Gradual lymphocyte activation with IL-12 partial agonist STK-026 maintains anti-tumor efficacy and escapes acute NK-mediated cytokine release and toxicities associated with WT IL-12

Michael Totagrande, Ievgen Koliesnik, Cindy Buffone, Ryan Burgess, Kim Quyen Tran, Michele Bauer, Bhargavi Jayaraman, Priyanka Balasubrahmanyam, Mohammad Ali, Henry Rosas, Deepti Chaturvedi, Deepti Rokkam, Somya Singh, Woei Chang, Heiko Greb, Navneet Ratti, Sandro Vivona, Patrick Lupardus, Rene de Waal Malefyt, Luis Zuniga, David B Rosen, Martin Oft, Robert A. Kastelein Synthekine, Menlo Park

Synthekine
ABSTRACT #964

ABSTRACT

Interleukin-12 (IL-12) is a pro-inflammatory cytokine composed of p35 and p40 subunits produced by antigen-presenting cells to stimulate Th1 cells, cytotoxic CD8 T cells, and NK cells. IL-12 has potent anti-tumor properties in multiple preclinical models. However, clinical applications of wild type IL-12 (WT IL-12) or WT IL-12 Fc fusions are hampered by early severe dose-limiting toxicities and later tachyphylaxis [1]. Preclinically, IL-12 toxicity is mediated by NK cell activation [2].

STK-026 is an Fc-fused human IL-12 partial agonist (STK-026) with diminished binding to IL-12Rb1. STK-026 exhibits selectivity for antigen activated T cells and avoids NK cell induced toxicities. Here we demonstrate that STK-026, or its murine surrogate mSTK-026, provides gradual, prolonged, and efficacious T cell and NK cell activation, achieving anti-tumor efficacy with minimal toxicity, while avoiding acute NK-cell mediated toxicity.

Pharmacokinetic/pharmacodynamic responses and overall toxicity to human STK-026 were evaluated in healthy cynomolgus macaques. The data demonstrate that STK-026 exhibits reduced release of IL-12 related toxicity associated .

NK-dependent toxicity limits the use of WT IL-12

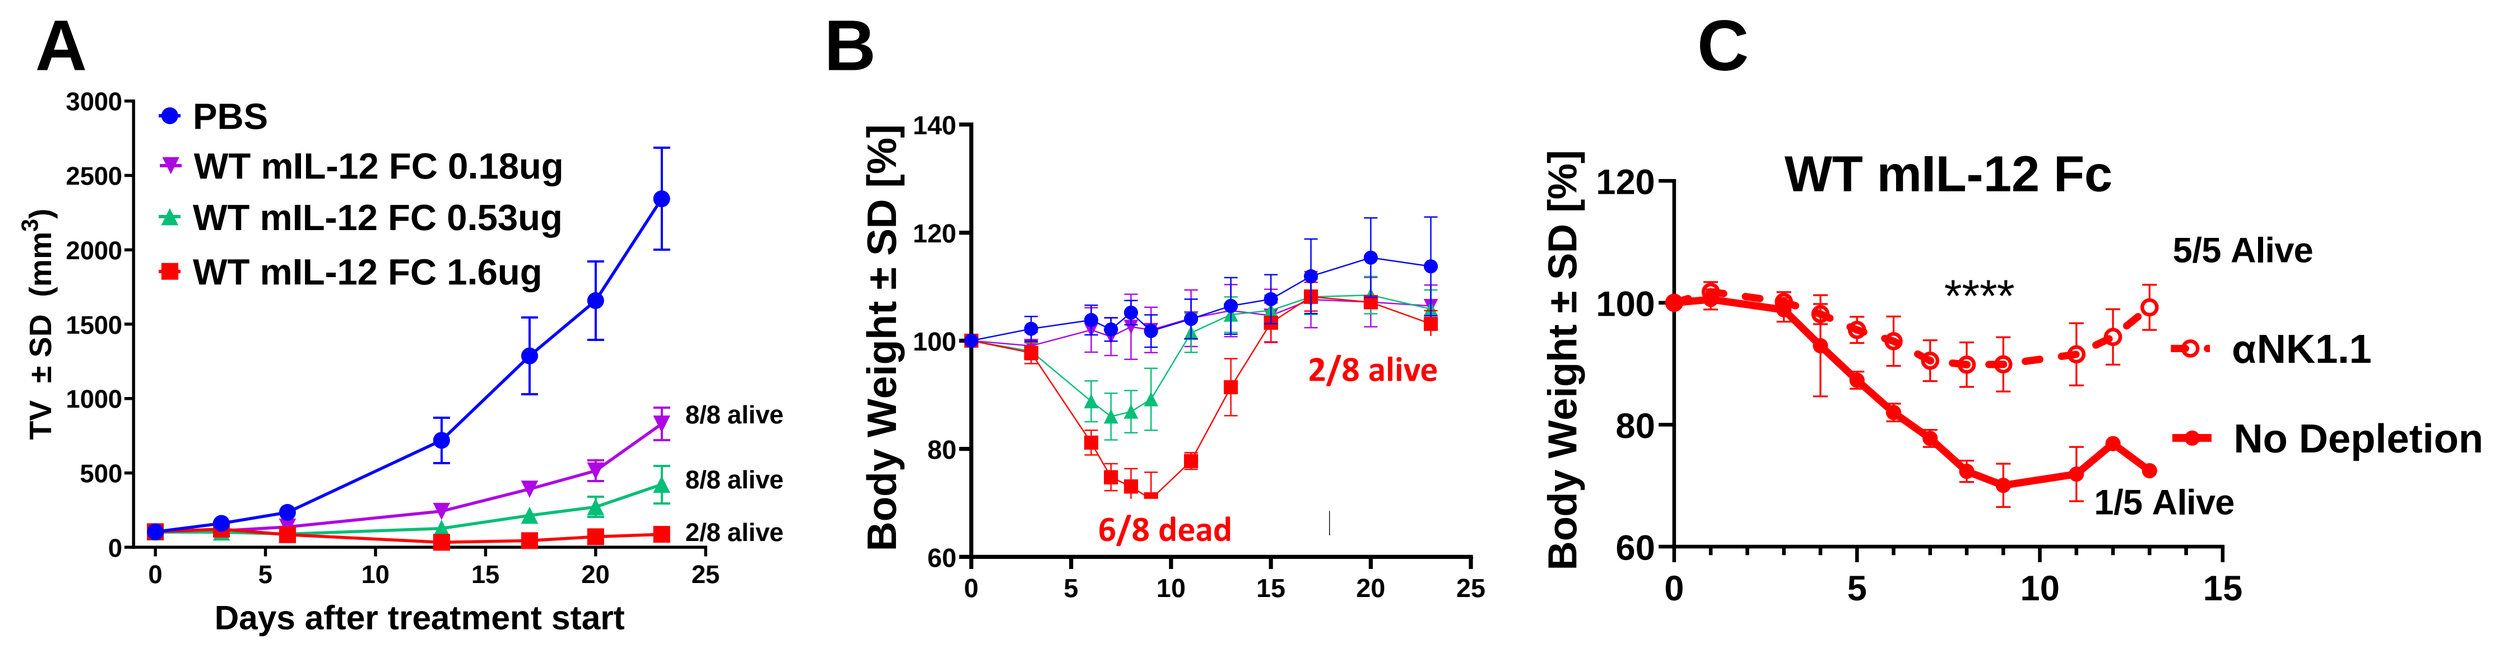
A
PBS
WT mIL-12 FC 0.18ug
WT mIL-12 FC 0.53ug
WT mIL-12 FC 1.6ug
8/8 alive
8/8 alive
2/8 alive
Days after treatment start
B
2/8 alive
6/8 dead
C
WT mIL-12 Fc
5/5 Alive
αNK1.1
No Depletion
1/5 Alive

Figure 1: Wild Type (WT) IL-12 Fc demonstrates dose-dependent efficacy in MC38 tumor model, but is associated with NK cell activity

A: MC-38 tumor volume (TV) and body weight of C57BL/6 mice treated with WT mIL-12-Fc. **B:** Dose-dependent effect of WT mIL-12-Fc on tumor volume and body weight. **C:** Body weight and survival of healthy C57BL/6 mice treated with 1.6 ug WT mIL-12-Fc 2x/week, with or without NK cell-depleting antibody (aNK1.1).

STK-026 design leverages T cell IL-12R upregulation

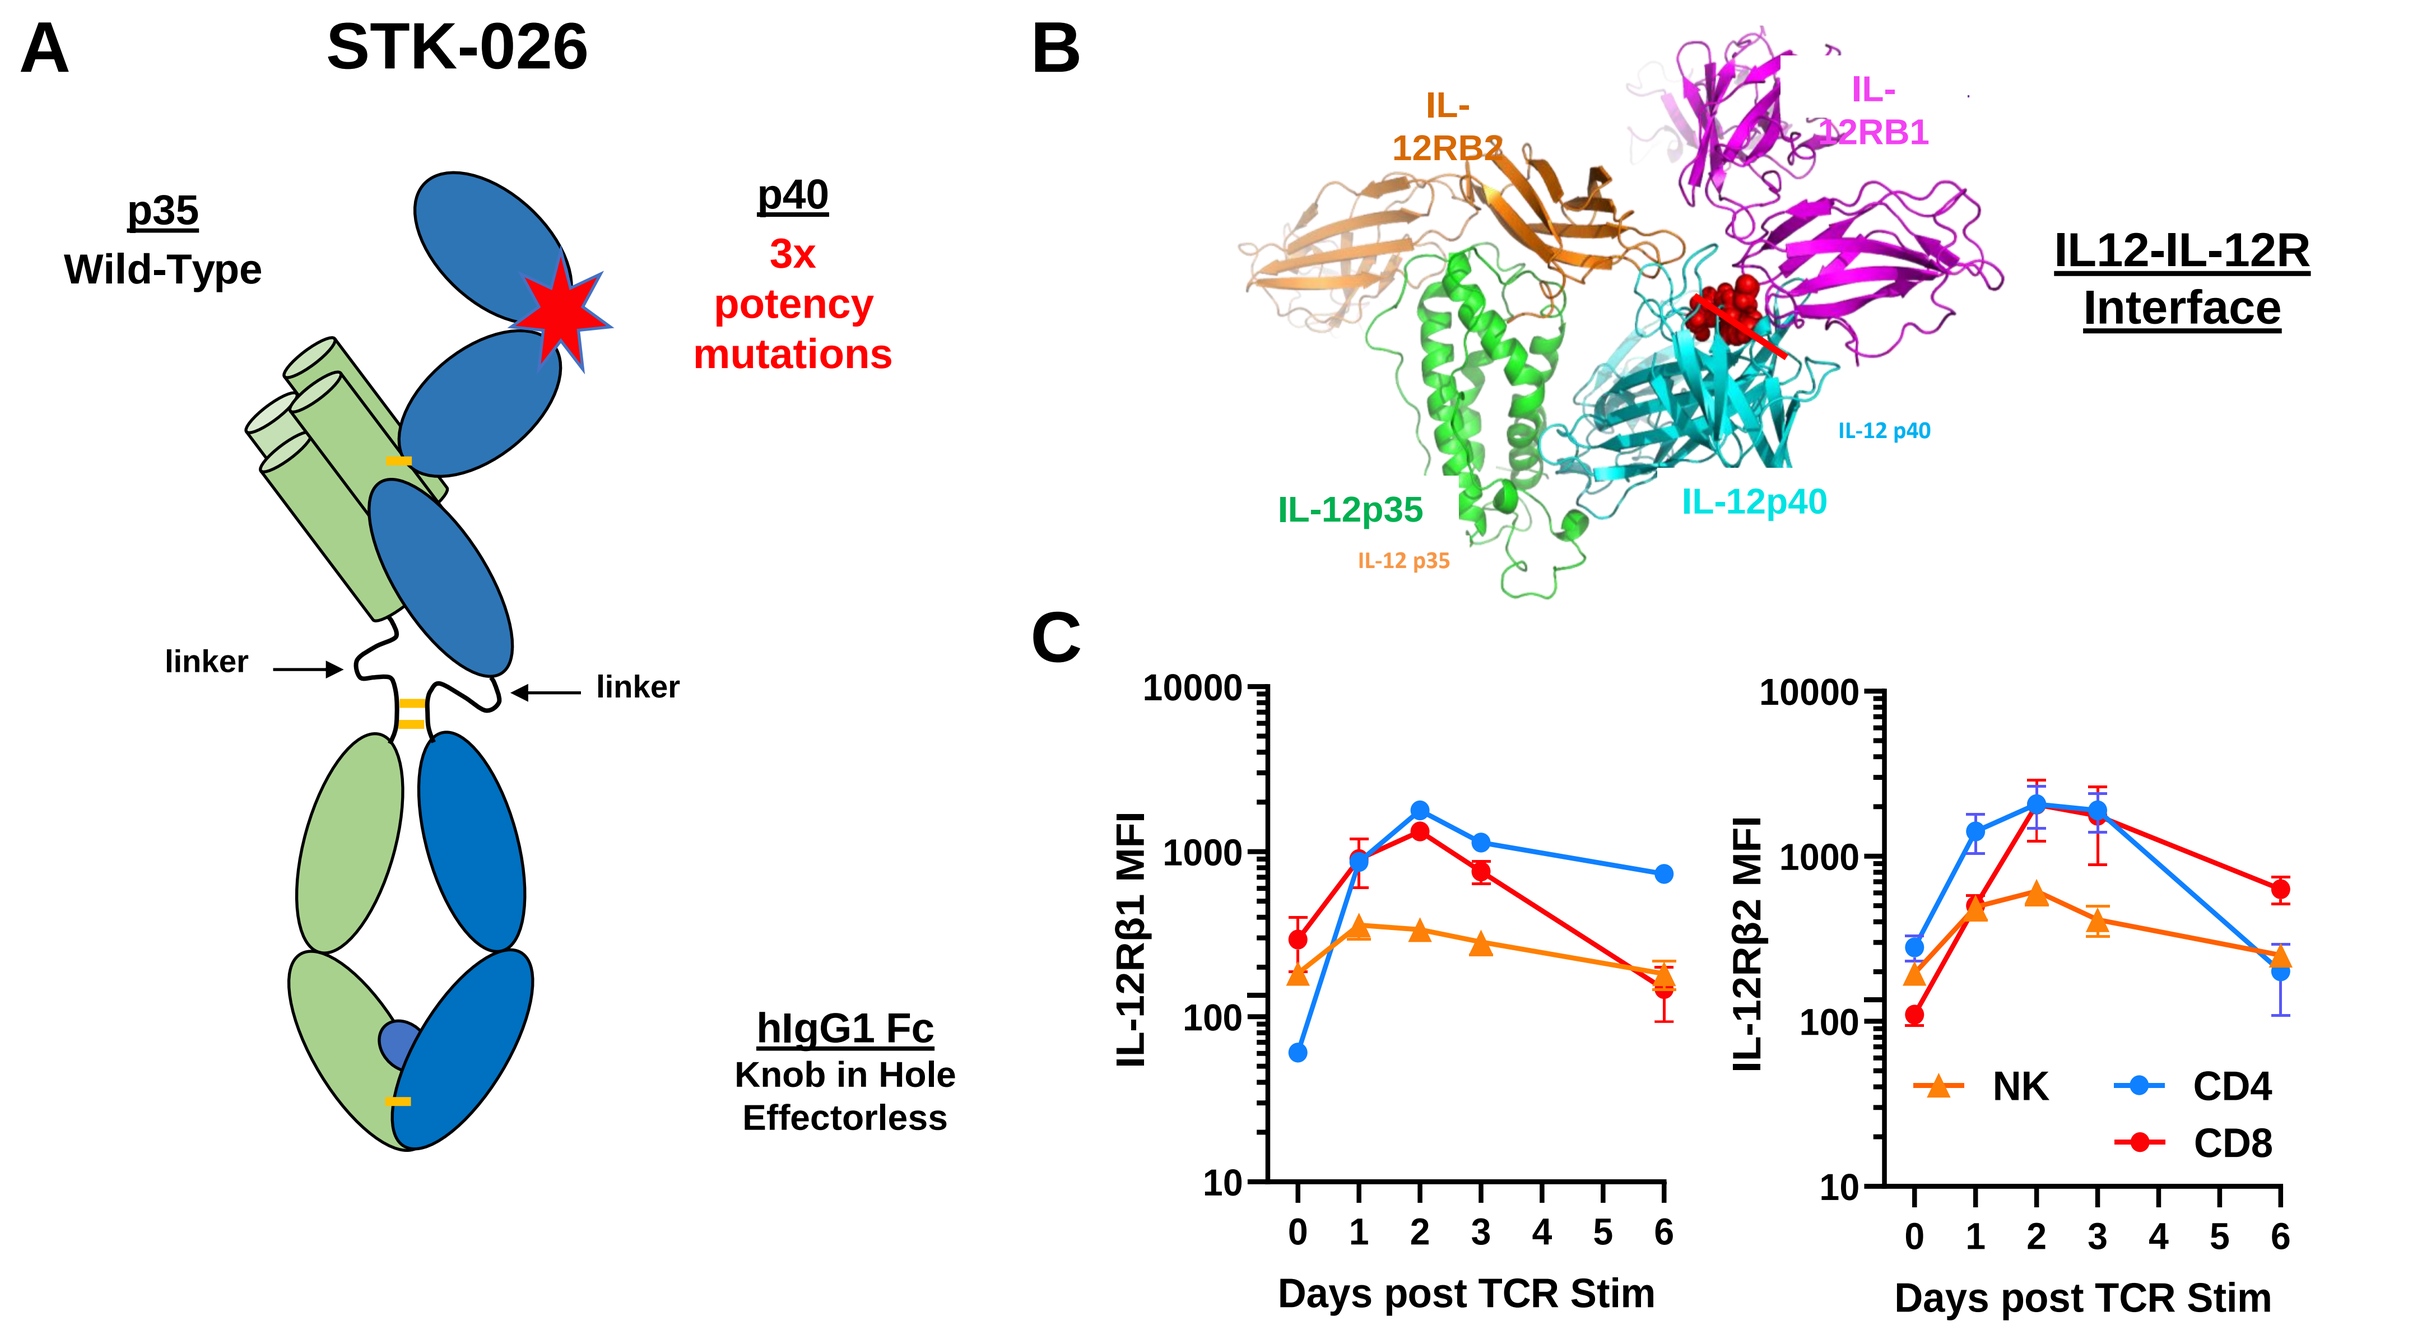
A
STK-026
p35
Wild-Type
p40
3x potency mutations
linker
linker
hIgG1 Fc
Knob in Hole
Effectorless
B
IL-12RB2
IL-12RB1
IL12-IL-12R Interface
IL-12p35
IL-12p40
C
Days post TCR Stim
NK
CD4
CD8

Figure 2: STK-026 is a human IL-12 partial agonist with reduced binding to IL-12Rb1

A: Representation of STK-026, highlighting mutations in p40, wild type p35, and half-life extension via effectorless KiH-Fc. **B:** Model of quaternary IL-12, IL-12R complex. **C:** Time course of IL-12Rb1 and IL-12Rb2 on CD3/CD28 stimulated human PBMCs.

STK-026 has reduced NK cell potency vs WT IL-12 Fc

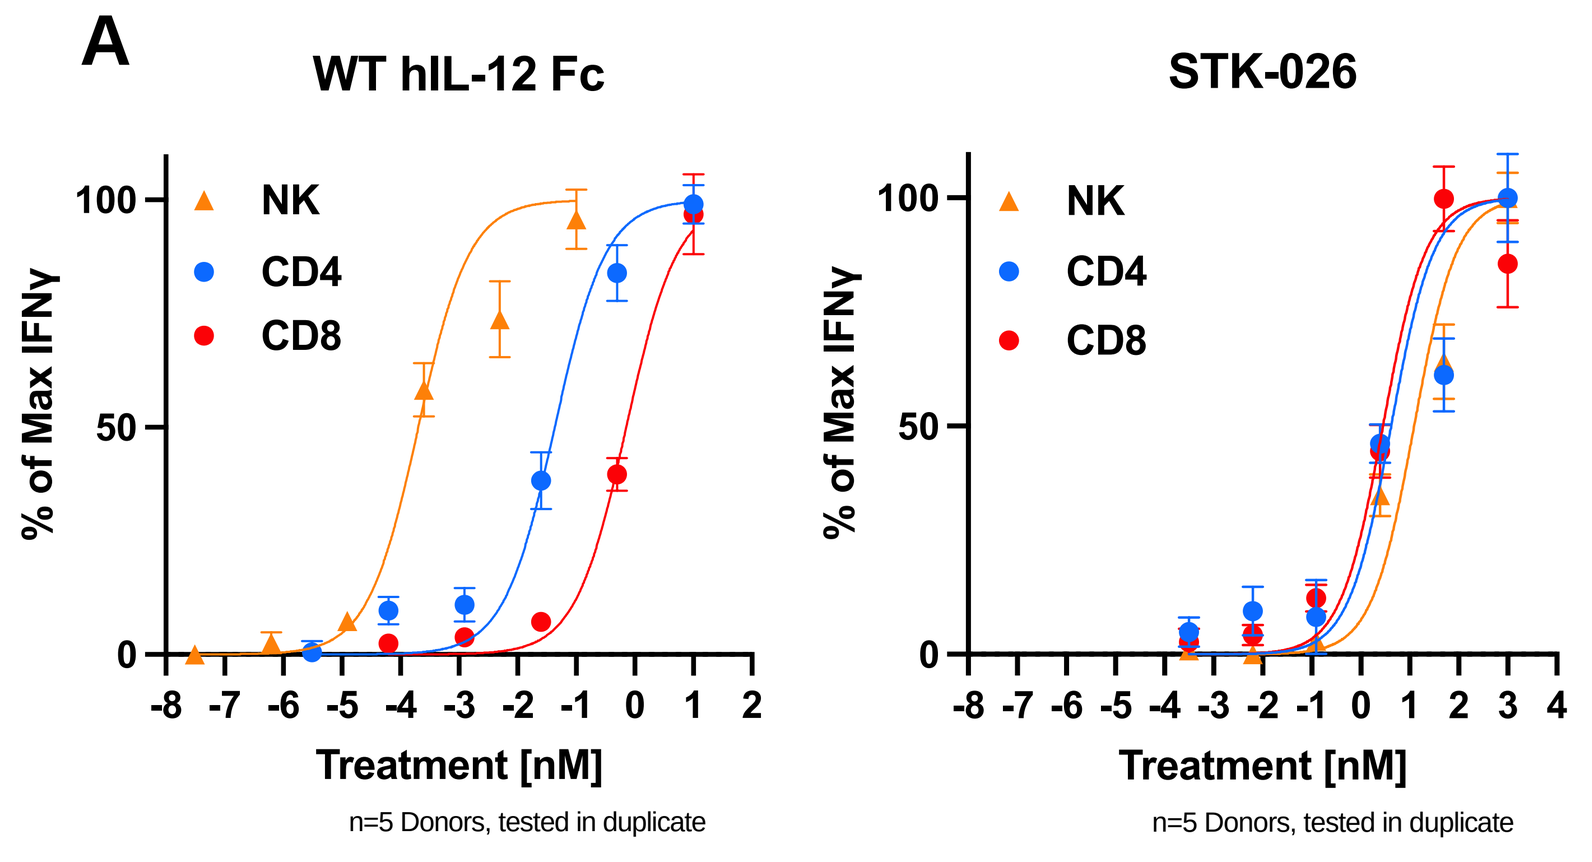
A
WT hIL-12 Fc
STK-026
NK
CD4
CD8
% of Max IFNγ
Treatment [nM]
n=5 Donors, tested in duplicate

Figure 3: STK-026 parity in NK and T cell responses

A: Interferon gamma (IFNγ) secretion by cells pre-stimulated with anti-CD3/anti-CD28 + IL-2, treated with IL-2 + WT IL-12 Fc or STK-026.

Mouse STK-026 maintains anti-tumor efficacy

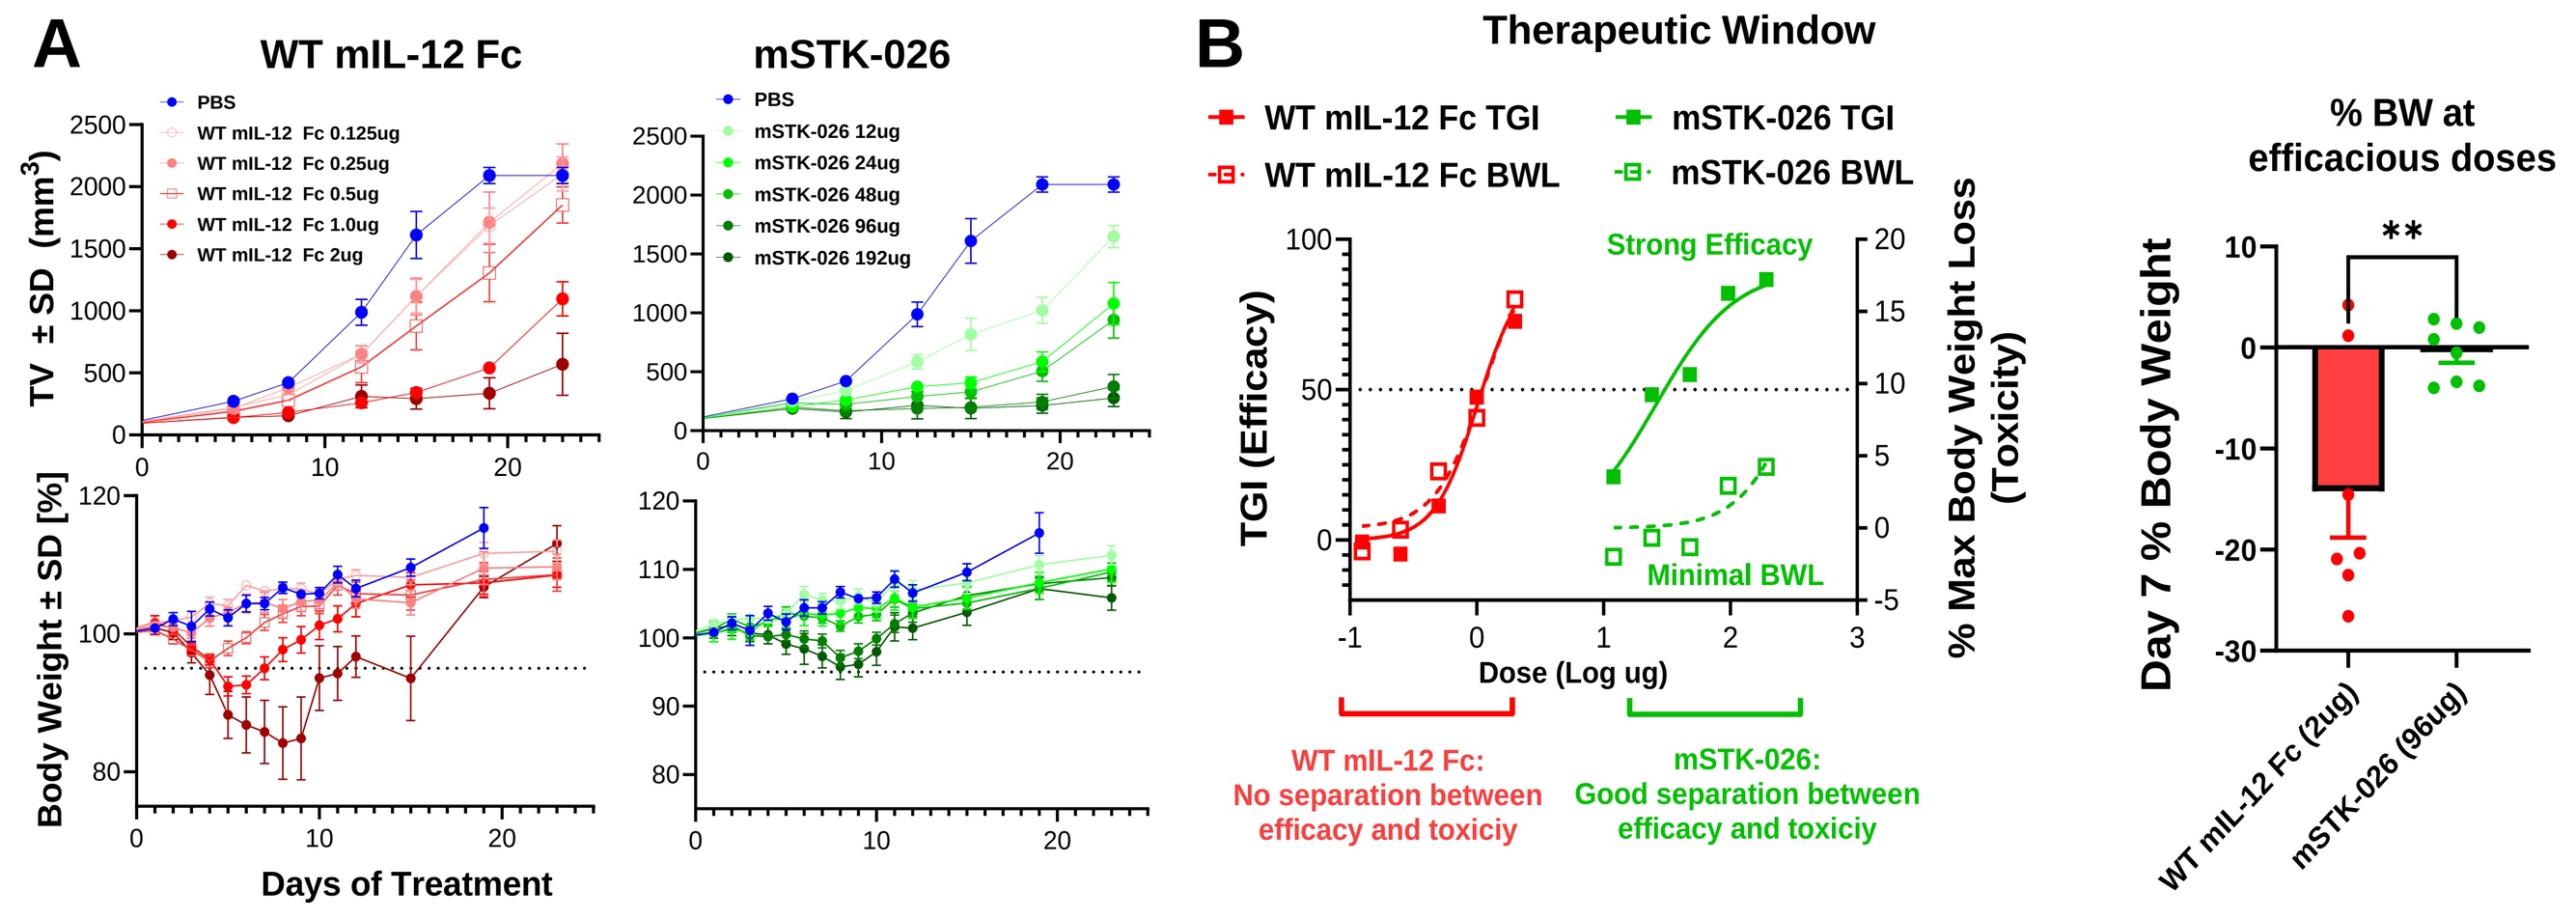
A
WT mIL-12 Fc
mSTK-026
Days of Treatment
B
Therapeutic Window
WT mIL-12 Fc TGI
mSTK-026 TGI
WT mIL-12 Fc BWL
mSTK-026 BWL
Strong Efficacy
Minimal BWL
Dose (Log ug)
WT mIL-12 Fc: No separation between efficacy and toxiciy
mSTK-026: Good separation between efficacy and toxiciy
% BW at efficacious doses

Figure 4: Mouse STK-026 maintains potent anti-tumor efficacy, without causing significant body weight loss at therapeutic doses

A: MC-38 tumor growth and body weights of tumor bearing mice across a dose titration of WT mIL-12 Fc and mSTK-026. **B:** Tumor Growth Inhibition (TGI) (study day 23) vs. max % body weight loss by dose (study day 7).

Mouse STK-026 avoids acute NK hyperactivation and allows gradual, sustained NK and CD8 T cell activation

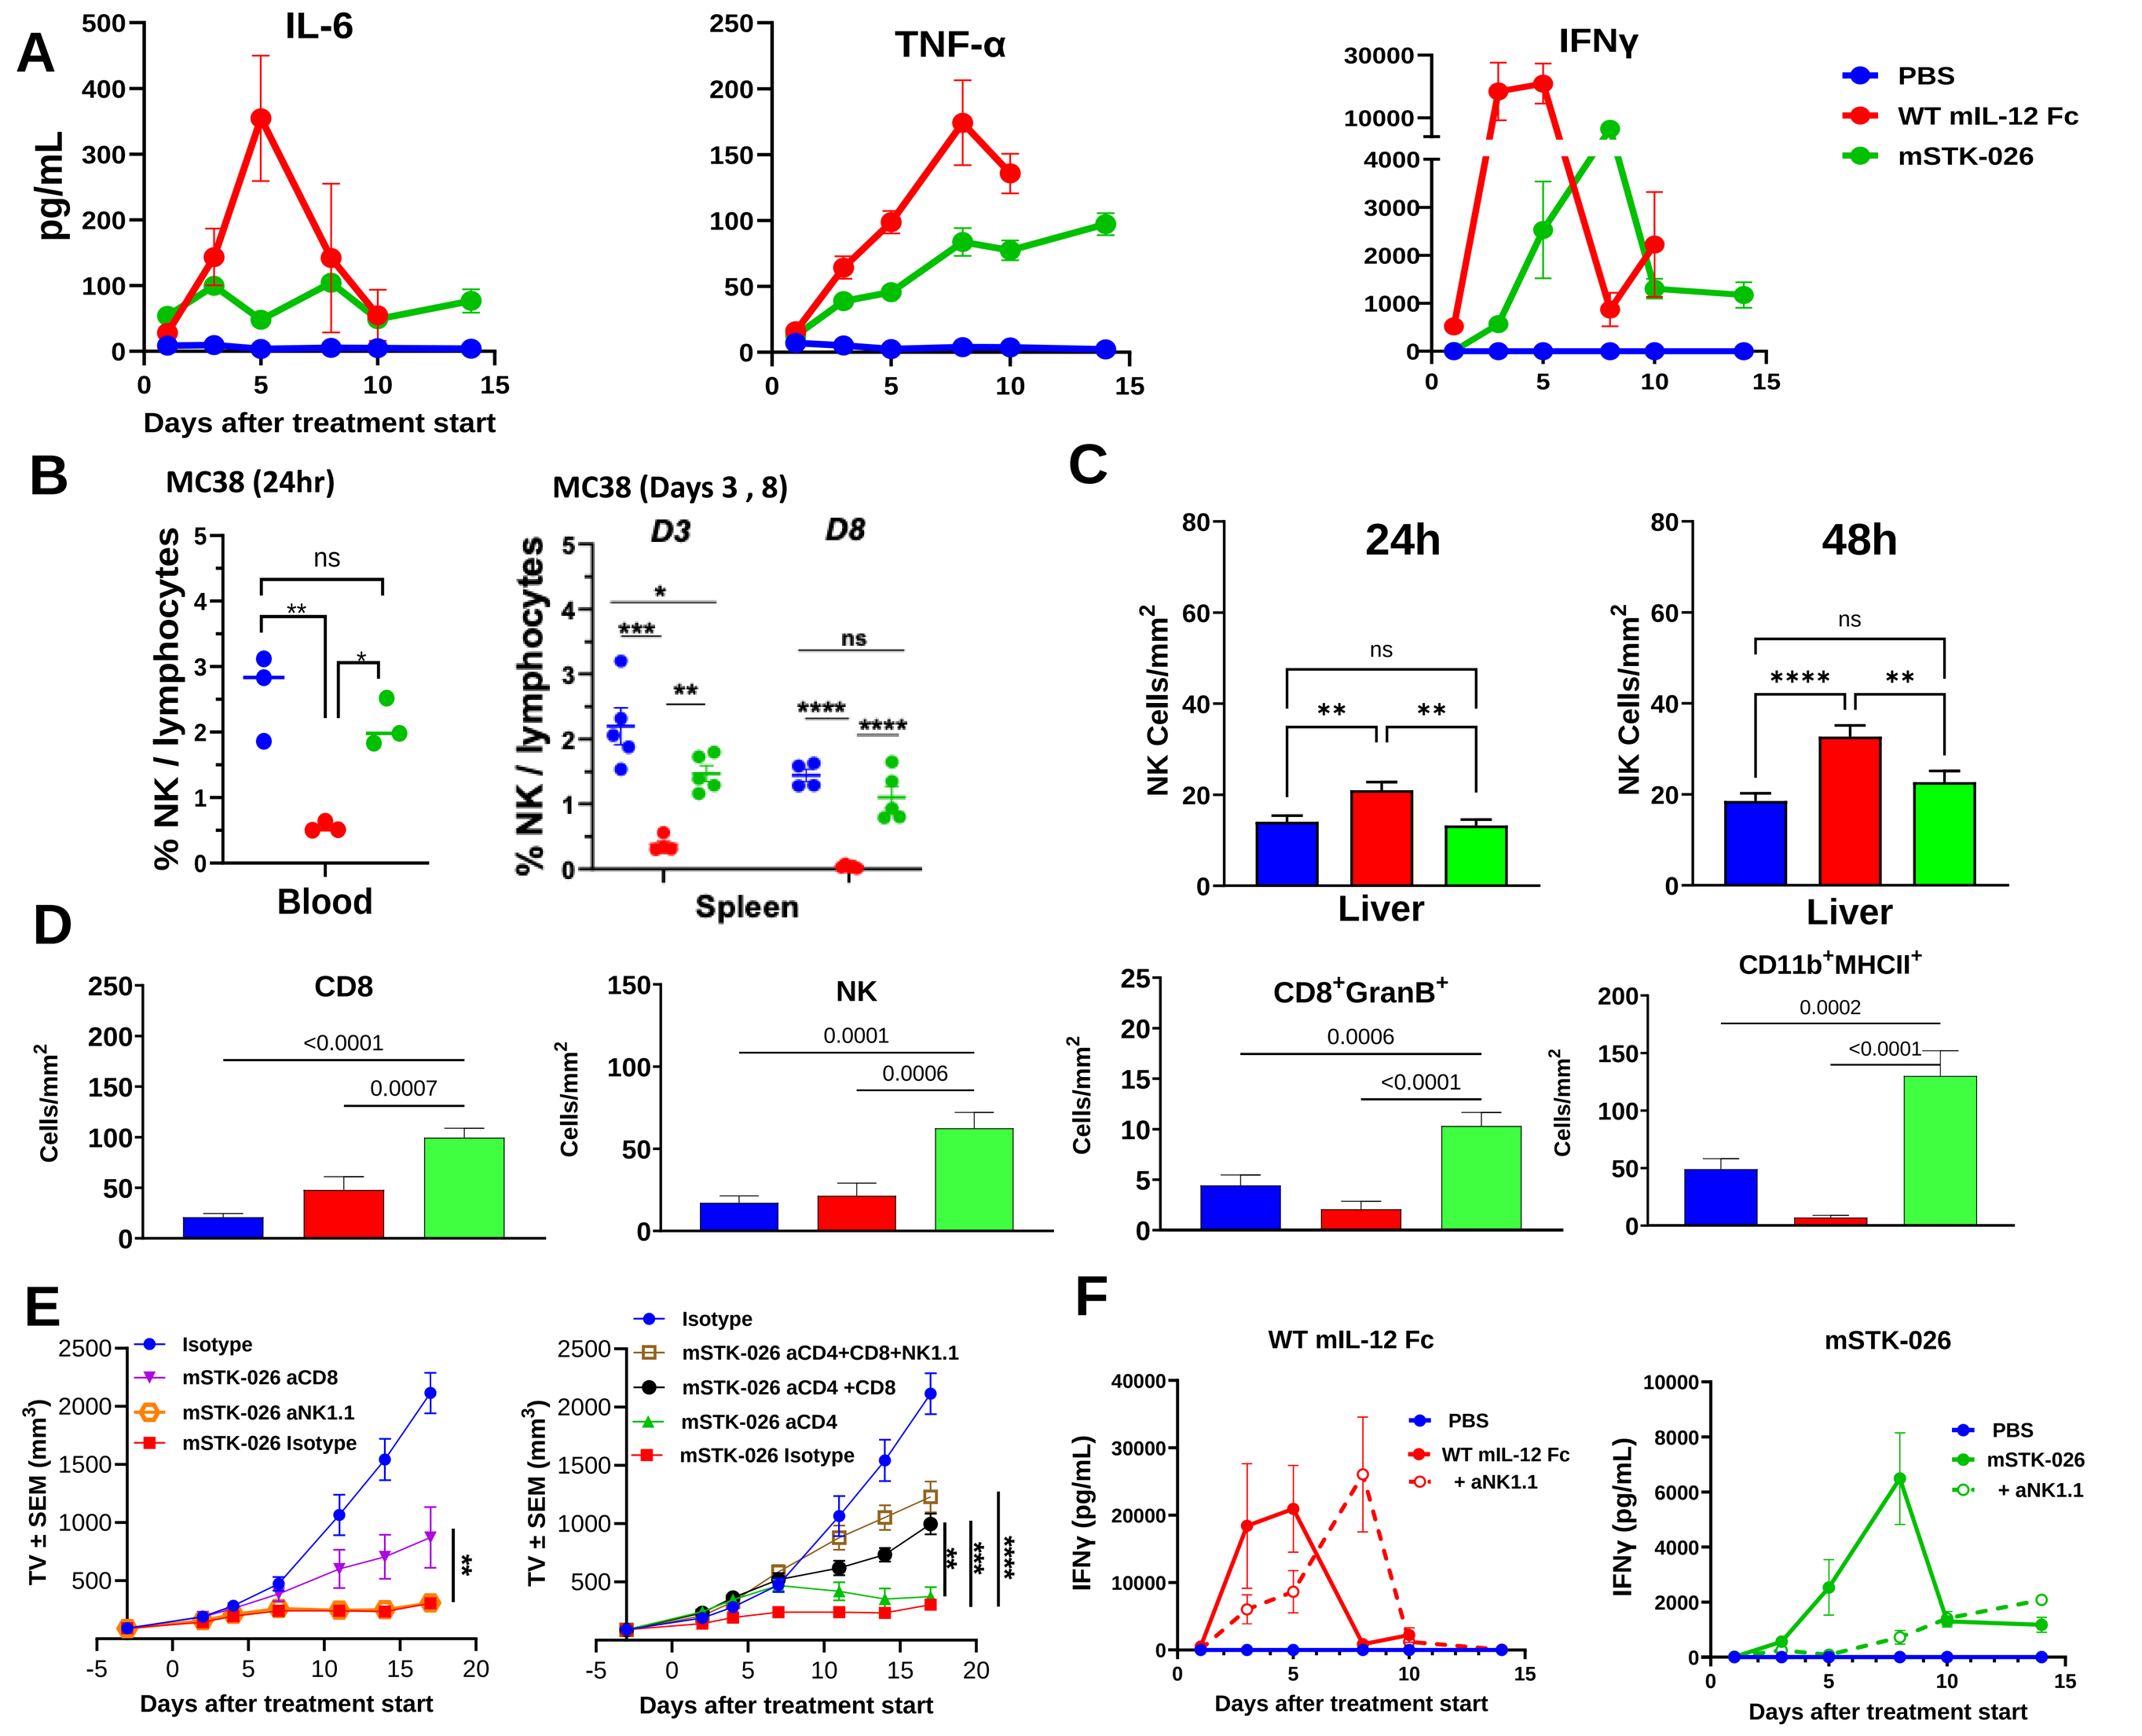
A
IL-6
TNF-α
IFNγ
PBS
WT mIL-12 Fc
mSTK-026
Days after treatment start
B
MC38 (24hr)
MC38 (Days 3 , 8)
Blood
Spleen
C
24h
48h
Liver
D
CD8
NK
CD8+GranB+
CD11b+MHCII+
E
F
WT mIL-12 Fc
mSTK-026

Figure 5: mSTK-026 avoids IL-12-induced cytokine release syndrome yet retains activation of T cells, NK cells and APCs

A: Cytokines of healthy C57BL/6 mice treated with mSTK-026 (96 ug/wk) or mIL-12 Fc (1.6 ug/wk). **B** and **C:** NK cell composition in blood, spleen, and liver. **D:** TILs from MC-38 tumors harvested at day 26. **E** and **F:** MC-38 tumor growth and serum IFNγ of mice treated with mSTK-026 and anti-NK1.1/CD4/CD8 depleting antibodies.

mSTK-026 reverses "cold" B16F10 tumor model by activating T cells and Macrophages

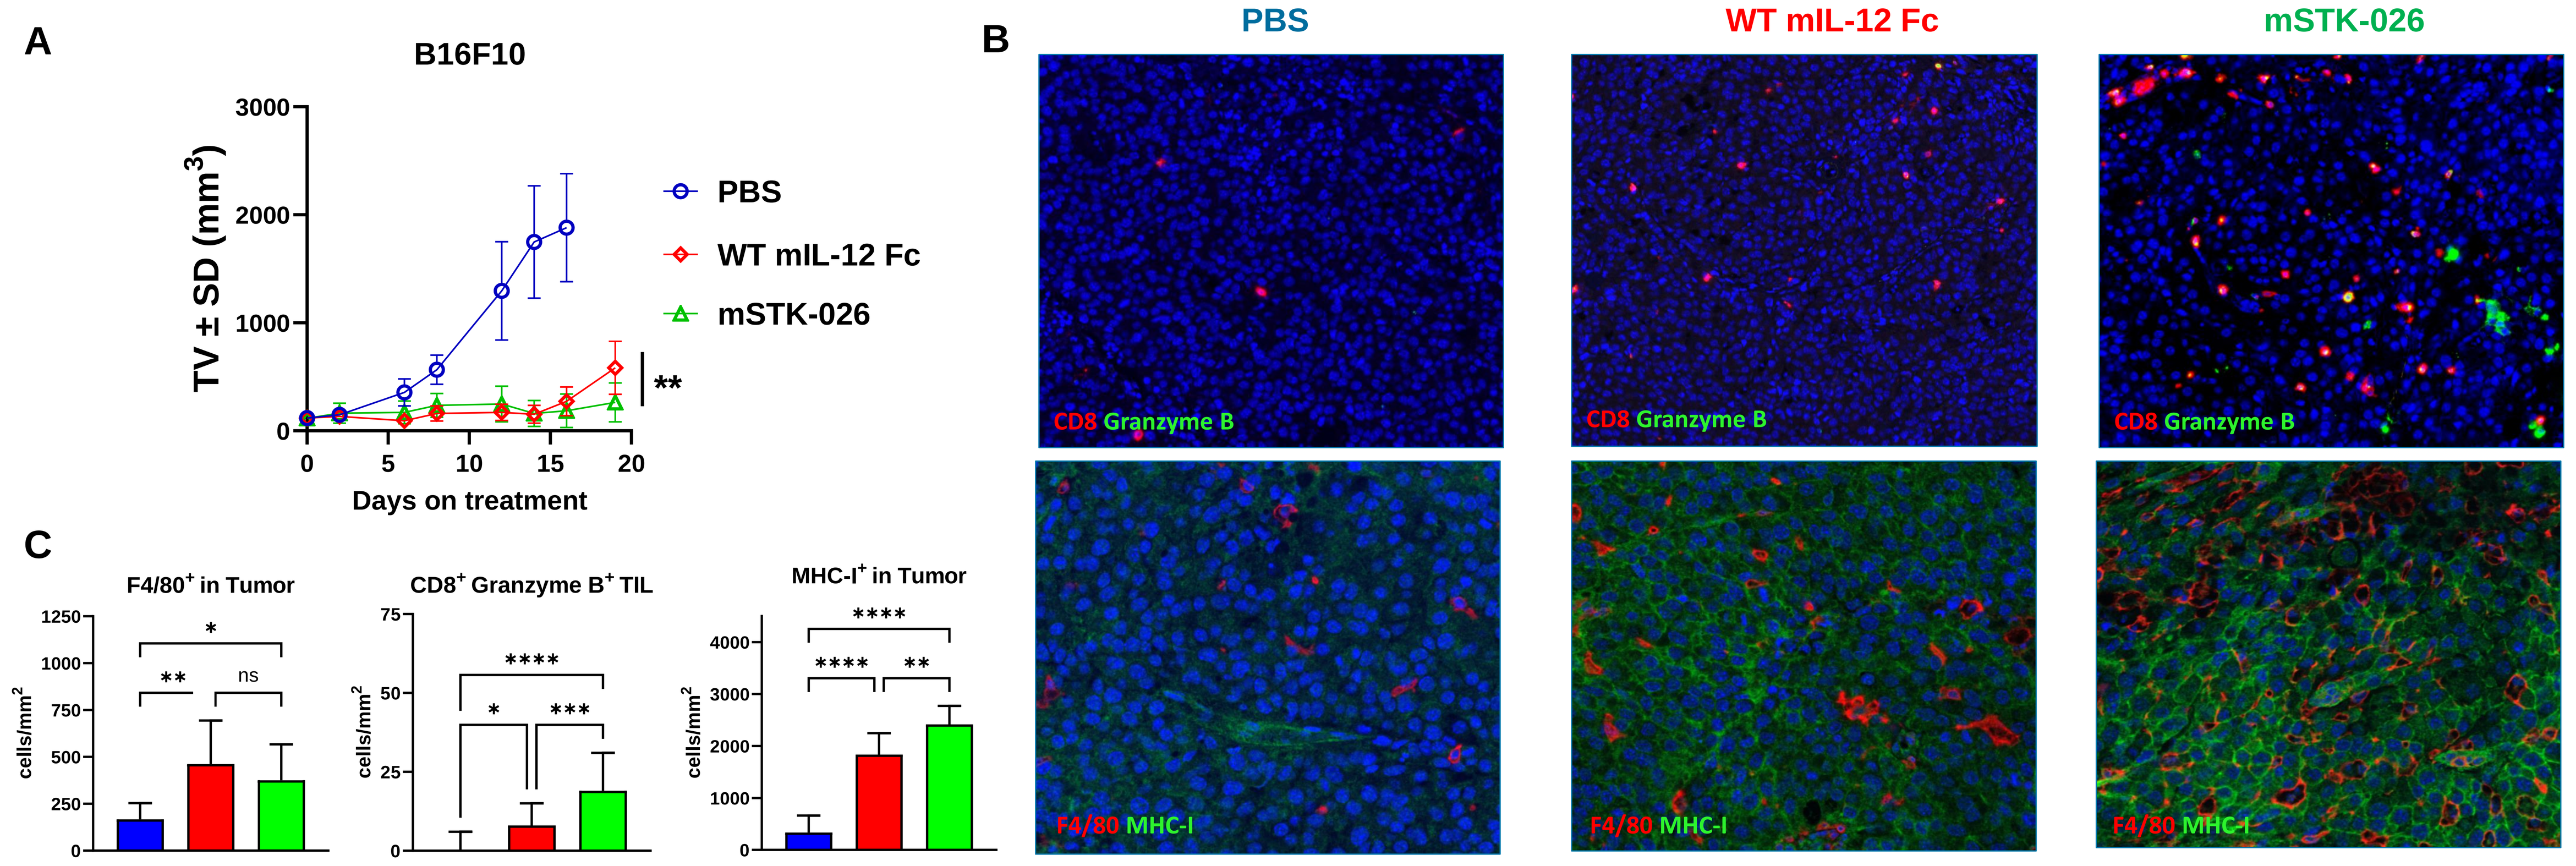
A
B16F10
PBS
WT mIL-12 Fc
mSTK-026
Days on treatment
B
PBS
WT mIL-12 Fc
mSTK-026
CD8 Granzyme B
F4/80 MHC-I
C
F4/80+ in Tumor
CD8+ Granzyme B+ TIL
MHC-I+ in Tumor

Figure 6: Mouse STK-026 effectively controls "cold" tumors by activating T cells, priming macrophages for MHC-I expression

A: B16F10 tumor growth of tumor bearing mice treated with WT mIL-12 Fc (0.5 ug/week) and mSTK-026 (48 ug/week). **B** and **C:** Immunohistochemistry image and TIL quantification from B16F10 tumors, illustrating elevated CD8 T cell and macrophage activation on treatment (day 19).

STK-026 induces gradual lymphocyte activation with reduced cardiac inflammation in NHP

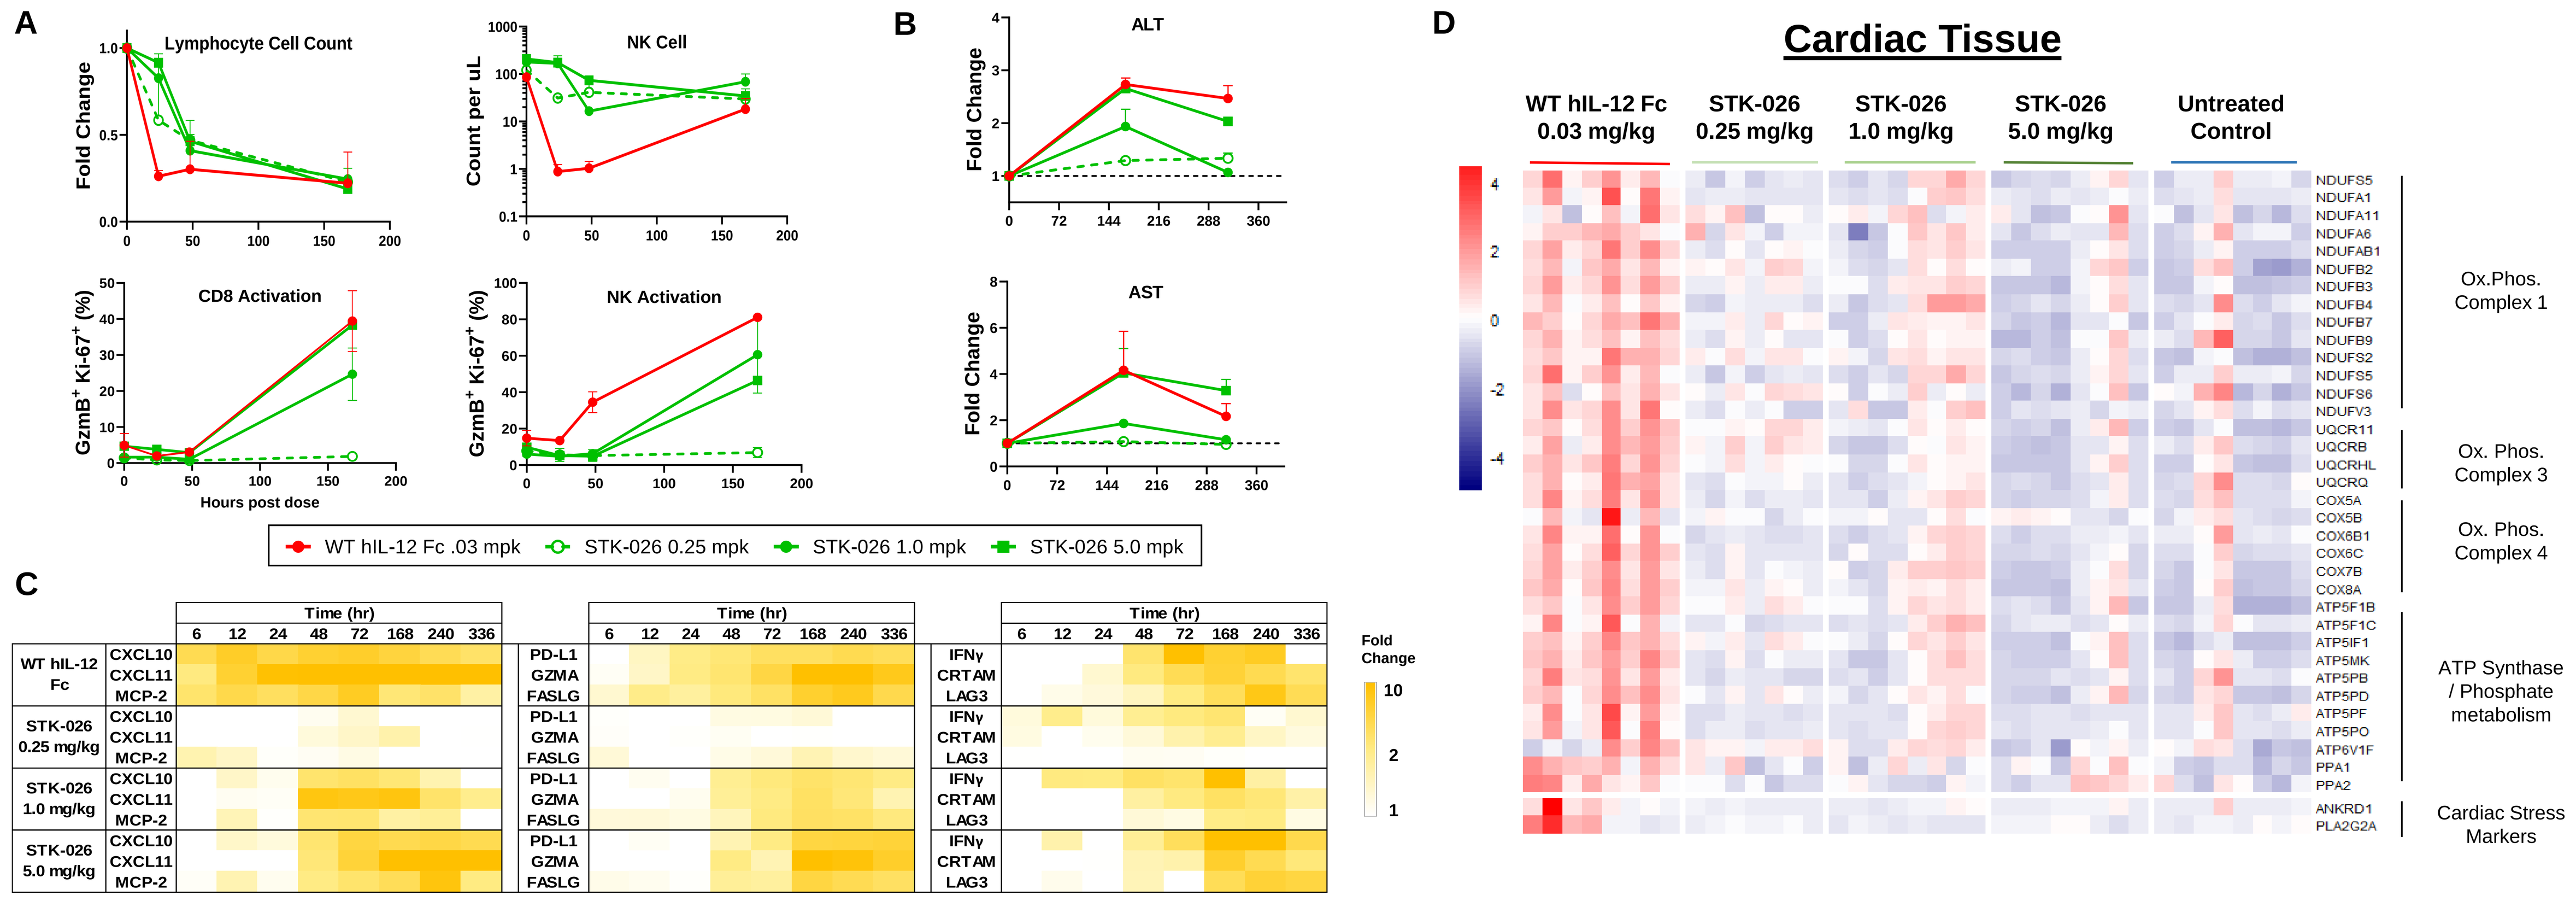
A
Lymphocyte Cell Count
NK Cell
CD8 Activation
NK Activation
Hours post dose
B
ALT
AST
WT hIL-12 Fc .03 mpk
STK-026 0.25 mpk
STK-026 1.0 mpk
STK-026 5.0 mpk
C
Time (hr)
Fold Change
D
Cardiac Tissue
WT hIL-12 Fc 0.03 mg/kg
STK-026 0.25 mg/kg
STK-026 1.0 mg/kg
STK-026 5.0 mg/kg
Untreated Control
Ox.Phos. Complex 1
Ox. Phos. Complex 3
Ox. Phos. Complex 4
ATP Synthase / Phosphate metabolism
Cardiac Stress Markers

Figure 7: STK-026 is active in cynomolgus macaques while avoiding toxicity associated with wild type IL-12

A: Lymphocyte, NK cell counts, markers of activation in blood from dose-range finding cynomolgus *in vivo* study (single dose, 1M/1F per group).
B: ALT/AST liver enzyme measurements during study. **C:** Serum cytokines, chemokines, and activation-induced proteins measured via Olink.
D. Nanostring analysis of heart tissues, highlighting genes associated with oxidative phosphorylation and cardiac stress (4 regions/animal).

Murine STK-026 activates T and NK cells and leads to myeloid cell reprogramming and antigen presentation

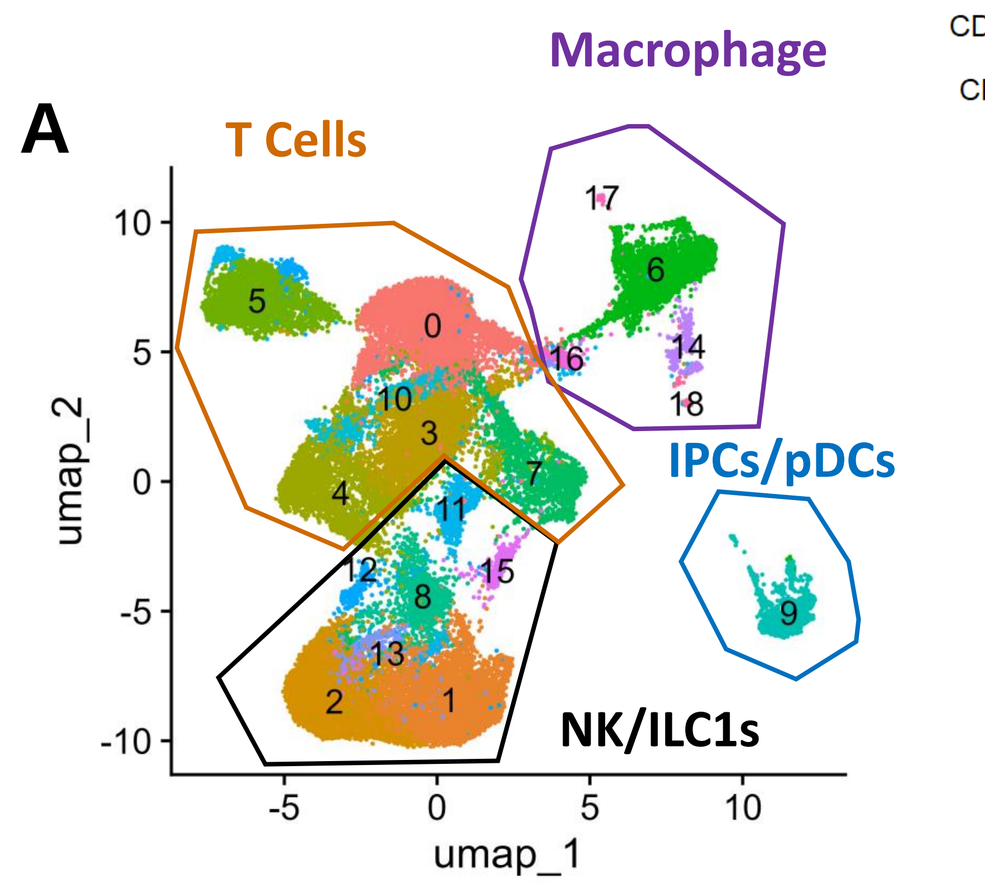
A
T Cells
Macrophage
IPCs/pDCs
NK/ILC1s
umap_1
umap_2

Treatment	PBS	IL-12	STK-026
# CD45+ TILs	12,211	11,863	11,696

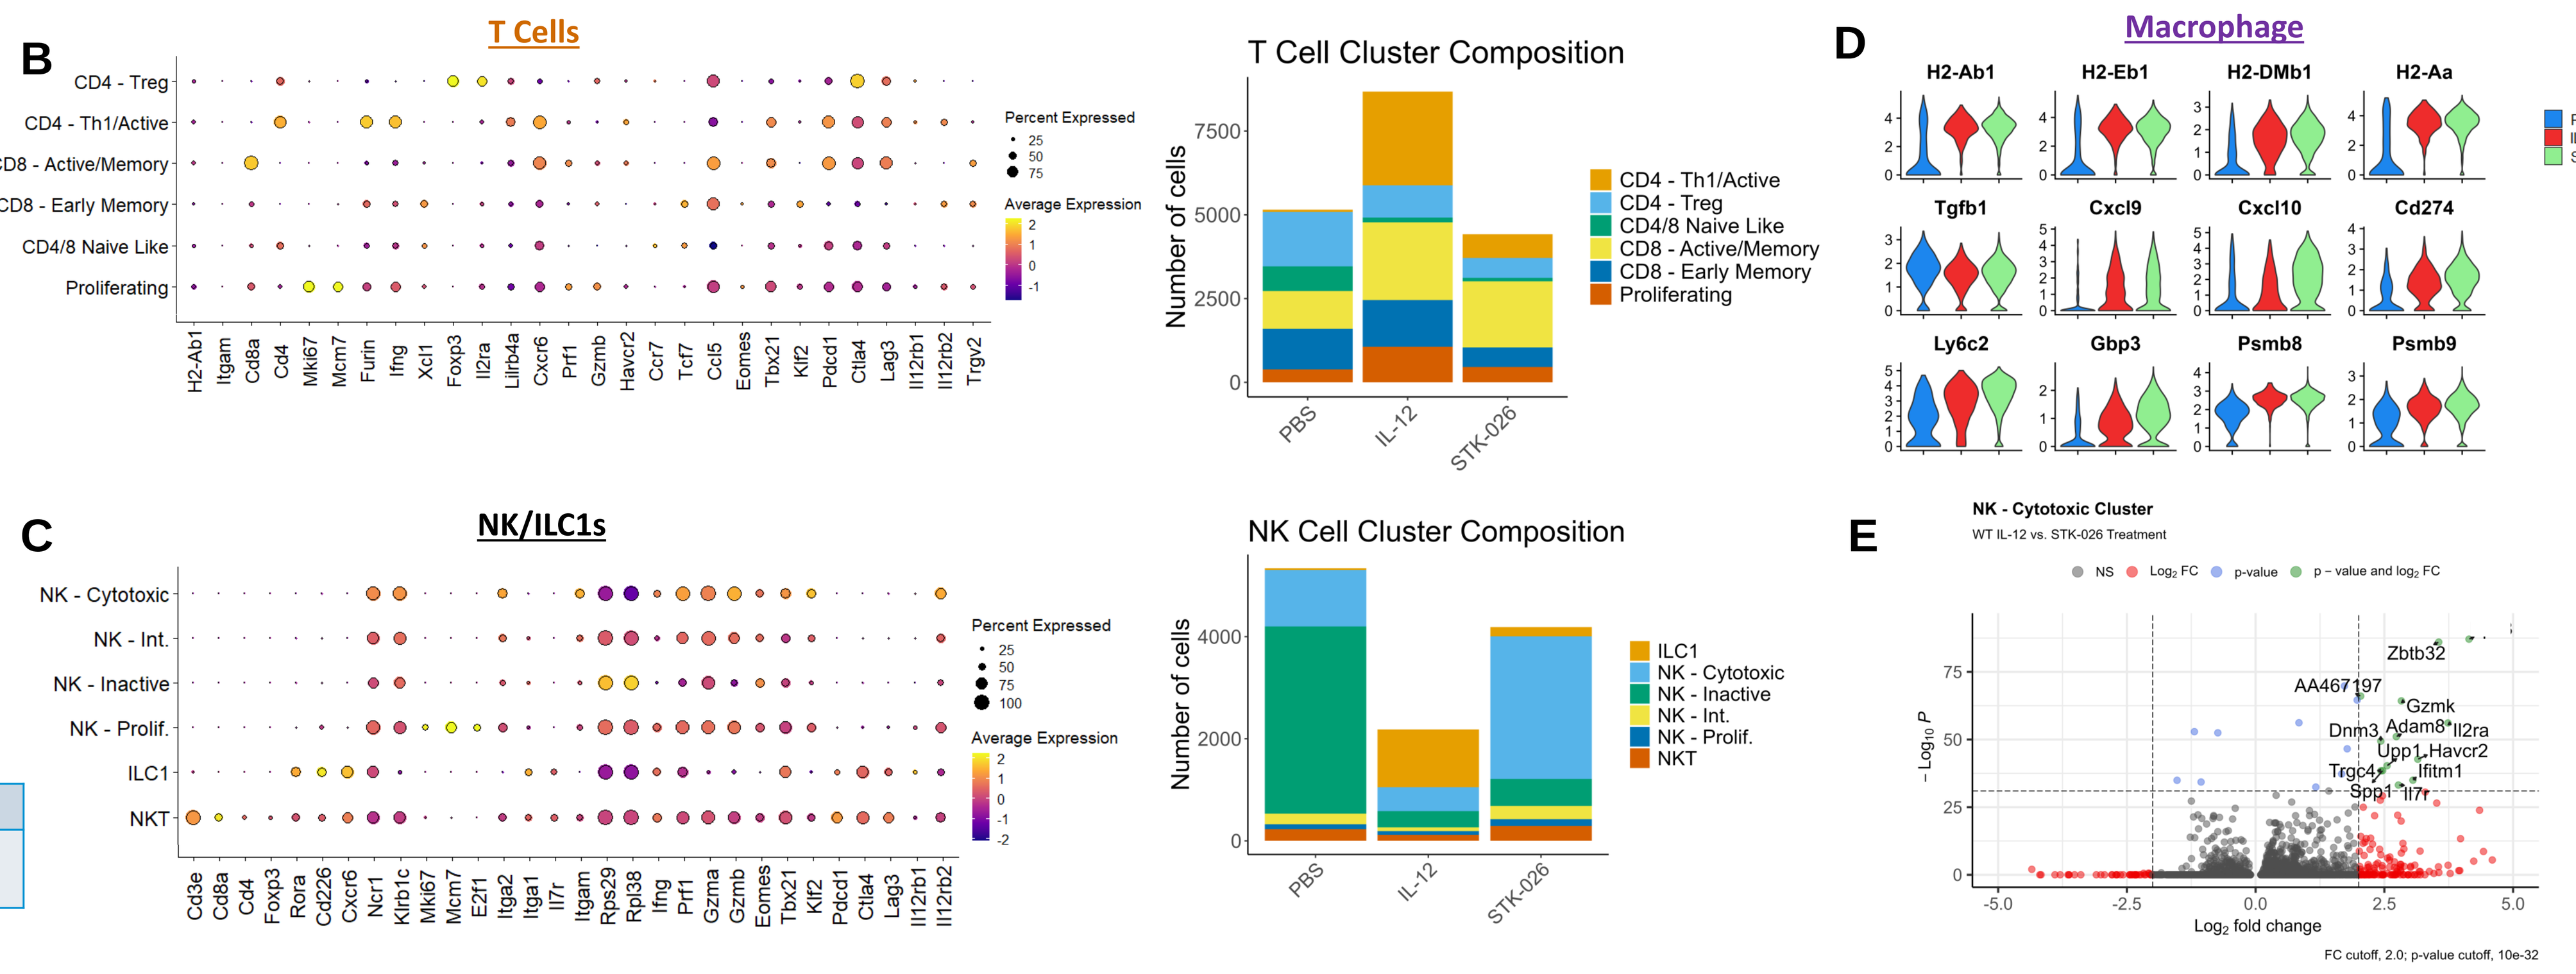
B
T Cells
T Cell Cluster Composition
C
NK/ILC1s
NK Cell Cluster Composition
D
Macrophage
E
NK - Cytotoxic Cluster
WT IL-12 vs. STK-026 Treatment

Figure 6. scRNA-Seq reveals activated NK and T cell phenotype, with increased myeloid reprogramming and antigen presentation
A: UMAP projection of combined tumor CD45+ cells (MC38, day 8 tumors); cell counts tabulated per treatment. **B:** T cell subcluster gene expression and total composition. **C:** NK cell and ILC1 subcluster gene expression and total composition. **D:** Cluster 6 (Mo) macrophage select gene expression associated with antigen presentation. **E:** Differential gene expression volcano plot of Cytotoxic NK cluster, WT mIL-12 Fc vs. STK-026.

Conclusions

1. **STK-026 is a human IL-12 partial agonist engineered with half-life extension.**
2. **Human STK-026 demonstrates parity in NK and T cell potency under inflammatory conditions.**
3. **mSTK-026 maintains the potent anti-tumor efficacy of WT mIL-12 without the acute cytokine storm associated body weight loss.**
4. **Single cell RNASeq analysis of MC38 TILs reveals activity consistent with IL-12, with reduced NK and T cell exhaustion, and increased effector cell survival and infiltration genes.**
5. **mSTK-026 effectively activates intratumoral CD4 and CD8 cells in "cold" tumor types, resulting in myeloid activation consistent with IFNγ secretion.**
6. **STK-026 avoids acute circulating lymphocyte activation in non-human primates and WT IL-12 associated cardiac toxicities, while maintaining measurable pharmacodynamic cytokines and chemokines.**

1) Atkins MB, *et al.* Phase I evaluation of intravenous recombinant human interleukin 12 in patients with advanced malignancies. Clinical Cancer Research. 1997; 3 (3): 409-17.
2) Carson W, *et al.* A fatal cytokine-induced systemic inflammatory response reveals a critical role for NK cells. J Immunology. 1999; 162 (8): 4943–4951.