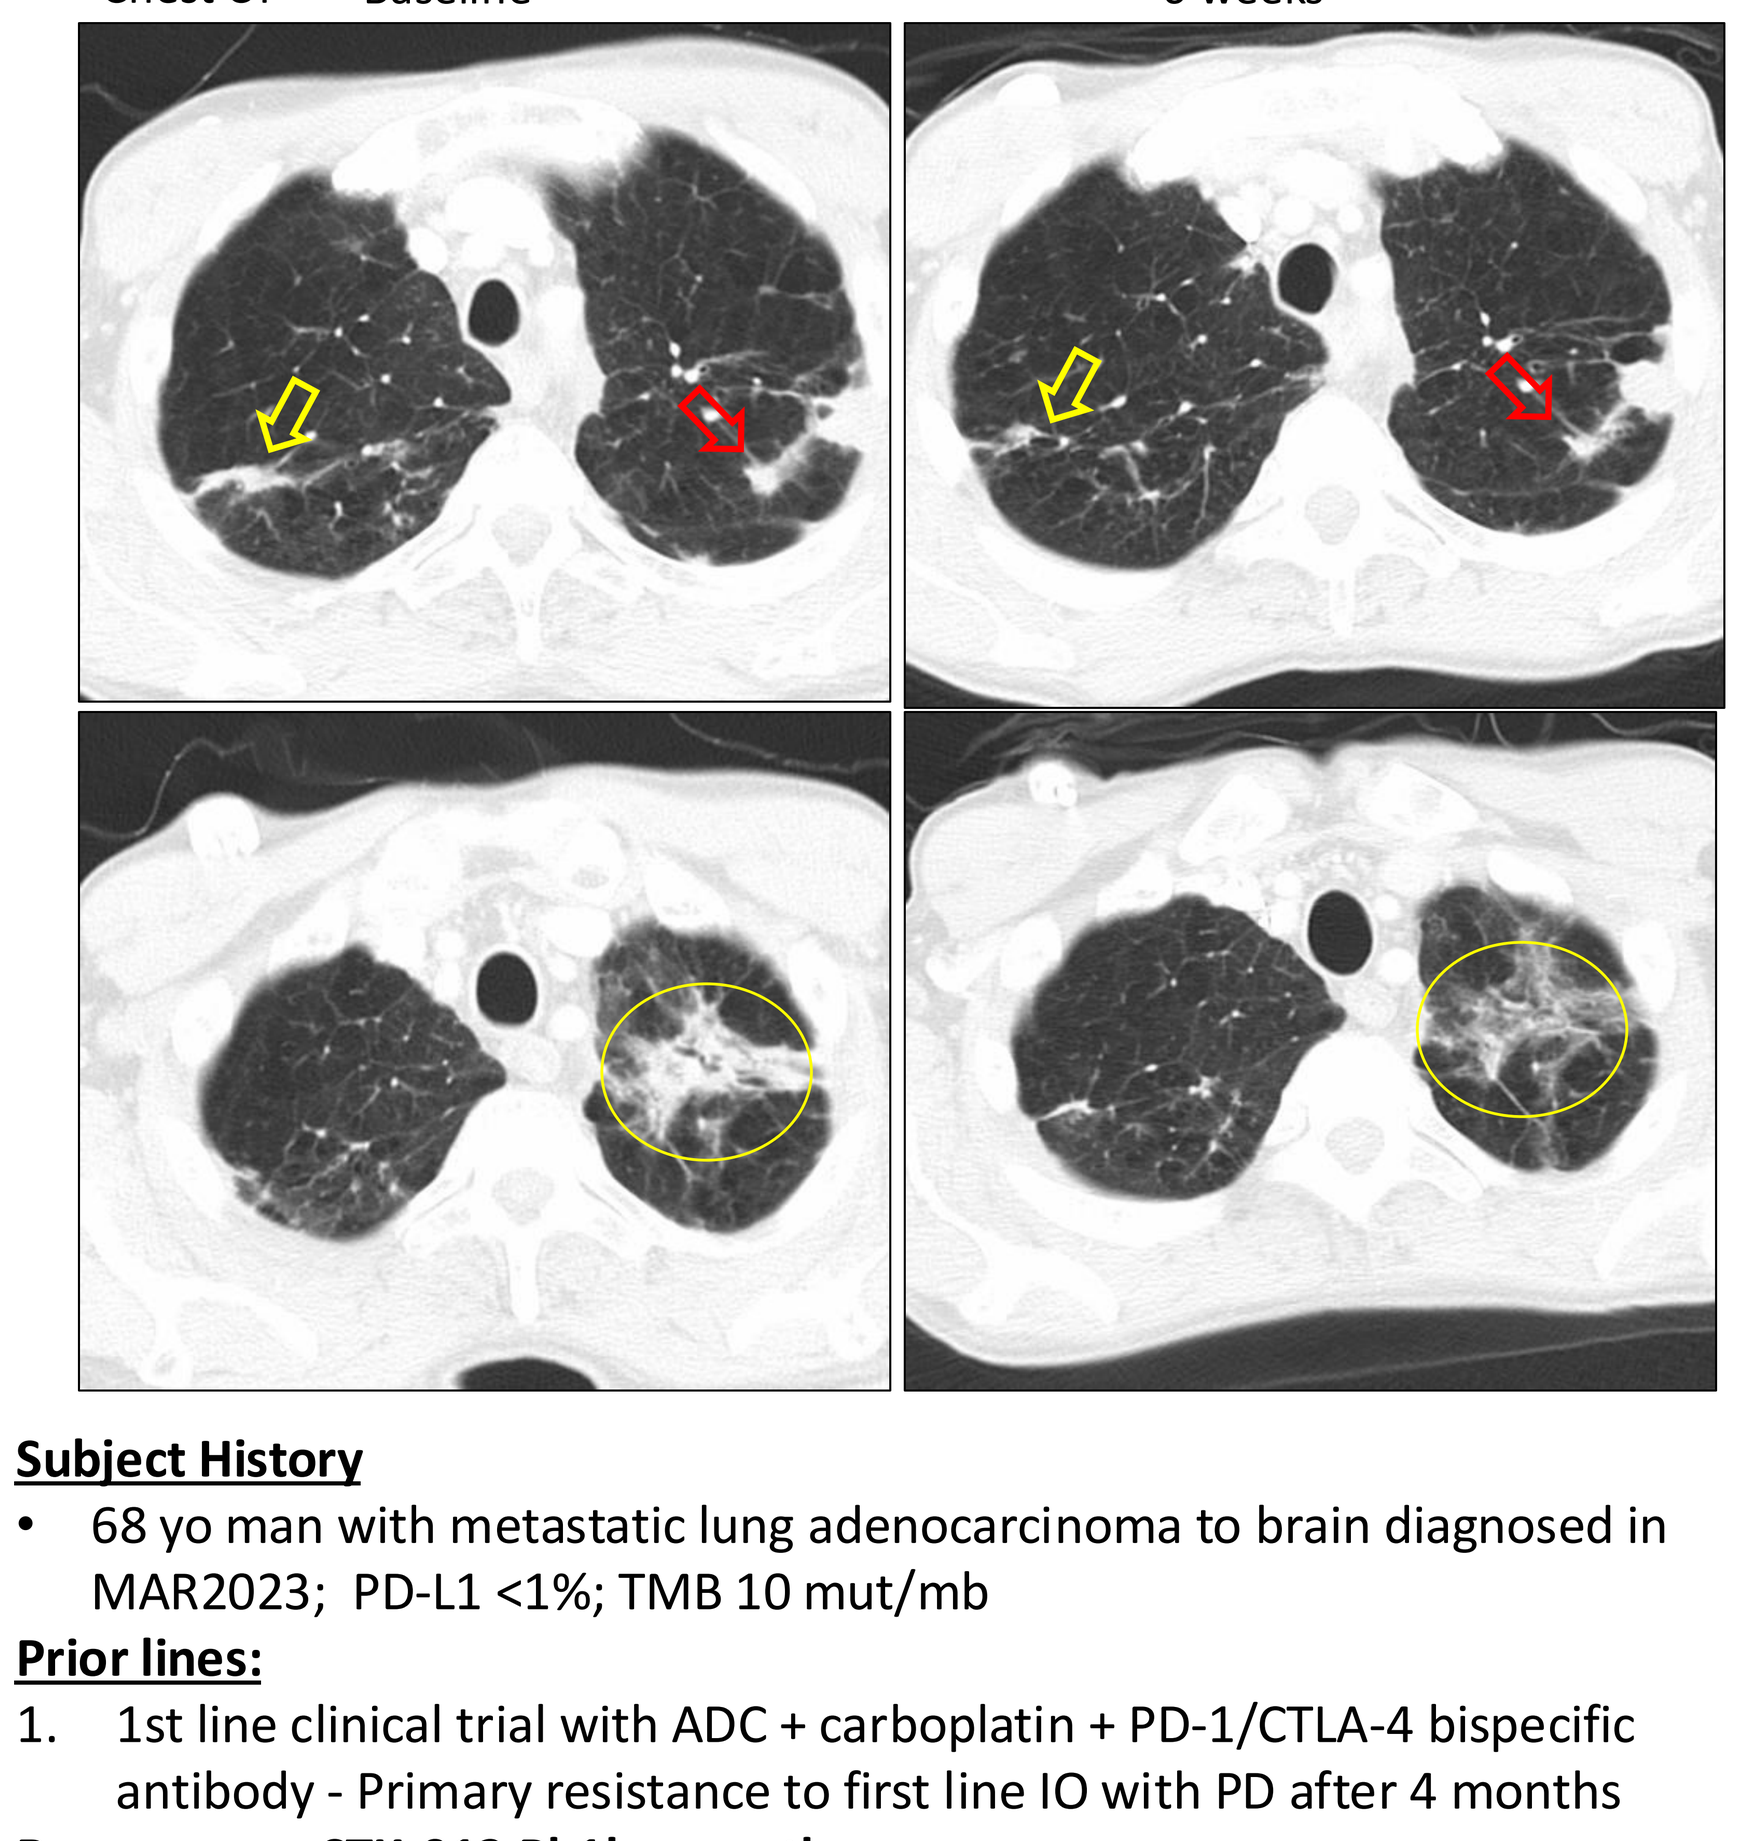
Chest CT
Baseline
6 weeks

Subject History

- 68 yo man with metastatic lung adenocarcinoma to brain diagnosed in MAR2023; PD-L1 <1%; TMB 10 mut/mb

Prior lines:

1. 1st line clinical trial with ADC + carboplatin + PD-1/CTLA-4 bispecific antibody - Primary resistance to first line IO with PD after 4 months

Response on STK-012 Ph1b monotherapy:

- CT at 6 weeks 28% response with treatment for 18 weeks
- Decline in tumor marker (CEA 218→141)

Conclusions

- STK-012 targets activated, CD25+ T cells, avoiding the majority of non-activated T cells and NK cells.
- mSTK-012 has improved anti-tumor efficacy compared to IL-2
- mSTK-012 significantly increases tumor infiltrating T cells (TILs), CD25+ TILs and tumor-antigen specific T cells compared to IL-2 and non-a-IL-2
- mSTK-012 avoids IL-2 associated capillary leak syndrome (CLS) and lethality

- In the ongoing STK-012-101 study, monotherapy STK-012 showed a favorable safety profile and no DLTs at high, durable exposures, long T1/2, and PD profiles significantly improved from aldesleukin and non-α IL-2 analogues.
- Preliminary PK-PD data support selectivity for IL-2R α/β, selective expansion of activated CD8 T cells, and dose dependent serum IFNγ increase.
- Preliminary efficacy with monotherapy STK-012 (3 partial responses in Phase 1a) was observed in subjects who progressed on/after prior immunotherapy.
- Preliminary evidence of activity was observed in the ongoing Phase 1b.
- Further development is warranted and enrollment in STK-012 monotherapy dose expansions (Phase 1b) in advanced NSCLC and ccRCC is ongoing.
- The trial is registered with Clinicaltrials.gov, NCT05098132.